Investigation of Bayesian inference in predicting tissue concentrations using RStan

Shan Pan, Stefano Zamuner, Shuying Yang

Clinical Pharmacology Modelling & Simulation, GSK, Stevenage, United Kingdom

Background & Objectives

- A physiologically based pharmacokinetic (PBPK) model representing physiological profiles may not be available due to limited data in tissues or organs, even in animal studies.
- Minimal PBPK models, generically lumped full PBPK models, can be an alternative to evaluate target tissue concentrations.
- In this work we aimed to investigate predictions of tissue concentrations using Bayesian approach, for compound X that is intended for the treatment of inflammatory liver disease.

Methods

- A full PBPK model in GastroPlus® for compound X (**Figure 1**) was available from a pre-clinical study in cynomolgus monkeys, with the liver as the site of elimination.
- Using the full PBPK model scaled to humans, 1,000 subjects aged between 20 and 80 were simulated in R with ordinary differential equations [1].
- In the liver, volume and blood flow were correlated and decreased by 20-40% with increase in age; approximately one-fold decline in liver function with increase in age was considered [2].
- Distributions of volume, blood flow and clearance in the liver were obtained and referred to as true parametric distributions.
- Random sampling from the 1,000 subjects was conducted for the following two testing scenarios: (1) plasma data only, and (2) plasma and liver data. In each scenario, both sparse and rich samples were considered for 10 and 100 subjects, respectively.
- Bayesian modelling with a classical PK model or a minimal PBPK [3] was performed in RStan, with prior distributions defined on tissue volume, blood flow and clearance.
- In RStan, analytical solutions for both classical PK and minimal PBPK models were implemented, with the default MCMC sampling settings: nChains=4, nPost=1000, nBurn=1000, nThin=1.
- Trace plots of parameters and log likelihood were monitored, ensuring a stable convergence over iterations for all chains.
- Individual predictions from the classic PK or minimal PBPK model were plotted against plasma or plasma plus liver concentrations as a measure of goodness of fit.
- Posterior distributions of liver parameters, blood flow, volume and clearance, were compared with true parametric distributions in the PBPK stochastic simulation.

ODE for non-eliminating tissues: $V_{tissue} \times \frac{dC_{tissue}}{dt} = Q_{tissue} \times \left(C_{arterial} - \frac{C_{tissue}}{K_p/BP}\right)$

ODE for eliminating tissues: $V_{tissue} \times \frac{dC_{tissue}}{dt} = Q_{tissue} \times \left(C_{arterial} - \frac{C_{tissue}}{K_p/BP}\right) - CL_{int} \times \frac{C_{tissue}}{K_p/BP} \times f_{unbound}$

