Dose projection and prediction of PK/PD response - a bench to bedside example for LY3023414

Sophie Callies[1], Elizabeth Smith LaBell[2], Enaksha Wickremsinhe[2], Vijay Reddy[2], Ji Lin[2], Gregory Donoho[2], Volker Wacheck[3], Johan Wallin[4]
Eli Lilly and Company[1,2,3,4] (France[1], USA[2], Austria[3], Sweden[4])

Background:

- LY3023414 (LY) is a dual PI3K/mTOR inhibitor.
- Dose-dependent tumor growth inhibition (TGI) by LY was observed in mouse xenograft models.
- Dose-dependent biomarker (p4EBP1) target inhibition (TI) by LY was observed in mouse xenograft models.

We hypothesized LY could lead in human to anti-tumor activity as observed in non-clinical models

Objectives:

- To support design of the first in human dose (FHD) study, aimed at defining the «biological-active-safe» dose range.
- «biological-active-safe» dose range = dose range leading to significant TI, some level of reduction in tumor size (TGI) and adequate tolerability.

Data available:

PK data collected in mice, rat and dog as shown in Table 1 and 2. TI and TGI data collected in mouse non-clinical models.

Table 1: Data available in Rat and Dog

	# of animals (IV,PO)	IV PK Dose, sample time	PO PK Dose – sample time
Rat SD[a]	0,18		5, 10, 20, 30 mg/kg 0.5, 1, 2, 4, 8, 24 h
Rat MD[a]	0,18		5, 15, 30 mg/kg 0.5, 1, 2, 4, 8, 24 h
Dog SD[a]	8,36	3 mg/kg 0.25,0.5,1,2,4,8,12 h	1,3,6,12,15,30 mg/kg 0.5, 1, 2, 4, 8, 12, 24 h
Dog MD[a]	0,36		1, 3, 6, 9 mg/kg 0.5, 1, 2, 4, 8, 12, 24 h

a SD single dose ; MD multiple dose

Table 2: Data available in Mice

Model	PK	Target inhibition (TI)	Tumor growth inhibition (TGI)
U-87 MG	√	√	√
NCI - H1975			√
786-O			√
PIK3CA E545K (Eμ-myc)			√

Methods:

Allometric scaling was used to predict human PK based on non-clinical PK data.
Mouse PK, TI and TGI data were modelled to determine the PK/PD relationship.
From this information, the biological effective dose range in human was predicted.
Assumption : Similar LY exposure (hence similar EC_{50}/AUC_{50}) would be needed in human and non-clinical species to trigger TI or TGI.
NONMEM (version VII) was used for the analysis.
Figure 1 explains the analysis strategy.

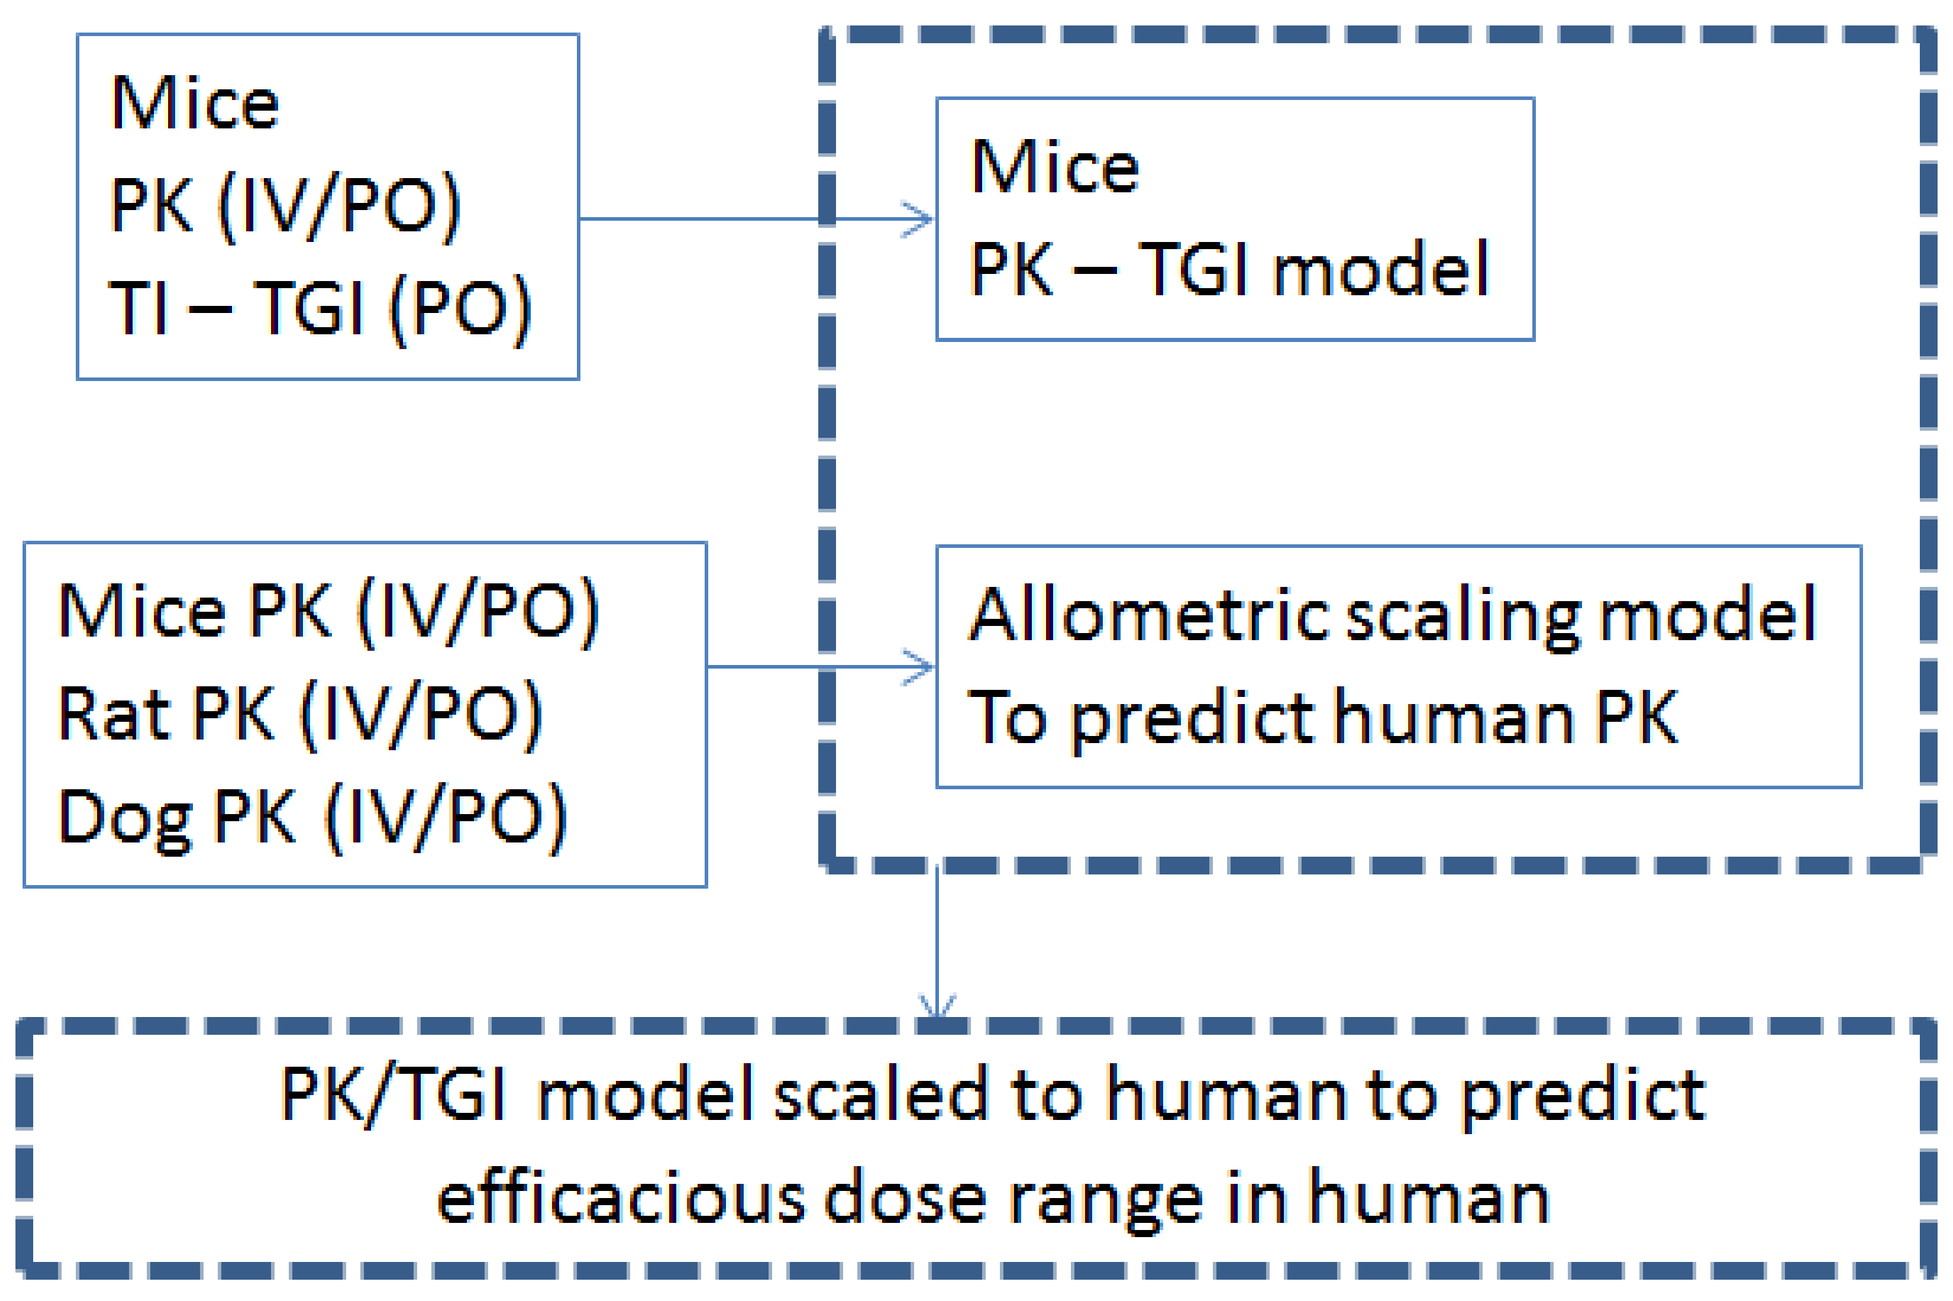
Mice
PK (IV/PO)
TI – TGI (PO)
Mice
PK – TGI model
Mice PK (IV/PO)
Rat PK (IV/PO)
Dog PK (IV/PO)
Allometric scaling model
To predict human PK
PK/TGI model scaled to human to predict
efficacious dose range in human

