Model Informed Drug Discovery and Development of Novel Treatments for Hyperuricemia: From Systems Pharmacology to Mechanistic PK/PD

Elena Soto[1], Barry Weatherley[1], Massimo Cella[2], Pinky Dua[2], Scott Marshall[1]
[1]Pharmacometrics, Pfizer, UK; [2]Biotherapeutics Research Clinical Pharmacology, Pfizer Neuroscience and Pain RU, Cambridge, UK

Objectives:

The rising population prevalence of hyperuricemia (1) has renewed interest in the purine metabolic pathway (PMP). Dua et al (2) recently published a system pharmacology model which has determined sensitivity of the pathway to mono and dual inhibitory effects, a representation of this model is shown in Fig 1. This work highlighted the potential therapeutic value of simultaneous inhibition of XO (xanthine oxidase: enzyme responsible for oxidation of xanthine to uric acid (UA)) and a transporter responsible for the reabsorption of UA. This therapeutic strategy is currently being explored via combination therapy (3) or as a target for a dual inhibitor (4). The aim of this work is to develop a mechanistic PK/PD model to characterise the interaction between these points in the PMP in terms of impact on rate and extent of changes in serum and urinary concentrations of xanthine and UA.

Methods:

A PK/PD model was developed using NONMEM 7.2 based on the systems model but simplified and calibrated with healthy volunteer serum and urinary data from an internal compound and from literature data (5). The model consisted of 4 compartments, where xanthine will be either renally cleared or converted into UA prior to elimination to the UA urinary compartment (Fig 2). Drug effects were tested in the (*i*) UA formation as informed by the systems model, (*ii*) UA clearance as informed by the internal compound data, but also in the (*iii*) xanthine clearance as available literature indicated a XO inhibitor could increase xanthine clearance.

Simulations were performed assuming different daily doses of the internal compound for 10 days and assuming different magnitude of the drug effects in each potential route. For each of the scenarios simulated [(A) Strong UA clearance stimulation + weak UA formation inhibitor, (B) Weak UA clearance stimulation + strong UA formation inhibitor, (C) Weak UA clearance stimulation + strong xanthine clearance stimulation] the serum and urinary UA and xanthine profiles are shown.

Figure 3. Serum Uric Acid, serum xanthine, urinary uric acid and urinary xanthine time course profile for three different scenarios

A. Strong *UA clearance* stimulation + weak *UA formation* inhibition

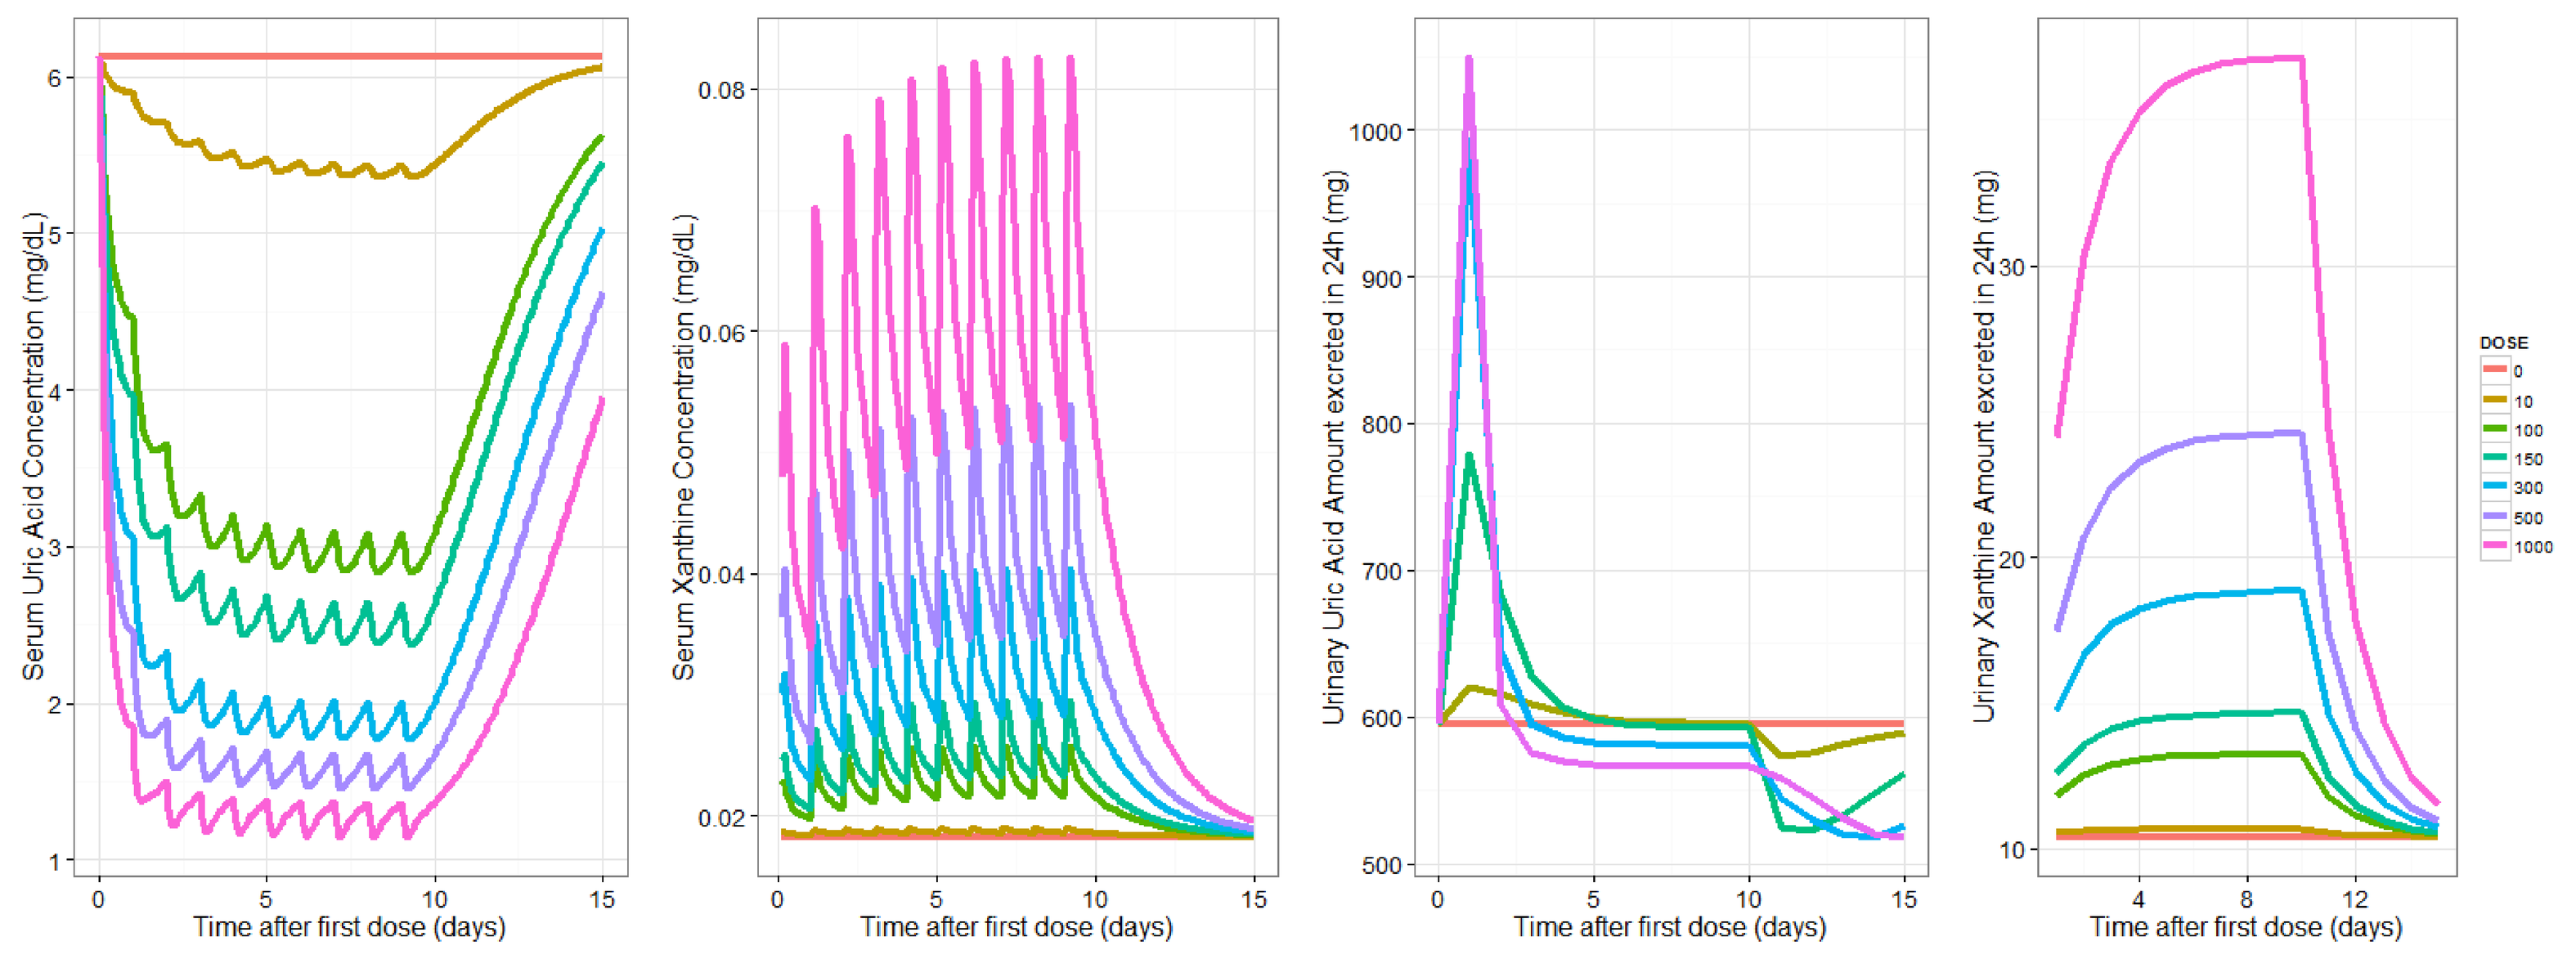
Serum Uric Acid Concentration (mg/dL)
Serum Xanthine Concentration (mg/dL)
Urinary Uric Acid Amount excreted in 24h (mg)
Urinary Xanthine Amount excreted in 24h (mg)
Time after first dose (days)
DOSE
0
10
100
150
300
500
1000

