Finerenone Dose-Exposure-Response for the Primary Kidney Outcome in FIDELIO-DKD Phase III: Population Pharmacokinetic and Time-to-Event Analysis

Paul van den Berg[1], Martijn Ruppert[1], Emir Mesic[1], Nelleke Snelder[1], Andreas Seelmann[2] · Roland Heinig[2] ·Amer Joseph[2] · Dirk Garmann[2] · Joerg Lippert[2] · Thomas Eissing[2]

[1]LAP&P Consultants Leiden, The Netherlands; [2] Bayer AG, Pharmaceuticals R&D, Germany. Contact: info@lapp.nl

Introduction

Finerenone reduces the risk of kidney failure in patients with chronic kidney disease (CKD) and type 2 diabetes (T2D) [1]. Phase III study FIDELIO-DKD investigated the efficacy and safety of finerenone compared to placebo on top of standard-of-care in patients with CKD and T2D [2]. Patients received either finerenone (starting dose depended on eGFR at screening) or placebo p.o. once daily and could be up- or down-titrated according to changes in serum potassium and eGFR and at the discretion of the investigator [2,3]. The primary endpoint of FIDELIO-DKD was a composite of (1) time to the first occurrence of kidney failure, (2) a sustained decrease of eGFR ≥ 40% from baseline over at least 4 weeks, or (3) renal death.

Objectives

- Characterize the PK of finerenone in patients from FIDELIO-DKD, including identification of covariate effects.
- Provide posthoc estimates for exposure-response analyses.
- Characterize the relationship between finerenone exposure and the time to reach the primary kidney endpoint, including investigation of selected prognostic factors (PFs).

Data & Methods

- Data from pivotal phase 3 trial FIDELIO-DKD included in analysis:
 - 5,674 subjects, randomised (1/1) to placebo or finerenone (10 mg/20 mg)
 - 5057 finerenone plasma concentrations (from 2284 subjects)
 - 1104 primary composite kidney events (504 / 2833 patients (17.8%) in the finerenone group and in 600 / 2841 patients (21.1%) in the placebo group) [4].
- Non-linear mixed-effects modelling (NONMEM 7.4.3)
- PopPK model development
 - Existing model used as starting point [5]
 - Stepwise covariate modelling
 - Simulations done to support labelling: (1) assess the magnitude and uncertainty of a single covariate effect and (2) to compare the finerenone exposure at steady-state in subgroups of interest taking combined individual covariate effects into account.
- Kidney TTE model development
 - Placebo model (CPH screening / lumping of PF, hazard shape + PF (full model + BWD)
 - Implement exposure-response (model-predicted individual PK drives effect on hazard)
 - Investigate potential effect of long-term sodium glucose cotransporter 2 inhibitor (SGLT2i) use on the effect of Finerenone.

Results

PopPK model

- A two-compartmental population PK model with volumes set equal and absorption through a series of transit compartments and first-order elimination adequately captured the finerenone concentration–time data in FIDELIO-DKD [6].
- Single covariate effects (Fig. 1) were generally contained within the equivalence range of 80–125% around typical $C_{max,md}$ and $AUC_{\tau,md}$.
- Simulations in subgroups of interest were also generally contained within the 80–125% range when comparing the median $C_{max.md}$ and $AUC_{\tau.md}$ values (see Fig. 2 for CKD subgroups).

