A Comparison of a Genetic Algorithm-Based Automated Search Algorithm to Standard Stepwise Approach for Population Pharmacokinetics using NONMEM

Robert R. Bies[1,2], Jeffry A. Florian Jr.[1], Bruce G Pollock[2], Kristin Bigos[3], Marci Chew[4], Yuyan Jin[1], Yan Feng[5],
Lon Schneider[6], Matt Muldoon[7], Steven Manuck[8], Mark E. Sale[9]

[1]Dept. of Pharmaceutical Sciences, Univ. of Pittsburgh, Pittsburgh, PA, USA [2]Centre for Addiction and Mental Health, Toronto, Ontario, Canada [3]Clinical Brain Disorders Branch, NIH, Bethesda, MD, USA
[4] Clinical Pharmacology, Pfizer Inc, New London, CT, USA [5]Strategic Modeling and Simulation Group, Bristol-Myers Squibb Co., Princeton, NJ, USA [6]Keck School of Medicine, USC, Los Angeles, CA, USA
[7]Department of Medicine, Univ. of Pittsburgh, Pittsburgh, PA, USA [8]Department of Psychology, Univ. of Pittsburgh, Pittsburgh, PA, USA, [9]Next Level Solutions, LLC, Raleigh, NC, USA

Objective

To compare, using the AIC, the selection of model structures, covariates, covariate relationships, and error structures in models chosen for five separate drugs using both stepwise or genetic algorithm (GA) approaches.

Introduction

- Model search strategies in pharmacokinetic/pharmacodynamic (PK/PD) modeling involve numerous decisions from the modeler
 - ⋆ selection of the appropriate model structure (compartments and parameterizations)
 - ⋆ covariates and covariate relationships
 - ⋆ error structure selection
- Discrete search space which can not be fully evaluated using conventional approaches
- Initial model structure and order of covariate inclusion can influence downstream results (Wade *et al.* 1994)
 - ⋆ error structures (additive, proportional)
 - ⋆ covariate relationships (proportional, power-law, Michaelis-Menten, Hill)
 - ⋆ addition of covariates and error relationships are not necessarily linear in reducing error
- Exploration of entire search space is impractical due to time constraints
- GA approach may help address these issues (Bies *et al.* 2006)
 - ⋆ capable of running without direct supervision
 - ⋆ criteria for assessing model accuracy essential

Methods

- The following drugs were evaluated: citalopram, perphenazine, olanzapine, quetiapine, and risperidone

Table 1: Data and stepwise model summary for the five compounds

Compound	# patients	# data points	Model structure	Covariates
citalopram	331	1324	2-comp., intravenous combined error	10
perphenazine	156	421	1-comp., oral proportional error	8
olanzapine	523	1527	1-comp., oral additive error	7
quetiapine	405	945	1-comp., oral additive error	7
risperidone	490	1236	1- or 2-comp. mixture model, intravenous combined error	7 3 CL groups

