A mechanistic PKPD model of early asthmatic response

Victor Sokolov[1], Tatiana Yakovleva[1], Kirill Zhudenkov[1], Anne-Kristina Mercier[3], Gabriel Helmlinger[2], Kirill Peskov[1], Ulf Eriksson[3]

M&S Decisions, Moscow, Russia[1], and Quantitative Clinical Pharmacology, AstraZeneca, Waltham, USA[2] and Gothenburg, Sweden[3]

1085-P

Abstract

This work was aimed at understanding the early asthmatic response to anti-leukotriene drug administration in allergen challenge studies.

The model describes the relationship between leukotriene (LT) inhibitor pharmacokinetics (PK), pre-challenge suppression of blood leukotriene B4 (LTB4), urinary leukotriene E4 (uLTE4) levels, and the reduction of Forced Expiratory Volume in 1 second (FEV1).

Introduction

- Leukotrienes are a family of inflammatory mediators produced by a variety of cells in many asthmatic subjects.
- In asthmatic patients with an activated LT cascade, inhibition of LT biosynthesis or action reduces acute and chronic symptoms including bronchoconstriction, edema, inflammation, and remodeling.
- Urinary LTE4 is an efficacy biomarker used in early phase clinical development, while FEV1 is an endpoint of choice in later phases.
- FEV1 is highly variable and dependent upon multiple factors, thus the prediction of FEV1 response based on early clinical biomarker data is a challenge.

Methods

- Published data from clinical Phase I and II studies of a 5-Lipoxygenase-activating-protein inhibitor (GSK2190915) and a 5-Lipoxygenase inhibitor (MK-0591) were used to build the model [1-3]. The model incorporates the early asthmatic response (within 2 hours after allergen challenge).
- Modeling was performed in the IQM Tools (replacing the SBTOOLBOX2 by IntiQuan - http://www.intiquan.com/). The model was based on a 2-compartment module for PK, an "Imax" transit compartment to describe the biomarker, and an indirect response function to describe the allergen challenge and the FEV1 endpoint estimation.

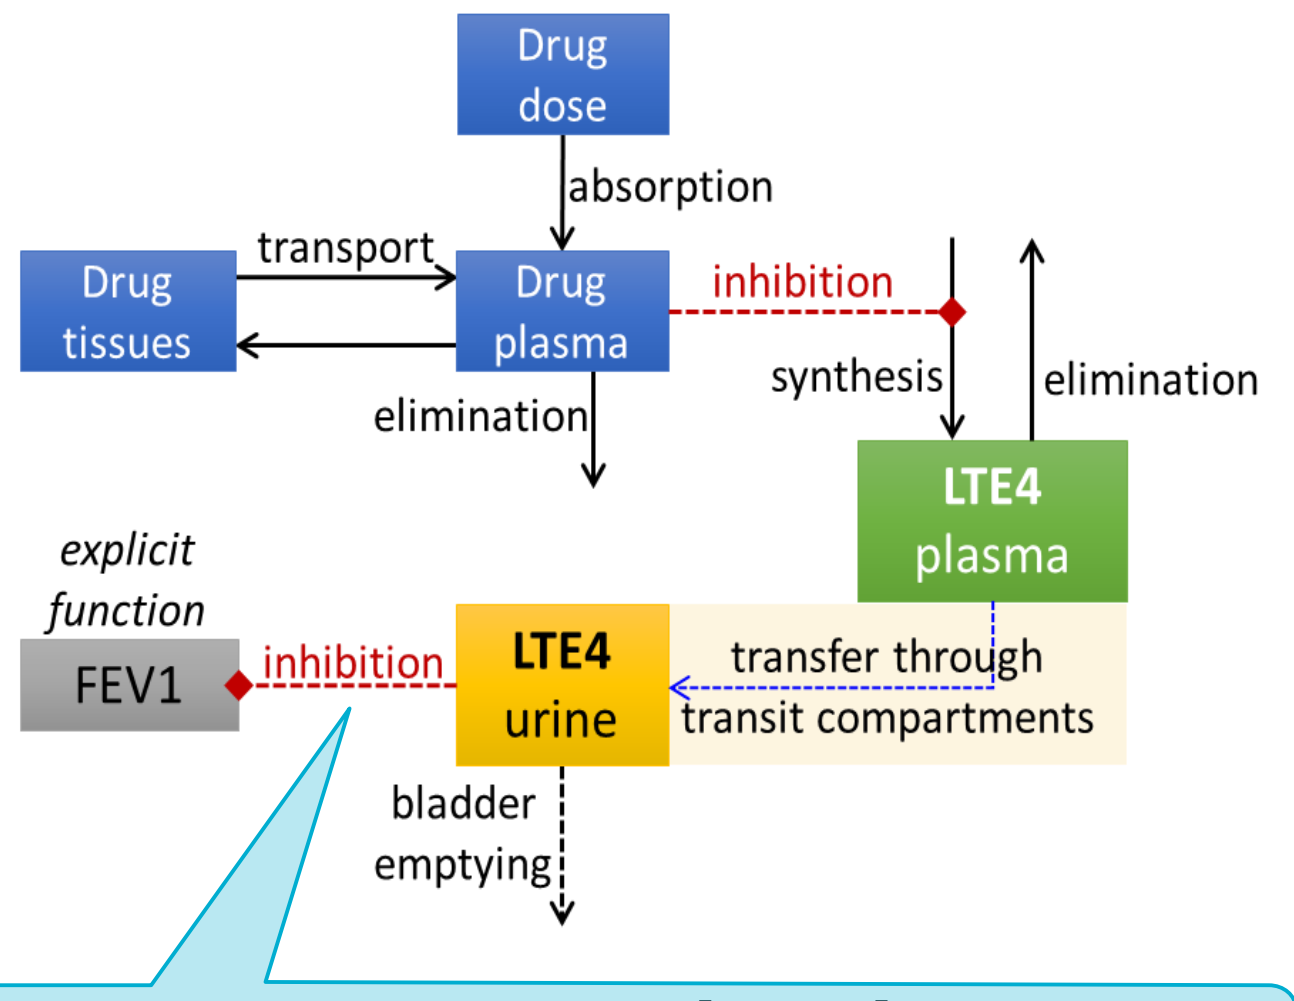
Drug dose
absorption
Drug tissues
transport
Drug plasma
inhibition
elimination
synthesis
elimination
LTE4 plasma
explicit function
FEV1
inhibition
LTE4 urine
transfer through transit compartments
bladder emptying