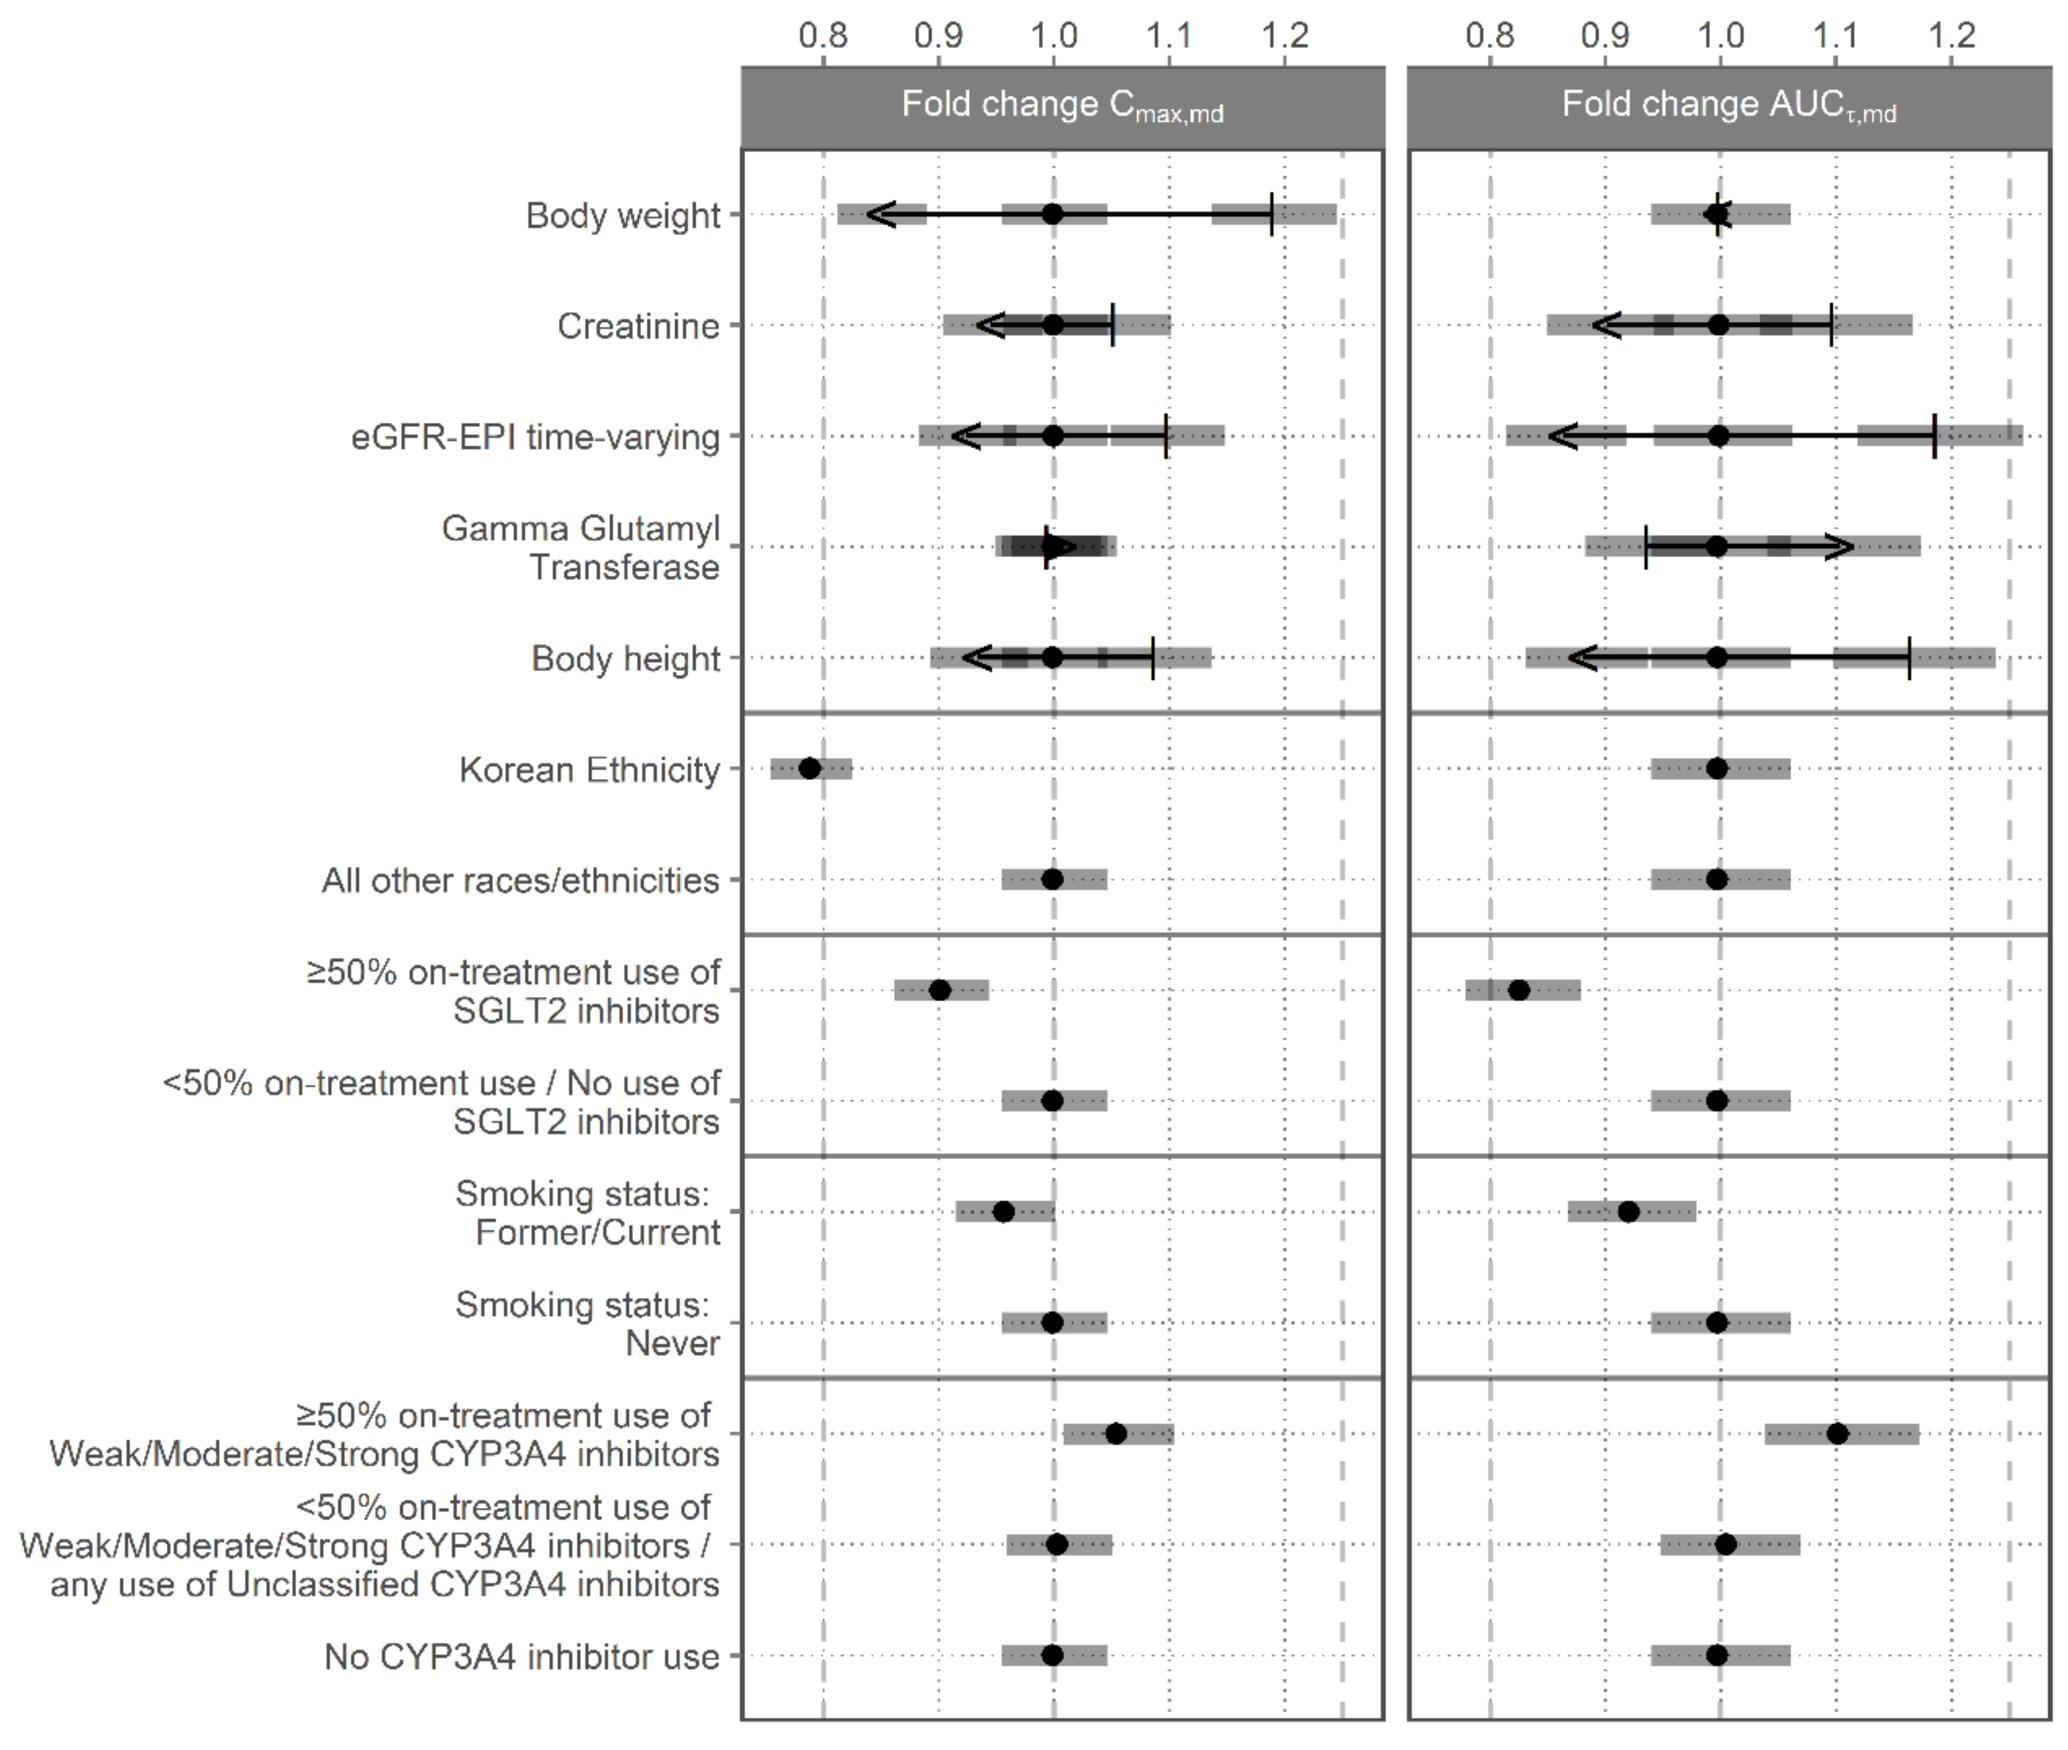
Fold change Cmax,md
Fold change AUCτ,md
0.8
0.9
1.0
1.1
1.2
Body weight
Creatinine
eGFR-EPI time-varying
Gamma Glutamyl Transferase
Body height
Korean Ethnicity
All other races/ethnicities
≥50% on-treatment use of SGLT2 inhibitors
<50% on-treatment use / No use of SGLT2 inhibitors
Smoking status: Former/Current
Smoking status: Never
≥50% on-treatment use of Weak/Moderate/Strong CYP3A4 inhibitors
<50% on-treatment use of Weak/Moderate/Strong CYP3A4 inhibitors / any use of Unclassified CYP3A4 inhibitors
No CYP3A4 inhibitor use

