Optimal Design for the improvement of sampling schedules of microdialysis studies

M. Scholz (1), N. Plock (1,2), C. Kloft (1)
(1) Martin-Luther-Universitaet Halle-Wittenberg, Abt. Klinische Pharmazie, Germany
(2) Nycomed. Pharmacometrics, Constance, Germany

Objectives

The methods of Optimal Design have been explored for various applications, but not for the design of sampling schedules in microdialysis (µD) studies. In contrast to plasma sampling, µD allows to determine the concentration of drugs or PD markers at the site of action. Microdialysate is continuously sampled over a longer time period which is divided in usually identical collection time intervals. Optimal Design usually focusses on sampling time *points* rather than time *intervals*, thus for µD studies a new approach has to be taken. In a first step the commonly used midinterval method (i.e. allocating measured µD concentrations to the middle of the sampling interval) was systematically investigated and assessed. The impact on accuracy when using the midinterval method was quantified exploiting the results of a developed model of a clinical µd trial with linezolid. For this purpose, simulation and estimation techniques were utilised.

Methods

Data base

34 IDs were included in a clinical trial with 600 mg linezolid bid [1]. 1176 unbound plasma and 2325 µD data after single dose and at steady state over 8 h each were determined. A population PK model was developed using NONMEM (Fig. 1: structural model, Tab. 1 & 2: estimated PK parameters).

Table 1: PK parameter values for simulation in BM & NONMEM

KA [1/h]	CL [L/h]	Q [L/h]	V2 [L]	V3 [L]	K24, K40 [1/h]
1.84	11.5	76.8	19.8	27	12.3
KIC [1/h]	IC50 [mg/L]	VAR	PC23	PC24	K23, K30 [1/h]
0.0027	0.1	0.567	1.05	1.07	50

Table 2: Used parameter values in NONMEM

CL [CV%]	V3 [CV%]	V2 [CV%]	V2/VAR	ω2VAR
49.8	20.5	37.1	0.573	6.36

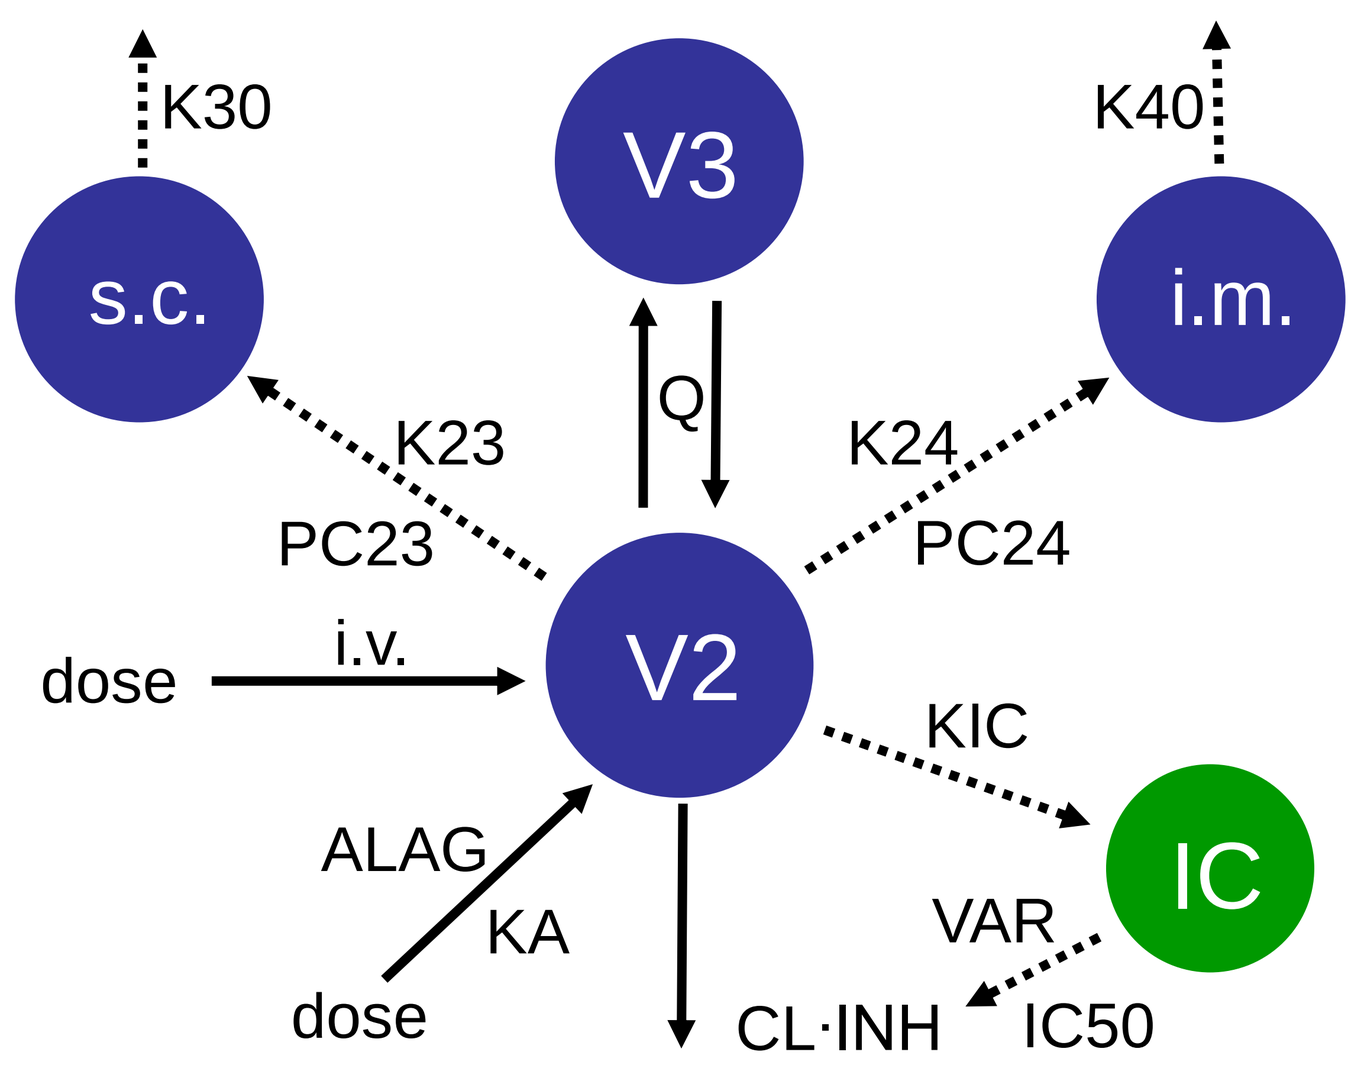
K30
V3
K40
s.c.
i.m.
K23
Q
K24
PC23
PC24
dose
i.v.
V2
KIC
ALAG
KA
IC
VAR
dose
CL·INH
IC50
Fig. 1: Schematic structural model

