Impact of study design for characterising PKPD covariance and nonlinearity in exposure-dichotomous response relationships

Rocío Lledó-García, Stefanie Hennig and Mats O. Karlsson
Department of Pharmaceutical Biosciences, Uppsala University, Sweden

Background and Objective

Characterisation of exposure-response for therapies where the clinical endpoint is a bivariate outcome generally involves two assumptions: (i) the random effect determining the dose-exposure relation is not related to any parameter of the exposure-response relation, and (ii) the drug effect is linear on the logit scale[1,2]. This study aims at assessing the possibility for randomised dose controlled trials (DCT) and concentration controlled trials (CCT) to characterise such phenomena, with a particular focus on drugs with narrow therapeutic index.

Methods

PKPD model: A simulation-based study was performed using NONMEM VI considering a hypothetical immunosuppressant agent with rejection as main efficacy endpoint. The PK-model was described by equations:

$$C_i = \frac{D_i}{CL_i} \qquad CL_i = \theta_{CL} \cdot e^{\eta_{CL}}$$

and the PD-relationship with a regression logistic model:

$$Logit_i = \theta_{B_X} + \theta_{S_X} \cdot C_i \qquad P_X = \frac{e^{Logit_i}}{1+e^{Logit_i}} \qquad \text{X= rejection}$$

Two alternative PD-models were considered:
a) PKPD covariance between CL and baseline or slope parameter:

$$Logit_i = \theta_{Brejection} + \theta_{Srejection} \cdot \left[\frac{CL_i}{CL}\right]^{\delta} \cdot C_i \quad Logit_i = \theta_{Brejection} \cdot \left[\frac{CL_i}{CL}\right]^{\delta} + \theta_{Srejection} \cdot C_i \quad \delta = 1, -1$$

b) Nonlinear exposure-response relationship:

$$Logit_i = \theta_{Brejection} + \theta_{Srejection} \cdot C_i^{\gamma} \qquad \gamma = 2, 3, 4$$

Simulation setup: the default considered as typical value for clearance (θCL), 20 L/h and 45% of IIV. As baselines and slopes: θBinf= -3.5, θSinf =15, θBrej= -1 and θSrej= -12.
As δ and γ the values shown in the equations. When γ took the values 1, 2, 3, 4, the θSrej took values of -12, -120, -1200 and -12000, respectively. So that, in all cases the covariance/non-linearity was inside the range of study and could be detected *a priori*

Study design: 3 randomized, cross-over designs with two dose/exposure levels were considered:
(i) DCT with two dose level targets;
(ii) TCCT with exposures that reflect the expected typical exposure in the corresponding DCT;
(iii) VCCT targeting the two exposure levels that result in equal exposure variability as the corresponding DCT.

A study size of 500 subjects was considered and four different ranges of target levels were explored[3].

When assessing the non-linearity, the CCTs designs targeted three levels of exposures inside the same range as the corresponding DCT, so that the VCCT still was equi-variable to the DCT.

Results and Discussion

With regards to precision and bias in parameter estimates: DCT and VCCT were superior for a (+) and (-) PKPD covariance between CL and baseline, respectively (Figure 1 & 2).

When the PKPD covariance existed between CL and slope, the VCCT design was the more precise regardless of the sign of the correlation (Figure 3). The VCCT and TCCT showed highest power to detect the correlation in all cases (results not shown).

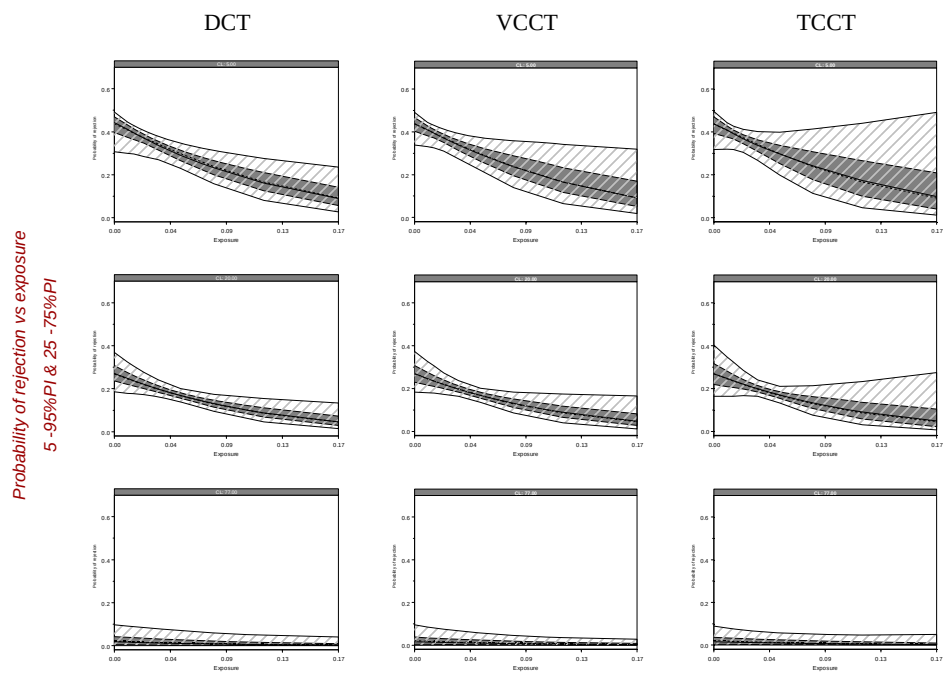
DCT
VCCT
TCCT
Probability of rejection vs exposure
5 -95%PI & 25 -75%PI