Figure 1. Forest plots illustrating the influence of the identified covariate effects on $C_{max,md}$ and $AUC_{\tau,md}$ relative to the median covariate value (continuous) or reference subgroup (categorical). Black dots: reference or the fold change relative to the reference. Whiskers shaped as an arrow to indicate the direction of the effect: PK parameter values at the 5th–95th percentiles of the covariate distribution. Gray bars: uncertainty shown as 5th – 95th percentiles of the simulated 5th percentile, median, and 95th percentile. Vertical dashed lines indicate unity and general acceptance range for equivalence of 0.8–1.25.

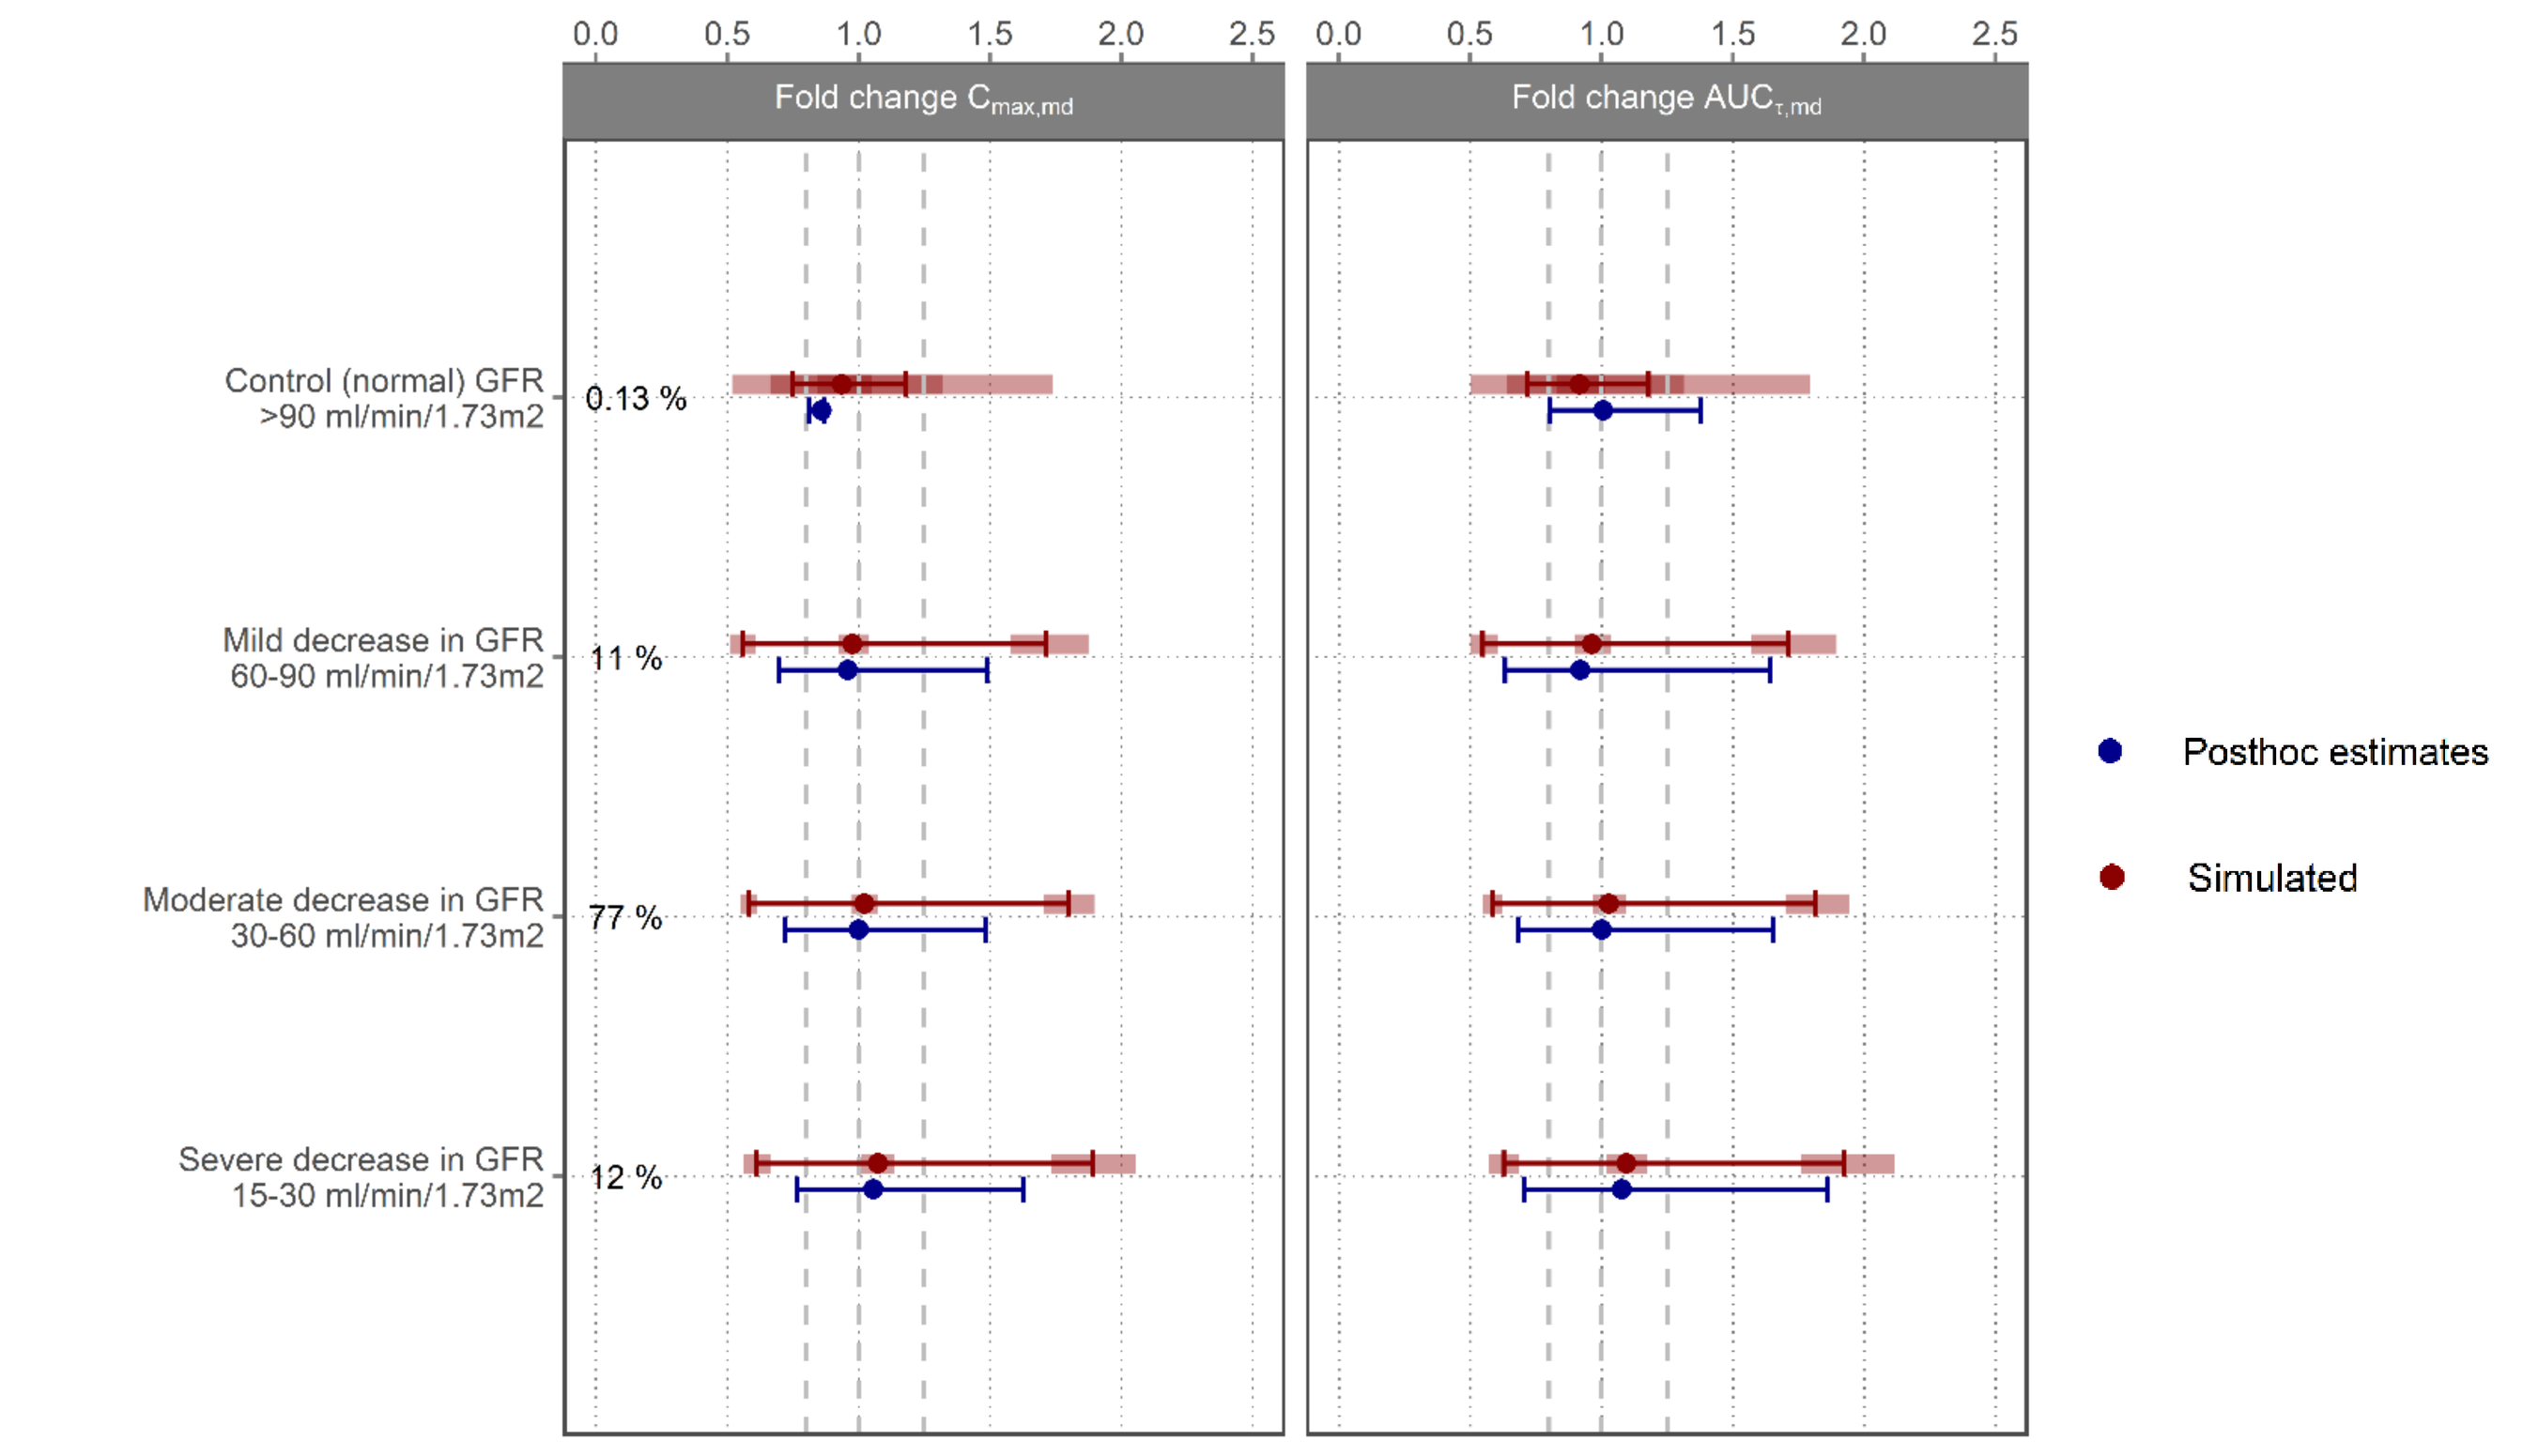
Fold change Cmax,md
Fold change AUCτ,md
0.0
0.5
1.0
1.5
2.0
2.5
Control (normal) GFR >90 ml/min/1.73m2
0.13 %
Mild decrease in GFR 60-90 ml/min/1.73m2
11 %
Moderate decrease in GFR 30-60 ml/min/1.73m2
77 %
Severe decrease in GFR 15-30 ml/min/1.73m2
12 %
Posthoc estimates
Simulated

