Prediction of Serotonin 2A Receptor (5-HT_{2A}R) Occupancy in Man from Nonclinical Pharmacology Data. Exposure vs. 5-HT_{2A}R Occupancy Modeling Helped to Design a Positron Emission Tomography (PET) Study in Healthy Male Subjects.

Hoeben, E.[1], Brochot, A.[1], Sinha, V.[2], de Boer, P.[3], Wuyts, K.[4], Bohets, H.[5], Scheers, E.[5], Langlois X.[6], Te Riele P.[6], and Lavreysen, H.[6]

Model Based Drug Development[1], Clinical Pharmacology[2], Experimental Medicine[3], Translational Sciences EDD[4], Drug Safety Sciences[5], Neuroscience Discovery[6], Janssen Research & Development a Division of Janssen Pharmaceutica N.V., Beerse, Belgium

Introduction

JNJ-40411813, which was characterized in non-clinical models as a positive allosteric modulator (PAM) of the metabotropic glutamate receptor 2 and a 5-HT_{2A}R antagonist, is currently in development for the treatment of disorders of the central nervous system and expected to have similar efficacy as mGlu2 agonists[1,2]. 5-HT_{2A}R occupancies in man were predicted based on *in vitro* and *in vivo* nonclinical pharmacology data in the rat. Predictions showed that binding to the 5-HT_{2A}R in man will be less than anticipated based on rodent data, when corrected for differences in free fraction and the different contributions of parent and metabolite in rat *vs.* man. To confirm and quantify 5-HT_{2A}R engagement at pharmacodynamic active dose levels, a Positron Emission Tomography (PET) study was performed in healthy male subjects[3].

Methods

in vitro assays

- [^{3}H]ketanserin (2nM) binding experiments were performed using membranes prepared from human 5-HT_{2A}R expressing NIH3T3 cells.
- To determine the antagonistic nature of JNJ-40411813 for the 5-HT_{2A}R, functional Ca^{2+} experiments (*i.e.* Inhibition of serotonin-induced Ca^{2+} signaling) were performed at CEREP.

in vivo 5-HT_{2A}R *occupancy*

- Male Wistar rats were dosed orally with JNJ-40411813 at 5 and 20 mpk. The 5-HT_{2A}R radioligand ([^{3}H]ketanserin – 10 µCi/animal) was injected intravenously 45 min before sacrifice at specific time points (1, 2, 4, 8 and 24 hours).
- Three rats were used per time point for plasma pharmacokinetics and *in vivo* 5-HT_{2A}R occupancy.
- Individual plasma concentrations were analysed for JNJ-40411813 and its major metabolite (M47) using a qualified research LC-MS-MS method.
- Dissection of prefrontal Cortex and Cerebellum. Tissue homogenized and counted in Tri-Carb liquid scintillation counter.

Exposure vs. 5-HT_{2A} *R occupancy modeling*

Human plasma concentrations of JNJ-40411813 at the anticipated clinical doses were predicted using an exploratory population PK model.

The plasma concentration *vs.* 5-HT_{2A}R occupancy modeling was performed using WinNonlin Professional (Version 5.2.1).

Human 5-HT_{2A} PET study

The concentration-related binding of JNJ-40411813 to the 5-HT_{2A}R was investigated in healthy male subjects (n=12) using [^{11}C]MDL 100,907 PET. [^{11}C]MDL 100,907 is the ^{11}C-labeled derivative of raclopride which is a selective 5-HT_{2A}R antagonist. The human PET study was conducted at the University Medical Center in Groningen.

To characterize 5-HT_{2A}R occupancy, single doses of JNJ-40411813 were administered over a dose range from 200 to 700 mg. 5-HT_{2A}R occupancy was calculated from PET scans performed around T_{max} (between 1 and 3 hours postdose) and optionally 24 hours postdose.

Results

In Vitro 5-HT_{2A} Activity of JNJ-40411813 and Major Metabolites in Rat and Man

In vitro	K_b (% effect)	5-HT_{2A} Binding IC50 (nM)
JNJ-40411813 Unchanged drug	1.1 µM (70%)	708
Major Rat metabolite M47	nd	102
Major Human metabolite M45	nd	>10000

JNJ-40411813 acts as an mGlu2 PAM with a potency of 147 nM, as determined using *in vitro* [^{35}S]GTPgS experiments in CHO cells expressing the human mGlu2 receptor.