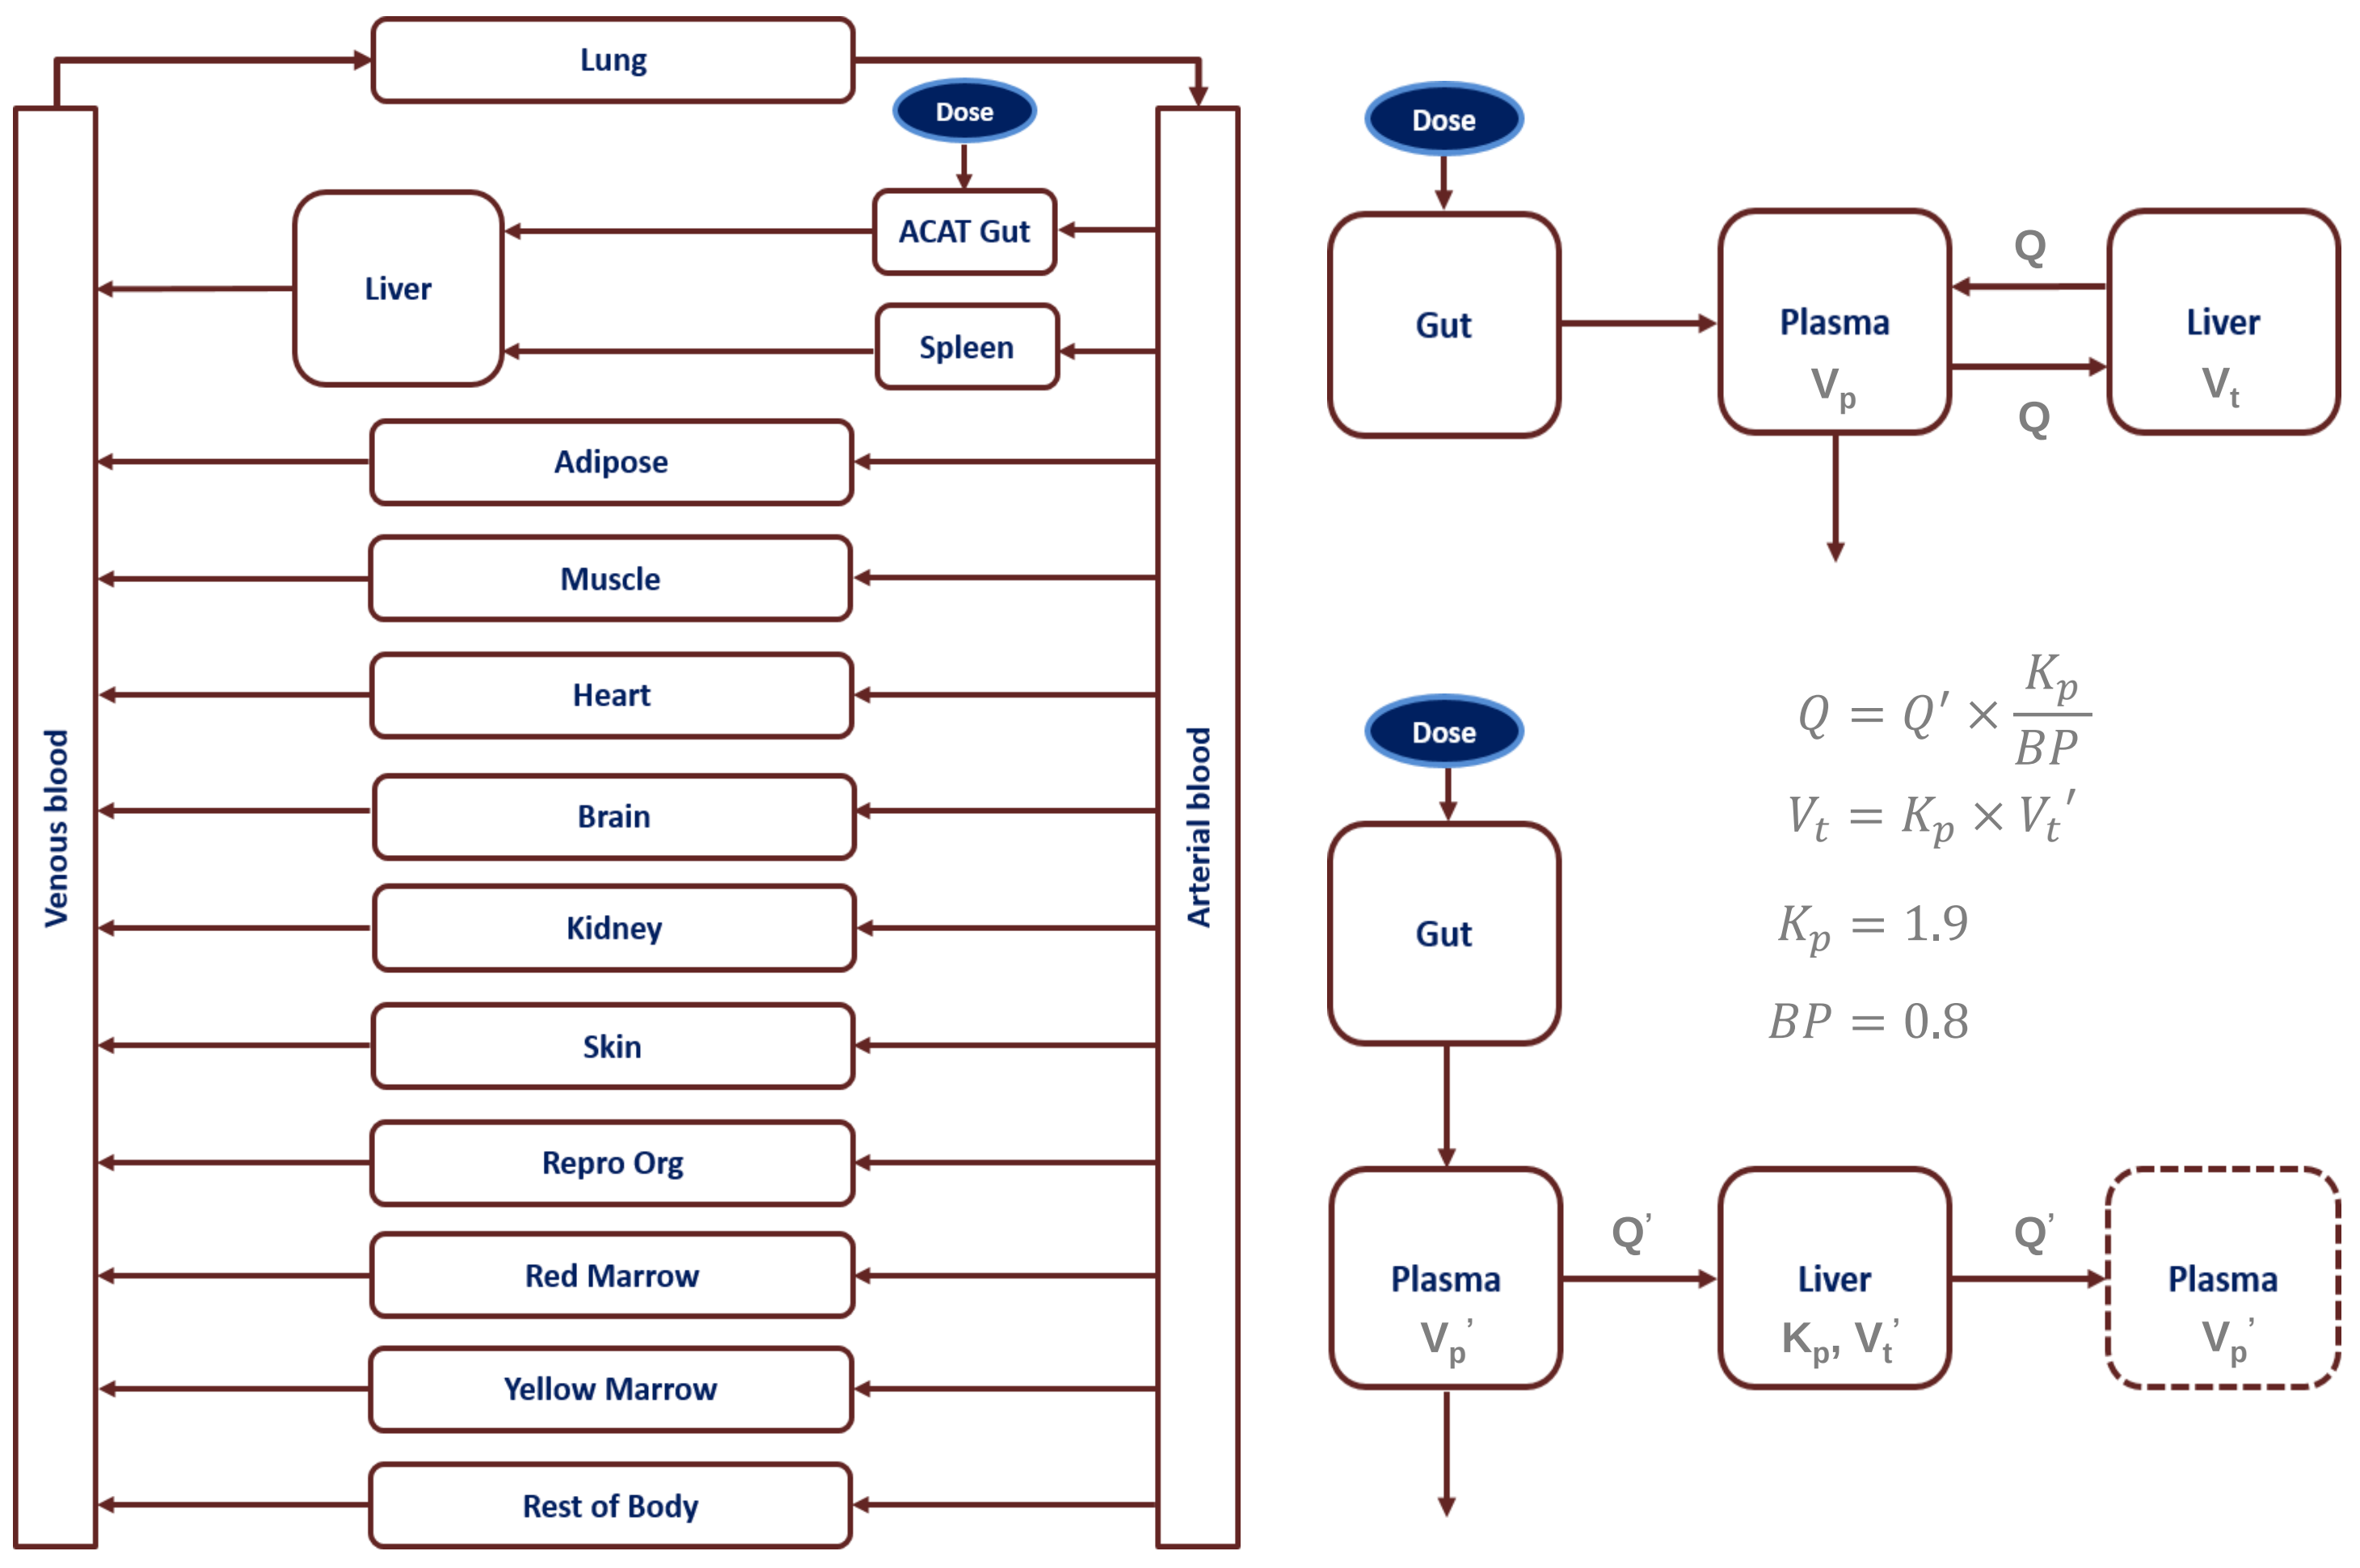
Lung
Dose
ACAT Gut
Liver
Spleen
Adipose
Muscle
Heart
Brain
Kidney
Skin
Repro Org
Red Marrow
Yellow Marrow
Rest of Body
Venous blood
Arterial blood
Gut
Plasma
Vp
Liver
Vt
Q
$Q = Q' \times \frac{K_p}{BP}$
$V_t = K_p \times V_t'$
$K_p = 1.9$
$BP = 0.8$
Q'
Vp'
Kp, Vt'

