Forest Laboratories, Inc.

Pharmetheus
Pharmacometrics Consulting Services

A Population PK/PD Analysis of Nebivolol and Valsartan Combination Therapy

Mats O. Magnusson[1], E. Niclas Jonsson[1], Chun Lin Chen[2], Timothy Carrothers[2], Parviz Ghahramani[2]

[1] Pharmetheus AB, Uppsala, Sweden | [2] Forest Research Institute, Jersey City, NJ, USA

Objectives

To develop population PK/PD models that describe the effects of placebo, nebivolol and valsartan as single drugs and as a fixed-dose combination (FDC) on diastolic and systolic sitting cuff-measured blood pressure (BP) and 24-hour ambulatory measured BP (ABPM), and to use the models to simulate possible combinations of dosages.

Background

- Nebivolol is a highly selective β1-blocker with vasodilating properties and is approved in the US for the treatment of hypertension. Nebivolol is a CYP2D6 substrate and a racemic mixture for which both the *d*- and *l*- enantiomers as well as the glucuronide metabolite are active. *d*-nebivolol's β receptor affinity is > 1000-fold higher than *l*-nebivolol.
- Valsartan is an angiotensin II receptor antagonist with high affinity to angiotensin II receptor type 1.
- Nebivolol and valsartan are being developed as a FDC for oral administration. The FDC doses are here presented as Nebivolol dose / Valsartan dose in mg, e.g. FDC 10/160.

Conclusion

Modeling and simulation of nebivolol and valsartan combination therapy demonstrate a partially additive effect on diastolic and systolic sitting cuff-measured BP as well as on diastolic and systolic 24-hour ambulatory measured BP among all FDC doses studied.

Data and Methods

- Model predicted exposures from population PK models of valsartan and nebivolol were used in this exposure-response analysis.
- The exposure-response population included 761 patients for the sitting cuff-measured BP and 746 patients for ambulatory BP.
- The BP lowering drug effects of nebivolol and valsartan were described by inhibitory E_{max} models with a parameter (α) to account for the interaction between the compounds [1].
- The diurnal rhythm in the ambulatory BP data was described by the sum of cosine functions [2].
- The model was used to simulate the BP reduction in treatment arms not included in the Phase 3 study.