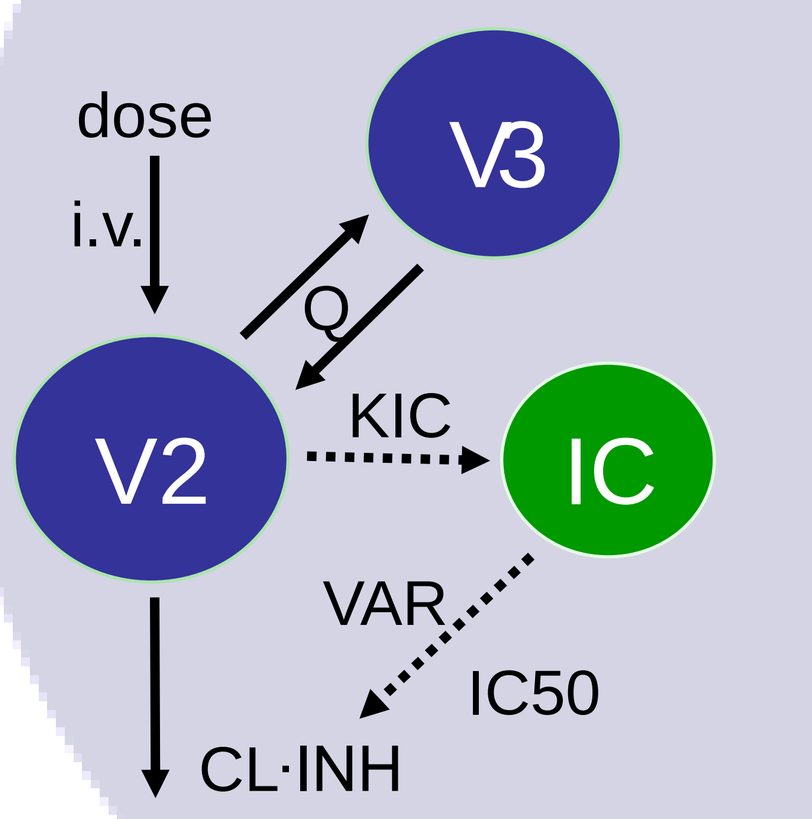
dose
i.v.
V3
Q
V2
KIC
IC
VAR
IC50
CL·INH
Fig. 3: Schematic simplified structural model

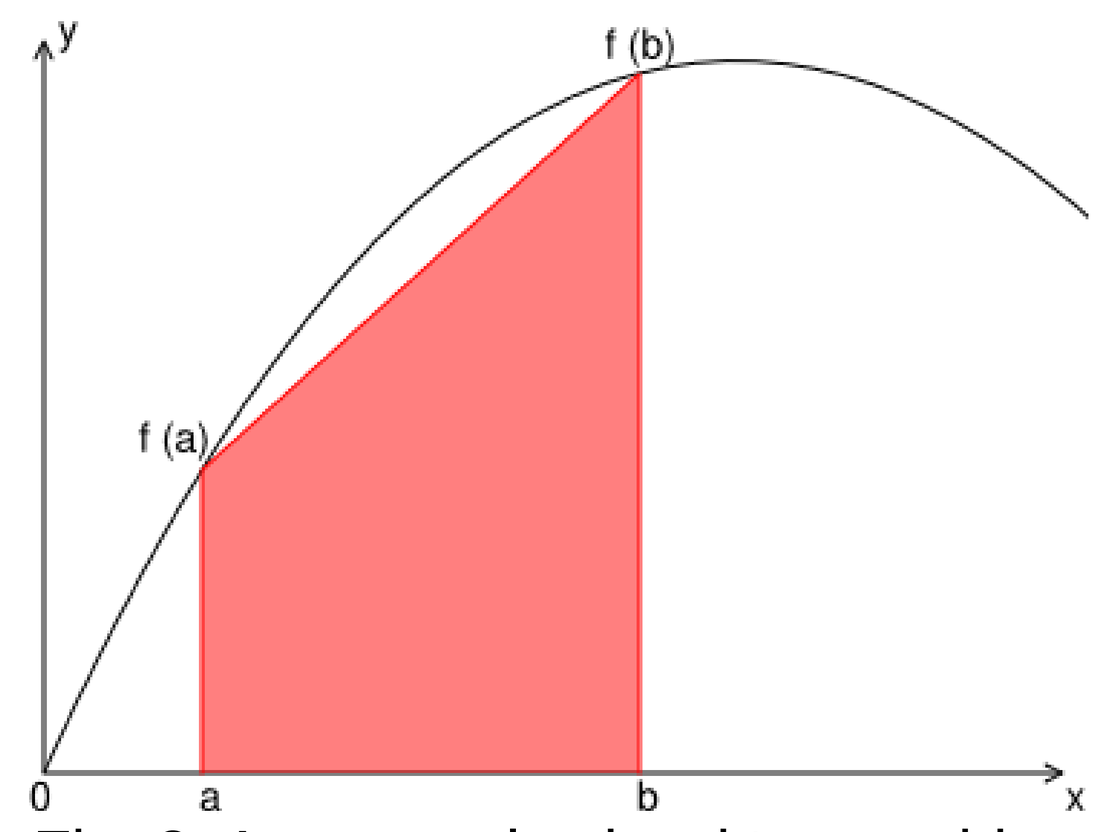
y
f (b)
f (a)
0
a
b
x
Fig. 2: An example chord trapezoid