Figure 1: Structure of the full PBPK model used in the study with oral administration (Left)
Structure of the classical PK model used in the study with oral administration (Right Top)
Structure of the minimal PBPK model used in the study with oral administration (Right Bottom)

Results

- Simulated concentration-time profiles from the full PBPK model for plasma and liver are shown in **Figure 2,** which are identical to the outputs reported in GastroPlus®.

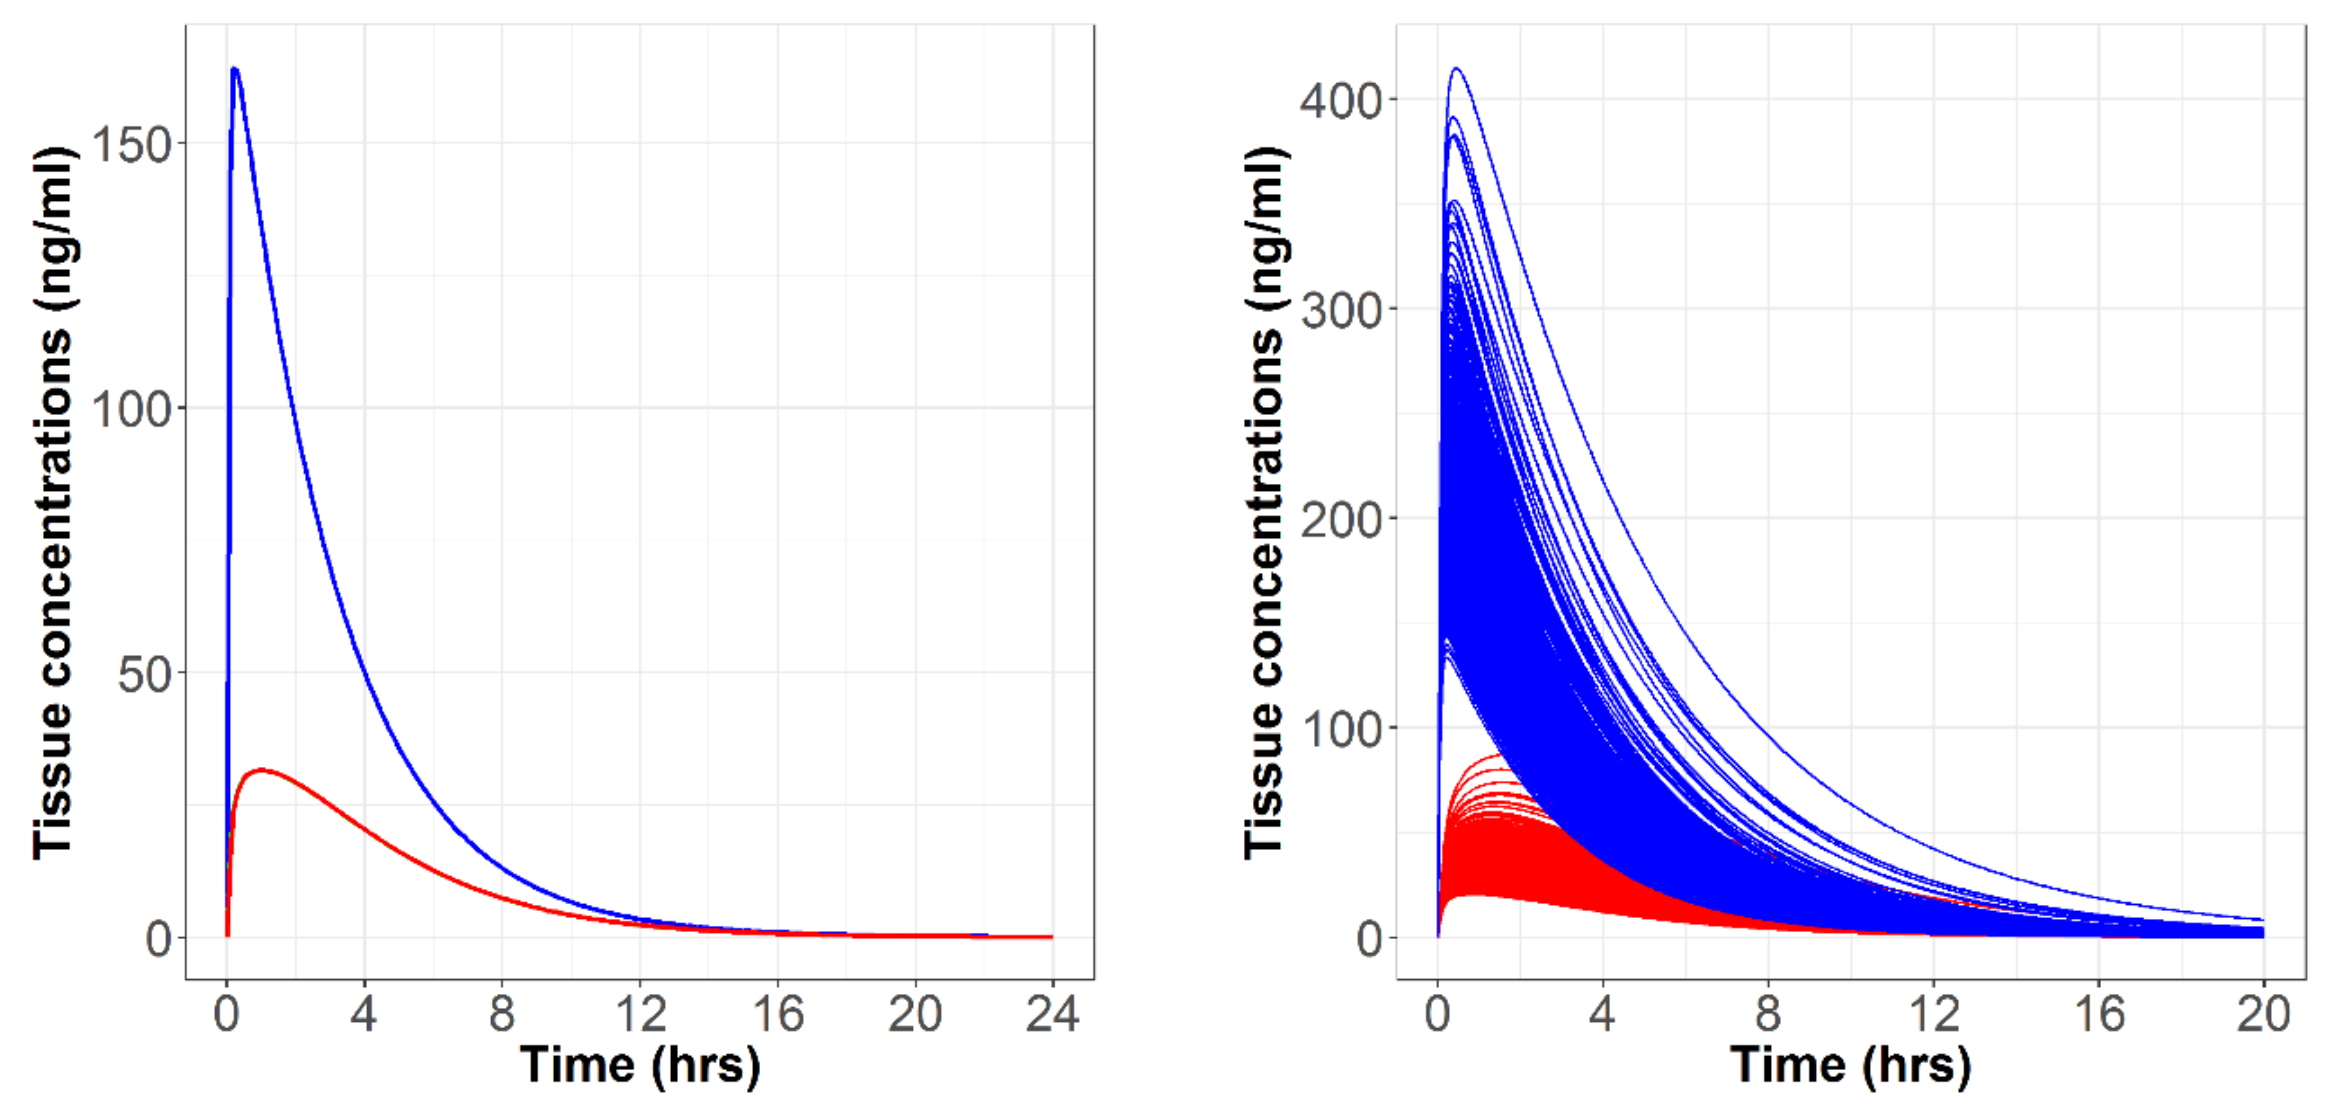
Tissue concentrations (ng/ml)
Time (hrs)