Results and Simulations

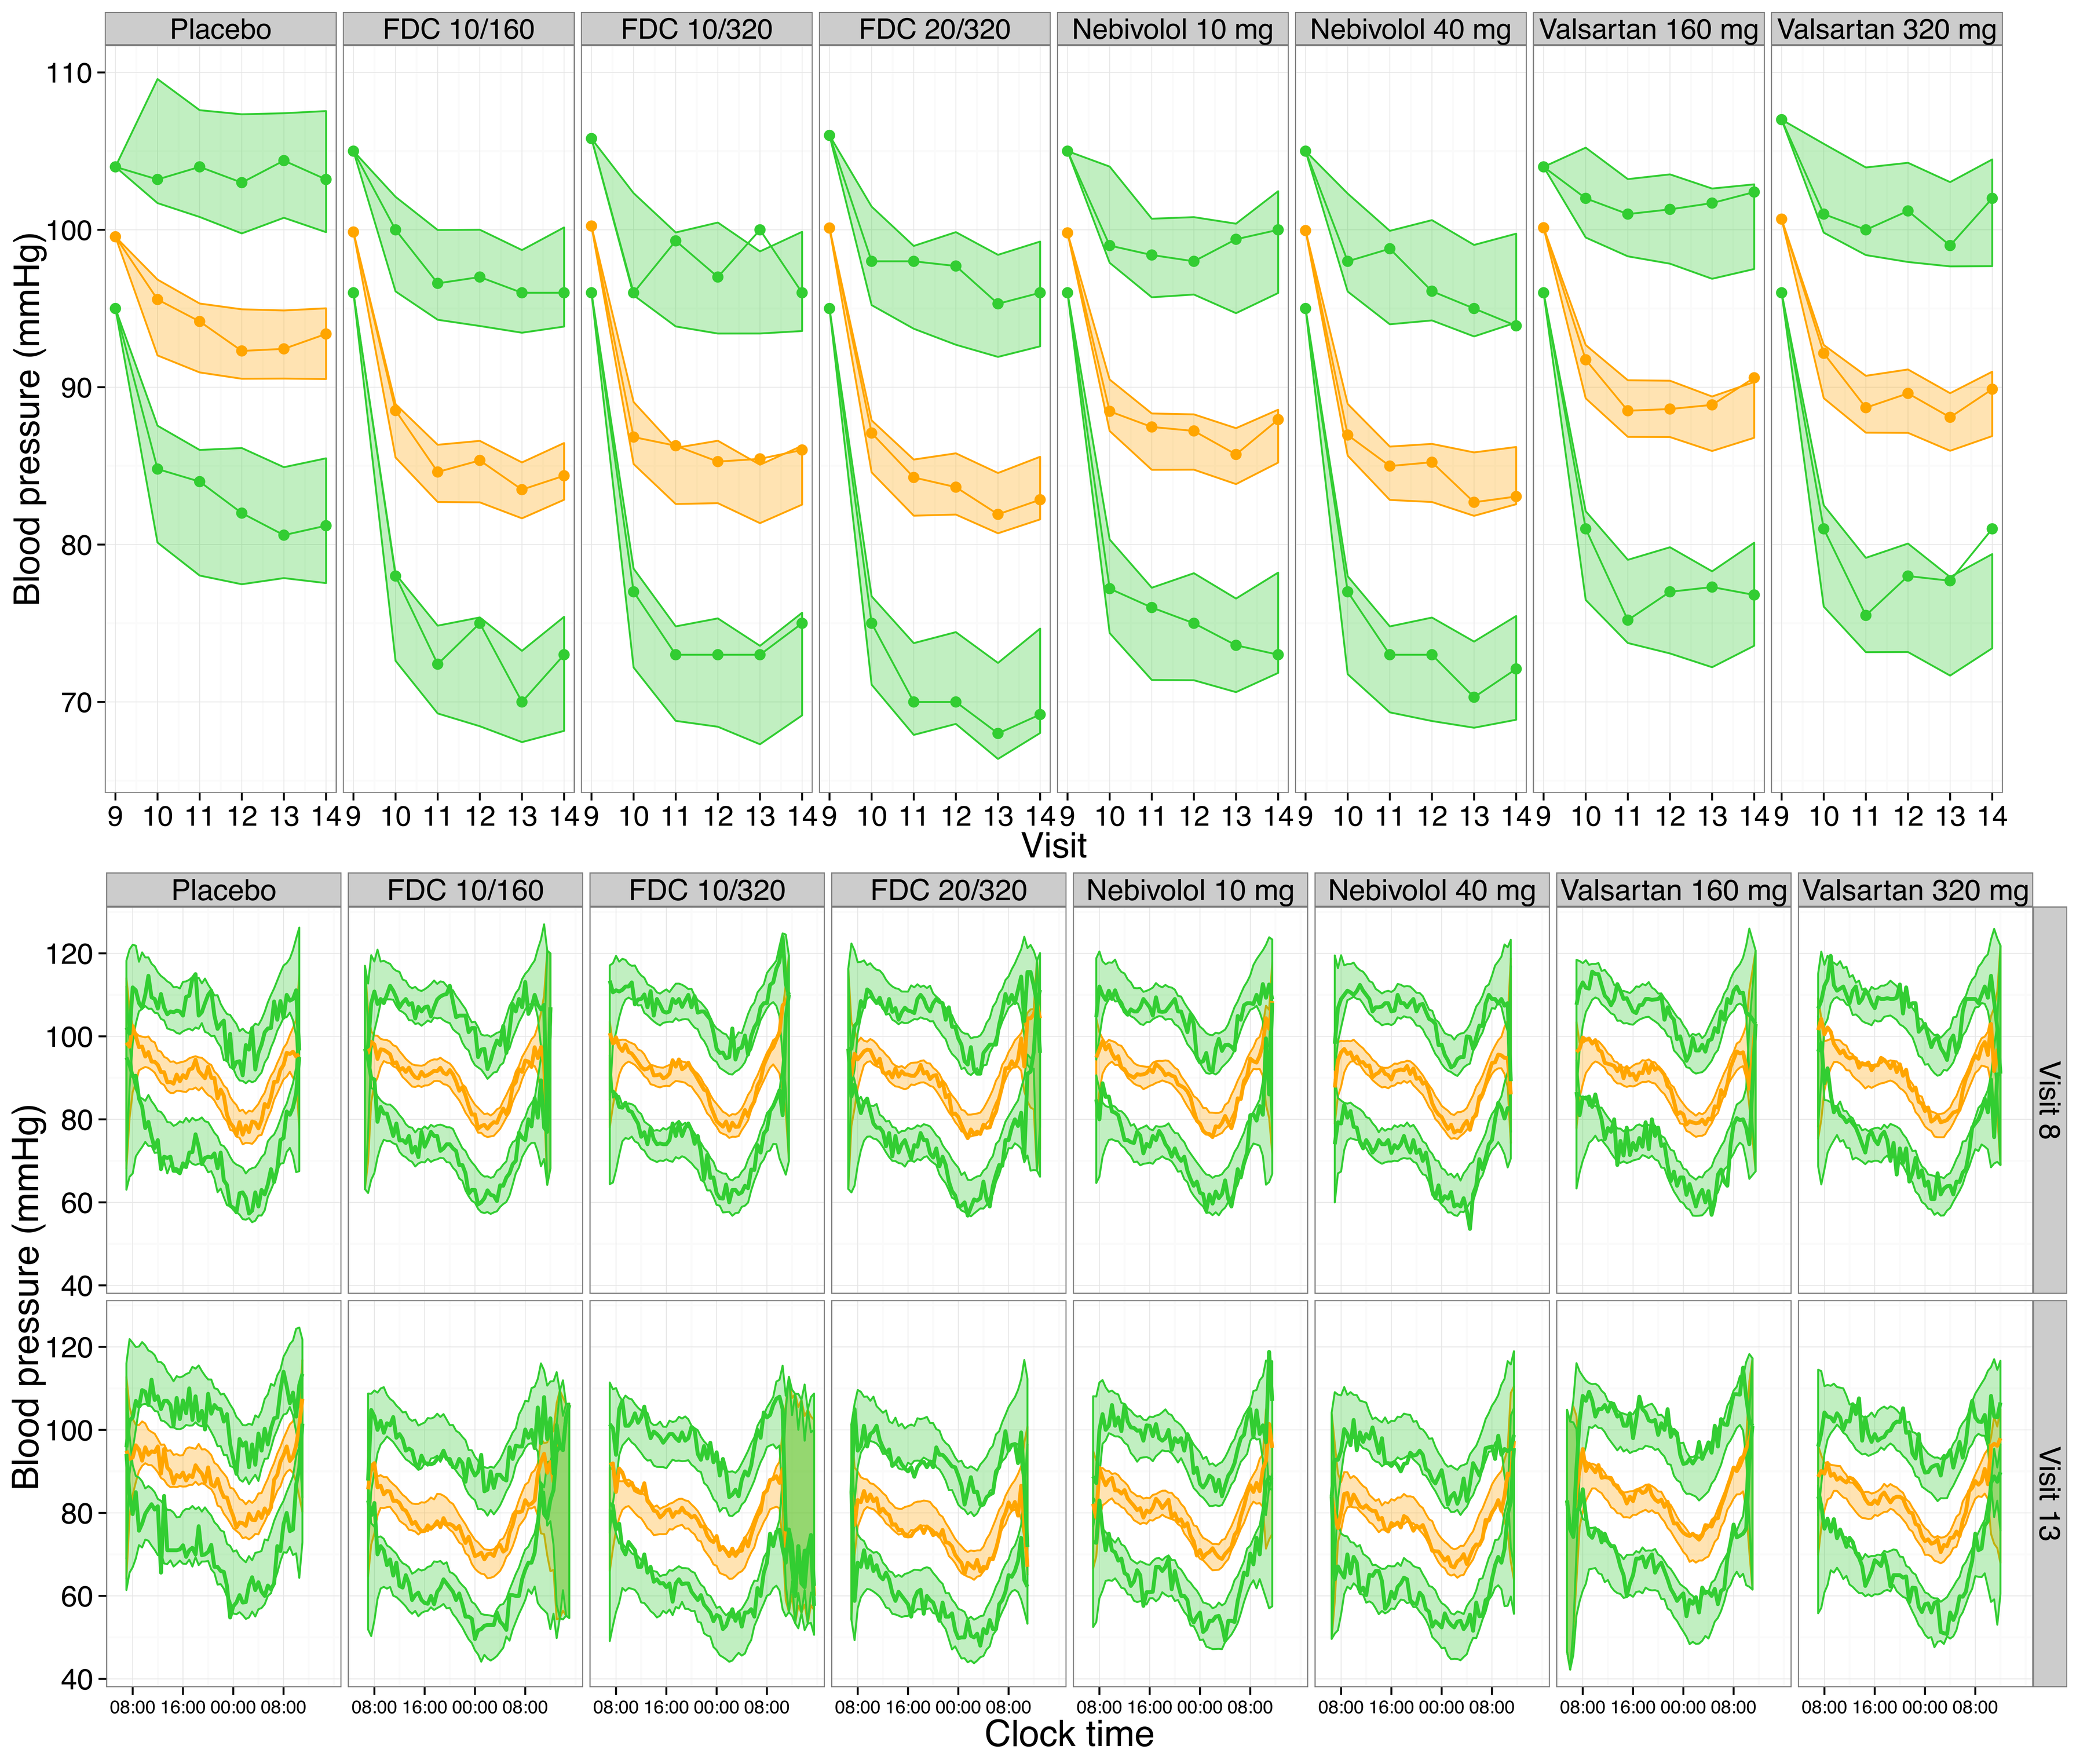

Placebo
FDC 10/160
FDC 10/320
FDC 20/320
Nebivolol 10 mg
Nebivolol 40 mg
Valsartan 160 mg
Valsartan 320 mg
Blood pressure (mmHg)
Visit
Clock time
Visit 8
Visit 13

Figure 1. *VPC of the final sitting cuff-measured (top panels) and ABPM (bottom panels) diastolic models versus visit number or clock time. Visit 8 is pre-treatment, Visit 13 is day 56. Solid lines represent the 10th, 50th and 90th percentiles, shaded areas are 95% confidence intervals of model predicted percentiles.*