JNJ-40411813 is a weak 5-HT_{2A}R antagonist. In rats JNJ-40411813 is rapidly metabolized to M47 which is a relatively potent and selective 5-HT_{2A}R antagonist.

nd = not determined

In vivo 5-HT_{2A}R Occupancy in Male Wistar Rats

The time-course of the 5HT_{2A}R occupancy in the rat after PO administration is in good correspondence with the plasma concentration time course of the major rat metabolite (M47).

Low contribution of unchanged drug (UD) to the *in vivo* 5-HT_{2A}R occupancy in the rat.

UD = unchanged drug
M47 = major metabolite in rats

PO: 5 mpk PO: 20 mpk

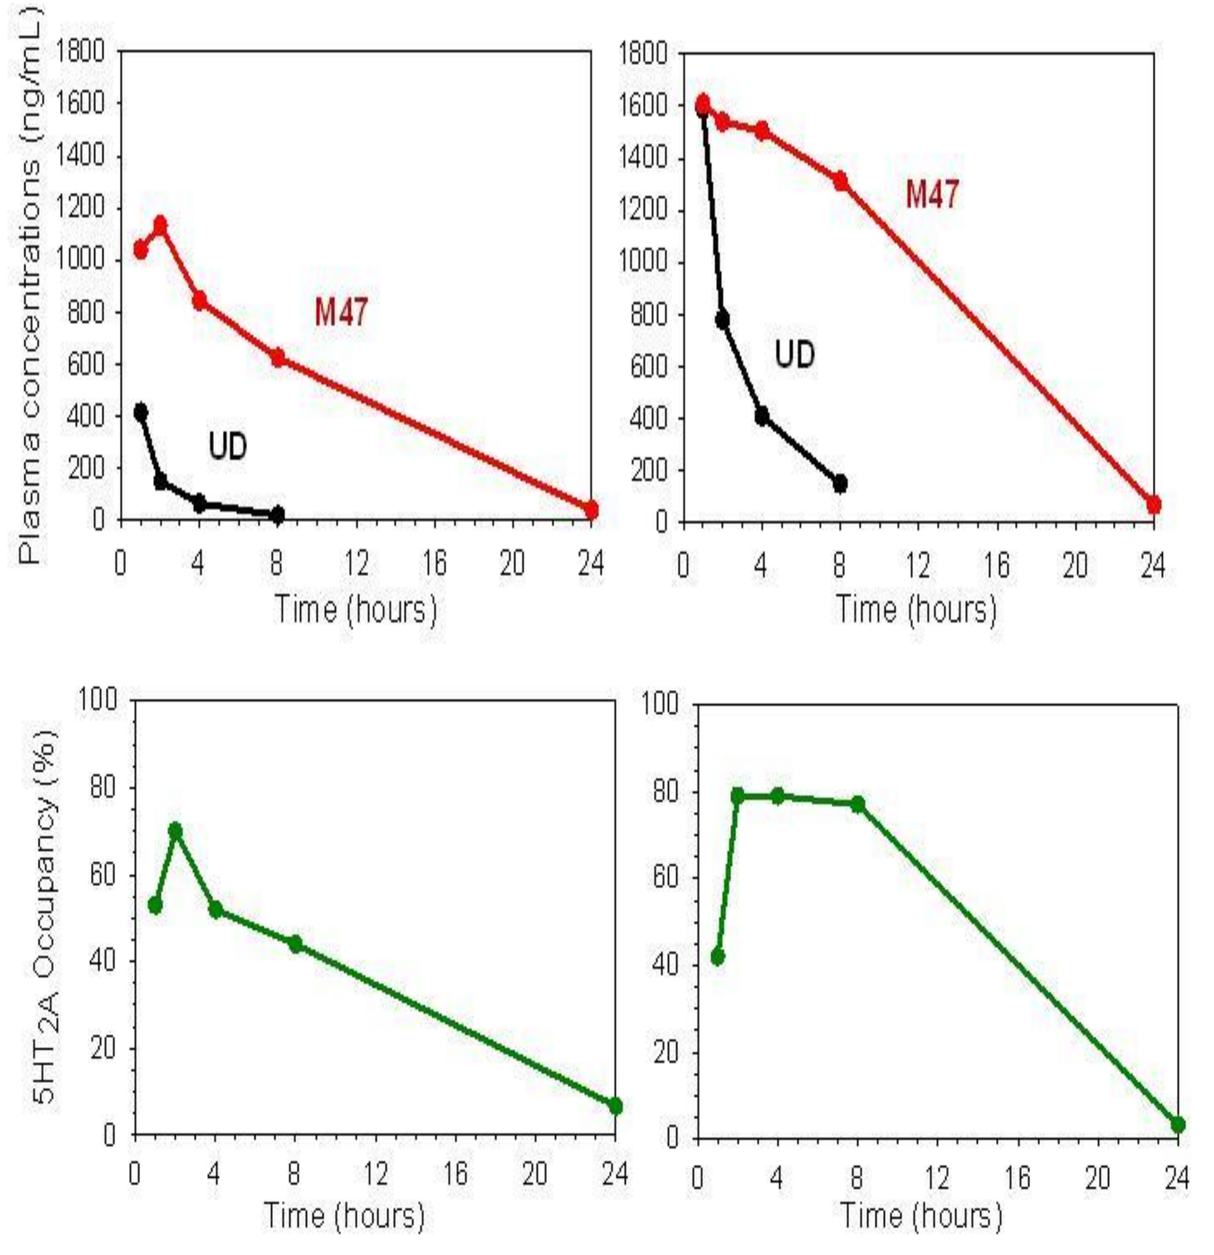
Plasma concentrations (ng/mL)
M47
UD
Time (hours)
5HT 2A Occupancy (%)

