Improved parameter estimation and design optimization for *In Vitro* ligand binding experiments

UPPSALA UNIVERSITET

C. Steven Ernest II[1,2], Andrew C. Hooker[1] and Mats O. Karlsson[1]

[1]Department of Pharmaceutical Biosciences, Uppsala University, Sweden.

[2]Eli Lilly and Company, Indianapolis, IN USA

Background and Objective

Estimation of ligand binding parameters to receptors *in vitro* constitutes a basic operation in many areas of scientific research. Therefore, efficient and accurate determination of these parameters is paramount for evaluation of ligand potency, interactions with other molecules, and prediction of *in vivo* performance. Previous analyses have demonstrated that simultaneous non-linear regression (SNLR) versus sequential non-linear regression (NLR) provided better approximation to the true values of ligand binding parameter estimates[1].

The aim of this study was to extend this previous work and compare SNLR and NLR with commonly encountered experimental error, specifically residual variability (RUV) of binding measurements, experiment to experiment variability (BEV) and non-specific binding (B_{NS}). Additionally, optimal design (ligand concentrations and sampling times) of these ligand binding experiments was examined using the optimal design software PopED 2.08 [2] for SNLR analysis.

Methods

Monte Carlo simulation and estimation were used to evaluate the performance of various NLR and SNLR methods for estimating ligand binding parameters. Data simulation and estimation were performed using FOCE(I) in NONMEM VI. Simulation followed by parameter estimation of ligand binding data was performed for equilibrium, dissociation, association and non-specific binding experiments using the following mathematical equations:

Equilibrium: $$B_{eq}=\sum_{r=1}^{n}\frac{B_{\max,r}*L}{\frac{k_{-r}}{k_r}+L}+\alpha*L$$

Dissociation: $$B_{dis,t}=\sum_{r=1}^{n}\frac{B_{\max,r}}{1+\frac{k_{-r}}{L*k_r}}*\left[1-e^{-(L*k_r+k_{-r})*t)}\right]*e^{-k_{-r}(t-\tau)}+\alpha*L$$

Association: $$B_{ass,t}=\sum_{r=1}^{n}\frac{B_{\max,r}}{1+\frac{k_{-r}}{L*k_r}}*\left[1-e^{-(L*k_r+k_{-r})*t)}\right]+\alpha*L$$

B_{NS}: $$B_{NS}=\alpha*L$$

Briefly, NLR was performed by estimating parameters from one experiment and then introducing one or more of the parameters k_1, k_{-1}, B_{max} or α as constants for subsequent analyses. SNLR was performed by fitting data simultaneously from each experiment.