Figure 1: PK/PD analysis strategy

Results:

Allometric scaling predicted an IV and PO LY clearance (CL, CL/F) of 41.9 (34.0-51.7) and 55.9 (45.3-68.9) L/h in human (mean (90% CI), assuming bioavailability (F) 0.75 and body weight 70 kg. The slope of that relationship was 0.88 (see Figure 2)

The non-clinical TGI versus exposure relationship was modelled using an Emax model. (see Figure 3)
Estimated LY IC_{50} of 95.1 ng/mL (CV 40 %), corresponding to daily exposure of 2280 ng*h/mL following a 3 mg/kg BID dose (minimally efficacious dose in mice) (see table 3).

Optimal efficacious doses in mice were determined to be 6 to 10 mg/kg BID leading to daily exposure of 5380 to 9110 ng*h/mL (Figure 3).

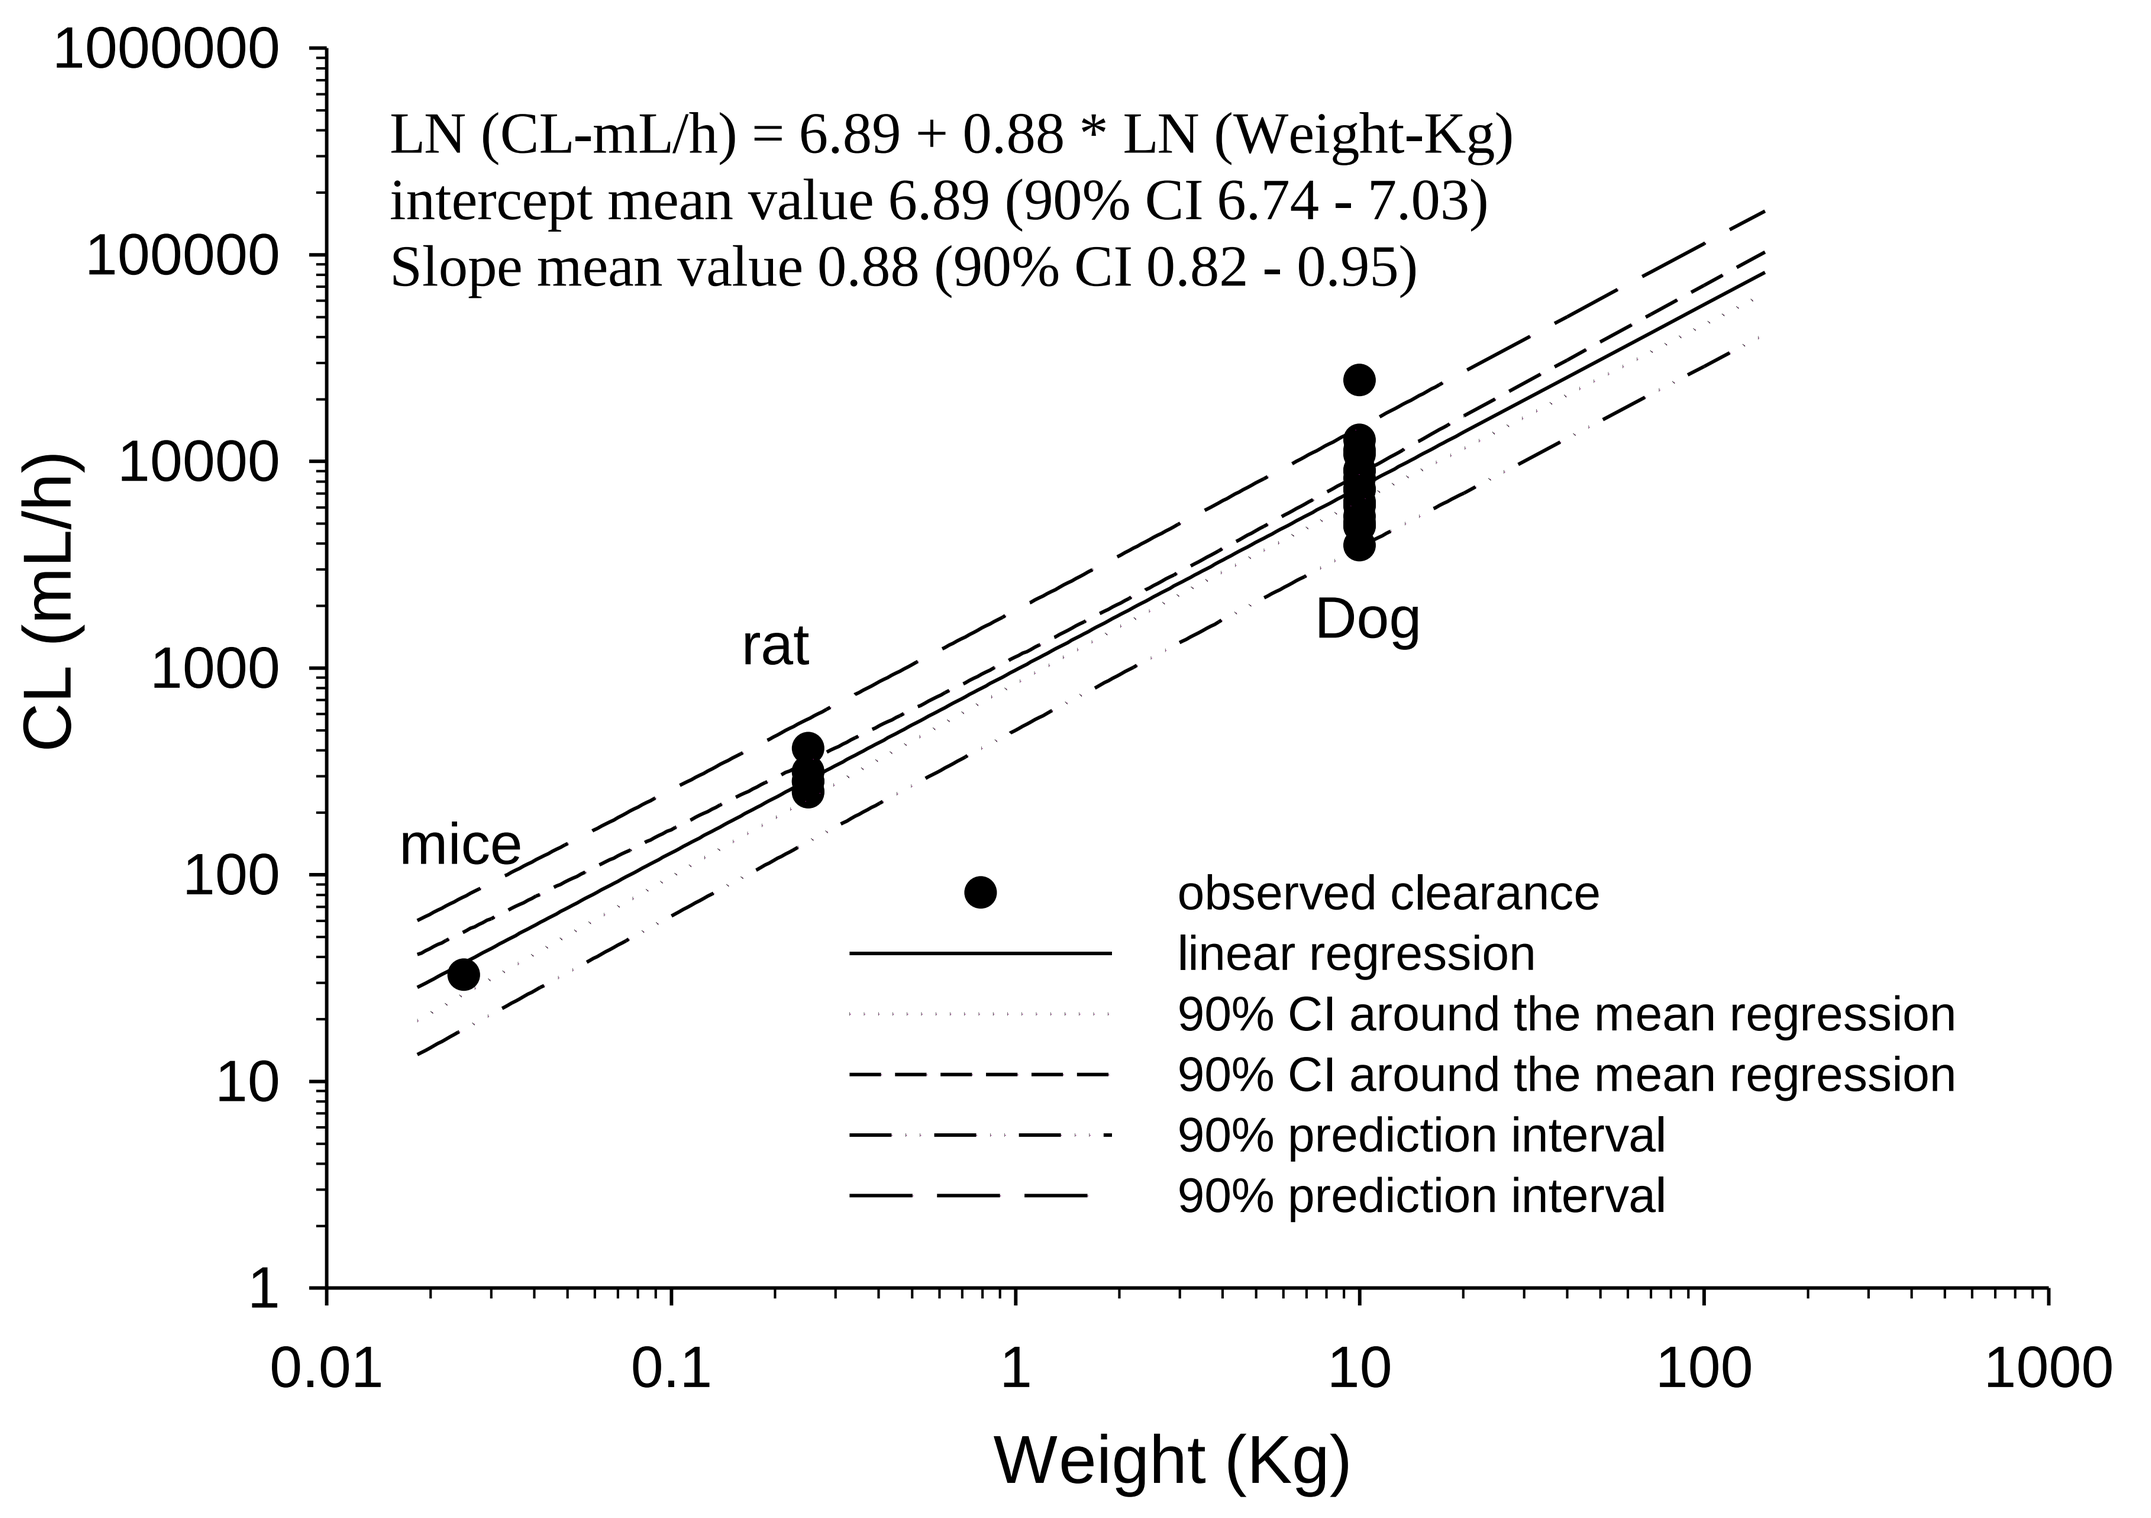
LN (CL-mL/h) = 6.89 + 0.88 * LN (Weight-Kg)
intercept mean value 6.89 (90% CI 6.74 - 7.03)
Slope mean value 0.88 (90% CI 0.82 - 0.95)
CL (mL/h)
Weight (Kg)
mice
rat
Dog
observed clearance
linear regression
90% CI around the mean regression
90% CI around the mean regression
90% prediction interval
90% prediction interval