- Search space was restricted to those originally considered during the initial model evaluation
 - ⋆ all model structures (compartments and/or mixtures), covariates, covariate relationships, and output error metrics
 - ⋆ 6 relationships for continuous covariates and 4 for discrete covariates
 - ⋆ 3 interindividual variability structures (exponentiated, proportional, additive)
 - ⋆ 3 residual unknown variability structures (additive, proportional, combined)
 - ⋆ risperidone analysis included a mixture model option
- Example code with token sets shown below

```
Number of models =
1
########### Begining of model # 1 ############
$PROB GA COVARIATE SEARCH 1CPTMOD OLANZAPINE COMBINED

$INPUT ID DATE=DROP TIME AMT RATE DV MDV CMT SS II SEX SMOK AA AGE WT

$DATA FINALDATASET8.CSV

$SUB ADVAN=ADVAN2 SS=SS2 TRANS2

$PK

    TVCL5 = THETA(1)
    TVCL4 = TVCL5 COVSEXCL(1)
    TVCL3 = TVCL4 COVSMOKCL(1)
    TVCL2 = TVCL3 COVAACL(1)
    TVCL1   = TVCL2 COVAGECL(1)
    TVCL = TVCL1 COVWTCL(1)
    CL   = TVCL CLERR(1)
    TVVE = THETA(2)
    TVVD  = TVVE COVSEXV(1)
    TVVC = TVVD COVSMOKV(1)
    TVVB = TVVC  COVAAV(1)
    TVVA = TVVB COVAGEV(1)
    TVV = TVVA COVWTV(1)
    V    = TVV VERR(1)
    TVKA = THETA(3)
    KA   = TVKA
    S2   = V

  IF(MDV.EQ.0.AND.TIME.GE.0.AND.TIME.LE.24) THEN
    PPCI = 1
  ELSE
   PPCI = 0
  END IF
  RP = IREP
$ERROR
   Y=F RESERR(1)
   IPRED=F
   OB = DV ; FOR NPDE PROBLEM 1
   Y= F RESERR(1)
   FSIM = Y ; FOR NPDE PROBLEM 2

$THETA
   (1,18,50)   ;CL COEFF
   (100,3000,8000)   ;V COEFF
   (0.05,0.5,5)     ;KA
   COVSEXCL(2)
   COVSMOKCL(2)
   COVAACL(2)
   COVAGECL(2)
   COVWTCL(2)
   COVSEXV(2)
   COVSMOKV(2)
   COVAAV(2)
   COVAGEV(2)
   COVWTV(2)
```

```
              COVAACL
N token sets =
              4
N tokens =
              2
Check syntax =
              False
IS IOV OMEGA =
              False
N IOV OMEGA =
              0
Token set #
              1
Token set #
              2
+AA*{THETA(C)}
{$THETA(C)=}(-4,0.01,4)
Token set #
              3
*(1+AA*{THETA(C)})
{$THETA(C)=}(-4,0.01,4)
Token set #
              4
*EXP(AA*{THETA(C)})
{$THETA(C)=}(-4,0.01,4)
Group stem =
              COVAGECL
N token sets =
              6
N tokens =
              2
Check syntax =
              False
IS IOV OMEGA =
              False
N IOV OMEGA =
              0
Token set #
              1
Token set #
              2
+AGE*{THETA(D)}
{$THETA(D)=}(-4,0.01,4)
Token set #
              3
*(1+AGE*{THETA(D)})
{$THETA(D)=}(-4,0.01,4)
Token set #
              4
*EXP(AGE*{THETA(D)})
{$THETA(D)=}(-4,0.01,4)
Token set #
              5
*{THETA(D)}*AGE/({THETA(E)}+AGE)
{$THETA(D)=}(-4,0.01,4){crlf}{$THETA(E)=}(-4,0.01,4)
Token set #
              6
*AGE**{THETA(D)}
{$THETA(D)=}(-4,0.01,4)
```

Genetic Algorithm

- GA fitness function penalties are shown in Figures 1 and 2
- Fitness of models was evaluated based on penalty function
 - ⋆ objective function plus penalties from the options
 - ⋆ combination of posterior predictive check (PPC) and normalized predictive distribution errors (NPDE)
 - ⋆ AIC, eigenvalues, objective function value, and other criteria
- Downhill search and niche evaluation every 5 generations
- Selection of the next population generation was based on a random selection from the previous generation, weighted according to fitness function values
- Niche membership selects all model pairs differing by $< N$ bits
 - ⋆ all bit combinations between the top models within a niche are then evaluated
- Model with the highest fitness carried to the next generation
- First-order estimation with interaction for all models
- NONMEM VI with Intel Fortran (VERSION 9.1)
- Performed on 10 Dual Core Computers
- GA search summary and search convergence are shown in Figures 4 and 5
- Stepwise versus GA performance was assessed using model predicted AIC