Exposure-5HT_{2A}R Occupancy Modeling in Rat

Model:

$$E = \underbrace{\frac{E_{max1} \cdot C_{UD}^{n1}}{EC_{50,UD}^{n1} + C_{UD}^{n1}}}_{\text{UD}} + \underbrace{\frac{E_{max2} \cdot C_{Met}^{n2}}{EC_{50,Met}^{n2} + C_{Met}^{n2}}}_{\text{Metabolite}}$$

E_{max} = maximal occupancy
EC_{50} = concentration reaching 50% occupancy
n = hill coefficient

Results:

Rat: $EC_{50,UD}$ = 5000 ng/mL | $EC_{50,Met}$ = 745 ng/mL

Man: Human fu corrected : $EC_{50,UD}$= 4167 ng/mL

Modeling experiments suggest that M47 significantly contributes to the 5-HT_{2A}R binding that is observed in rat. M47 is a very minor metabolite in humans and circulating levels are negligible. Therefore it may be hypothesized that in man 5-HT_{2A}R binding will be low.

Assumptions :
E_{max} fixed to 100 %,
n = 1
M45 is not contributing to the 5-HT_{2A}R occupancy

Predicted 5-HT_{2A}R occupancy in man around T_{max} at the anticipated clinical doses of 50 to 700 mg