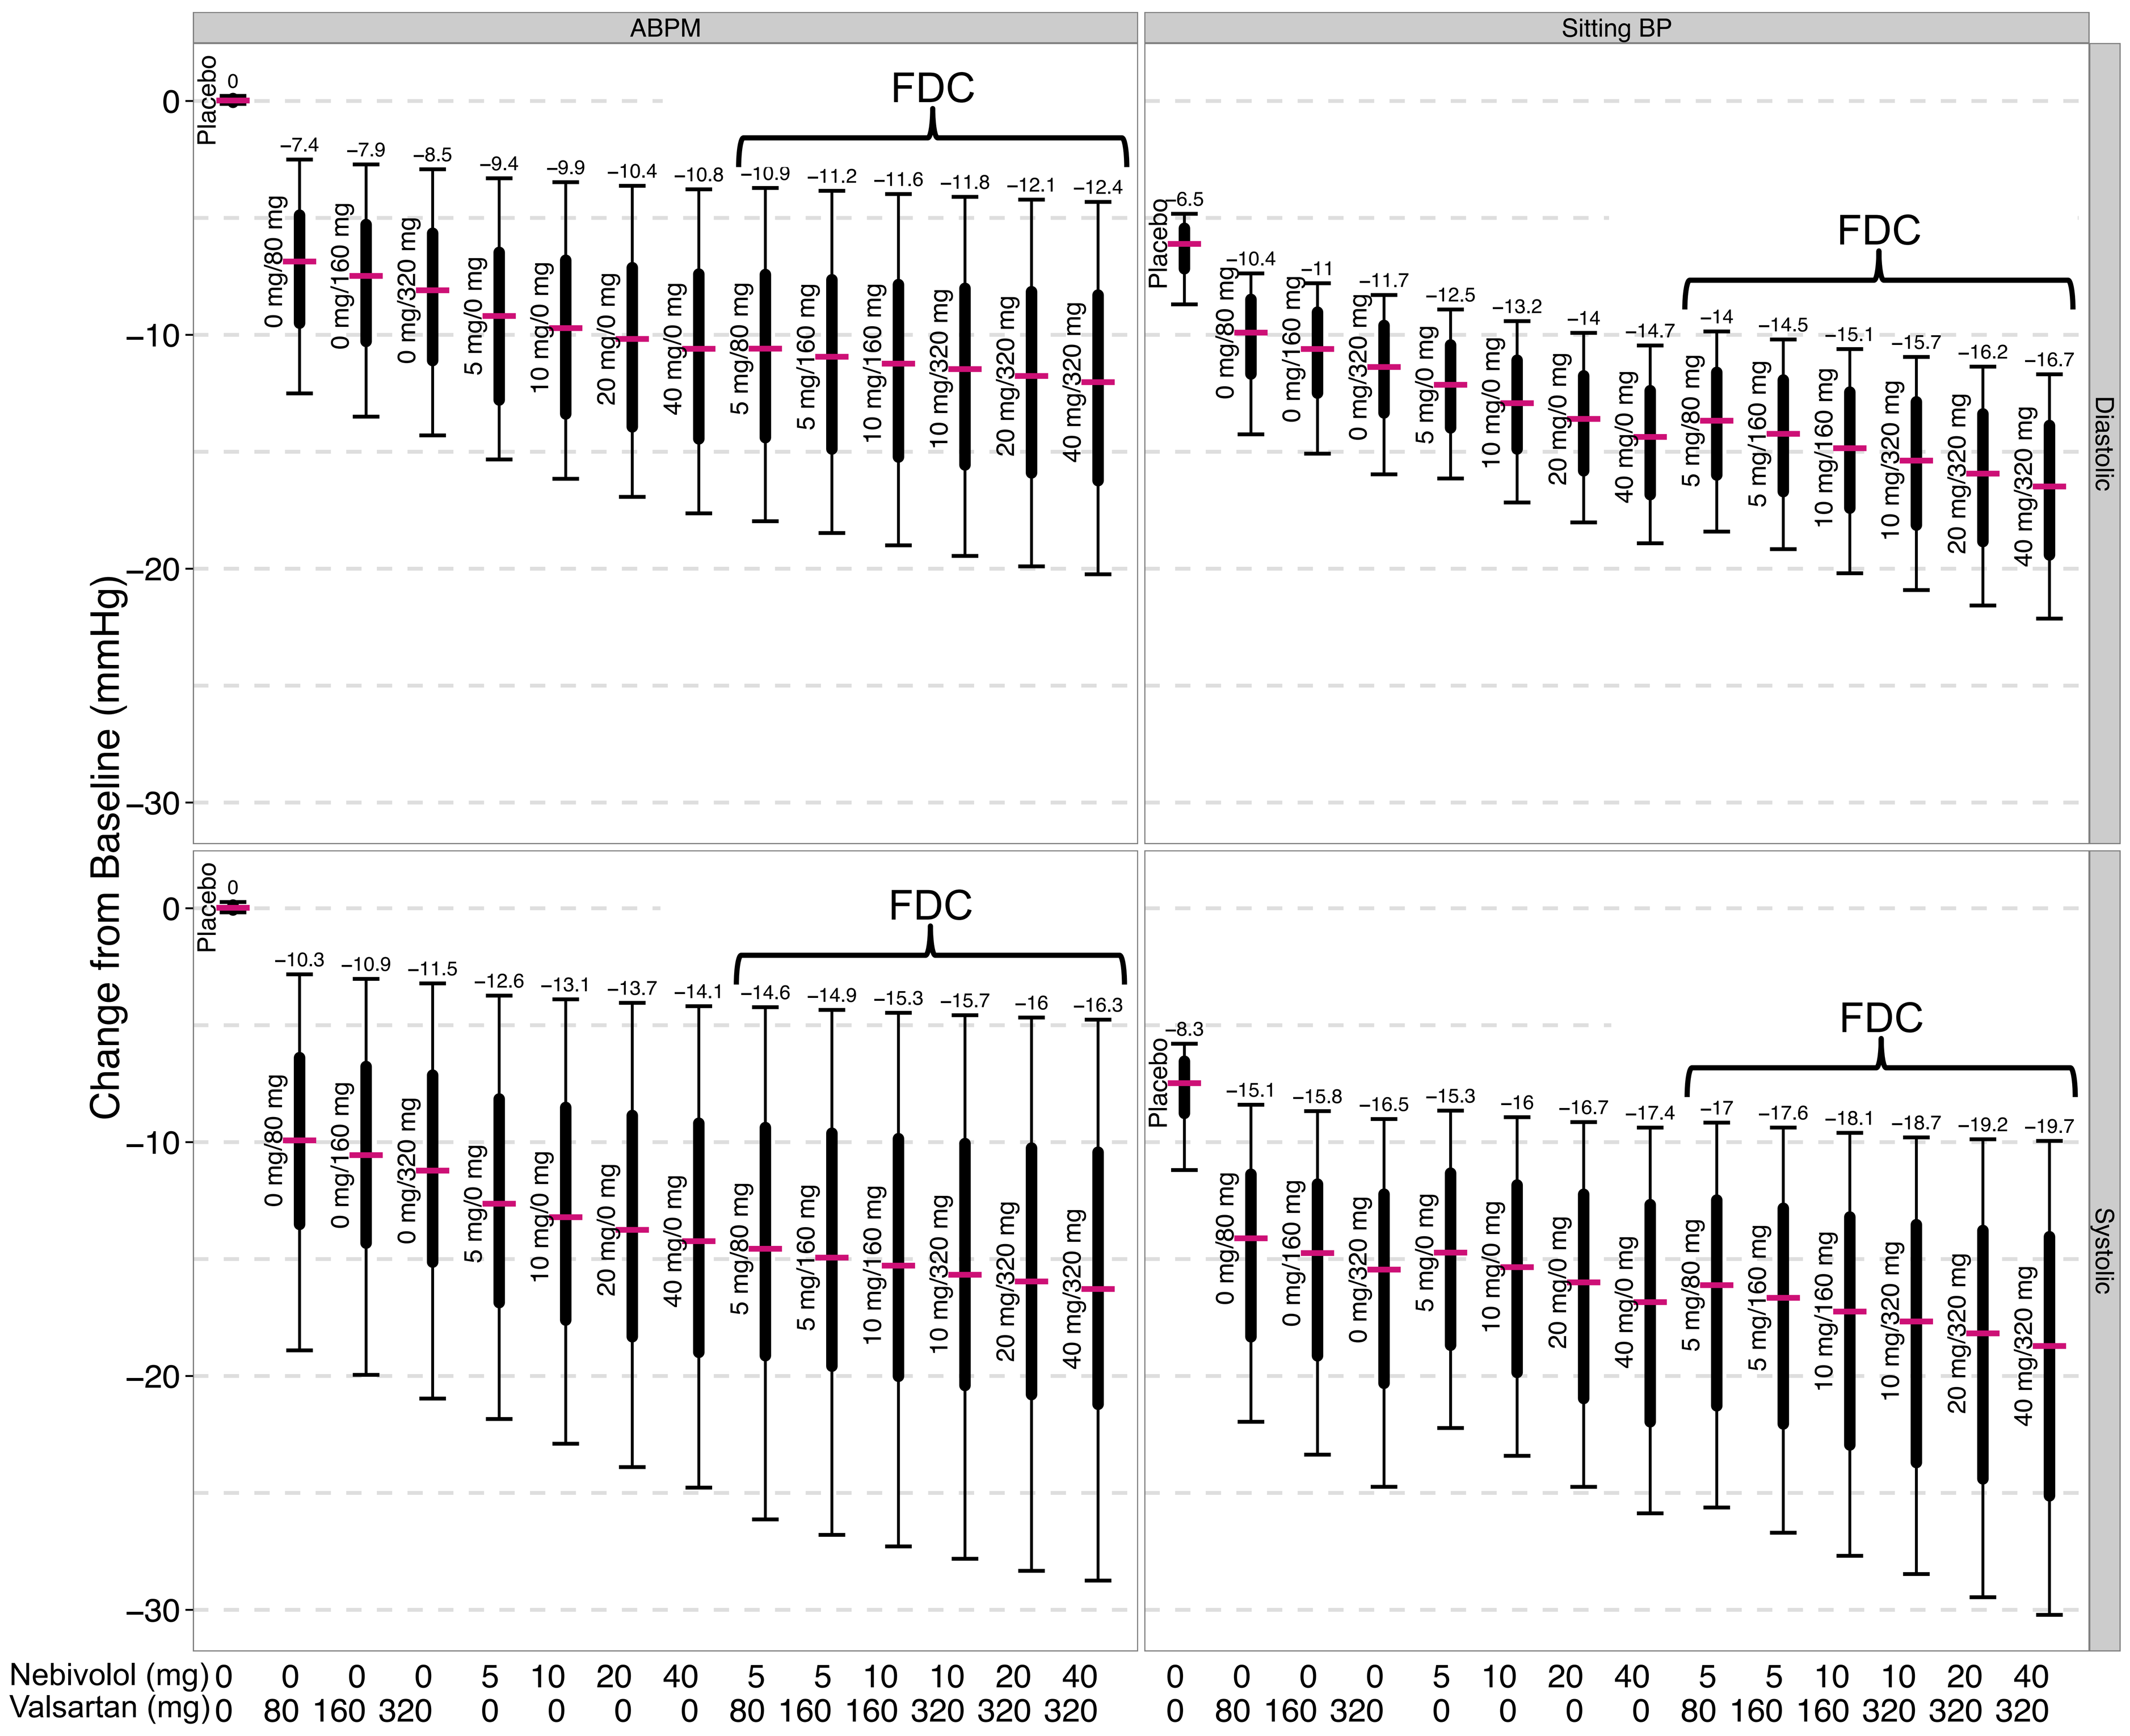

ABPM
Sitting BP
FDC
Diastolic
Systolic
Change from Baseline (mmHg)
Placebo
Nebivolol (mg) 0 0 0 0 5 10 20 40 5 5 10 10 20 40
Valsartan (mg) 0 80 160 320 0 0 0 0 80 160 160 320 320 320

Figure 2. *Predicted change from baseline to day 56 in ABPM (left panels) and sitting cuff-measured BP (right panels) in diastolic (top) and systolic (bottom) BP versus treatment. Numbers above the bars are means, dashes are 10th, 50th, and 90th percentiles, and thick lines are 25-75th range.*

The placebo response in sitting cuff measured BP was described by a mono-exponential function:

$$Placebo = \theta \times (1 - e^{(-k_{out\ placebo} \times PK_{time})})$$

The models for BP from ABPM were based on a sum of cosine functions to account for the diurnal rhythm in the data:

$$ABPM_{baseline,ij} = ABPM_{typical} + \sum_{1}^{n} AMP_{ijk} \times COS\left[(t - \tau_{ijk}) \times \frac{2\pi}{PER_k}\right]$$

The treatment effect of valsartan, nebivolol and the combination of the two compounds was estimated simultaneously by applying the interaction model below. Attempts were made to estimate the relative potencies of *d*-nebivolol, *l*-nebivolol and glucuronide. Due to the high correlation between the