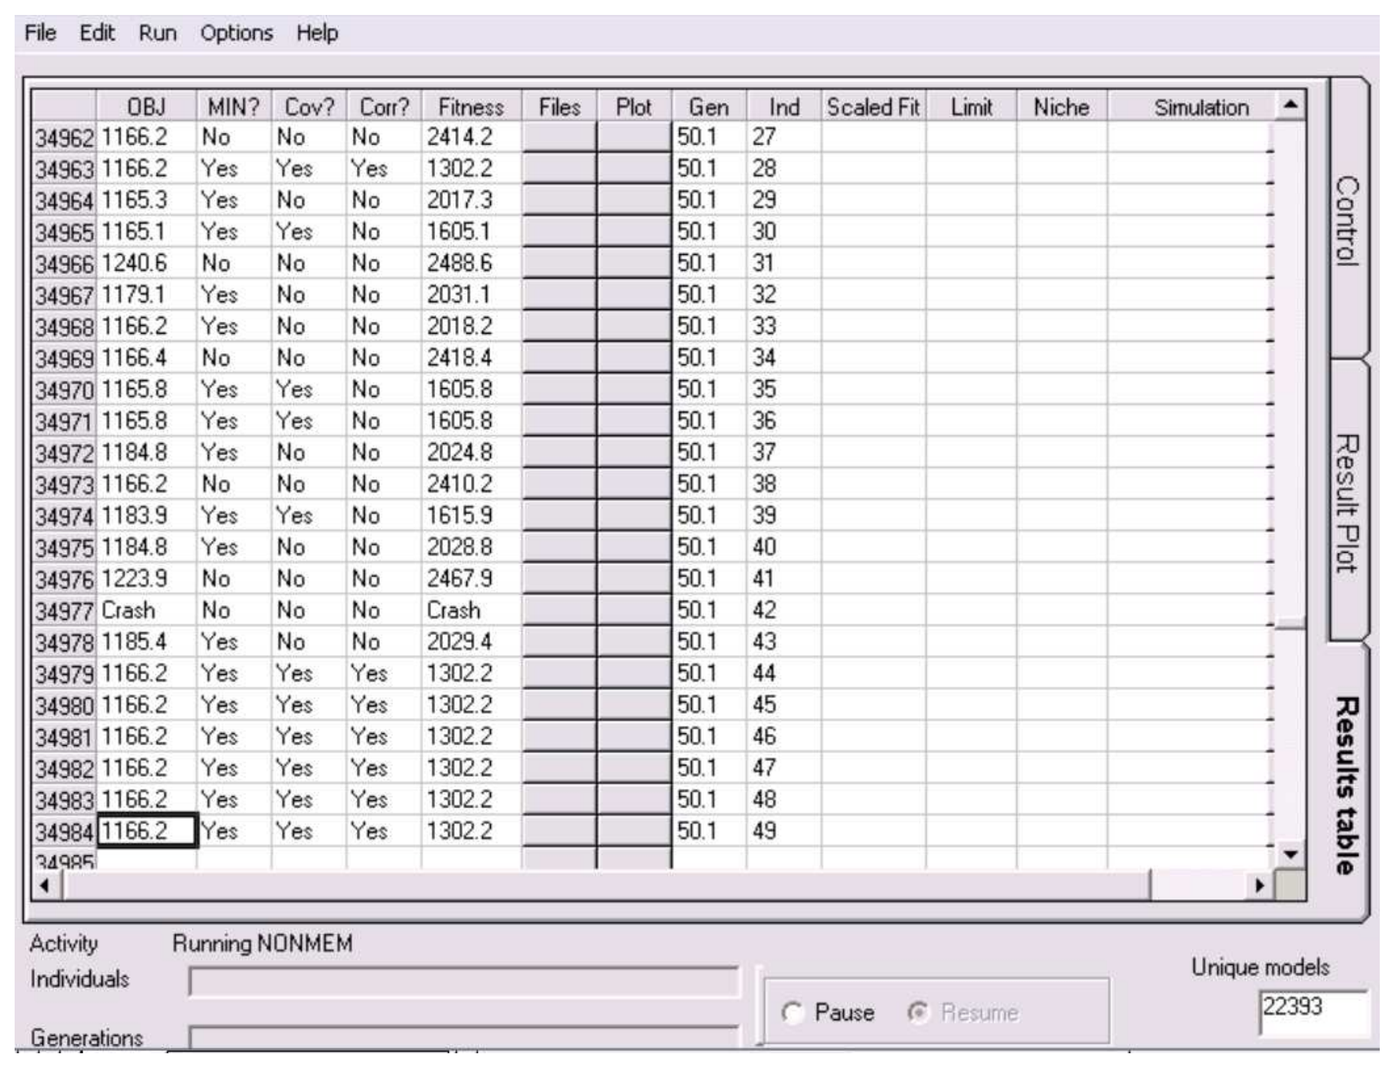
File Edit Run Options Help
OBJ MIN? Cov? Corr? Fitness Files Plot Gen Ind Scaled Fit Limit Niche Simulation
Control
Result Plot
Results table
Activity Running NONMEM
Individuals
Generations
Pause Resume
Unique models
22393