Calculation of relative error

To quantify the impact on accuracy using the midinterval method the relative error (RE) was calculated as the deviation of the 'hypothetically' measured µD concentration (C_m) at the middle of the sampling interval to the 'true', simulated µD concentration (C_s, Eq. 1). C_m was calculated from the AUC of the assumed µD sampling interval: For this purpose the AUC was approximated using the chord alternative of the trapezoidal rule (Fig. 2, Eq. 2). The step size h for the calculation of C_m was set to 0.005, equalling a time interval of 18 s for every chord trapezoid. The AUC calculated demonstrated negligible bias (<0.02% RE).

Eq. 1
$$RE = \left(\frac{c_m - c_s}{c_s}\right) \cdot 100$$

Eq. 2
$$c_m = \frac{1}{b-a} \cdot \sum_{i=1}^{n} \frac{h}{2} \cdot \left(f(a_i) + f(b_i)\right) \quad \text{with} \quad a_i = a + h \cdot (i-1), \quad b_i = a + h \cdot i$$

Simulation of typical conentration-time profiles

In Berkeley Madonna (BM) typical concentration-time profiles for single p.o. and i.v. administrations were simulated based on the model and parameters in Fig. 1 and Tab. 1. For the 30 min infusion, s.c. µD sampling was assumed. For p.o., µD sampling in the peripheral compartment (V3) was assumed and different KA values were investigated. These simulations were used to investigate the relation between µD sampling interval duration (15, 30, 45, 60, 90, 120 min) and RE of C_m.

Simulation of concentration-time profiles of a population

Concentration-time profiles of 100 IDs after single dose i.v. infusion of 600 mg linezolid were simulated based on the model and parameters in Fig. 1 and Tab. 1 in NONMEM. Additionally interindividual variability was implemented (Tab. 2). Based on the simulated dataset two new datasets were developed with a 15 min sampling interval duration (scenario A) and a 30 min interval duration (scenario B): Tab. 3 shows the time points and the corresponding intervals of the first hour which were replaced by the calculated µD concentration (C_m). At the bottom, sampling points and intervals after the first hour (not replaced by C_m, i.e. C_s) are listed being identical for both scenarios. The PK parameters of the 2 new datsets were estimated in NONMEM assuming C_m and C_s as observed concentrations (C_{obs}). A simplified structural model (Fig. 3) was used since only i.v. adminstrations were considered and µD sampling in the peripheral compartment (V3) was assumed. This simulation was used to investigate the impact of the biased C_m on the estimation results.

Table 3: Sampling schedules of scenario A & B

	Scenario A				Scenario B	
Intervals [h]	0-0.25	0.25-0.5	0.5-0.75	0.75-1*	0-0.5	0.5-1*
Sampling points [h]	0.125	0.375	0.625	0.875#	0.25	0.75#

*identical sampling points: 1.25, 1.75, 2.25, 2.75, 3.25, 3.75, 4.25, 4.75, 4.75, 5.25, 5.75, 6.25, 6.75, 7.25, 7.75, 8.5, 9.5, 10.5, 11.5
identical intervals: 1-1.5, 1.5-2, 2-2.5, 2.5-3, 3-3.5, 3.5-4, 4-4.5, 4.5-5, 5-5.5, 5.5-6, 6-6.5, 6.5-7, 7-7.5, 7.5-8, 8-9, 9-10, 10-11, 11-12

Results

Relation between interval duration and RE

The RE of C_m in relation to the duration of the first µD sampling interval after 600 mg linezolid is shown for an infusion over 30 min (Fig. 4) and an oral dose (Fig. 5). Due to a complex pattern of RE values for i.v. additional interval durations (6, 9, 12, 75, 105 min) were calculated. The RE values of the investigated interval durations were interpolated leading to a full profile between 6 and 120 min interval duration: The minimum of RE was reached at an interval duration close to 9 min, the maximum at an duration of approx. 60 min for a 'typical' linezold ID. Fig. 5 demonstrates that the larger KA the shorter the interval duration attributed with the smallest RE.