Results

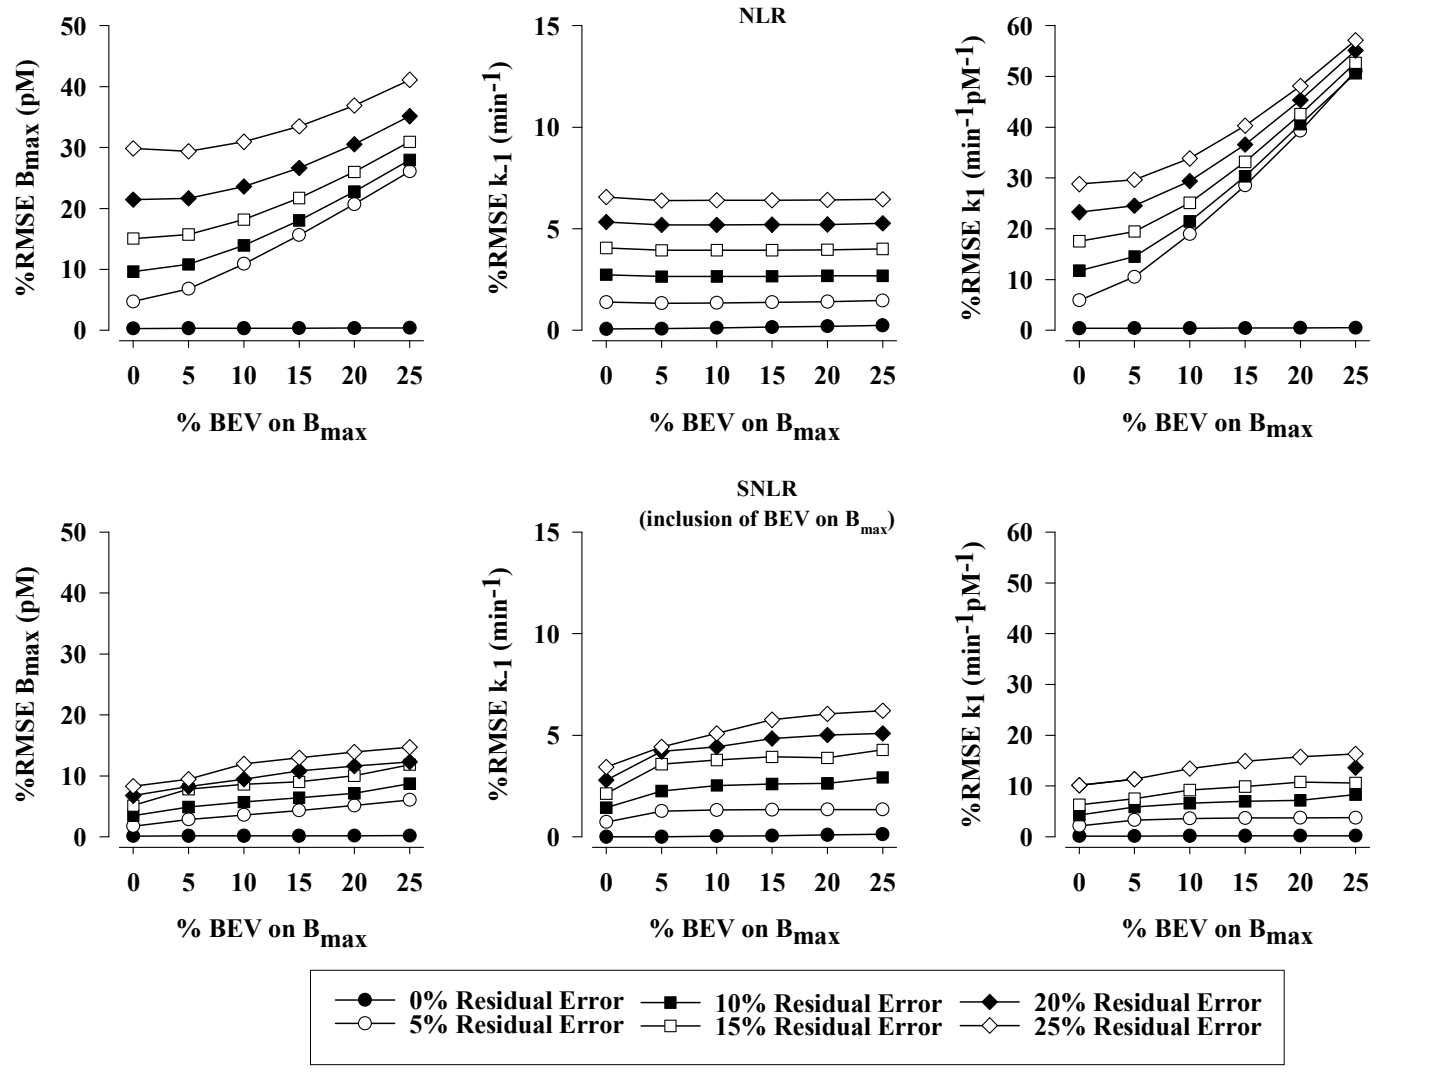
NLR
SNLR
(inclusion of BEV on Bmax)
%RMSE Bmax (pM)
%RMSE k-1 (min-1)
%RMSE k1 (min-1pM-1)
% BEV on Bmax
0% Residual Error
5% Residual Error
10% Residual Error
15% Residual Error
20% Residual Error
25% Residual Error

Figure 1. *Percent RMSE of B_{max}, K_{-1} and K_1 estimates using the NLR and SLNR methods with BEV on B_{max}.*

Across the span of residual error supplied in these simulations, both methods typically provided unbiased model based parameter estimates (<1%) except for estimate of B_{max} using NLR (<8%). SNLR showed higher parameter precision than NLR. If BEV is ignored in estimation of parameters both NLR and SNLR demonstrate reasonably comparable increases in bias of parameters. The inclusion of BEV parameters in SNLR estimation provided substantially decreased bias in estimates and improved precision of parameter estimates (Fig. 1).