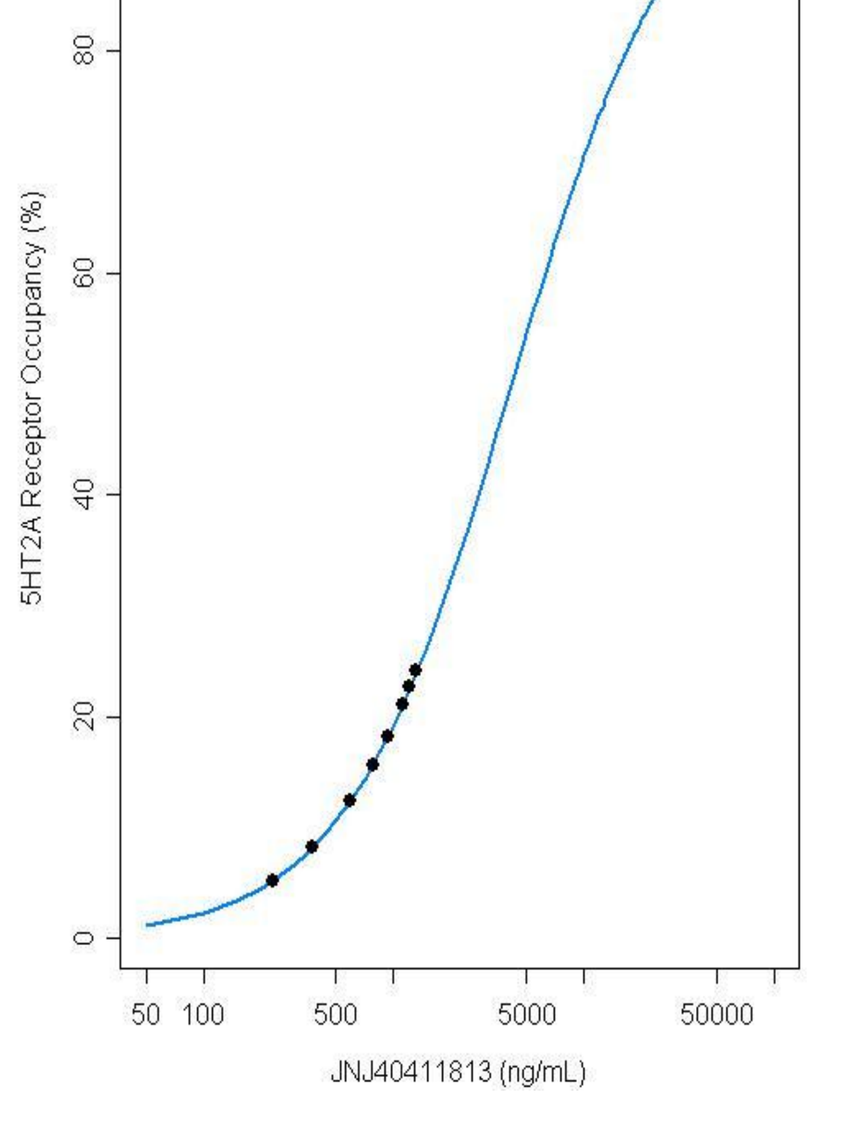
5HT2A Receptor Occupancy (%)
JNJ40411813 (ng/mL)

Prediction of 5-HT_{2A}R Occupancy in Man Following Single Oral Doses

Dose (mg)	50	100	200	300	700
Conc @ 2h* (95% CI)	232 (115-382)	371 (173-593)	591 (290-937)	777 (382-1246)	1323 (663-2080)
5HT_{2A} RO (95% CI)	5.3 (2.7-8.4)	8.2 (4-12)	12 (7-18)	16 (8-23)	24 (14-33)

* Mean 2h-concentrations based on simulation from exploratory pop PK model.