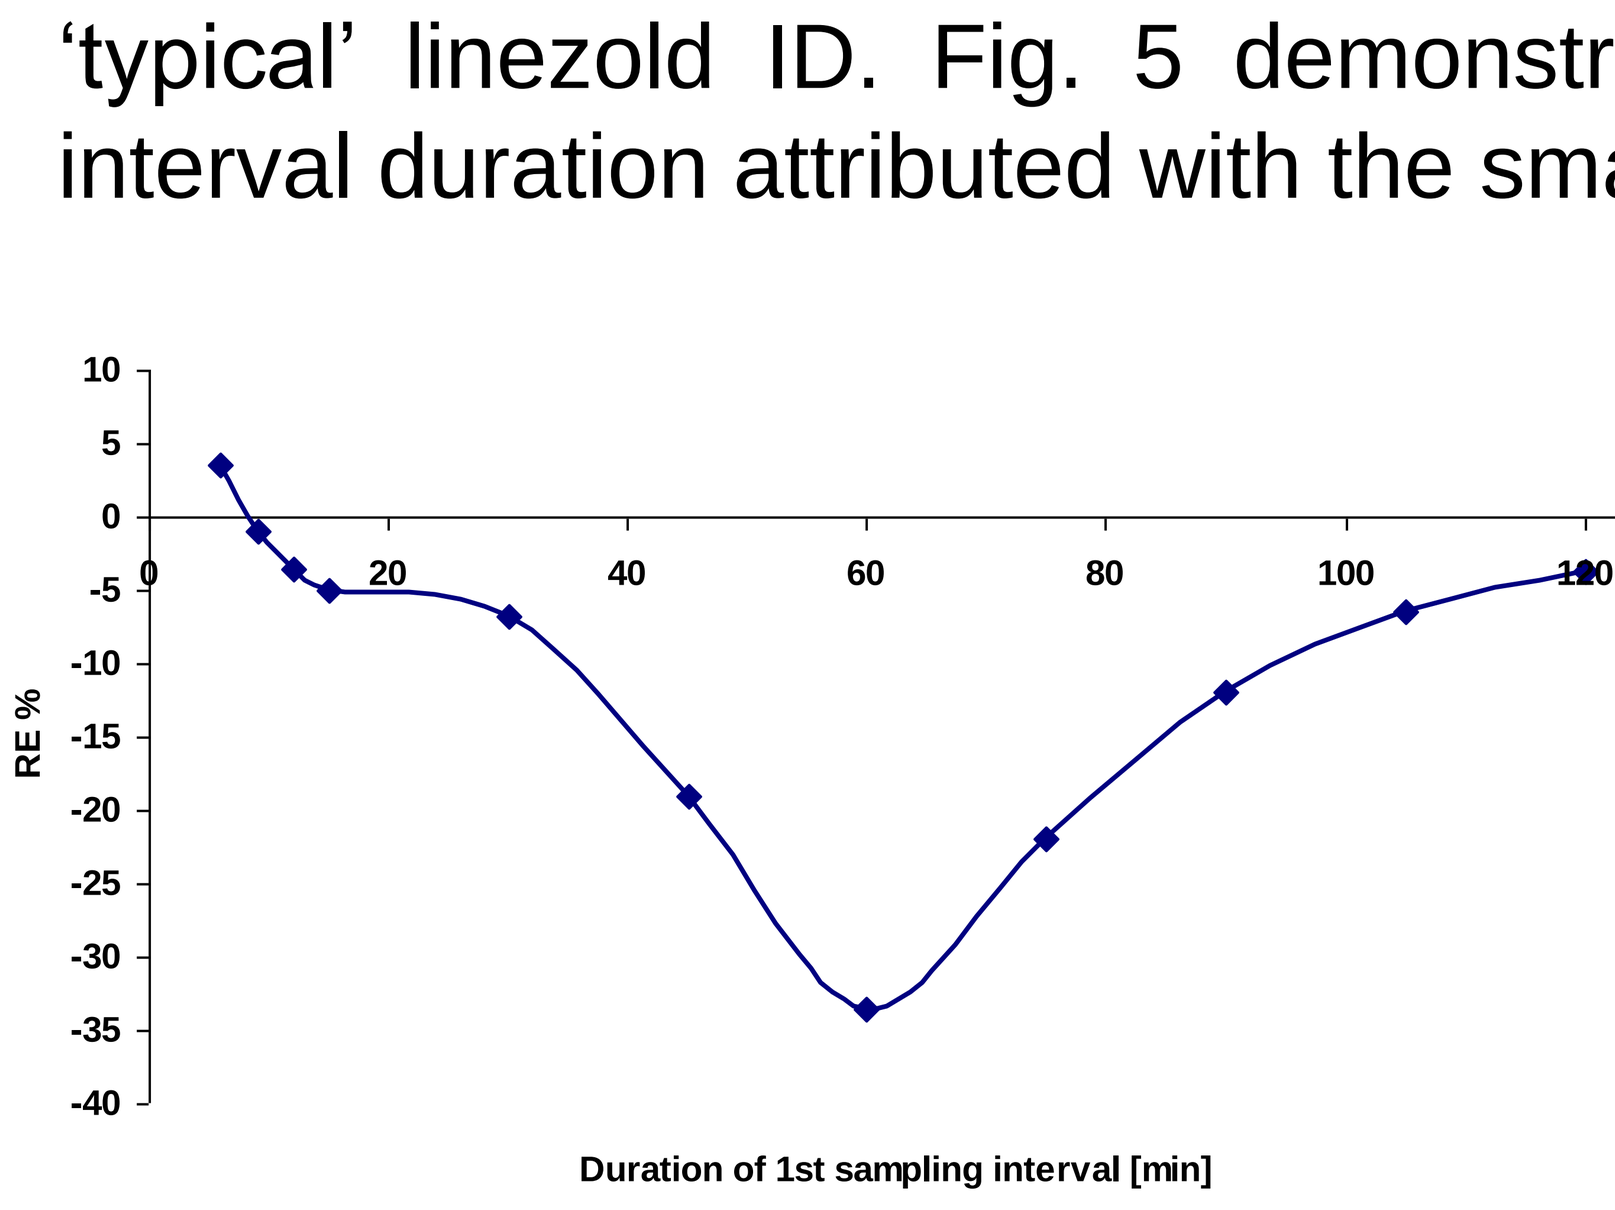
RE %
Duration of 1st sampling interval [min]
Fig. 4: Profile of RE (i.v.)