B. Weak *UA clearance* stimulation + strong *UA formation* inhibition

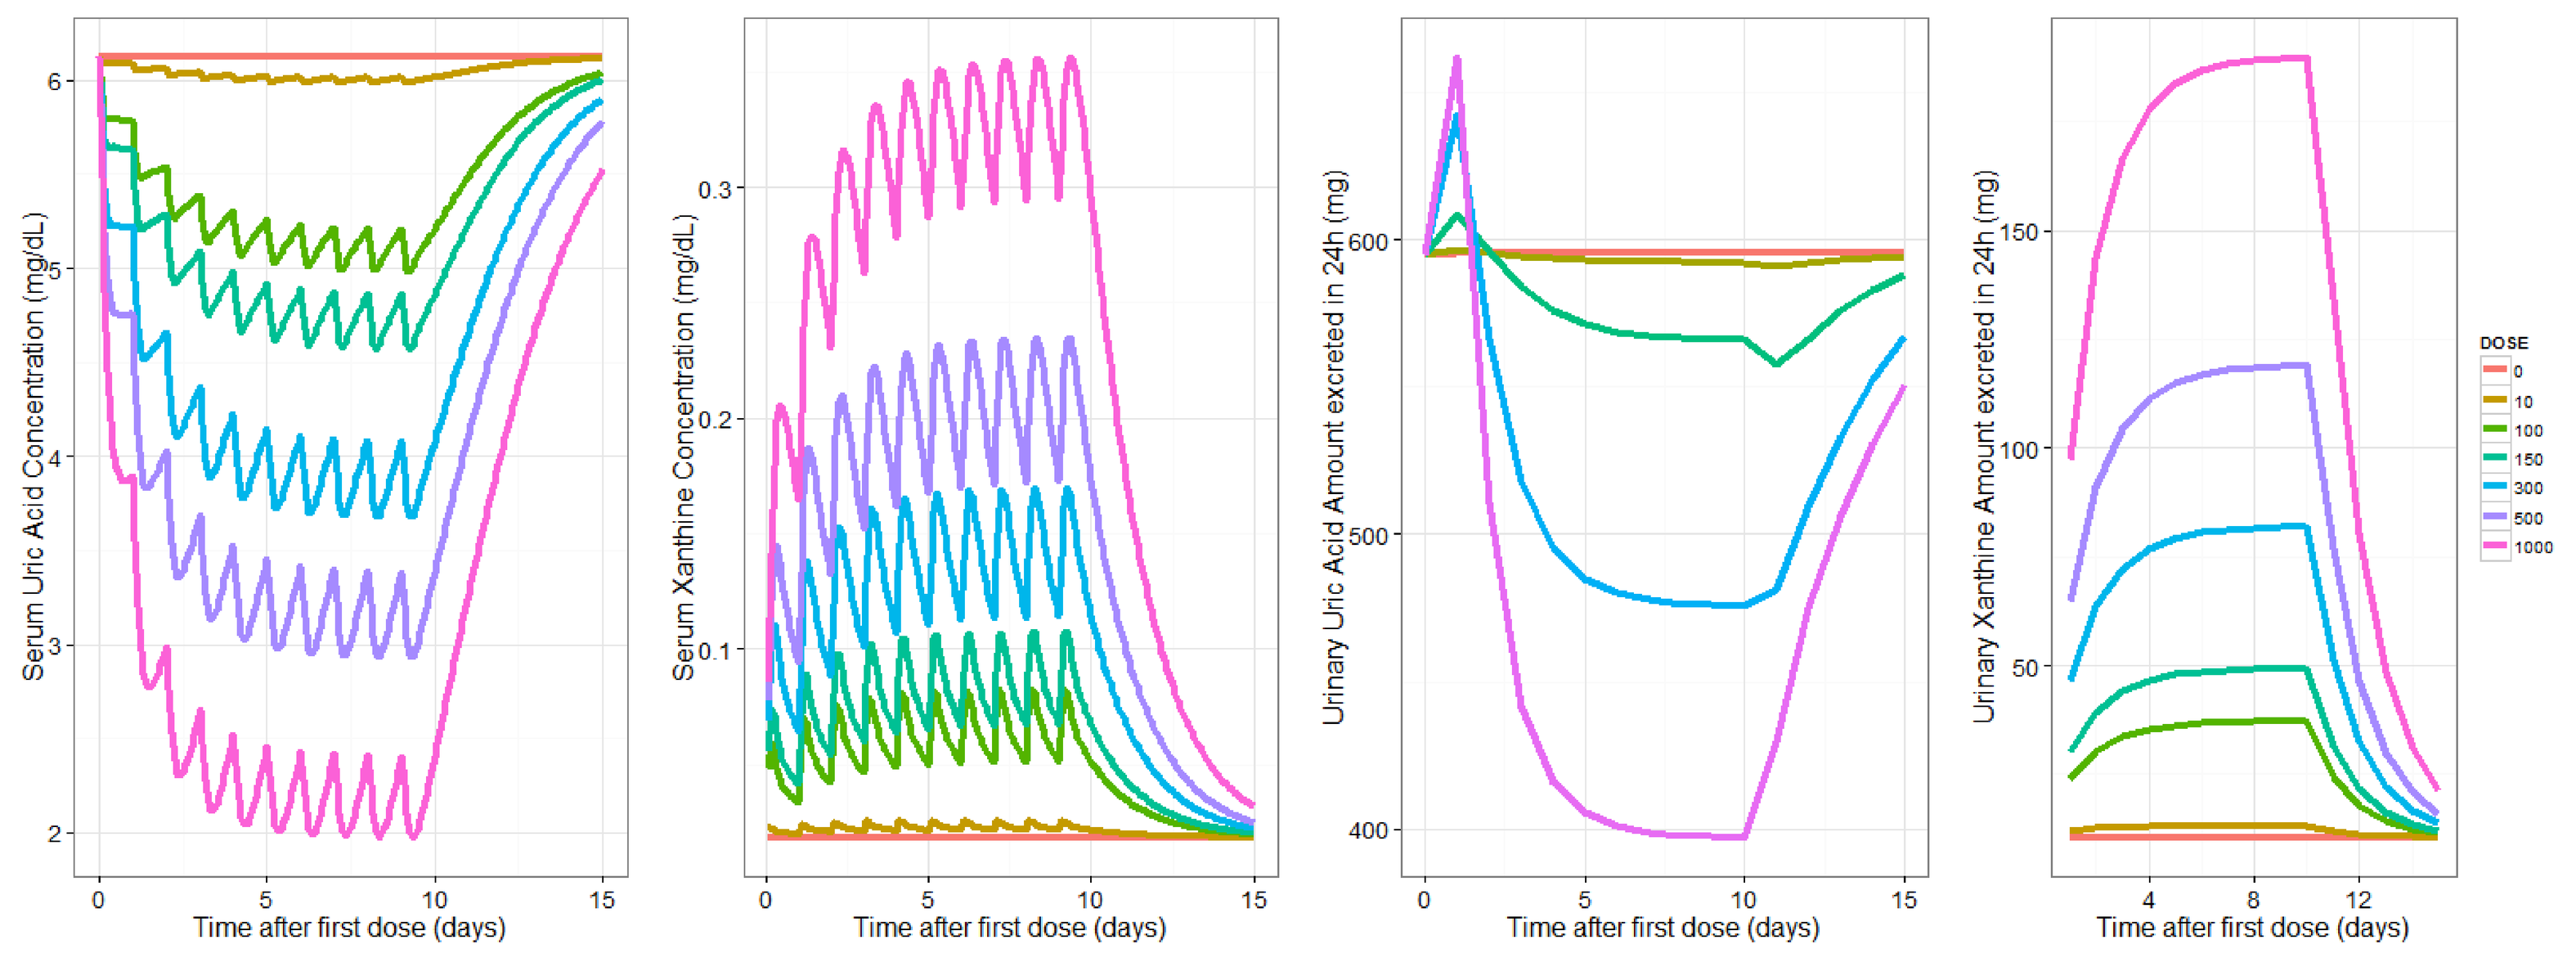
Serum Uric Acid Concentration (mg/dL)
Serum Xanthine Concentration (mg/dL)
Urinary Uric Acid Amount excreted in 24h (mg)
Urinary Xanthine Amount excreted in 24h (mg)
Time after first dose (days)
DOSE
0
10
100
150
300
500
1000