Figure 4: GA search summary

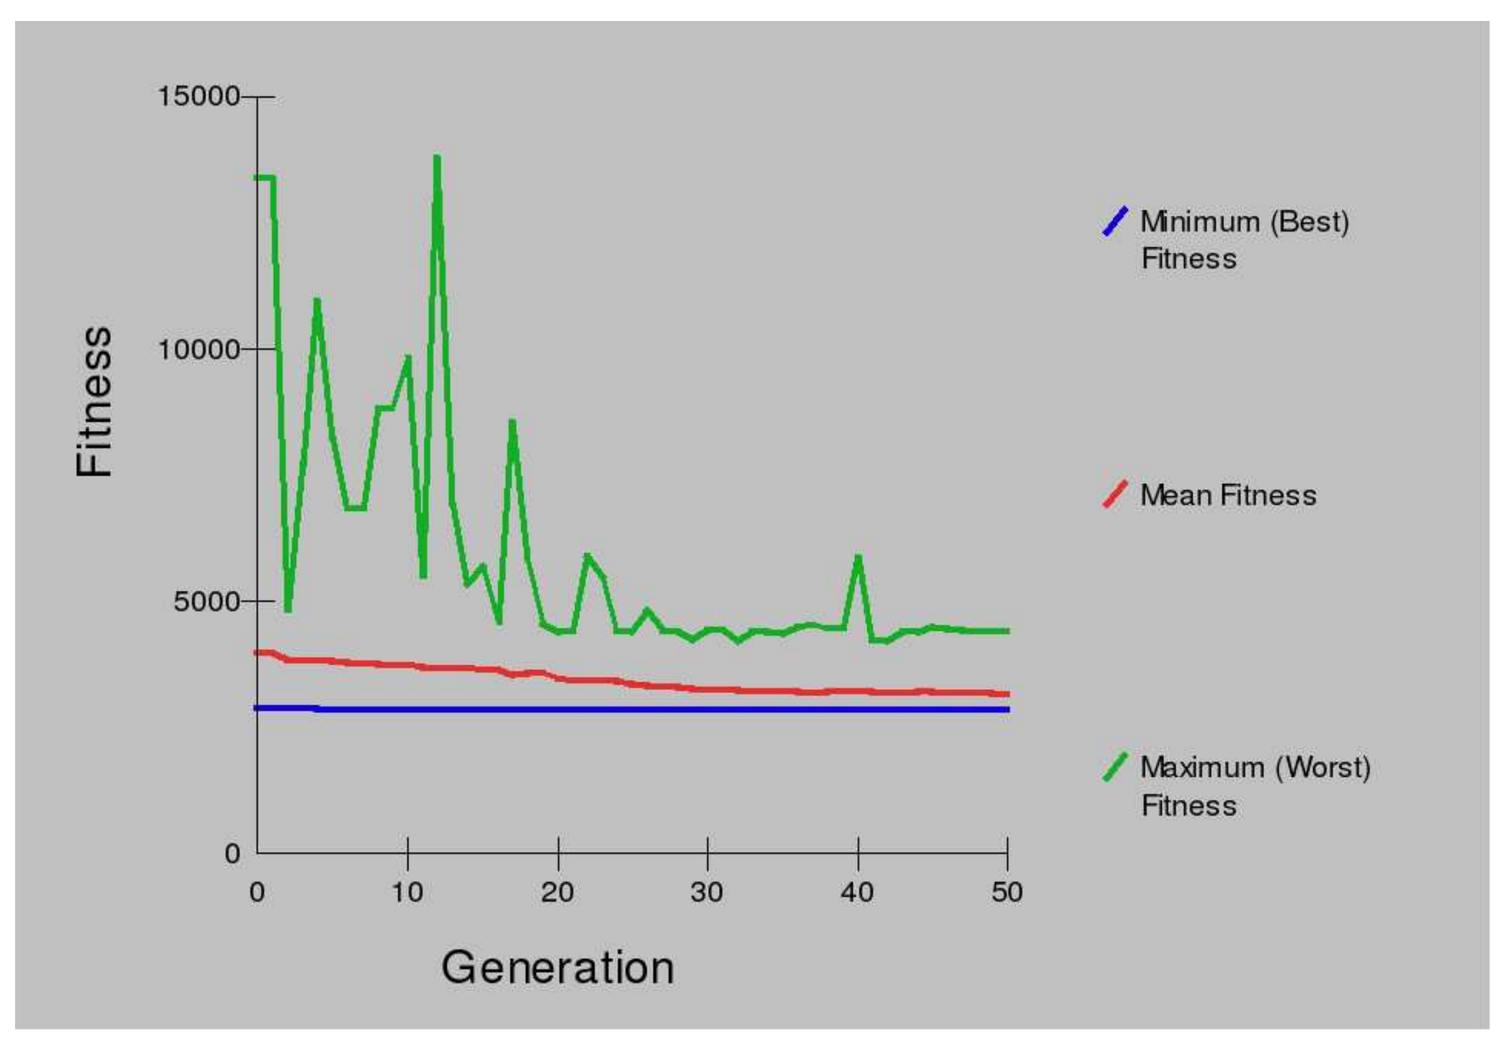
Fitness
15000
10000
5000
0
0 10 20 30 40 50
Generation
Minimum (Best) Fitness
Mean Fitness
Maximum (Worst) Fitness