$$FEV1 = (1 + k_{eff} * uLTE4) * D * \left[\frac{k_1}{k_1 - k_2}\right] * \left[e^{-k_2 t} - e^{-k_1 t}\right]$$

Fig. 1 Mechanistic PKPD model scheme and FEV1 equation

The following results are focused on the FEV1-uLTE4 compartments of the model.

Results

FEV1 dynamics after an allergen challenge

FEV1 levels typically drop, in response to LT production inhibition and as seen in provocation studies which make use of a specific allergen [4].

Using study design criteria [4] to drive the model, uLTE4 levels were observed to decrease to zero, following pre-challenge administration of an anti-LT compound.

uLTE4 levels recovered, with a delay, even after most of the inhibitor compound had been removed from the circulation [5,6].

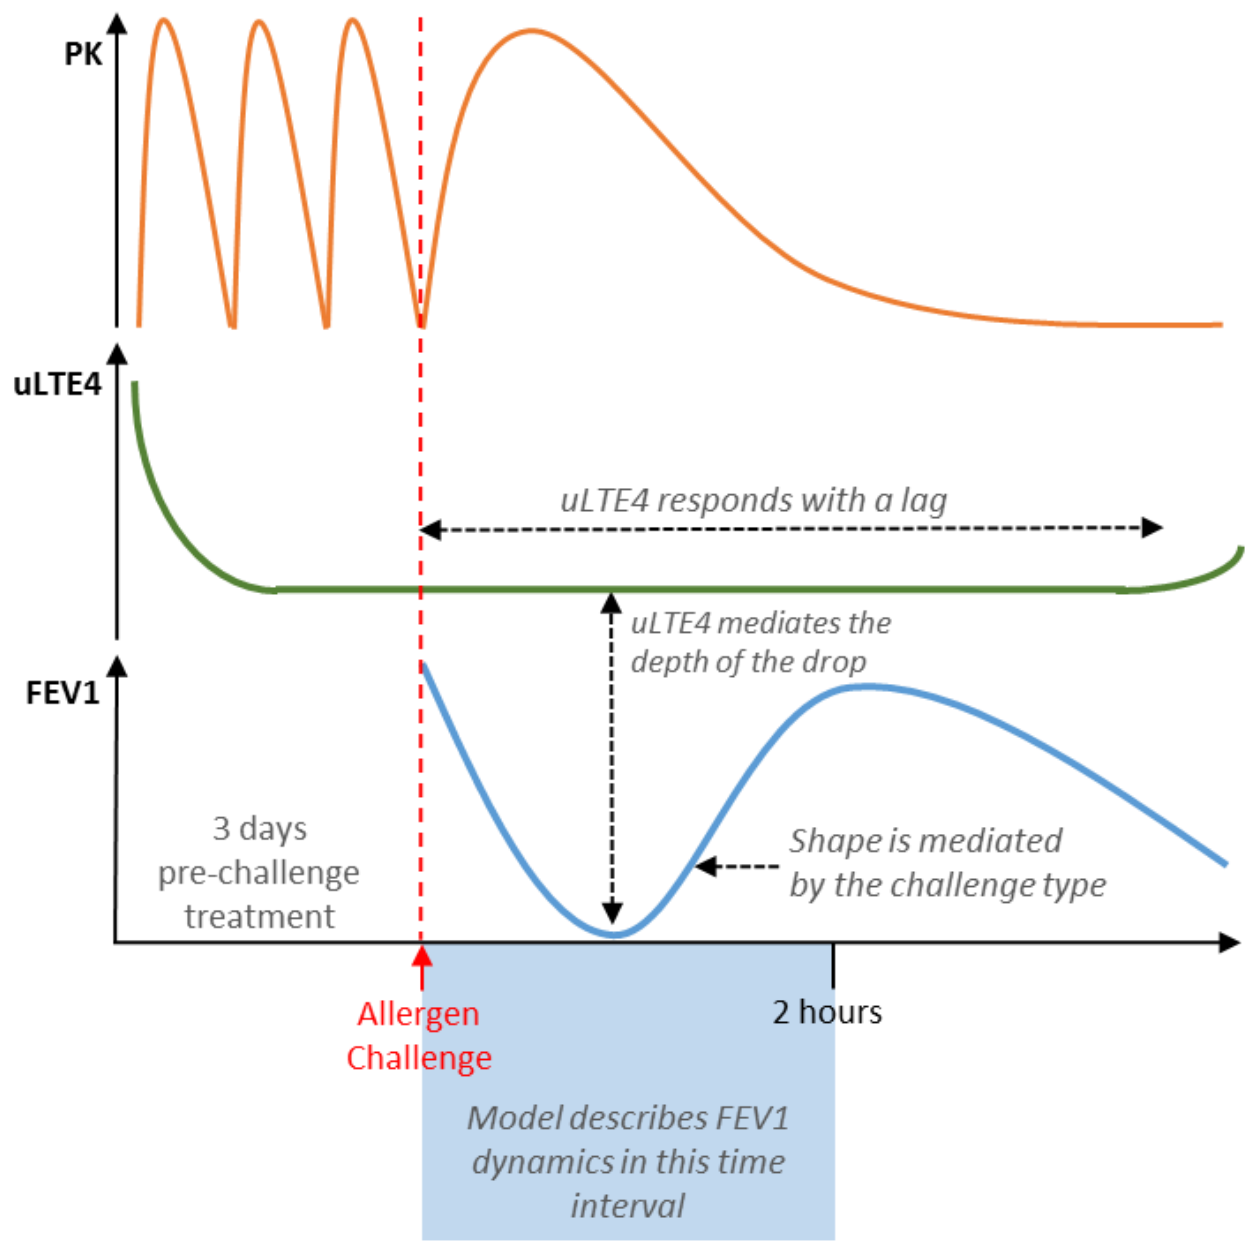
PK
uLTE4
FEV1
uLTE4 responds with a lag
uLTE4 mediates the depth of the drop
3 days pre-challenge treatment
Shape is mediated by the challenge type
Allergen Challenge
2 hours
Model describes FEV1 dynamics in this time interval

Fig. 2 PK, uLTE4 and FEV1 trends in allergen challenge studies.

uLTE4 regulates how strong FEV1 will decrease after allergen challenge thus responsible for the maximal drop of FEV1.

FEV1 parameter fitting based on GSK2190915 and MK-0591 treatment data

The model reproduced uLTE4 suppression by 10-450 mg of GSK2190915 [5] and 50-500 mg of MK-0591 [6], thus demonstrating a quantitative link between pre-challenge uLTE4 levels and FEV1 within the first 2 hours following an allergen challenge.