CL-Brej; CL: 5,20,77 L/h

Figure 1. *5 -95% &, 25-75% predicted intervals (PI), together with the predicted median and true probability of having rejection at different exposures. Based on information gained from DCT and CCTs, when the range of study was 0.5 & 1mg and a (+) covariance between CL-Brej was considered.*

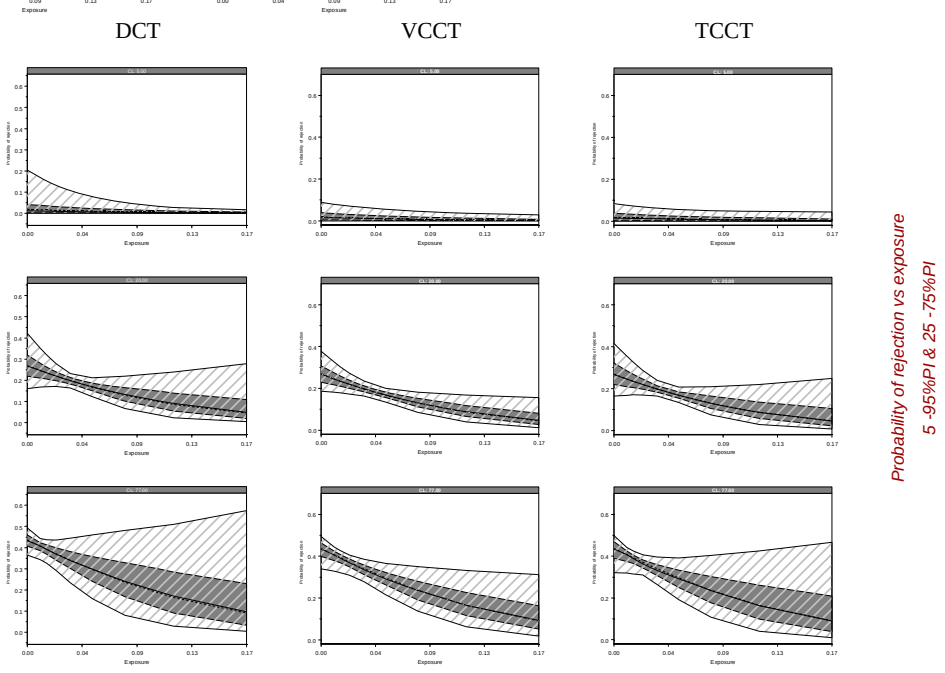
DCT
VCCT
TCCT
Probability of rejection vs exposure
5 -95%PI & 25 -75%PI

CL-Brej; CL: 5,20,77 L/h

Figure 2. *5 -95% &, 25-75% PI, together with the predicted median and true probability of having rejection at different exposures. Based on information gained from DCT and CCTs, when the range of study was 0.5 & 1mg and a (-) covariance between CL-Brej was considered.*

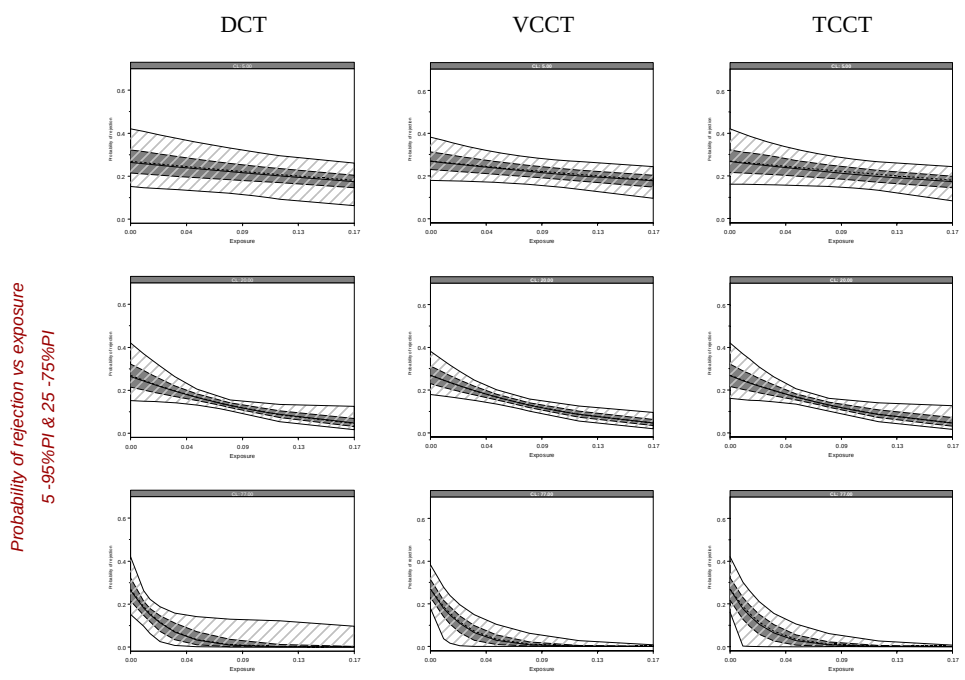
DCT
VCCT
TCCT
Probability of rejection vs exposure
5 -95%PI & 25 -75%PI