Figure 2: PK – Allometric scaling relationship

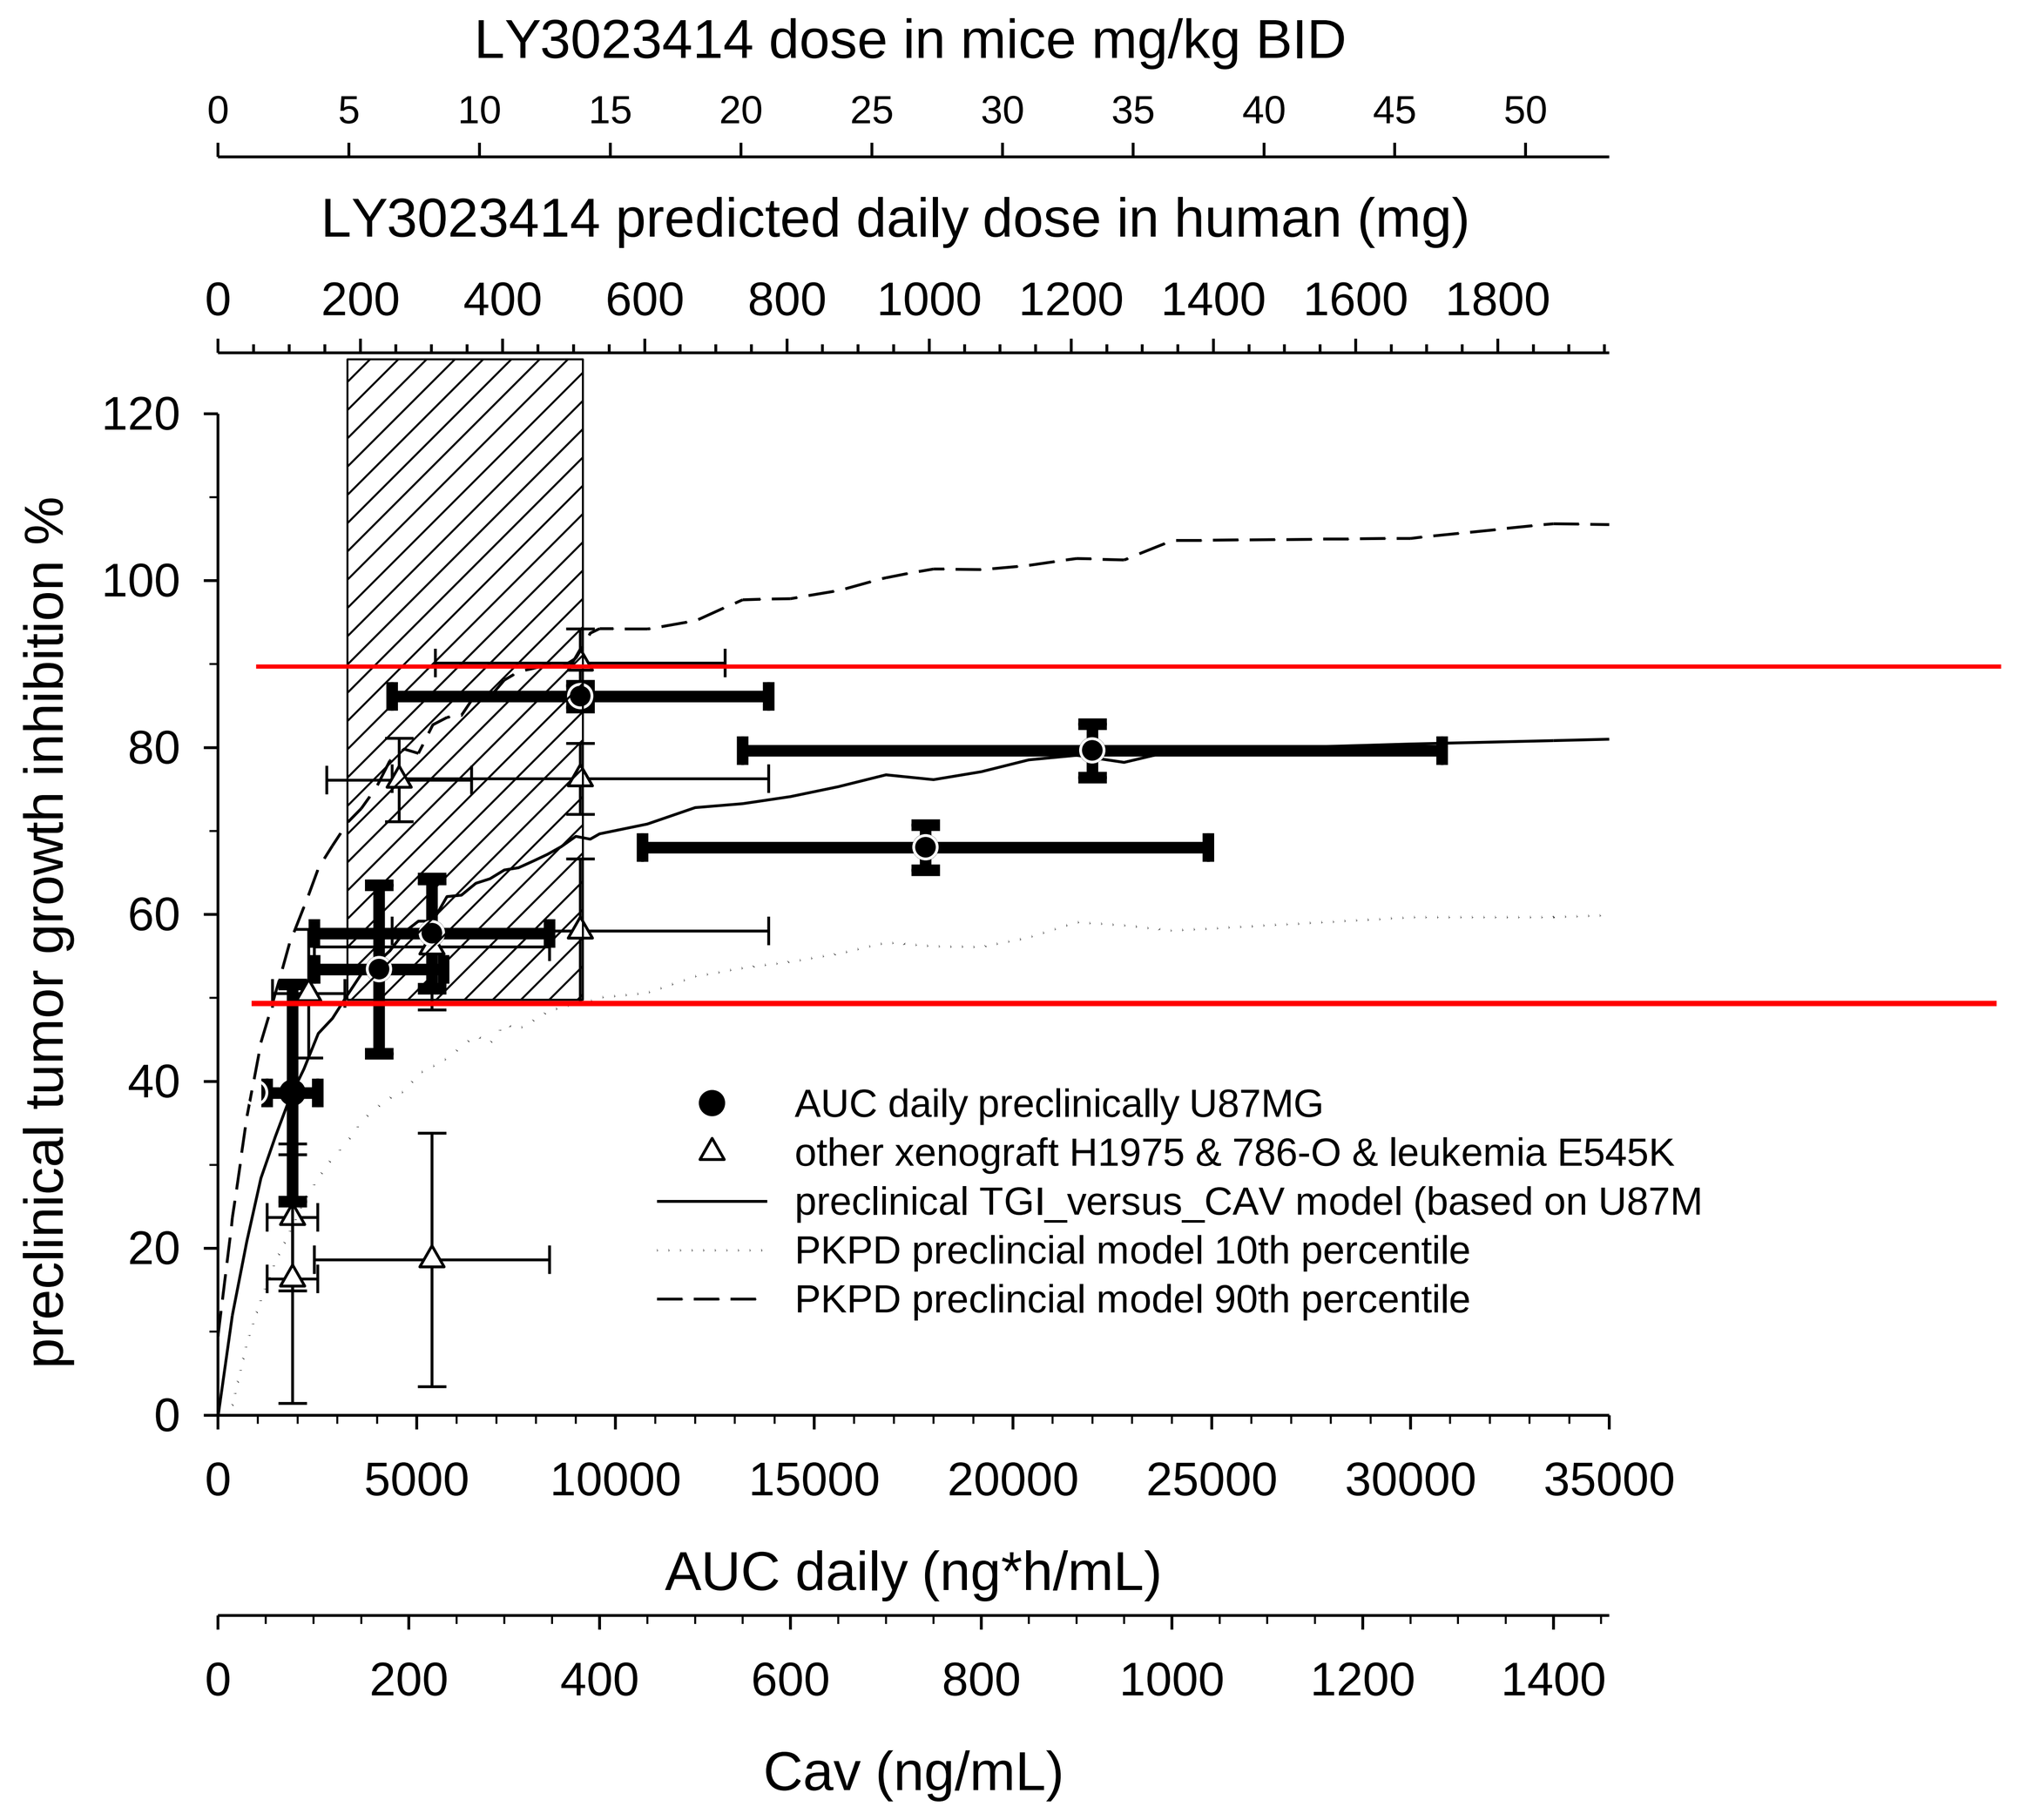
LY3023414 dose in mice mg/kg BID
LY3023414 predicted daily dose in human (mg)
preclinical tumor growth inhibition %
AUC daily preclinically U87MG
other xenograft H1975 & 786-O & leukemia E545K
preclinical TGI_versus_CAV model (based on U87M
PKPD preclincial model 10th percentile
PKPD preclincial model 90th percentile
AUC daily (ng*h/mL)
Cav (ng/mL)