C. Week *UA clearance* stimulation + strong *xanthine clearance* stimulation

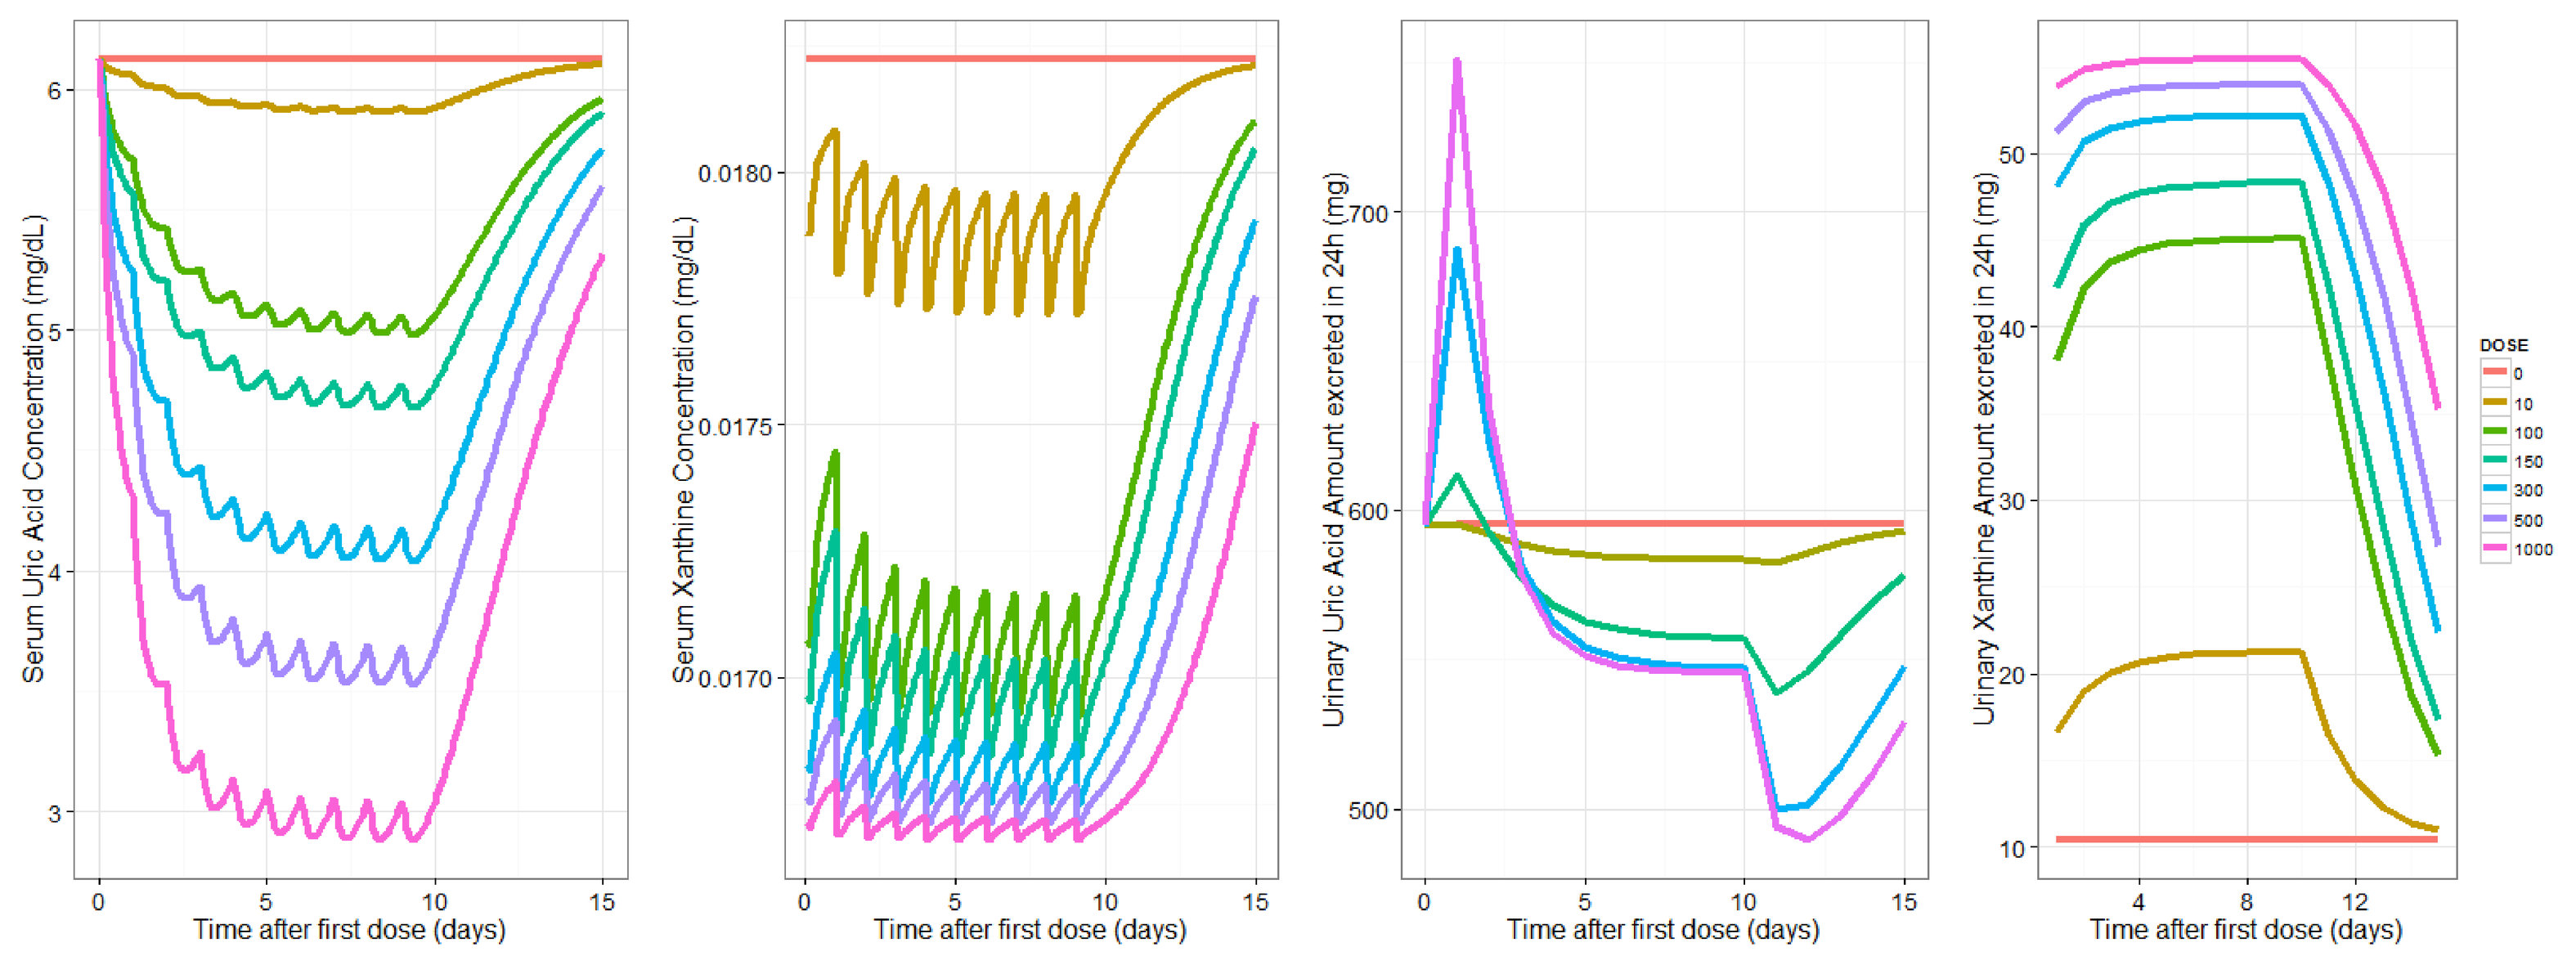
Serum Uric Acid Concentration (mg/dL)
Serum Xanthine Concentration (mg/dL)
Urinary Uric Acid Amount excreted in 24h (mg)
Urinary Xanthine Amount excreted in 24h (mg)
Time after first dose (days)
DOSE
0
10
100
150
300
500
1000