plasma concentration of the three compounds, the data did not support estimation of the relative contribution of *d*-nebivolol, *l*-nebivolol and glucuronide. Therefore, the predicted plasma concentrations of *d*-nebivolol were used to estimate their combined effect.

$$E = \frac{E_{max} \times \left([\frac{C_{d\text{-neb}}}{EC_{50,d\text{-neb}}}] + [\frac{C_{val}}{EC_{50,val}}] + [\frac{\alpha \times C_{d\text{-neb}} \times C_{val}}{EC_{50,d\text{-neb}} \times EC_{50,val}}] \right)^{\gamma}}{1 + \left([\frac{C_{d\text{-neb}}}{EC_{50,d\text{-neb}}}] + [\frac{C_{val}}{EC_{50,val}}] + [\frac{\alpha \times C_{d\text{-neb}} \times C_{val}}{EC_{50,d\text{-neb}} \times EC_{50,val}}] \right)^{\gamma}}$$

$$BP = BP_{baseline} - E - Placebo \qquad ABPM = ABPM_{baseline} - E$$

The maximum estimated drug effects were 13 mmHg for sitting cuff-measured BP, and 14 and 19 mmHg for diastolic and systolic ABPM, respectively. Statistically significant exposure-response relationships were identified for both monotherapies as well as the FDC, suggesting increasing efficacy with increasing drug exposures, both in monotherapy and in combination.