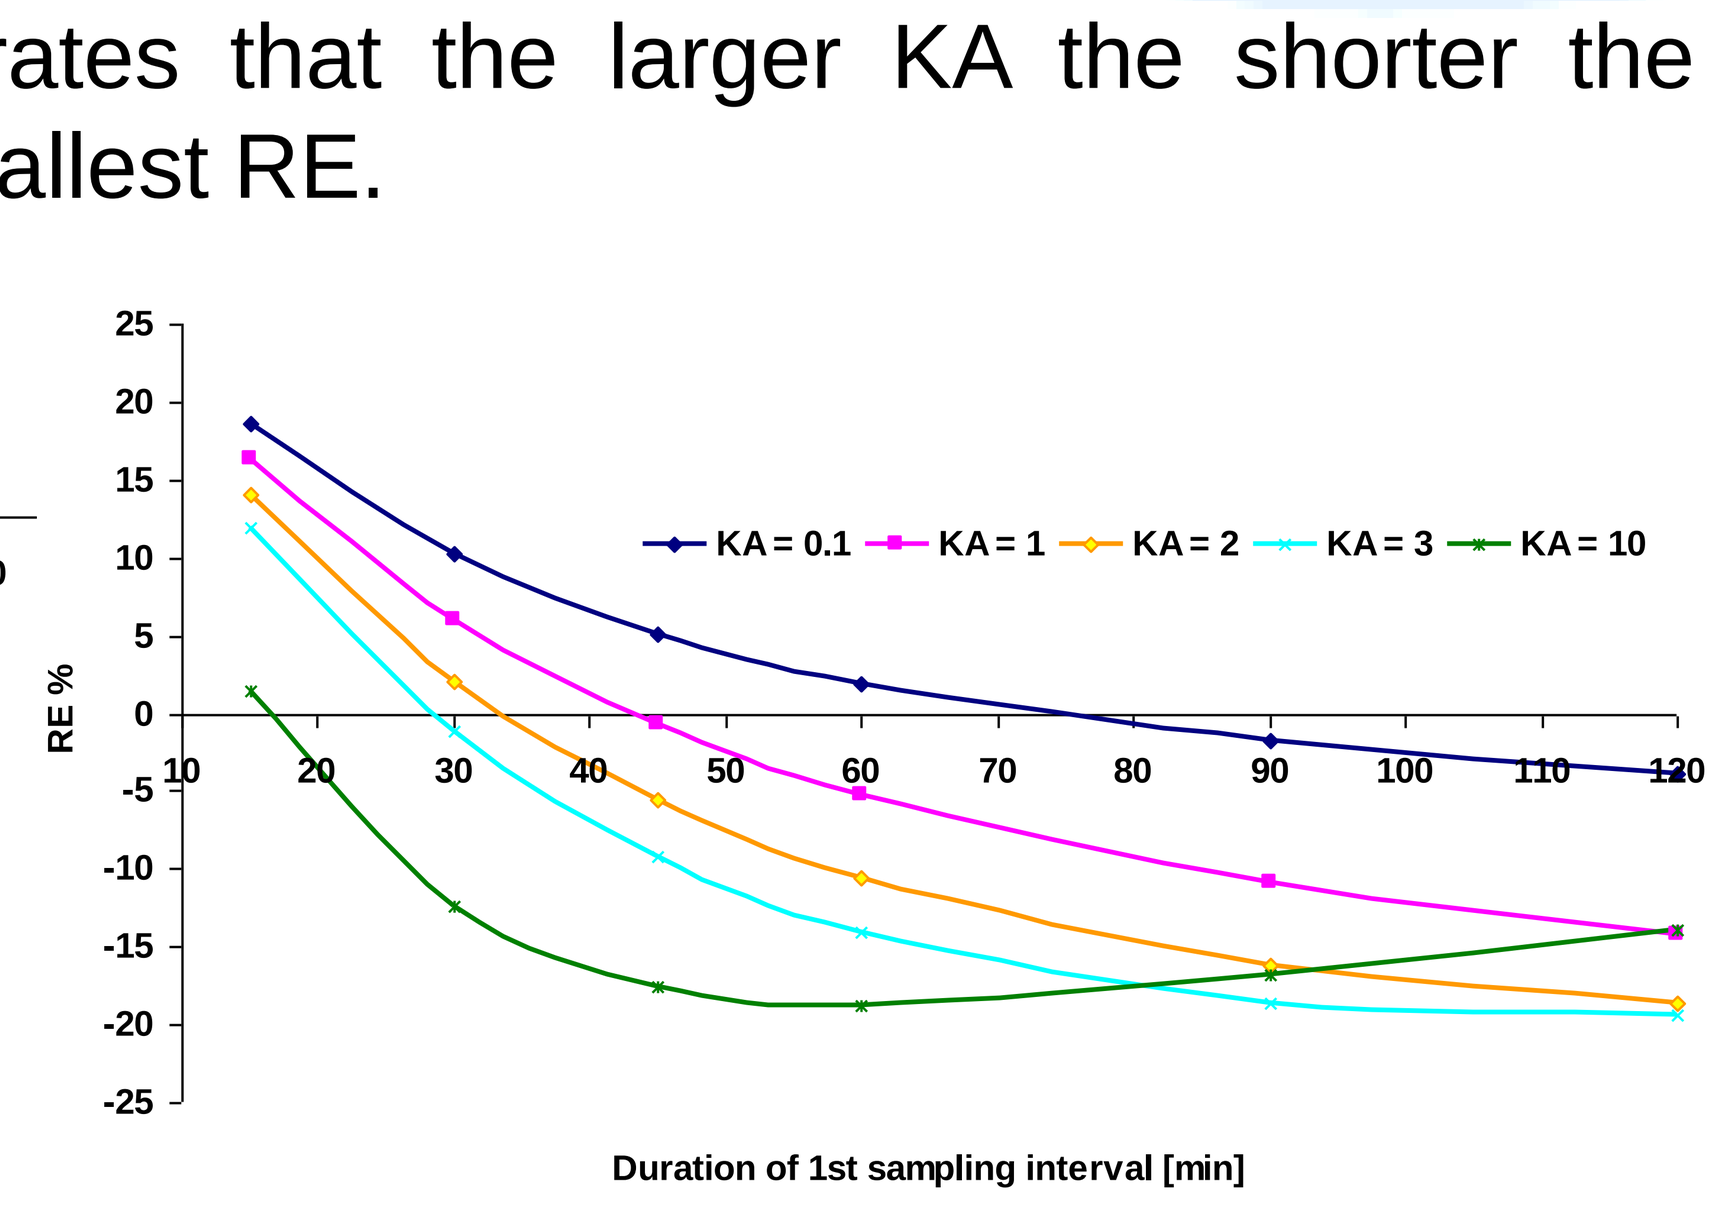
KA = 0.1
KA = 1
KA = 2
KA = 3
KA = 10
RE %
Duration of 1st sampling interval [min]
Fig. 5: RE profiles for different values of KA