CL-Srej; CL: 5,20,77 L/h

Figure 3. *5 -95% &, 25-75% PI, together with the predicted median and true probability of having rejection at different exposures. Based on information gained from DCT and CCTs, when the range of study was 1 & 2mg and a (+) covariance between CL-Srej was considered.*

To characterise a nonlinearity in exposure-response, DCT was more precise and less biased in the parameter estimates as well as showing higher power. However, the VCCT was almost as precise (Figure 4).

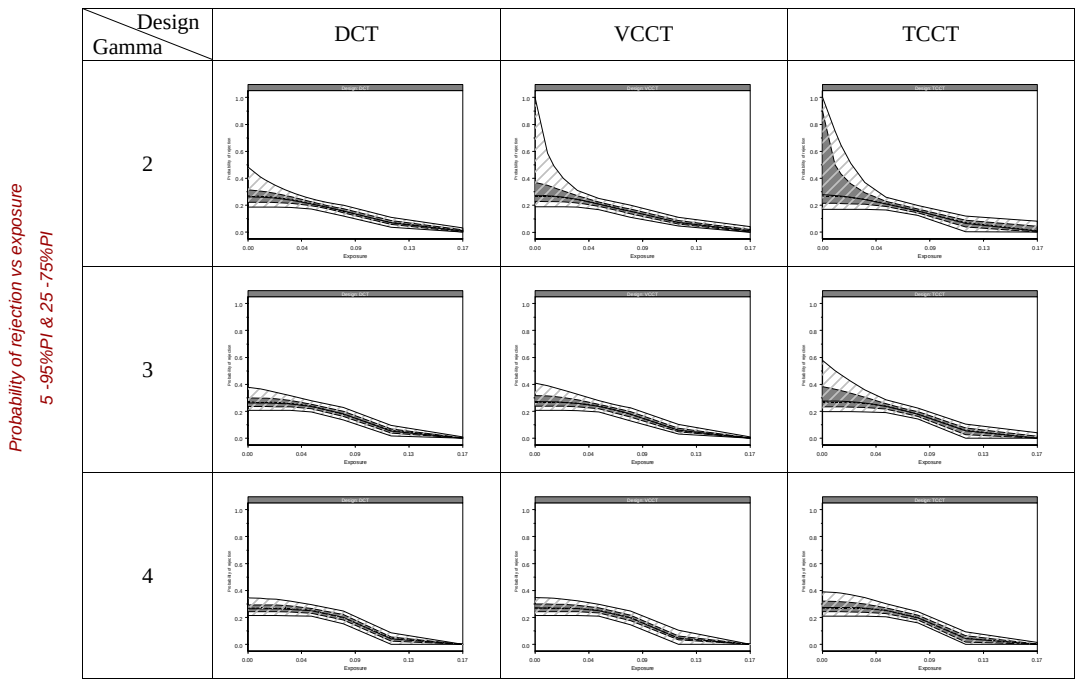
Design
Gamma
DCT
VCCT
TCCT
2
3
4
Probability of rejection vs exposure
5 -95%PI & 25 -75%PI

Figure 4. *5 -95% &, 25-75% PI, together with the predicted median and true probability of having rejection at different exposures. Based on information gained from DCT and CCTs, when the range of study was 1-1.5-& 2mg for the different degrees of non-linearity (γ =2,3,4).*

To achieve a >90% power to detect either a PKPD covariance or nonlinearity, studies involving >500 patients would be required (not shown).

Conclusion

For drugs with narrow therapeutic index either a VCCT or DCT have shown to be more informative designs to describe the exposure-response relationship when there is a PKPD covariance in the parameters, whereas a DCT seems to be more informative when describing nonlinear relationships between exposure and response. However, typical studies of this type would not have enough power to reliably detect such relationships regardless of design.

Acknowledgments

R.Ll.G. was supported by a grant from Pfizer, Sandwich, U.K

Reference

1. Hale, M.D., et al. Clin Pharmacol Ther, 1998. 64(6): p. 672-83.
2. Yassen, A., et al. Pharm Res, 2008. 25(1): p. 183-93.
3. Lledó-García R et al. PAGE 17 (2008) Abstract 1326 [www.page-meeting.org/?abstract=1326]