Figure 1

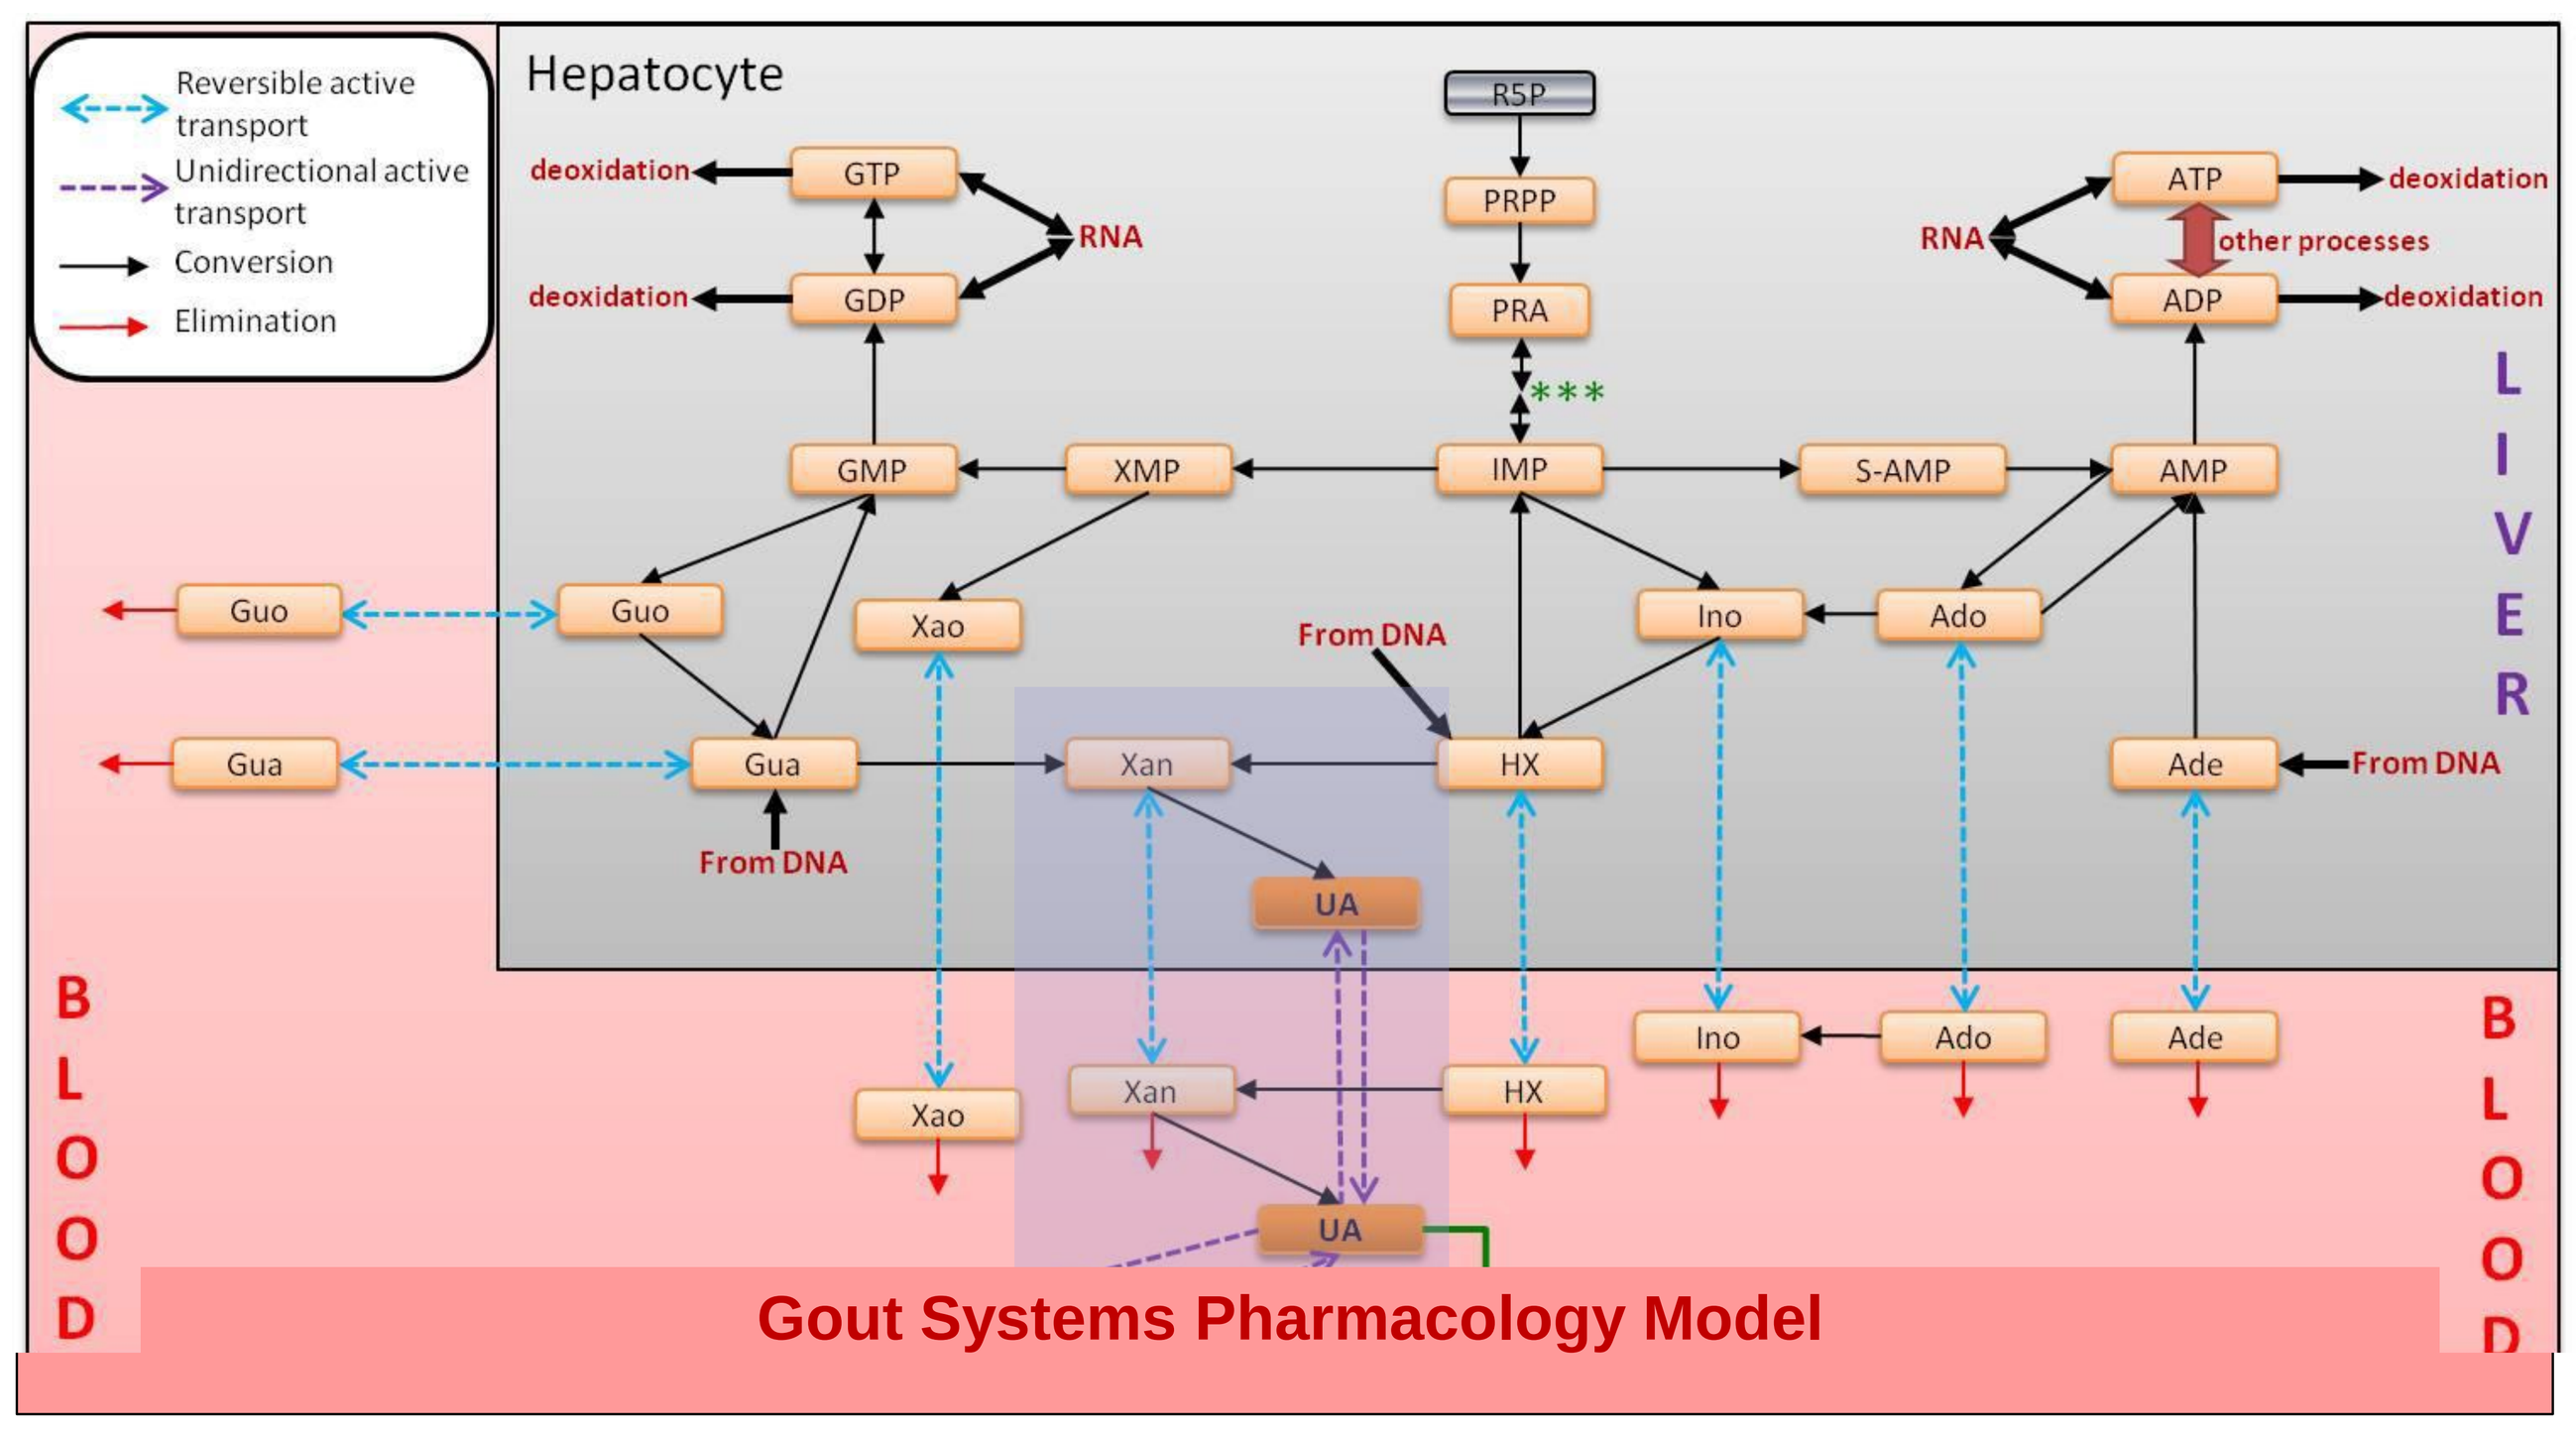
Reversible active transport
Unidirectional active transport
Conversion
Elimination
Hepatocyte
R5P
PRPP
PRA
IMP
GTP
GDP
GMP
XMP
S-AMP
AMP
ATP
ADP
RNA
deoxidation
other processes
Guo
Gua
Xao
Xan
HX
UA
Ino
Ado
Ade
From DNA
LIVER
BLOOD
Gout Systems Pharmacology Model