The parameter α was estimated to be positive in all sitting cuff-measured and ABPM models, and this effect was further characterized as partially additive, i.e. the total effect was greater than for the monotherapies but less than the sum of the effects of the two drugs.

The placebo effect increased over time to 6.13 mmHg and 7.55 mmHg in the sitting cuff-measured diastolic and systolic models, respectively. No placebo effect was seen in the ABPM data.

The final model described the data well as demonstrated in the VPCs in Figure 1 (systolic models showed similar goodness-of-fit).

The model-predicted change from baseline in BP demonstrated that all studied FDC doses had greater BP reductions compared to their monotherapy components in a partially additive manner (Figure 2). An exposure-response relationship was also evident between the FDC dose groups.

[1] Greco WR, Park HS and Rustum YM, 1990, Application of a new approach for the quantitation of drug synergism to the combination of cis-diamminedichloroplatinum and 1-beta-D-arabinofuranosylcytosine. Cancer Res **vol. 50**: 5318–5327.

[2] Hempel G, Karlsson MO, de Alwis DP, Toublanc N, McNay J and Schaefer HG, 1998, Population pharmacokinetic-pharmacodynamic modeling of moxonidine using 24-hour ambulatory blood pressure measurements. Clin Pharmacol Ther **vol. 64**: 622–635.