Figure 3: PK/PD relationship LY concentration - TGI

Table 3: PK/PD (TGI) relationship – model parameters

PD TGI model parameters	Mean estimate	%SEE[a]
LY Cav50 ng/mL	95.1	3.04
Emax % TGI	87.7	1.77
Add Res Err (i.e +/- SD) as % TGI	7.4	1.69

a SEE standard error on the estimate

Design First in human dose (FHD):

Human PK prediction:
LY dose range of 100-150 and 250-600 mg daily were predicted to produce in human daily exposure equivalent to exposure in mice leading to minimal- and optimal anti-tumor activity, respectively (Figure 3).
The proposed dose range for FHD was 20 mg (starting dose) to 600 mg maximum daily dose.

FHD primary objective : determine a recommended phase II dose.
FHD secondary objective: Assess safety, PK/PD properties and to document antitumor activity.

Dose Escalation Scheme: 20, 40, 80, 150, 225, 325 450 mg QD, and 150, 200, 250 mg BID.

Observed human CL/F was 64 L/h (CV 45.6%, 90%CI 60-70, n=38) after repeated doses in the 20 to 325 mg dose range.
This is comparable to the predicted CL/F of 55.9 L/h (90%CI 45.3-68.9) (Figure 4).

A two compartmental model, with first order absorption, adequately fit LY clinical exposure. (Table 5)
A sigmoidal Emax model adequately fit the TI (p4EBP1 inhibition) versus LY exposure relationship with mean AUCdaily50 and Caverage50 (leading to 50 % of maximal target inhibition) of 1090 ng*h/mL and 45.4 ng/mL, respectively (CV=44%, n=19) (Table 6 and Figure 5).

Table 4: LY Clinical exposure – Predicted & observed
(interim data cut of – 26 sept 2014)

LY Dose	Daily dose	Predicted Daily AUC [a]	N	Observed daily AUC
mg	mg	ng*h/mL		ng*h/mL
20 QD	20	358 [290-442]	3[b]	89 – 164 – 293
40 QD	40	716 [581-883]	3[b]	442 – 742 – 1041
80 QD	80	1431 [1161-1766]	3[b]	646 – 867 – 1574
150 QD	150	2683 [2177-3311]	3[b]	2986 – 3409 – 3843
225 QD	225	4025 [3266-4967]	3[b]	1833 – 1966 – 8569
150 BID	300	5367 [4354-6623]	3[b]	3314 – 3698 – 5066
325 QD	325	5814 [4717-7174]	7[c]	5577 (38) [4276-7281]
200 BID	400	7157 [5806-8830]	15[c]	7334 (28) [6432-8364]
450 QD	450	8050 [6531-9934]	3[b]	11865 – 24595 - 51982
250 BID	500	8945 [7257-11038]	4[b]	8714 – 9616 – 11026

a AUC reported as geomean 90% CI
b individual observed exposure reported; @ 250 BID only 3/4 patients with PK data
c AUC reported as geomean (CV [90% CI]