Figure 2. PKPD model

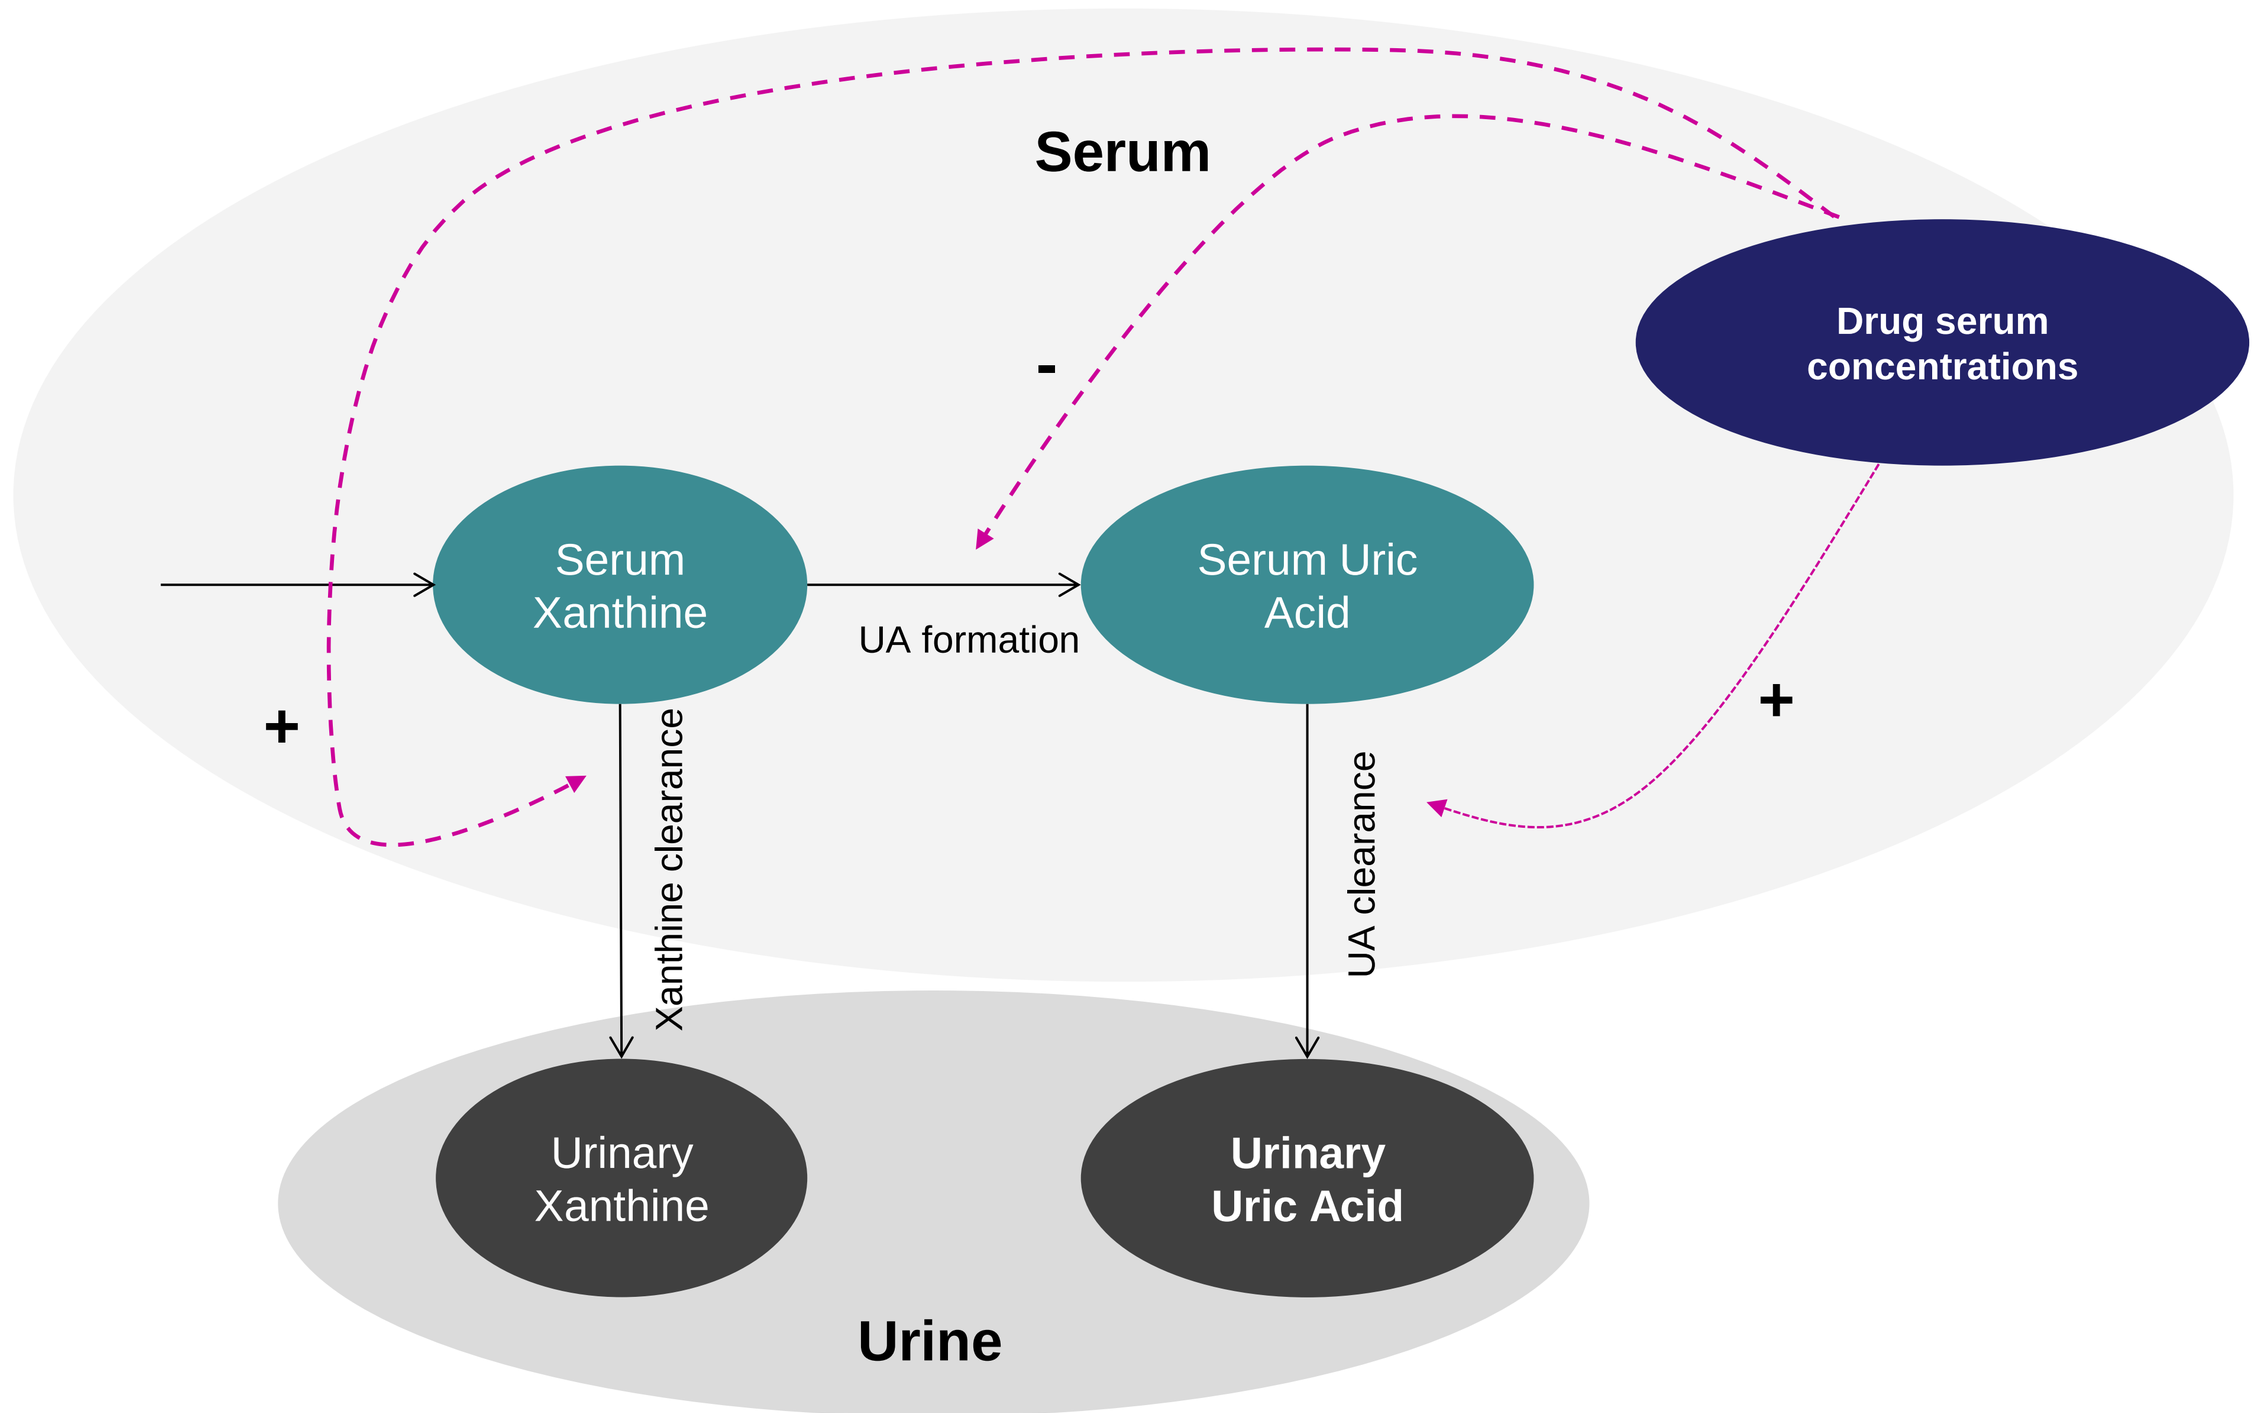
Serum
Drug serum concentrations
Serum Xanthine
Serum Uric Acid
UA formation
Xanthine clearance
UA clearance
Urinary Xanthine
Urinary Uric Acid
Urine