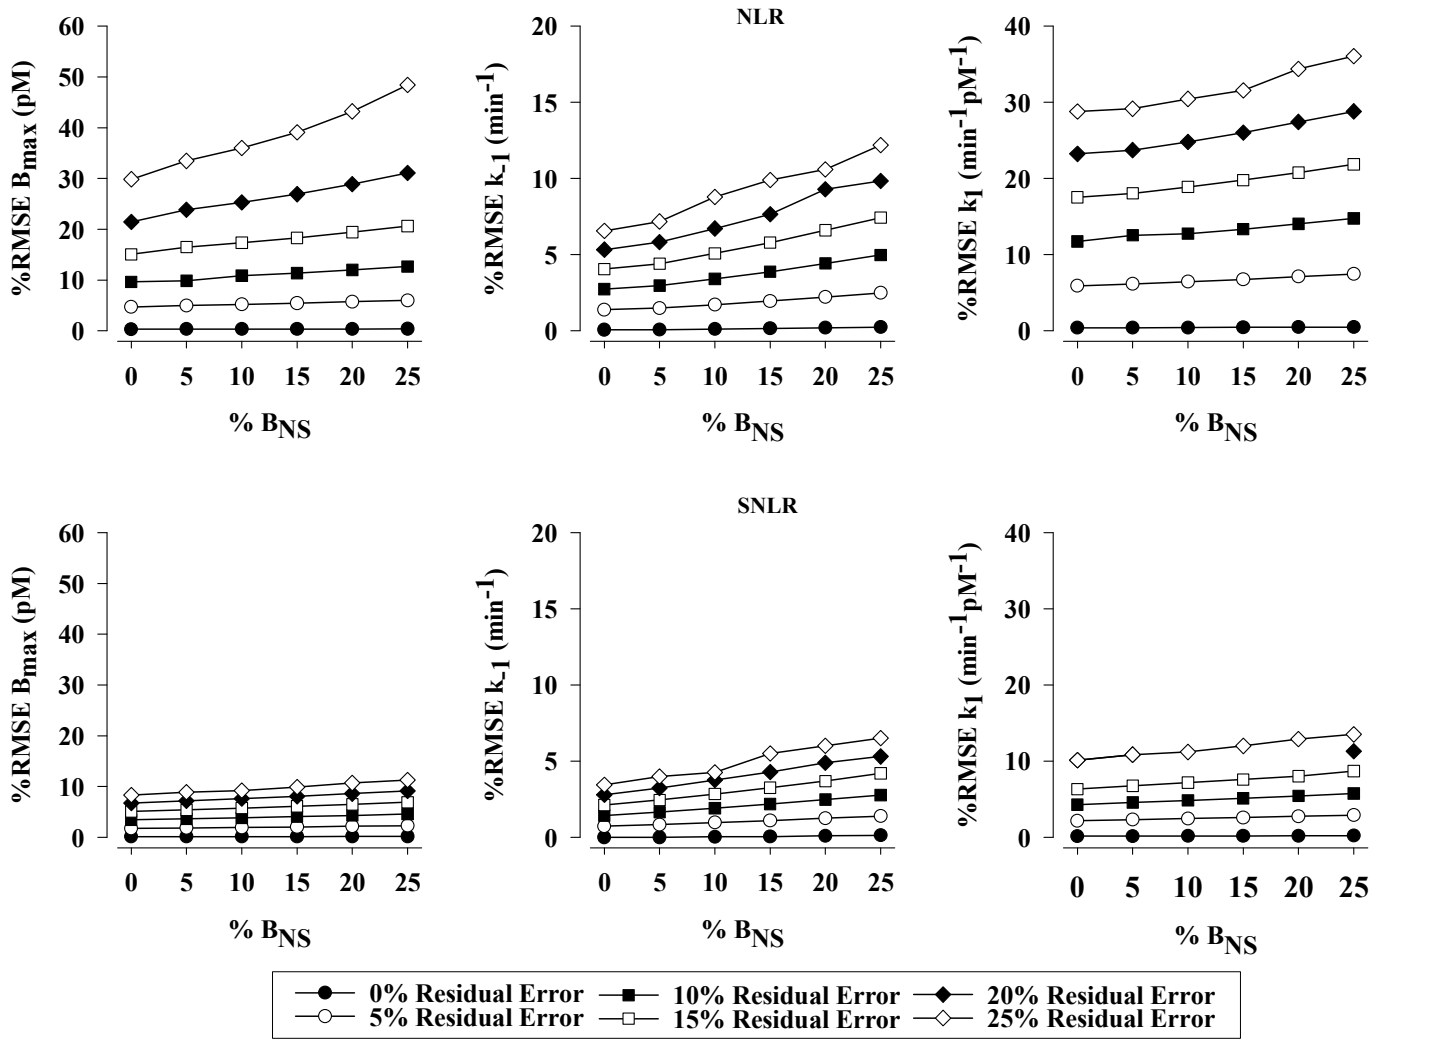
NLR
SNLR
%RMSE Bmax (pM)
%RMSE k-1 (min-1)
%RMSE k1 (min-1pM-1)
% BNS
0% Residual Error
5% Residual Error
10% Residual Error
15% Residual Error
20% Residual Error
25% Residual Error

Figure 2. *Percent RMSE of B_{max}, K_{-1} and K_1 estimates using the NLR and SLNR with addition of α for B_{NS}.*

Subtraction of B_{NS} from total binding measurements leads to significantly biased estimates of all parameters for both NLR and SNLR (not shown). Parameters estimates are improved by estimating specific binding α; SNLR preformed considerably better than NLR (Fig. 2).