Human 5-HT_{2A}R PET Results

Predicted *vs.* Observed 5-HT_{2A}R Occupancy

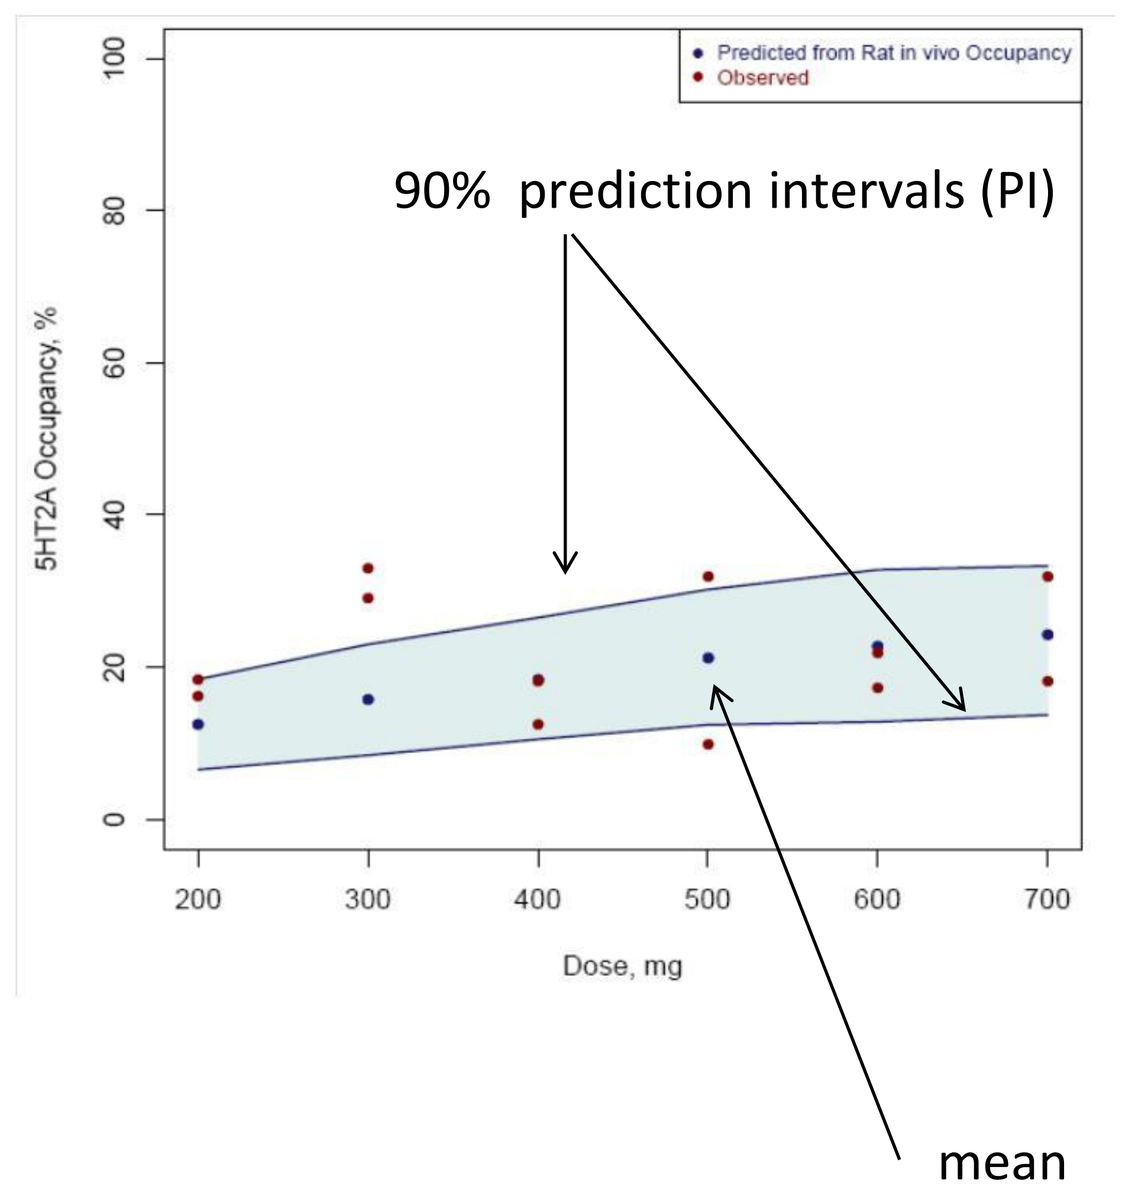
Predicted from Rat in vivo Occupancy
Observed
90% prediction intervals (PI)
5HT2A Occupancy, %
Dose, mg
mean