Table 1. Parameter fitting, FEV1 equation.

Parameter name	Value	Dimension
k_{eff}	0.0085	
D	0.569	L
k_1	13.14	hour^{-1}
k_2	0.786	hour^{-1}

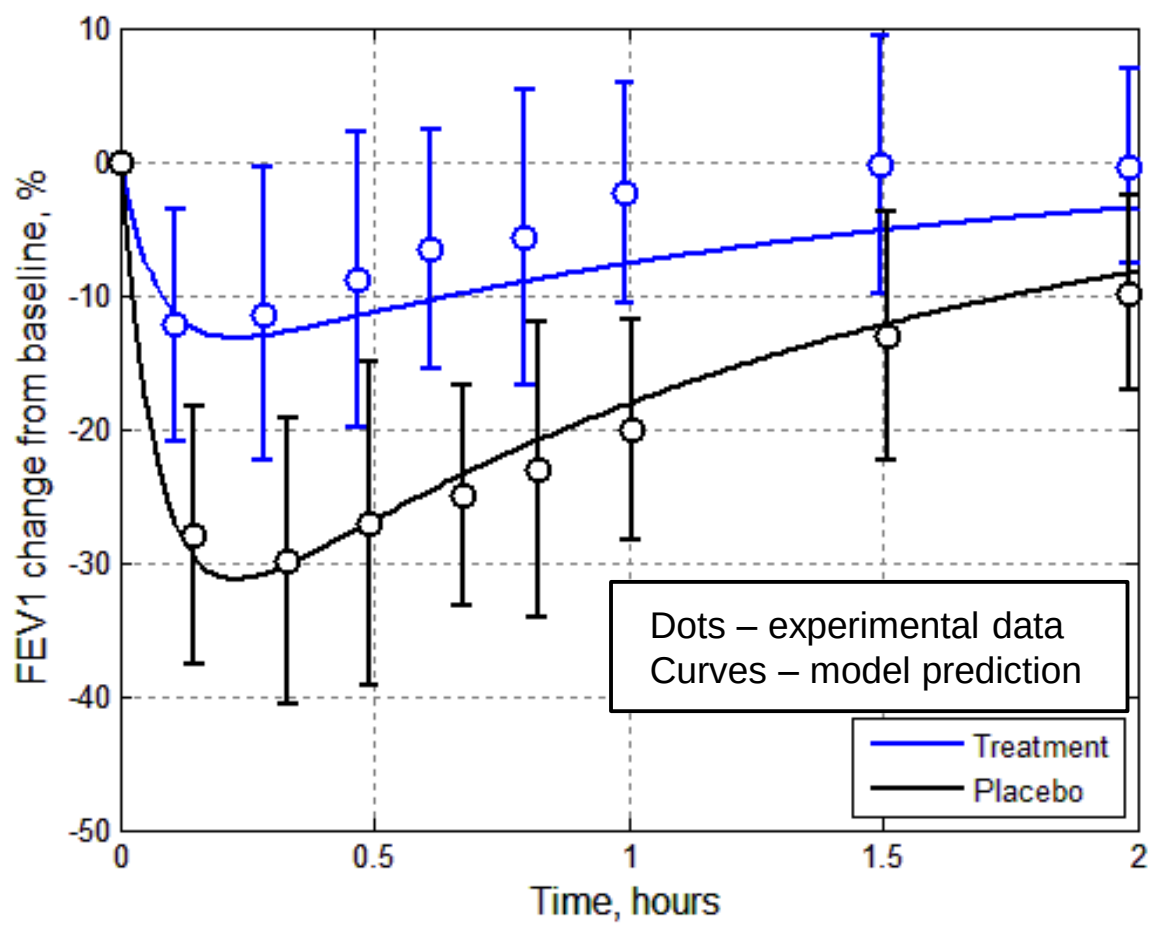
FEV1 change from baseline, %
Time, hours
Dots – experimental data
Curves – model prediction
Treatment
Placebo

Fig. 3 First two hours of FEV1 dynamics after allergen challenge under 250 mg MK-0591 treatment.