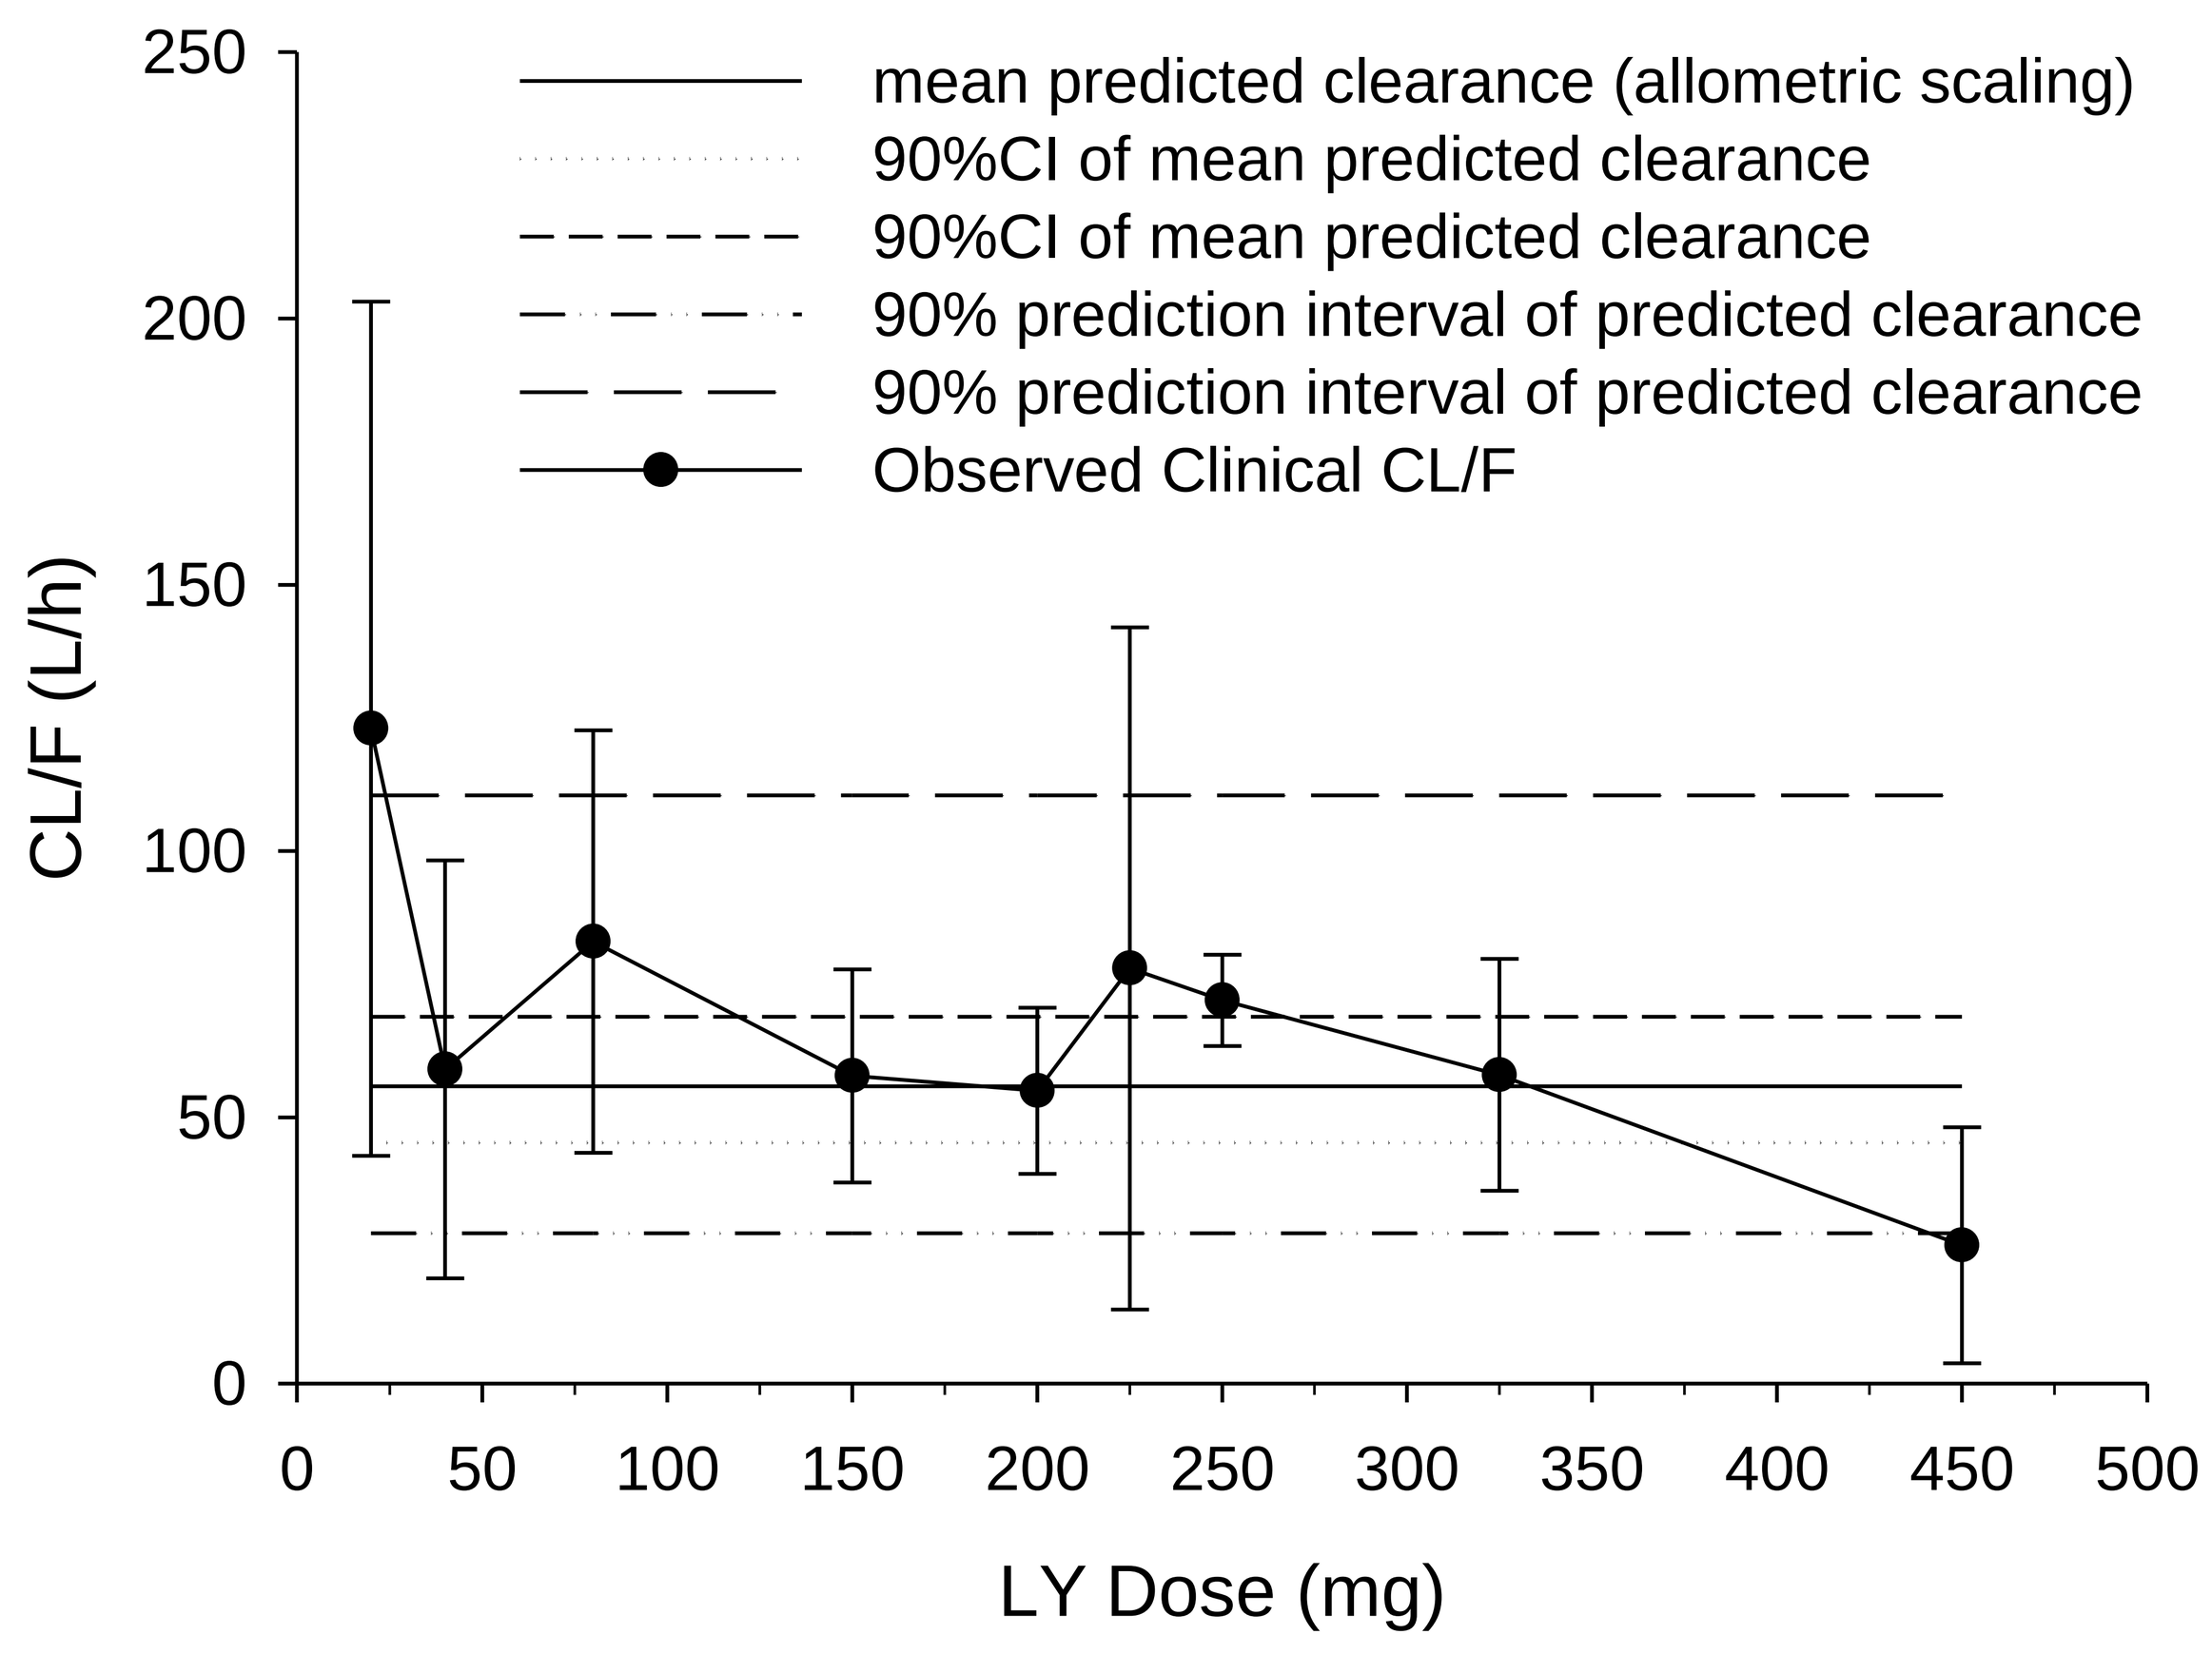
mean predicted clearance (allometric scaling)
90%CI of mean predicted clearance
90%CI of mean predicted clearance
90% prediction interval of predicted clearance
90% prediction interval of predicted clearance
Observed Clinical CL/F
CL/F (L/h)
LY Dose (mg)