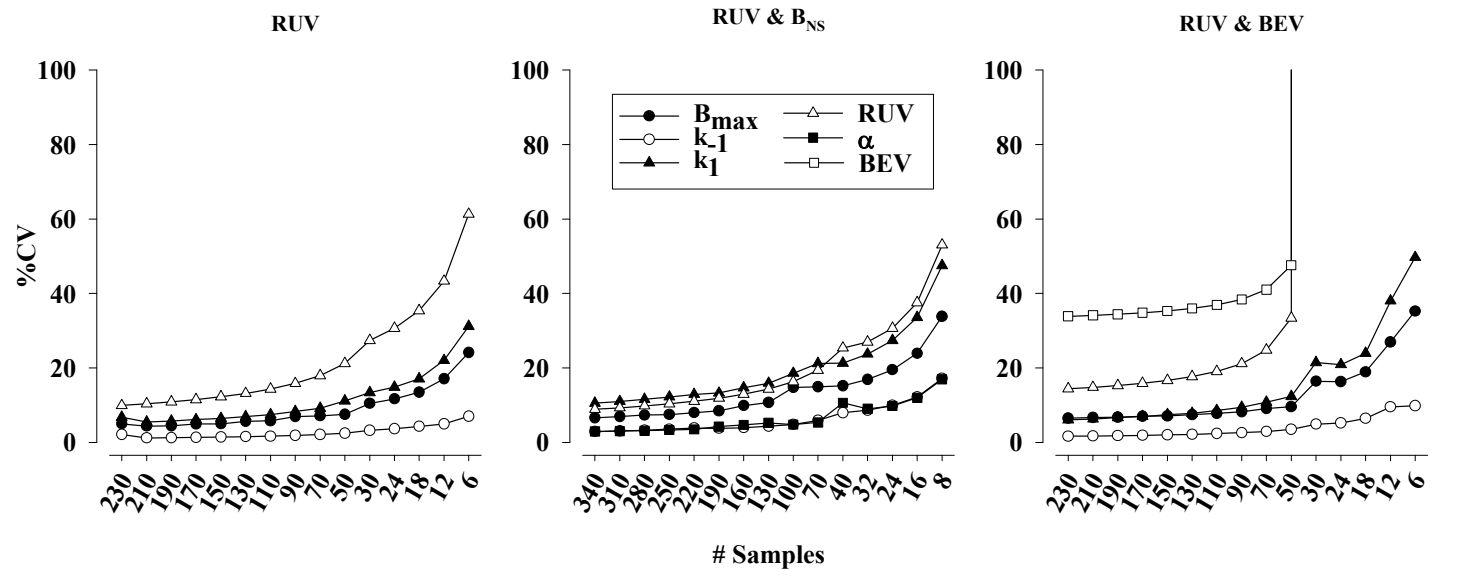
RUV
RUV & BNS
RUV & BEV
%CV
Bmax
k-1
k1
RUV
α
BEV
Samples

Figure 3. *Expected %CV for ligand binding parameters based on D-optimality from PopED with RUV, B_{NS} and BEV.*

Optimization of the design setup demonstrated that a reduced total number of samples provided relatively the same information as that with full sampling (Fig. 3). Additionally, optimizing the experimental design reduced the needed number of different ligand concentrations (similar to dose groups) from five to two.

Conclusions

Overall, SNLR provided superior parameter estimation in both precision and accuracy compared to NLR. In addition, substantial improvement can be made to the design of these experiments enabling a large reduction (>50%) in the samples/ligand concentrations needed to estimate parameters with high certainty.

References

1. *Karlsson M.O. and Neil A. (1988) Estimation of Ligand Binding Parameters by Simultaneous Fitting of Association and Dissociation Data: A Monte Carlo Simulation Study. Mol Pharmacol 35:59-66.*
2. *PopED, version 2.08 (2008) http://poped.sf.net.*