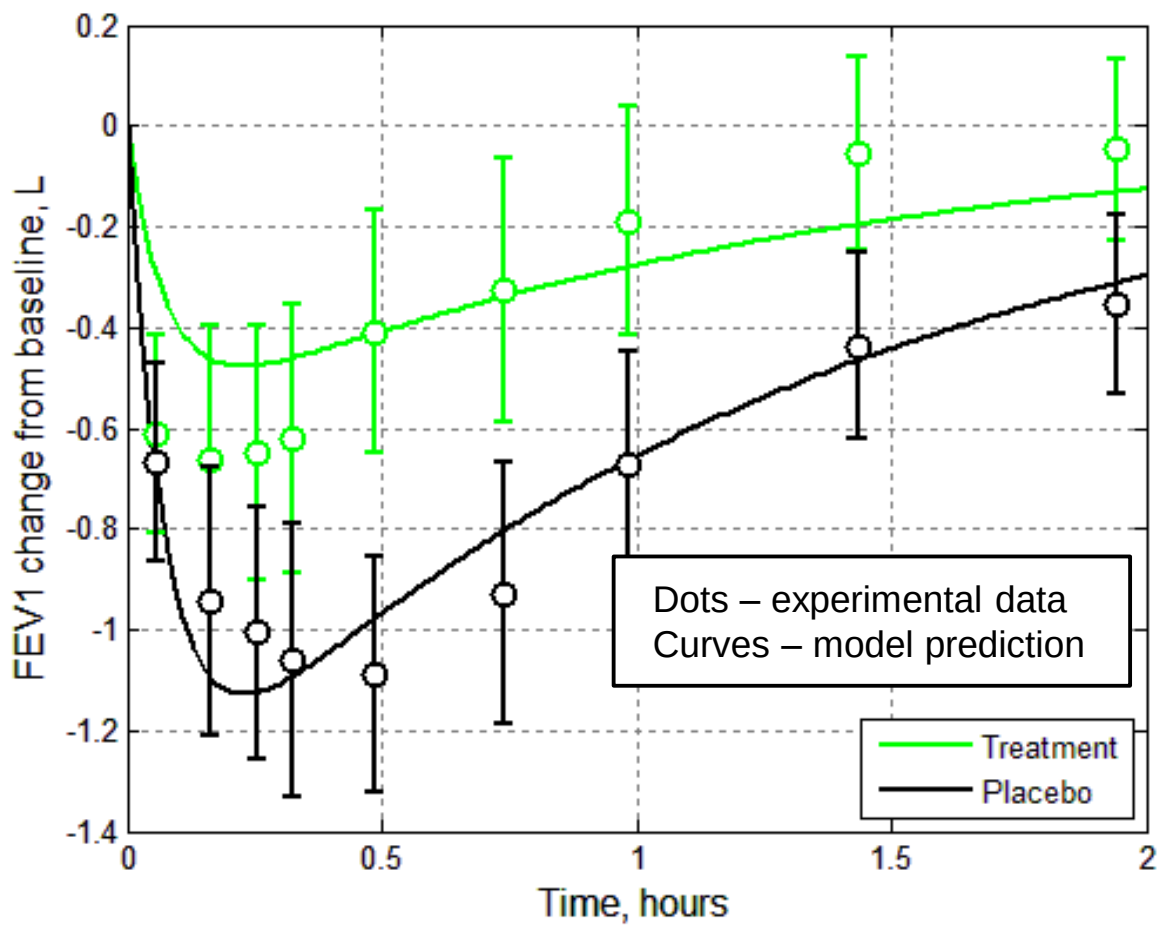
FEV1 change from baseline, L
Time, hours
Dots – experimental data
Curves – model prediction
Treatment
Placebo

Fig. 4 First two hours of FEV1 dynamics after allergen challenge under 100 mg GSK2190915 treatment.

Validation of FEV1 response under GSK2190915 treatment data set

Predictions of FEV1 levels were in full agreement with the data reported for the 10-50 mg GSK2190915 studies [3]. A maximal FEV1 drop of -0.5 to -0.8L was achieved *vs*. baseline. Model predictions were in close agreement with reported measurements.