Figure 2: Simulated concentration-time profiles from the full PBPK (–liver –plasma) (Left) and individual concentration-time profiles for 1,000 subjects aged between 20 and 80 (–liver –plasma) (Right)

- Based on linear decline of liver parameters with increase in age (i.e. 5% every 10 years for liver blood flow and volume, and 10% every 10 years for liver clearance), normal distributions from 1,000 subjects were fitted as shown in **Figure 3**.

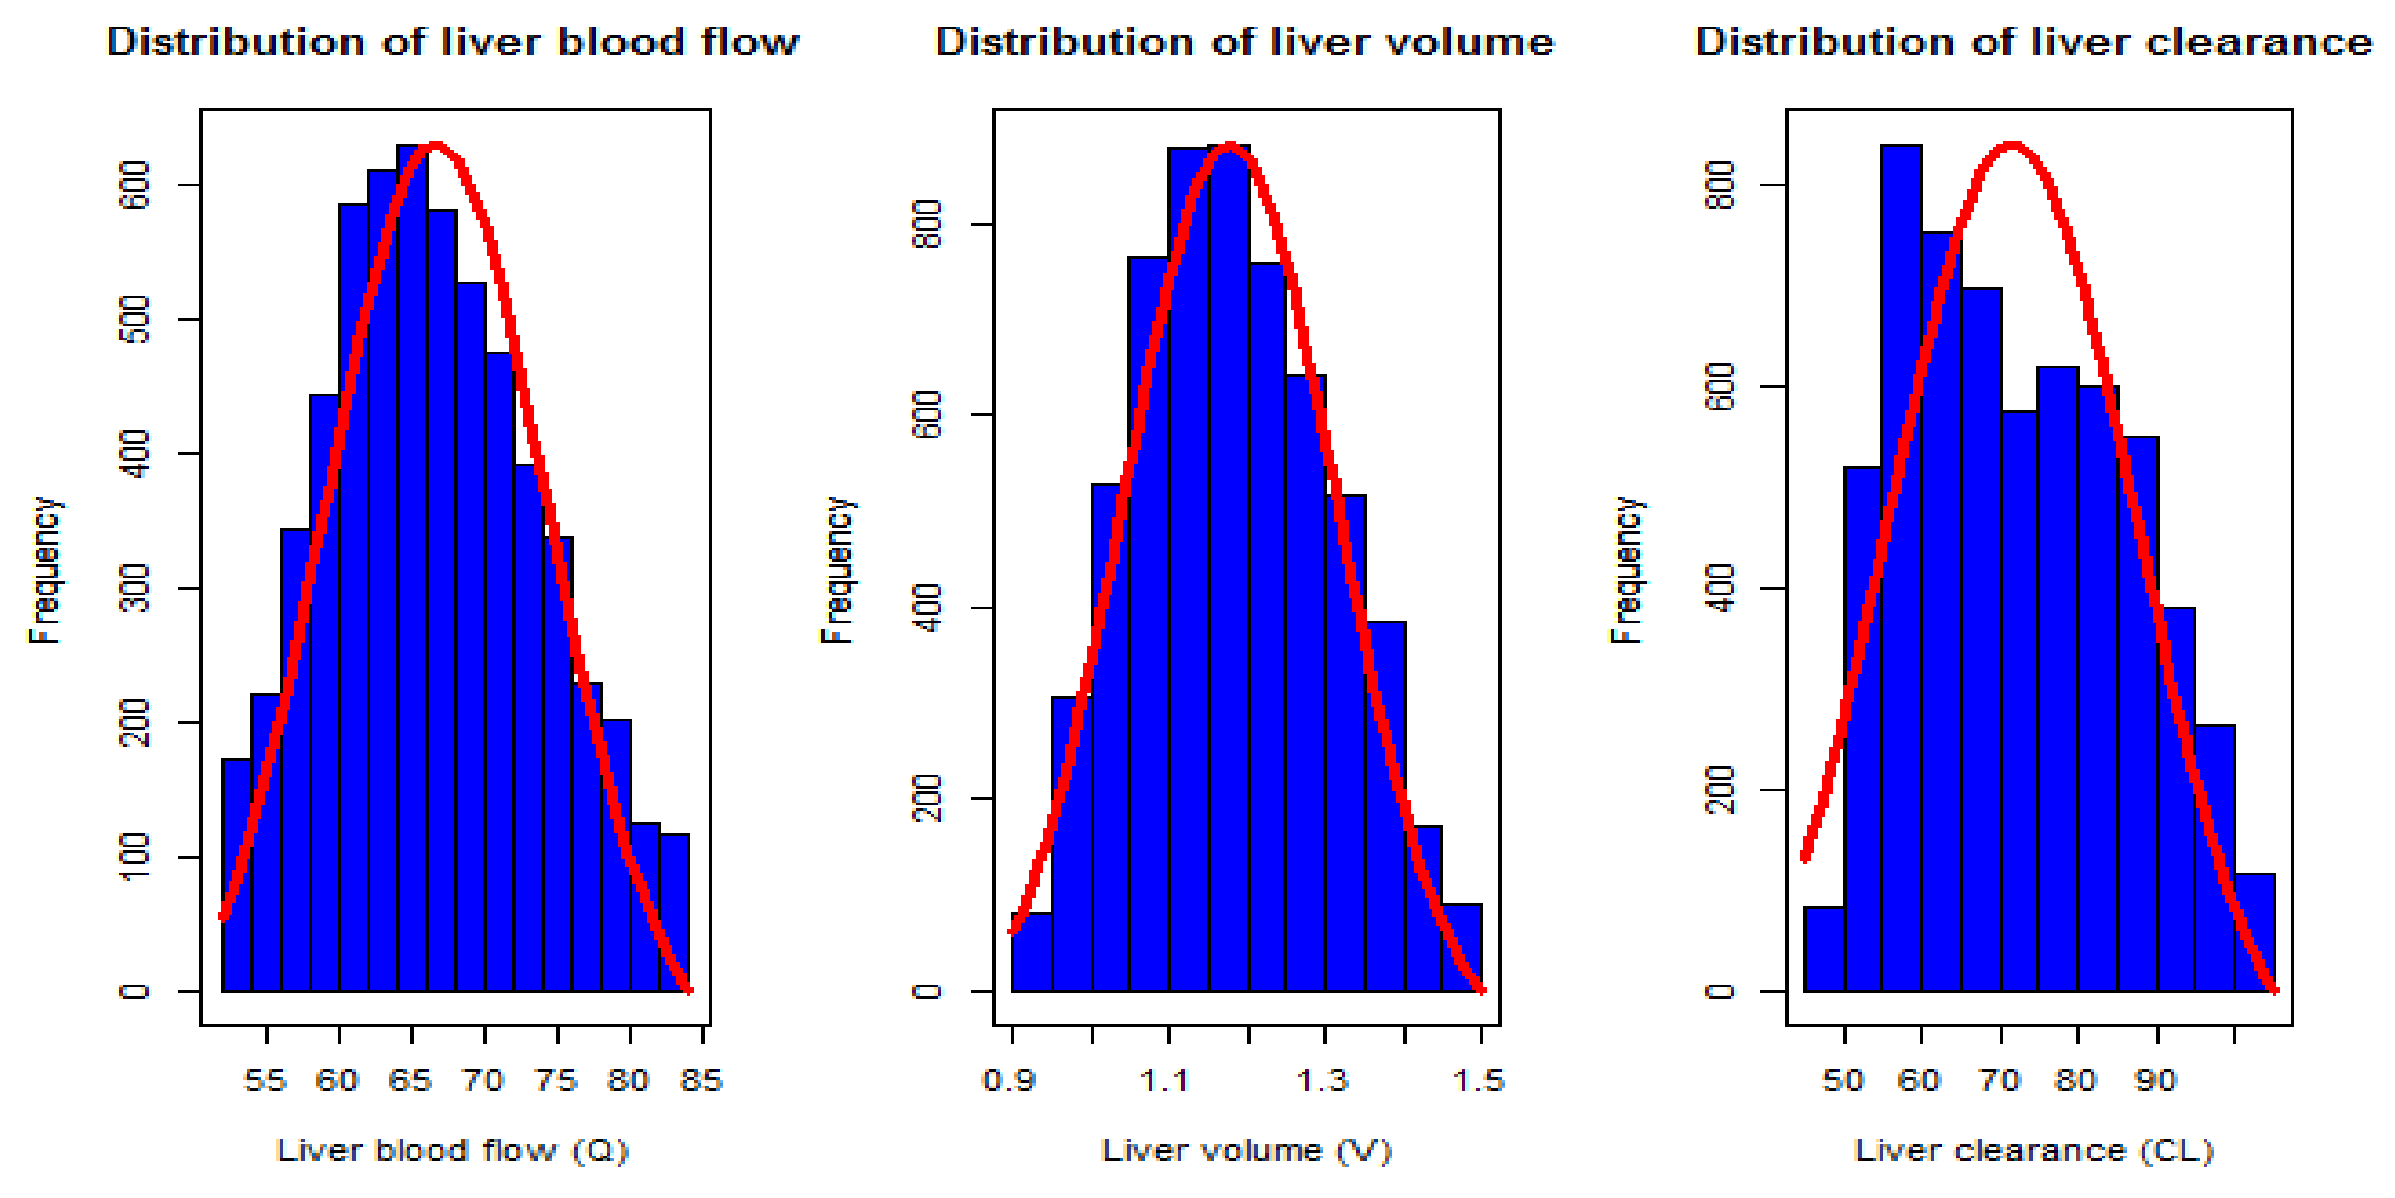
Distribution of liver blood flow
Distribution of liver volume
Distribution of liver clearance
Frequency
Liver blood flow (Q)
Liver volume (V)
Liver clearance (CL)