Impact of biased C_m on estimation results

Tab. 4 shows min., max. and median RE values of C_m of the 100 IDs simulated. The median RE of the 1st interval duration in scenario A was nearly twice as large as the one in scenario B. The median REs of the 2nd, 3rd and 4th interval in scenario A were significantly smaller than the one of the 2nd interval in scenario B.

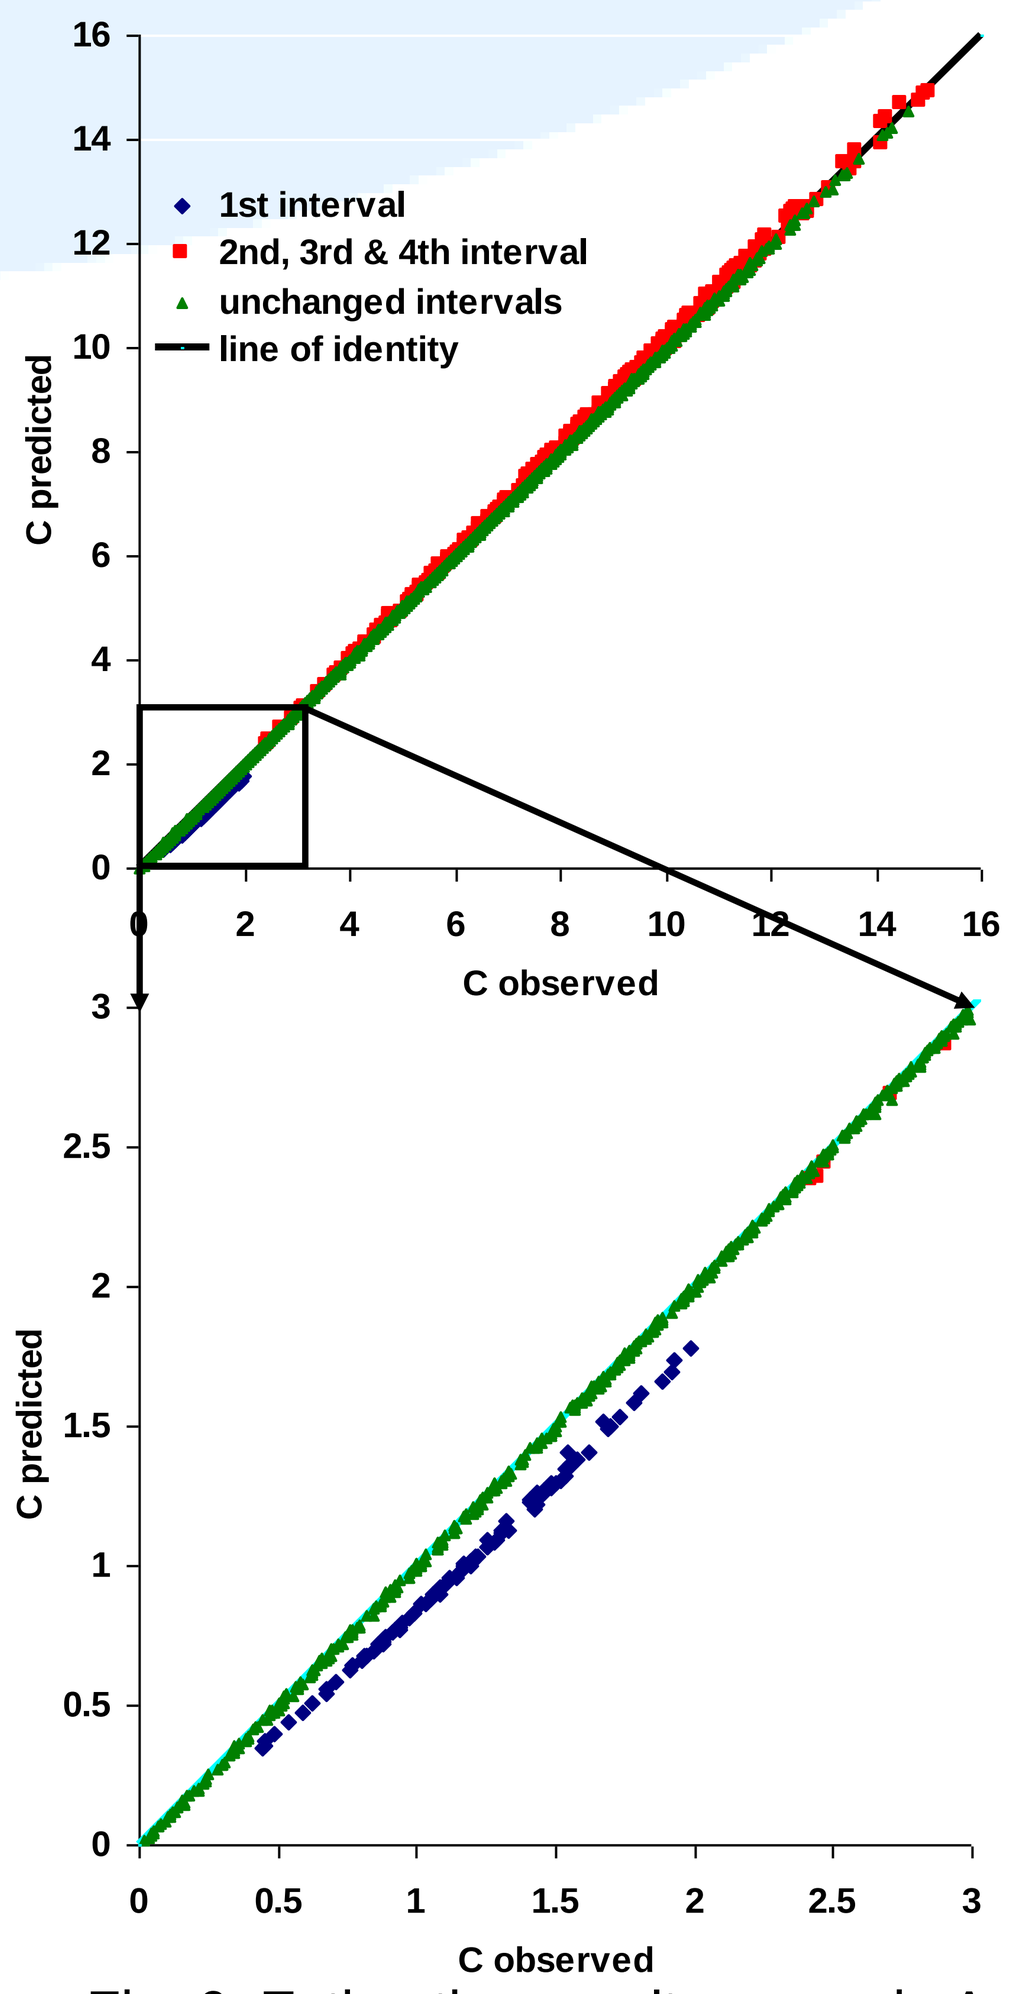
1st interval
2nd, 3rd & 4th interval
unchanged intervals
line of identity
C predicted
C observed
Fig. 6: Estimation results scenario A