Observed Plasma Concentration of JNJ-40411813 *vs.* 5-HT_{2A}R Occupancy

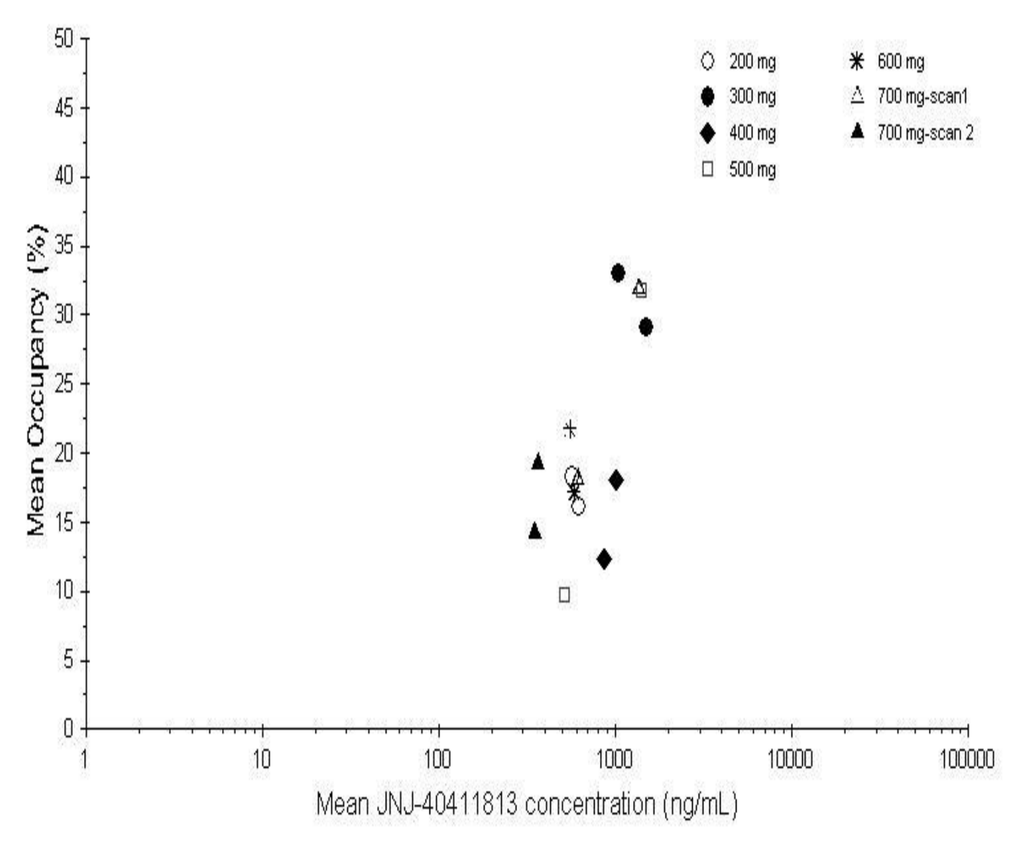
Mean Occupancy (%)
Mean JNJ-40411813 concentration (ng/mL)

The observed 5-HT_{2A}R occupancy falls within the 90% PIs. At clinical doses of 200 to 700 mg, predicted 5-HT_{2A}R occupancy in man ranged between 10% and 25%. The human PET data confirmed minimal 5-HT_{2A} R ocupancy by JNJ-40411813 in man, which is not expected to be clinically relevant. Maximal 5-HT_{2A}R occupancy of only 33% was reached with the studied doses. Due to this low occupancy, the estimated plasma concentrations associated with 50% 5-HT_{2A}R binding could not be reached.

Conclusions

5-HT_{2A}R occupancy could be predicted in man based on nonclinical pharmacology data in the rat, taking into account the difference in free fraction and the different contributions of parent and metabolite in rat *vs.* man.

At clinical doses, predicted 5-HT_{2A}R occupancy in man was low and in good agreement with observed 5-HT_{2A}R occupancy. A better strategy would have been to confirm the model by testing two doses only and based on outcome to stop the study or add additional doses to test 5-HT_{2A}R occupancy *vs.* exposure relationship.

This modeling work illustrates the "translatability" of *in vitro* and *in vivo* preclinical information to 5-HT_{2A}R occupancy in man.

References

[1] Swanson CJ et al. Metabotropic glutamate receptors as novel targets for anxiety and stress disorders. Nat. Rev. Drug. Discov. 2005; 4:131–144.
[2] Patil ST et al. Activation of mGlu2/3 receptors as a new approach to treat schizophrenia: a randomized Phase 2 clinical trial. Nat. Med. 2007; 13:1102–1107.
[3] Ito H et al. PET imaging of central 5-HT2A receptors with carbon-11-MDL 100,907. J. Nucl. Med. 1998; 39(1):208-214.

Acknowledgments The authors acknowledge the valuable contribution of all people involved in the generation of the results. This work is the result of a cross functional collaboration involving many people from Model Based Drug Development, Clin Pharm, Neuroscience Discovery, Translational Sciences EDD, Drug Safety Sciences, Experimental Medicine, and Compound Development Team. We would like to thank Addex Therapeutics for their involvement in the development of JNJ-40411813.