Figure 2. Forest plots comparing the $C_{max,md}$ and $AUC_{\tau,md}$ between eGFR groups relative to the overall population. The % indicates the percentage of subjects within each group when compared to the total population of subjects in analyzed data. Red dots and whiskers: median and 5th – 95th percentiles of the $C_{max,md}$ and $AUC_{\tau,md}$ ratio using a subject with median $C_{max,md}$ and $AUC_{\tau,md}$ as reference. The red areas indicate the uncertainty (5th – 95th percentiles of the simulated 5th percentile, median, and 95th percentile). Blue dots and whiskers represent the median and 5th – 95th percentiles of the $C_{max,md}$ and $AUC_{\tau,md}$ ratio using a subject with median $C_{max,md}$ and $AUC_{\tau,md}$ as reference, based on the posthoc estimates. Vertical dashed lines indicate unity and the 0.8 – 1.25 lines.

Kidney TTE model

- A Weibull hazard model with model predicted finerenone plasma concentration driving the exposure-response (E_{max} model) adequately captured the kidney composite TTE data (Fig. 3).
- Half-maximal effect concentration and the maximal hazard decrease were estimated at 0.166 µg/L and -36.1%, respectively.
- PFs (treatment-independent) included a low eGFR and a high urine albumin-creatinine ratio increasing the risk, while concomitant sodium-glucose transport protein 2 inhibitor (SGLT2i) use decreased the risk.