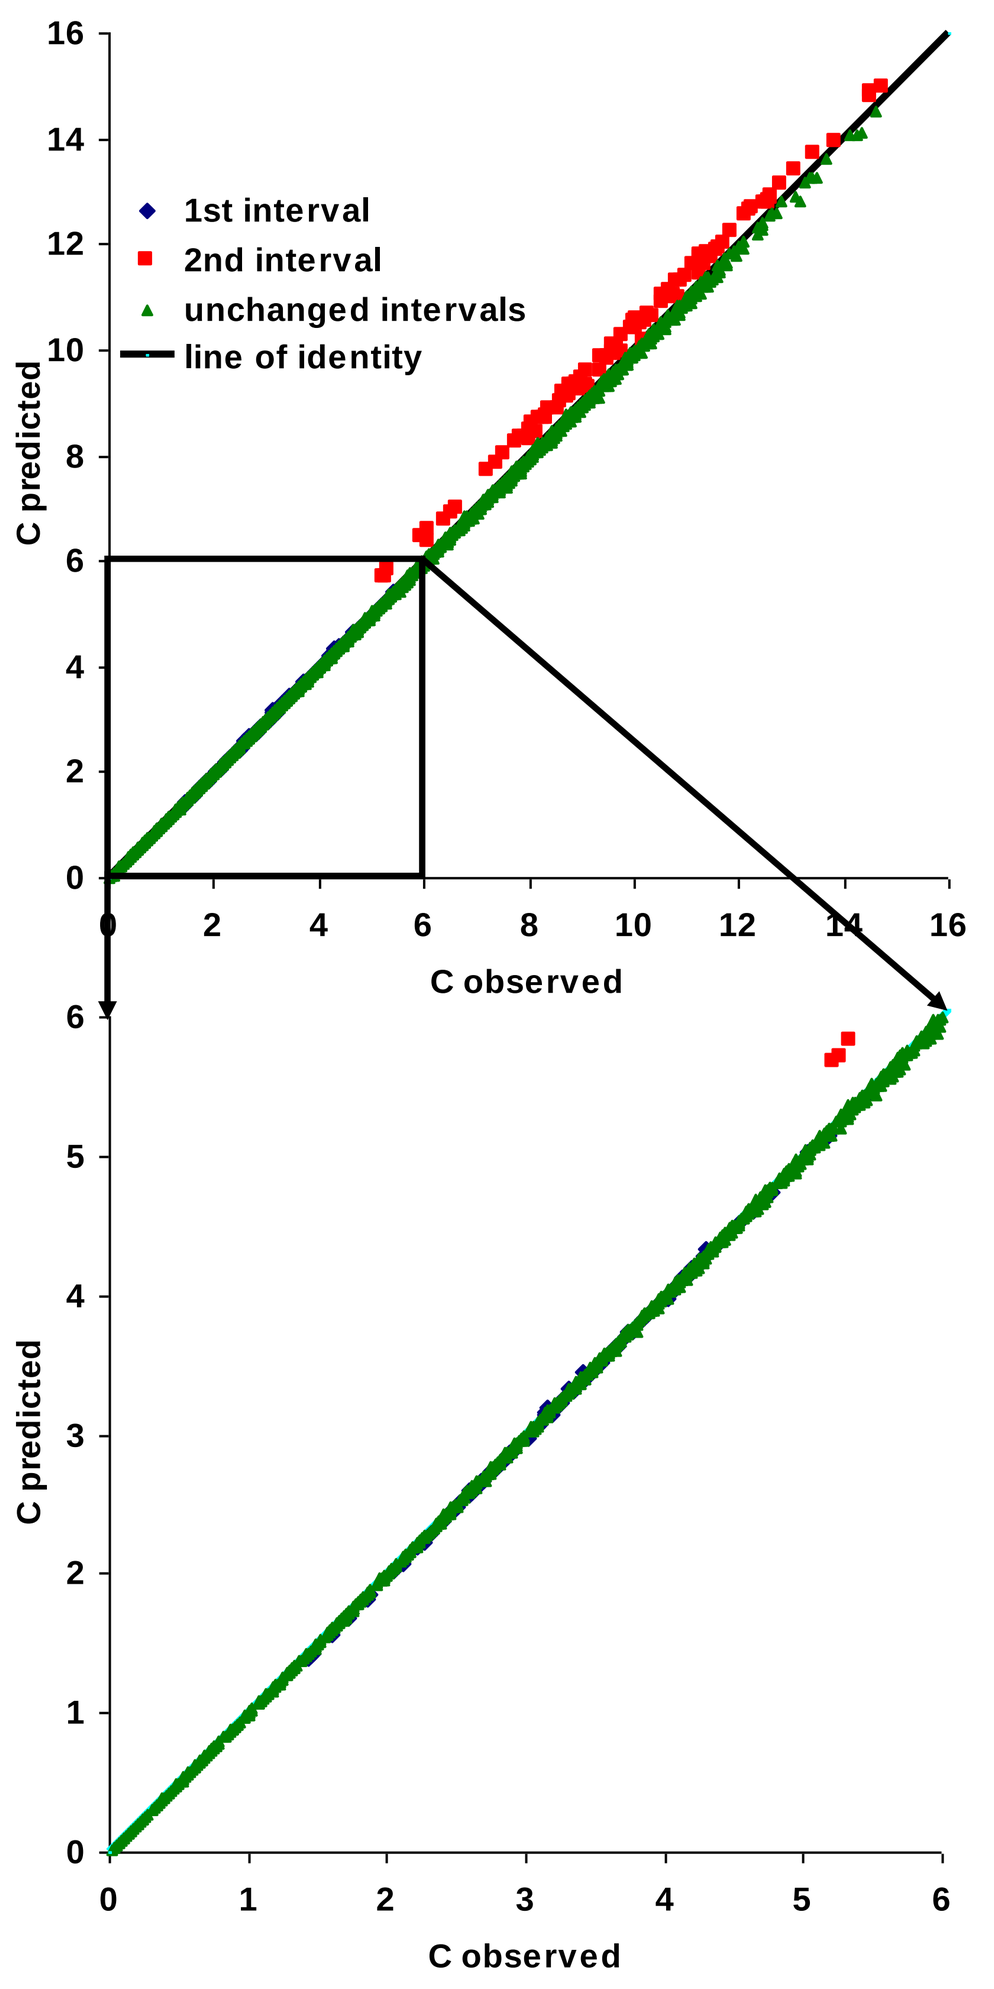
1st interval
2nd interval
unchanged intervals
line of identity
C predicted
C observed
Fig. 7: Estimation results scenario B