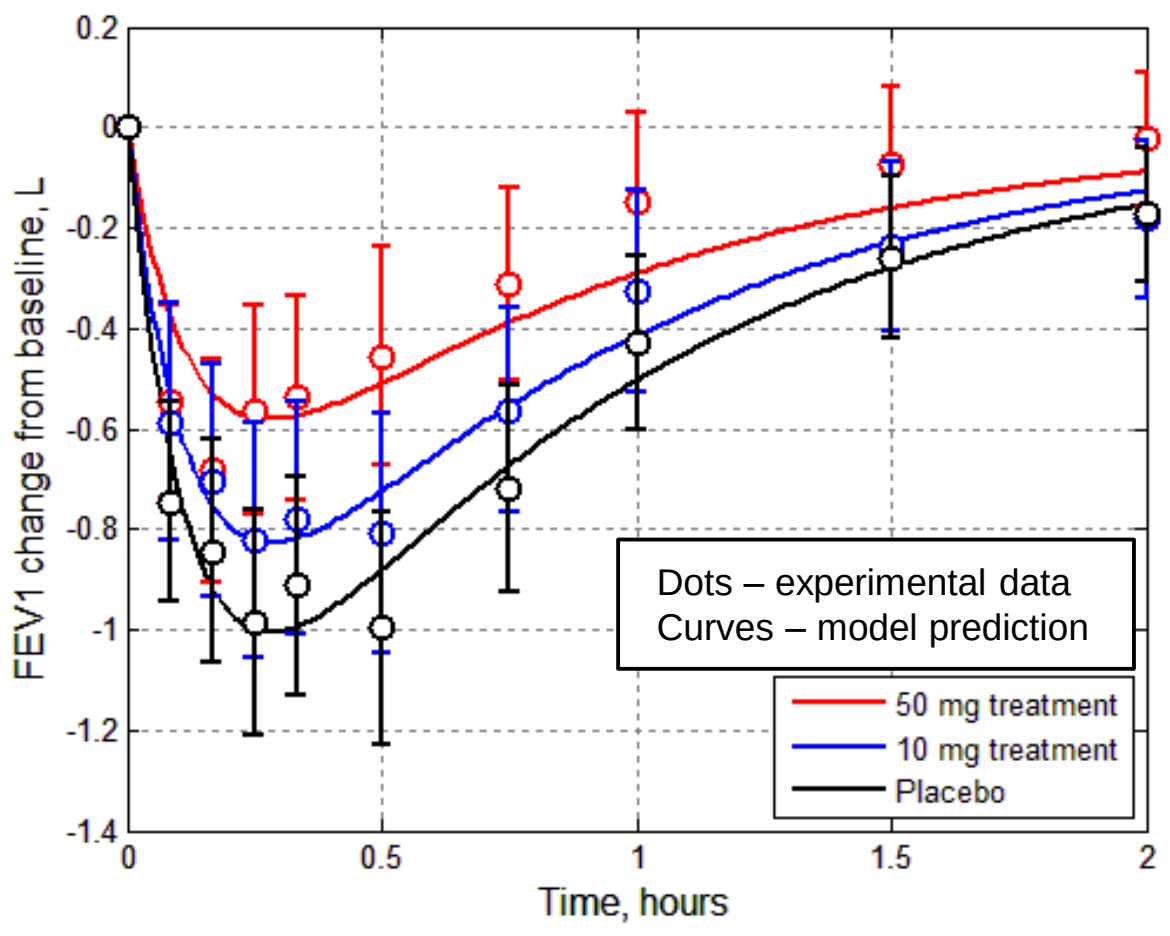
FEV1 change from baseline, L
Time, hours
Dots – experimental data
Curves – model prediction
50 mg treatment
10 mg treatment
Placebo

Fig. 5 First two hours of FEV1 dynamics after allergen challenge under 10 mg and 50 mg GSK2190915 treatment.

In summary, two data sets were used for parametrization of FEV1 response to an allergen challenge [5,6], while a third independent data set was successfully used for validation of the model [3].

Conclusions

- We developed a PKPD model describing the relationship between LT-inhibitor PK, pre-allergen-challenge uLTE4 levels, and early post-allergen-challenge FEV1.
- Such a model may be applied towards the prediction of FEV1 response in Phase II allergen challenge studies, based on Phase I compound PK and uLTE4 suppression data.

References

1. Diamant et al. 1995, J Allergy Clin Immunol
2. Kent et al. 2013, Clin Exp Allergy
3. Singh et al. 2013, Int J Gen Med
4. Kuziemski et al. 1998, Pol Merkur Lekarski
5. Bain et al. 2013, Br J Clin Pharmacol
6. Uematsu et al. 1995, Br J Clin Pharmacol

Supported by AstraZeneca IMED Biotech Unit

Victor Sokolov, PAGE 2016, 10 June 2016