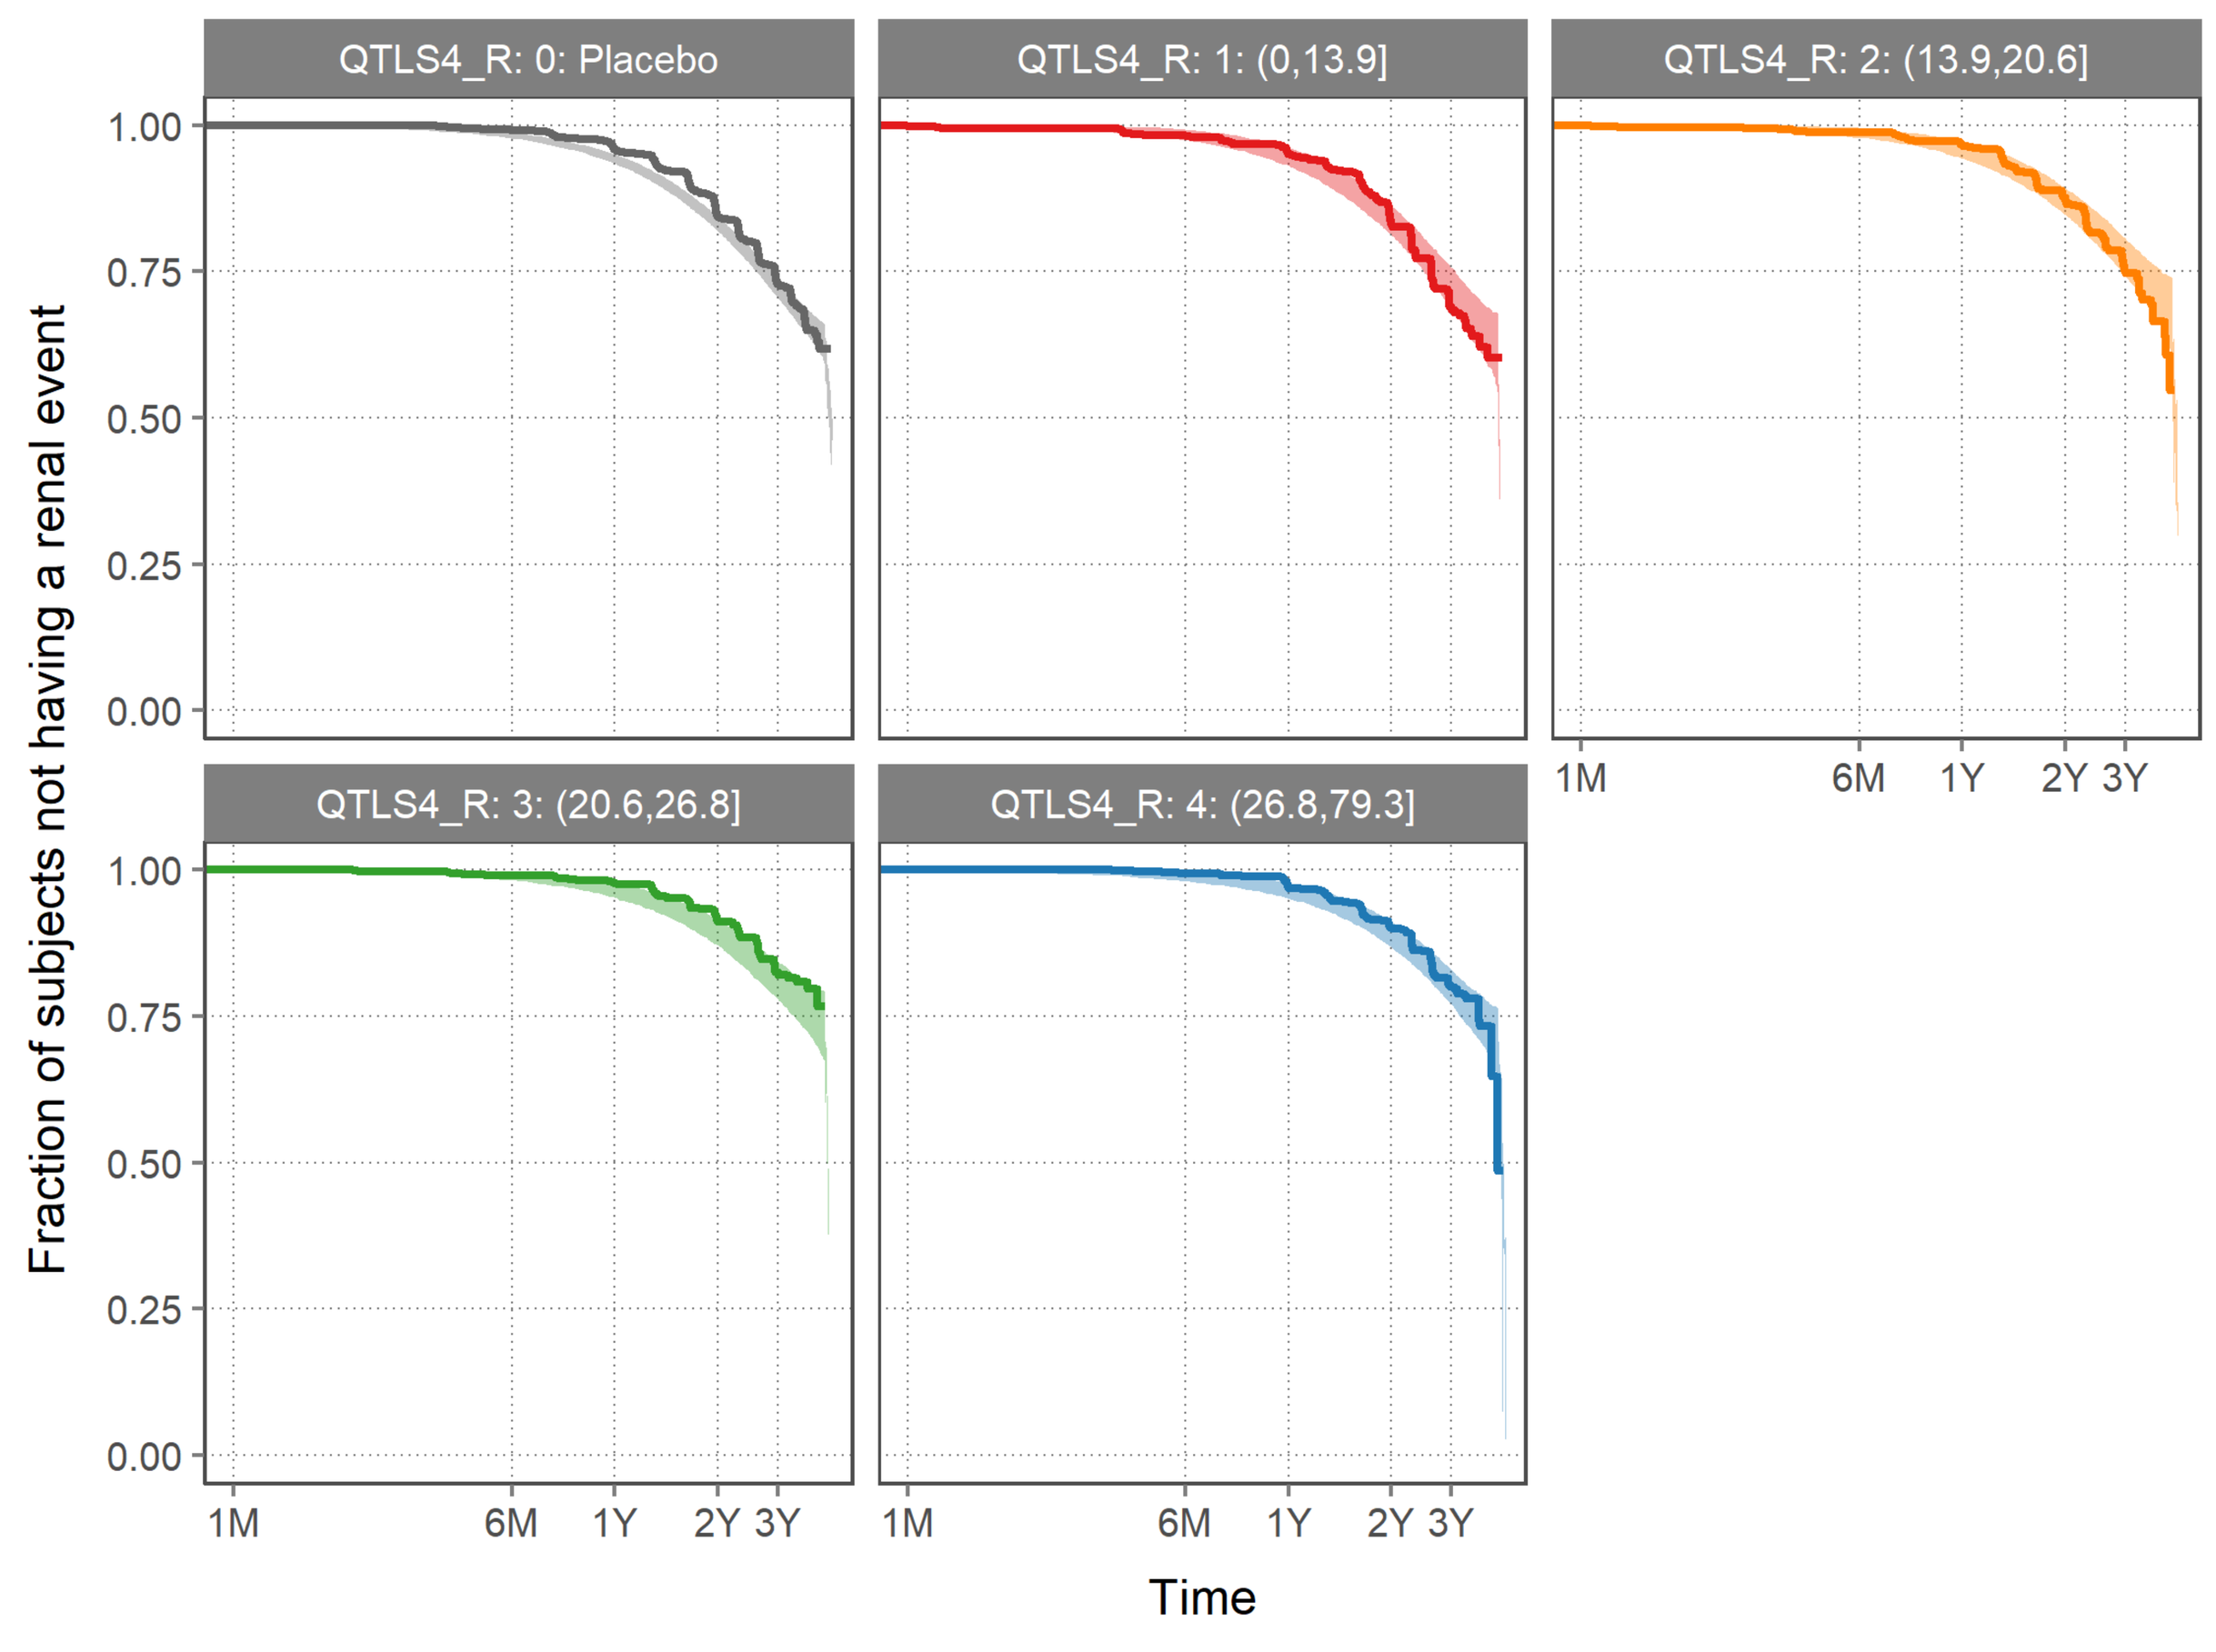
QTLS4_R: 0: Placebo
QTLS4_R: 1: (0,13.9]
QTLS4_R: 2: (13.9,20.6]
QTLS4_R: 3: (20.6,26.8]
QTLS4_R: 4: (26.8,79.3]
Fraction of subjects not having a renal event
1.00
0.75
0.50
0.25
0.00
1M
6M
1Y
2Y 3Y
Time