Tab. 4: RE values of C_m of 100 IDs simulated

	Interval (time point) [h]	min	median	max
Scen. A	0-0.25 (0.125)	12.02	18.40	24.19
	0.25-0.5 (0.375)	-0.01	0.38	1.29
	0.5-0.75 (0.625)	-1.54	-1.86	-2.12
	0.75-1 (0.875)	-0.06	-0.29	-0.51
Scen. B	0-0.5 (0.25)	4.34	10.10	17.60
	0.5-1 (0.75)	-2.08	-3.30	-3.87

In scenario A (Fig. 6) the µD concentrations of the 1st interval were underestimated while those of the 2nd -4th interval were overestimated. In scenario B (Fig. 7) the predicted concentrations of the 1st interval very closely matched C_{obs}, although C_{obs} which equals C_m is attributed with an RE of 10% (leading to a deviation in Q of 27%). In contrast, the concentrations of the 2nd interval were overestimated.

References

[1] Plock N, Buerger C, Kuester K, Joukhadar C, Kljucar S, Kloft C. A Population Pharmacokinetic Model for the Simultaneous Description of Linezolid Tissue and Plasma Disposition in Healthy Volunteers and Septic Patients, PAGE 15 (2006) Abstr 886 [www.page-meeting.org/?abstract=886].

Conclusion

Prerequisites for methodological investigations of Optimal Design for µD studies were generated and the special characteristics of this sampling technique explored. For the linezolid population PK model investigated, the relation between the µD sampling interval duration and the RE value was determined identifying favourable sampling interval durations. First results suggest an impact of the RE on the estimation of PK parameter - to comprehensively describe the impact of biased concentrations on parameter estimation further investigations have to be carried out. These analyses will give useful information about the utilisation of µD as an attractive tool to monitor target-site exposure in patients.

CLINICAL PHARMACY