Results:

Simulations showed that the three potential drug mechanisms explored have similar effects in the serum UA profile, however very different effects in the urinary profiles. A strong UA clearance effect, produces fast and large increase in the amount of UA present in urine which then returns to baseline levels (or below due to the UA formation inhibitor effect) as seen in Fig 3A. The rapid increase in UA in urine can be reduced by the presence of a stronger UA formation inhibitor (Fig 3B) that not only decreases the peak UA in urine during the first days but also reduces the UA amounts in urine below baseline levels. However the strong UA formation inhibitor produces a significant increase in the xanthine amount in urine. A strong xanthine clearance stimulation effect will produce urinary profiles similar to the UA formation inhibition however the increase in the urinary xanthine will be much faster than the UA formation inhibition. As high levels of UA in urine are not desirable further simulation work was done to explore other possibilities to reduce urinary UA in the first days of treatment; Figure 4 shows how titration could also reduce the UA peak.

Figure 4. Urine Uric Acid time profiles with and without titration

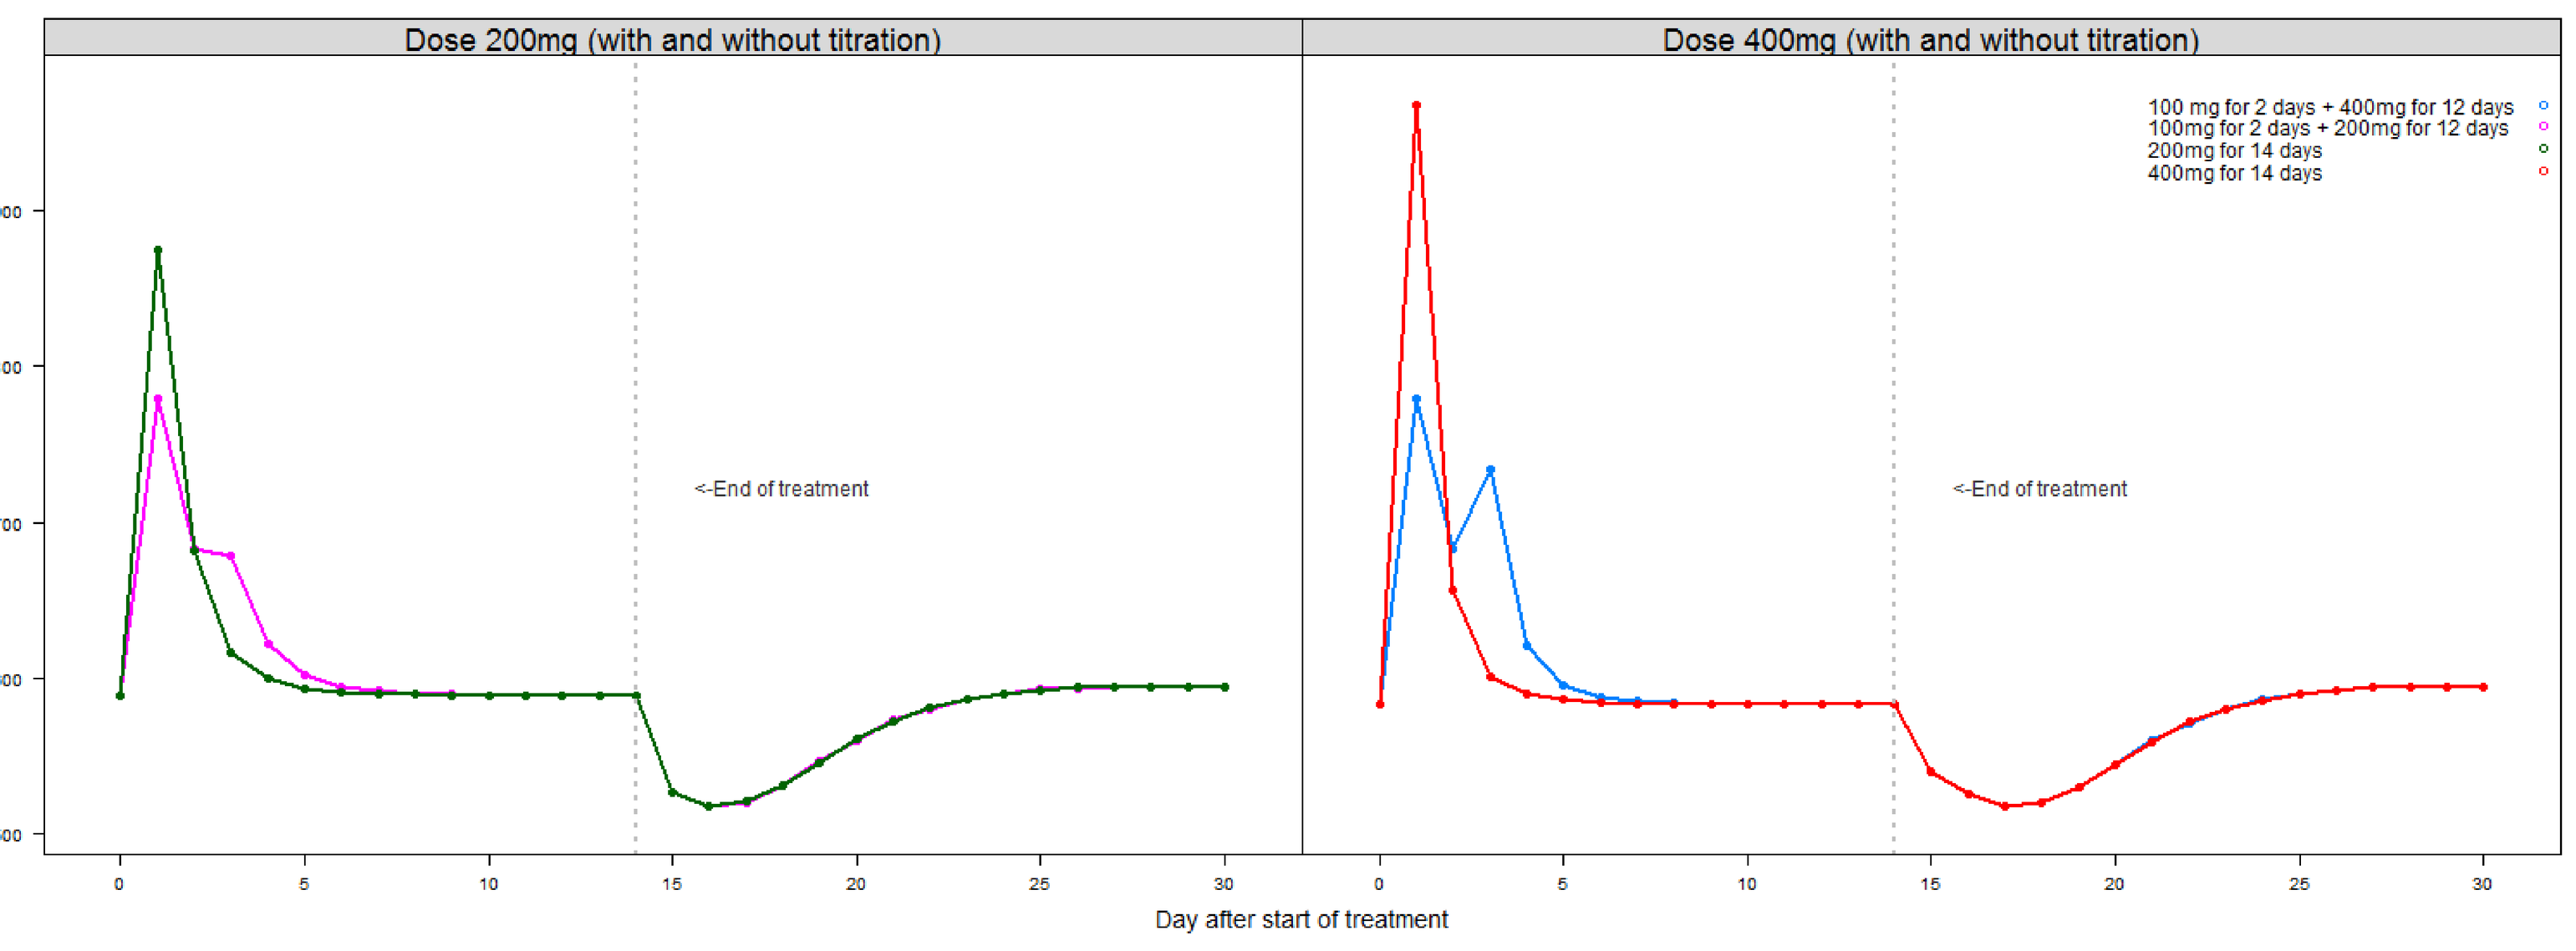
Dose 200mg (with and without titration)
Dose 400mg (with and without titration)
100 mg for 2 days + 400mg for 12 days
100mg for 2 days + 200mg for 12 days
200mg for 14 days
400mg for 14 days
<-End of treatment
Uric Acid excreted in Urine every 24hours (mg)
Day after start of treatment

Conclusions:

- Simulations provided greater insight into the interaction between key mechanisms and how balancing those may help optimize treatment with existing and emerging mono therapies while providing targets for future dual inhibitor development.
- A mechanistic PK/PD model was built based on systems pharmacology information, literature information and internal data. It has explored a renal component that the systems pharmacology model had not explored at this time. Further work in the systems pharmacology model can be based on the renal component exploration that has been done with the PK/PD model.
- This case study also illustrates how systems pharmacology can help define specific research questions which can be explored by focused mechanistic PK/PD modelling, which can ultimately be fed back into the wider systems pharmacology model.

References
1. Yanyan Zhu, ARTHRITIS & RHEUMATISM Vol. 63, No. 10, October 2011, pp 3136–3141.
2. Dua et al, A systems pharmacology of purine metabolism to explore novel options for the treatment of gout, 7th Noordwijkerhout Symposium on Pharmacokinetics, Pharmacodynamics and Systems Pharamcology. April 23-25 2014, in Noordwijkerhout, The Netherlands.
3. Xie et al, Relationship between the Dose of Urate Lowering Therapies and Serum Uric Acid in Healthy Volunteers and Gout Patients: A Model Based Meta-Analysis (MBMA), PAGE 2015.
4. Warrell Raymond P et al, Profound Hypouricemia Induced in Human Subjects By Novel Bifunctional Inhibitors of Xanthine Oxidase and URAT1. American College of Rheumatology meeting abstracts, abstract number 830, 2014.
5. Khosravan et al, Pharmacokinetics, Pharmacodynamics and Safety of Febuxostat, a non-purine selective inhibitor of xanthine Oxidase, in a Dose Escalation Study in Healthy Subjects. Clin Pharmacokinetics 2006, 45 (8): 821-841.