Figure 3. VPC of the kidney time-to-event model. Next to the fit of the placebo data (gray), the finerenone-treated subjects were divided into four exposure quartiles based on average concentration until (censored) event. The QTLS4_R numbers at the top indicate the exposure ranges in µg/L. Thick lines indicate the observed KM curves, where, owing to the visit-related nature of the event, the left corner of each step should be used to evaluate the goodness of fit relative to the ribbon, which indicates the 95% Prediction Interval.

Analyses indicate independent and additive effects of finerenone and SGLT2is on the primary kidney endpoint.

- SGLT2i use reduced the risk of a kidney event independent of finerenone treatment by 50.7% (95% confidence interval 29.8–71.6).
- No significant effect of SGLT2i use on the effect of finerenone was found.
- Results strengthened by recently published results on exposure-response analyses for the biomarkers/surrogates UACR and eGFR [7]:

Typical Subject Simulation Results	Finerenone 10 mg OD	Finerenone 20 mg OD	SGLT2i	Finerenone 20 mg OD + SGLT2i
Change in chronic eGFR slope vs placebo (%)	-27.3	-36.9	-56.1	-71.5

Conclusion

- Either covariate effects or multivariate forward-simulations in subgroups of interest were contained within the equivalence range of 80–125% around typical exposure indicating that these were not clinically relevant in FIDELIO-DKD.
- Finerenone effects on kidney outcomes approached saturation towards 20 mg once daily.
- SGLT2i use provided additive benefits [6].

[1] Agarwal R, et al. Nephrol Dial Transplant. 2020.
[2] Bakris GL, et al. Am J Nephrol. 2019;50(5):333–44.
[3] Goulooze B, et al. Clin Pharmacokinet. 2022;61(3):451-462.
[4] Bakris GL, et al. N Engl J Med 2020;383:2219-29.
[5] Snelder N, et al. Clin Pharmacokinet. 2020;59(3):359–70.
[6] Van den Berg P, et al. Clin Pharmacokinet. 2022;61(3):439-450.
[7] Goulooze SC, et al. Clin Pharmacokinet. 2022; https://doi.org/10.1007/s40262-022-01124-3