Figure 4: CLss/F versus dose

Table 5: LY3023414 PK parameters (based on interim data)

LY PK parameters	Mean Estimate (SEE%) [a]	IIV % (SEE%) [a]
CL/F (L/h)	69.9 (6.9)	52.3 (33.7)
V1/F (L)	71.3 (24.4)	171 (36.1)
Q/F (L/h)	5.68 (31.7)	NC
V2/F (L)	55.4 (16.0)	81.1 (68.1)
KA (1/h)	0.477 (10.3)	32.7 (41)
Prop Res Err (%)	57.5% (14.4)	

a SEE standard error on the estimate, IIV inter-individual variability expressed as CV coefficient of variation %.

Table 6: LY3023414 PK/TI PK/PD parameters

PD TI model parameters	Mean Estimate (SEE%) [a]	IIV % (%SEE) [a]
LY AUC50 ng.h/mL	1090 (15.5)	43.7 (86.9)
Emax % TI	100 FIX	
GAM	1.10	
Add Res Err +/- SD (% TI) [a]	14.4 (29.5)	

a SEE standard error on the estimate, IIV inter-individual variability expressed as CV coefficient of variation %, additive residual error expressed as SD standard deviation of mean % TI.

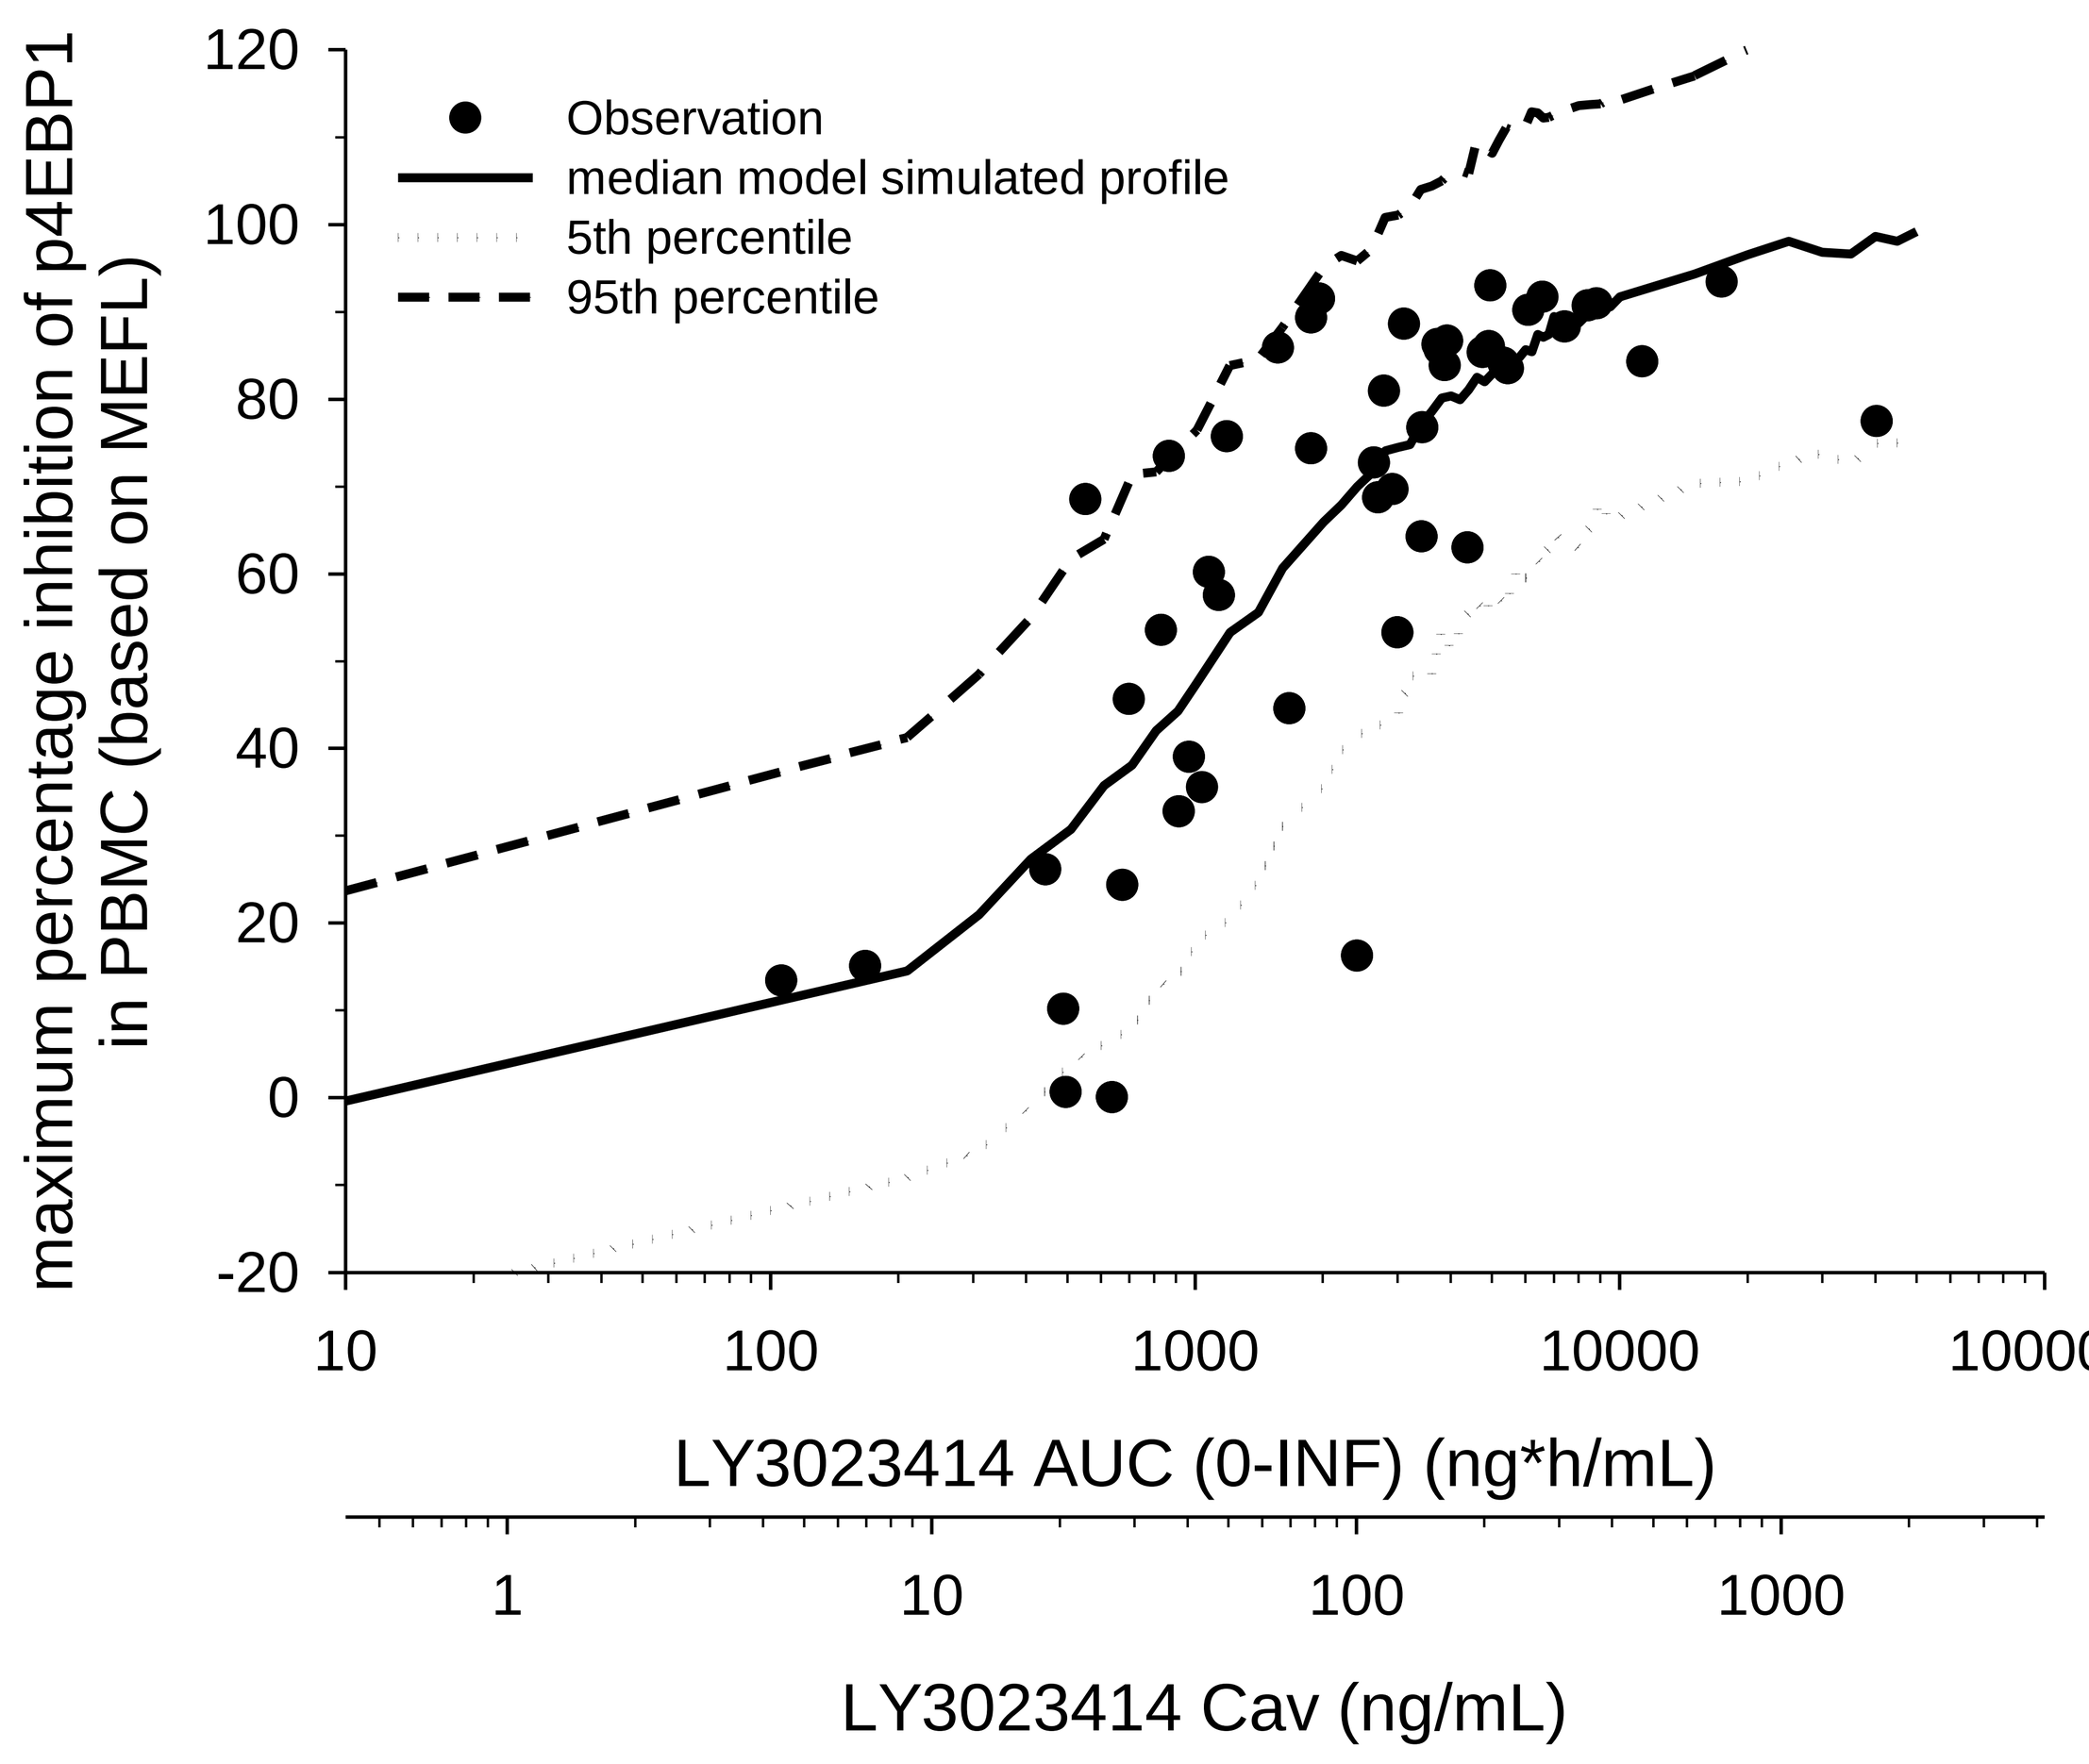
Observation
median model simulated profile
5th percentile
95th percentile
maximum percentage inhibition of p4EBP1
in PBMC (based on MEFL)
LY3023414 AUC (0-INF) (ng*h/mL)
LY3023414 Cav (ng/mL)

Note : MEFL Molecules of Equivalent Fluorescein PBMC Peripheral Blood Mononuclear Cells

Figure 5: PK/TI relationship

Conclusion:

The integration of the non-clinical data using modelling approach, enabled the prediction with good accuracy of the biological active dose range of LY in human FHD study.

- Range for predicted optimal active daily dose = 250 – 600 mg QD.
- Range for observed «biological-active-safe» dose in human = 300 to 400 mg QD

- «biological-active-safe» = dose range showing significant target inhibition and tolerated
- Preliminary anti-tumor activity was observed in patients treated a « biological active-safe» dose range