Figure 3: Distributions of liver blood flow, liver volume and clearance from 1,000 subjects aged between 20 and 80, considering the physiological change of liver with increase in age as suggested in the literature [2]

- When plasma concentrations were available only, sparse samples from 100 subjects and rich samples from 10 subjects gave similar parameter estimates in RStan, as shown in **Figure 4**.

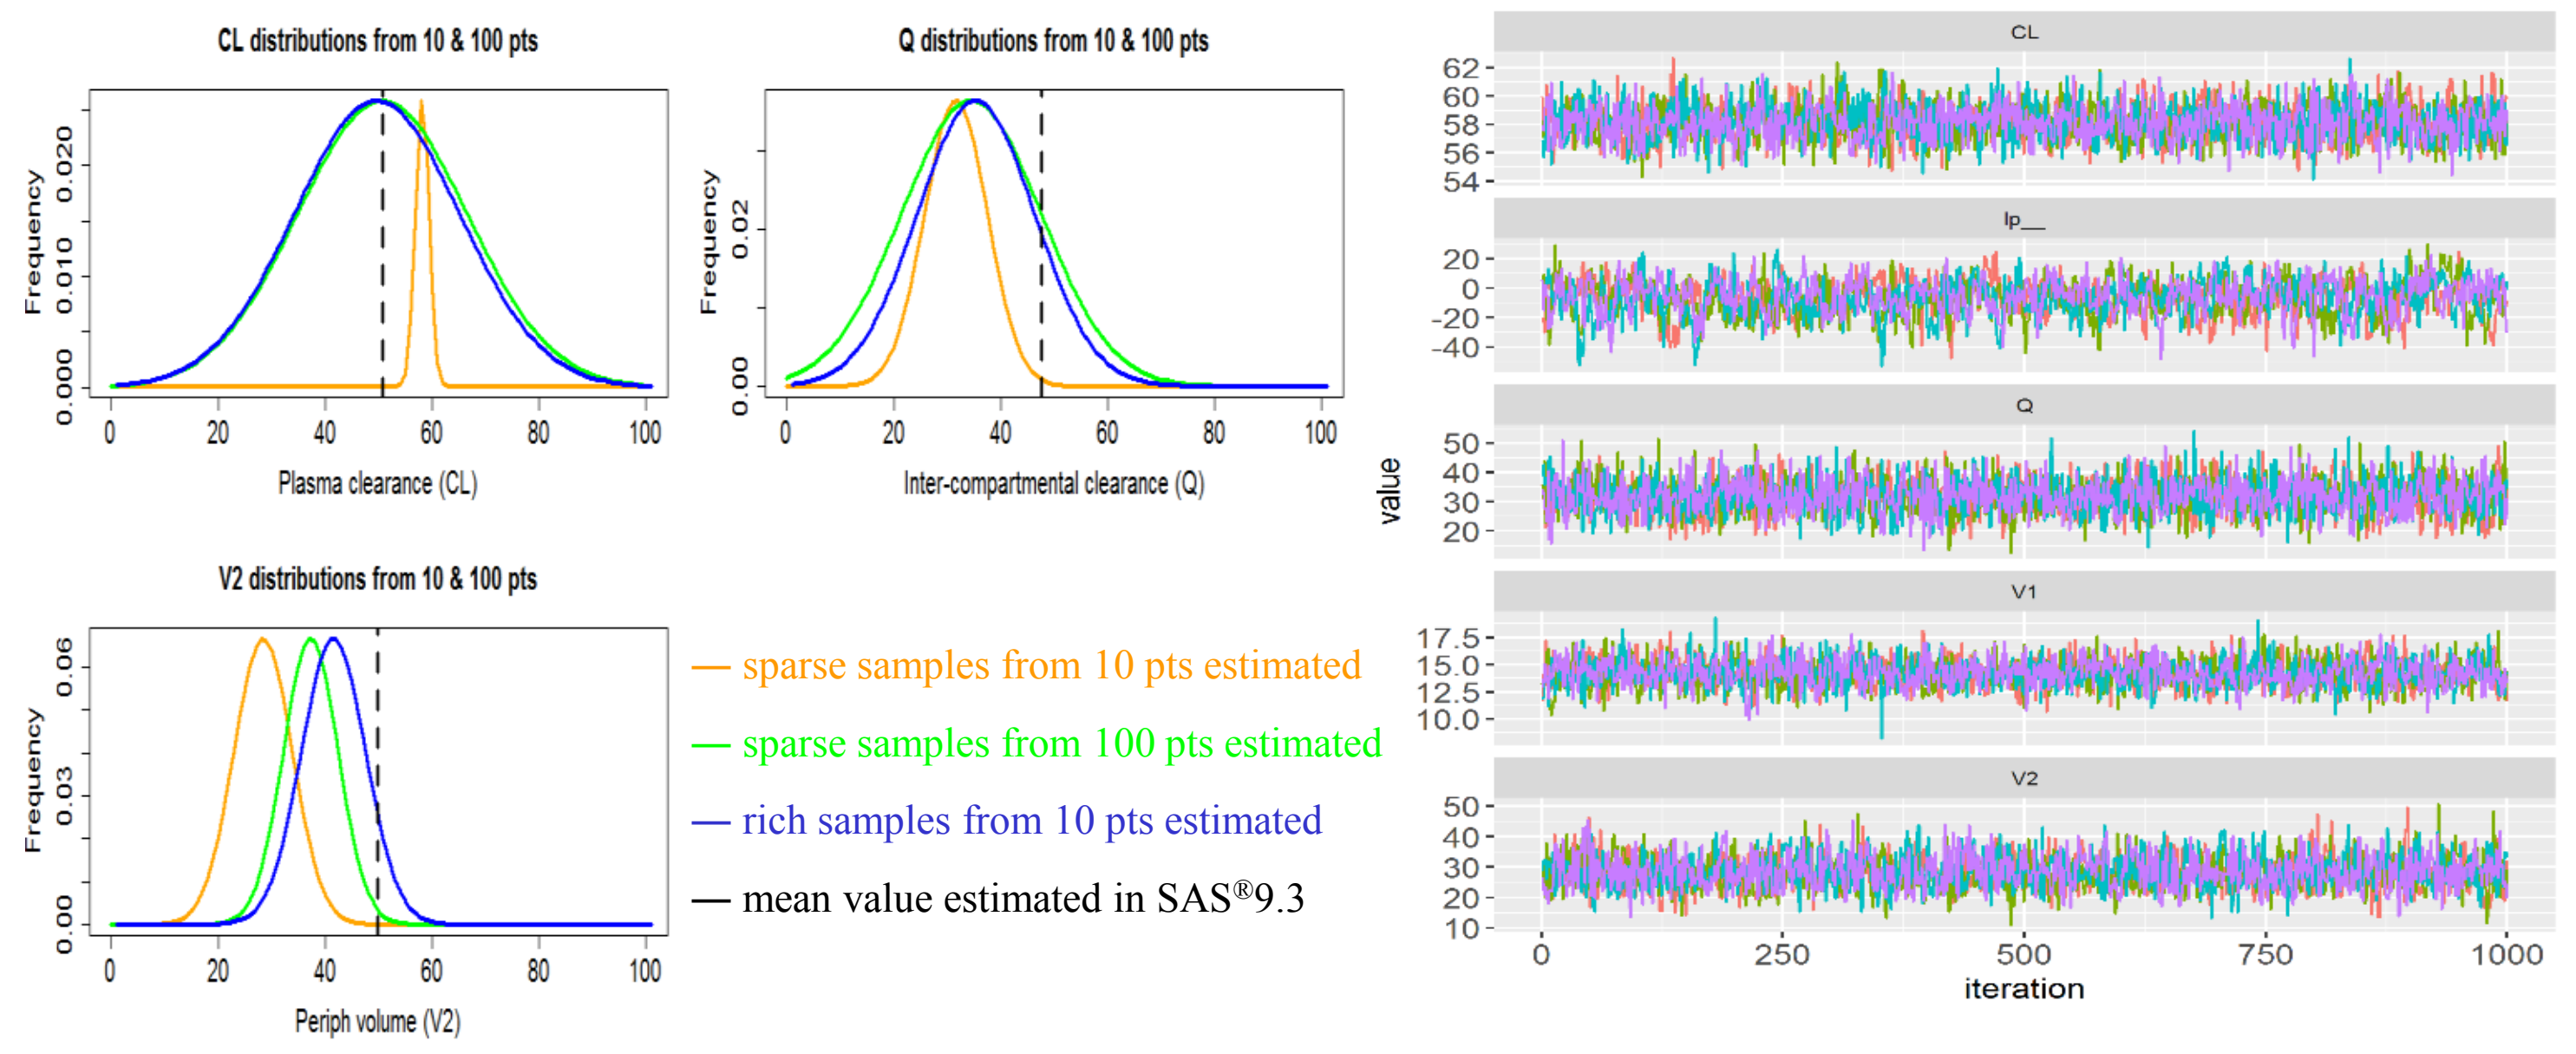
CL distributions from 10 & 100 pts
Q distributions from 10 & 100 pts
V2 distributions from 10 & 100 pts
Frequency
Plasma clearance (CL)
Inter-compartmental clearance (Q)
Periph volume (V2)
— sparse samples from 10 pts estimated
— sparse samples from 100 pts estimated
— rich samples from 10 pts estimated
— mean value estimated in SAS®9.3
CL
lp__
Q
V1
V2
value
iteration

Figure 4: Posterior distributions of parameters from classical PK model with plasma concentrations only (Left) and one example of trace plots for model parameters and log likelihood (Right)

Conclusions

- The current study explored scenarios where Bayesian interference may be useful in predicting drug concentrations in plasma or tissue.
- Estimation with the minimal PBPK in RStan using both plasma and liver concentrations is currently under investigation and correction.

References

[1] Jones and Rowland-Yeo. CPT Pharmacometrics Syst Pharmacol. 2013;2:e63
[2] Schmucker DL. Exp Gerontol. 2005;40(8–9):650-9
[3] Cao&Jusko. J Pharmacokinet Pharmacodyn. 2012;39(6):711-23