Figure 5: GA search convergence profile

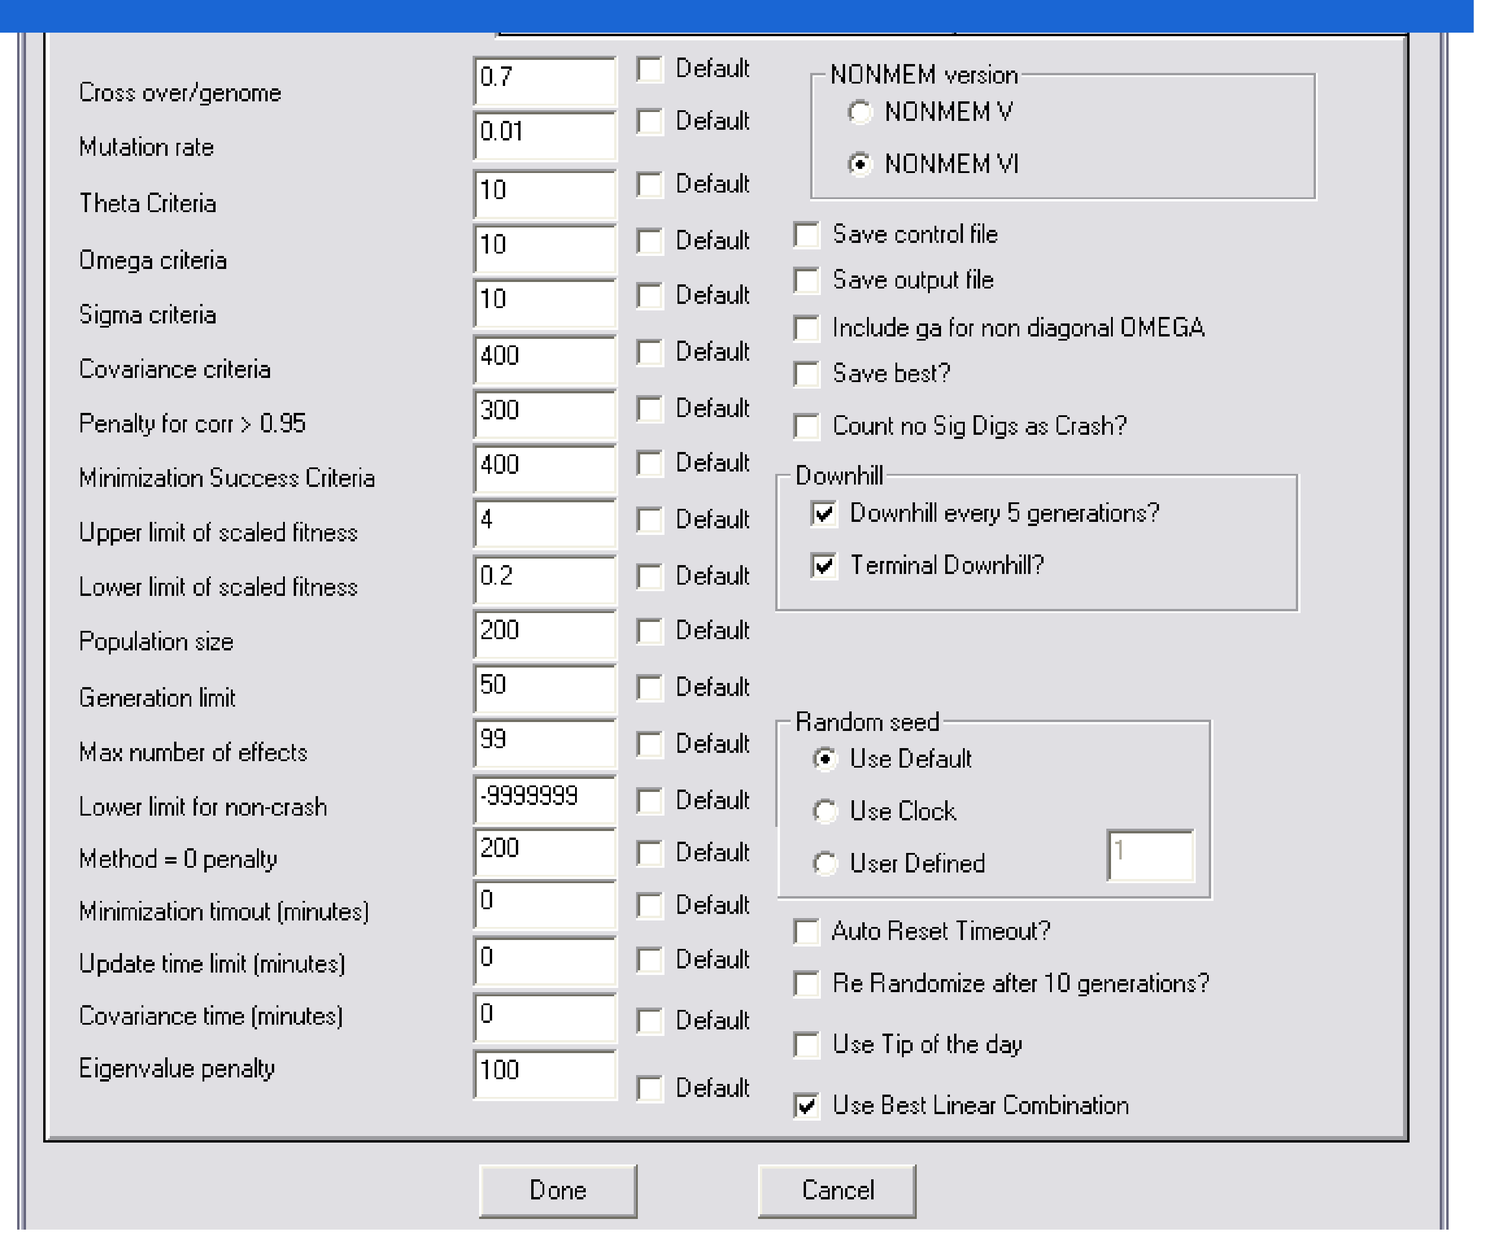
Cross over/genome 0.7
Mutation rate 0.01
Theta Criteria 10
Omega criteria 10
Sigma criteria 10
Covariance criteria 400
Penalty for corr > 0.95 300
Minimization Success Criteria 400
Upper limit of scaled fitness 4
Lower limit of scaled fitness 0.2
Population size 200
Generation limit 50
Max number of effects 99
Lower limit for non-crash -9999999
Method = 0 penalty 200
Minimization timout (minutes) 0
Update time limit (minutes) 0
Covariance time (minutes) 0
Eigenvalue penalty 100
Default
NONMEM version
NONMEM V
NONMEM VI
Save control file
Save output file
Include ga for non diagonal OMEGA
Save best?
Count no Sig Digs as Crash?
Downhill
Downhill every 5 generations?
Terminal Downhill?
Random seed
Use Default
Use Clock
User Defined
Auto Reset Timeout?
Re Randomize after 10 generations?
Use Tip of the day
Use Best Linear Combination
Done
Cancel

Figure 1: GA fitness function options

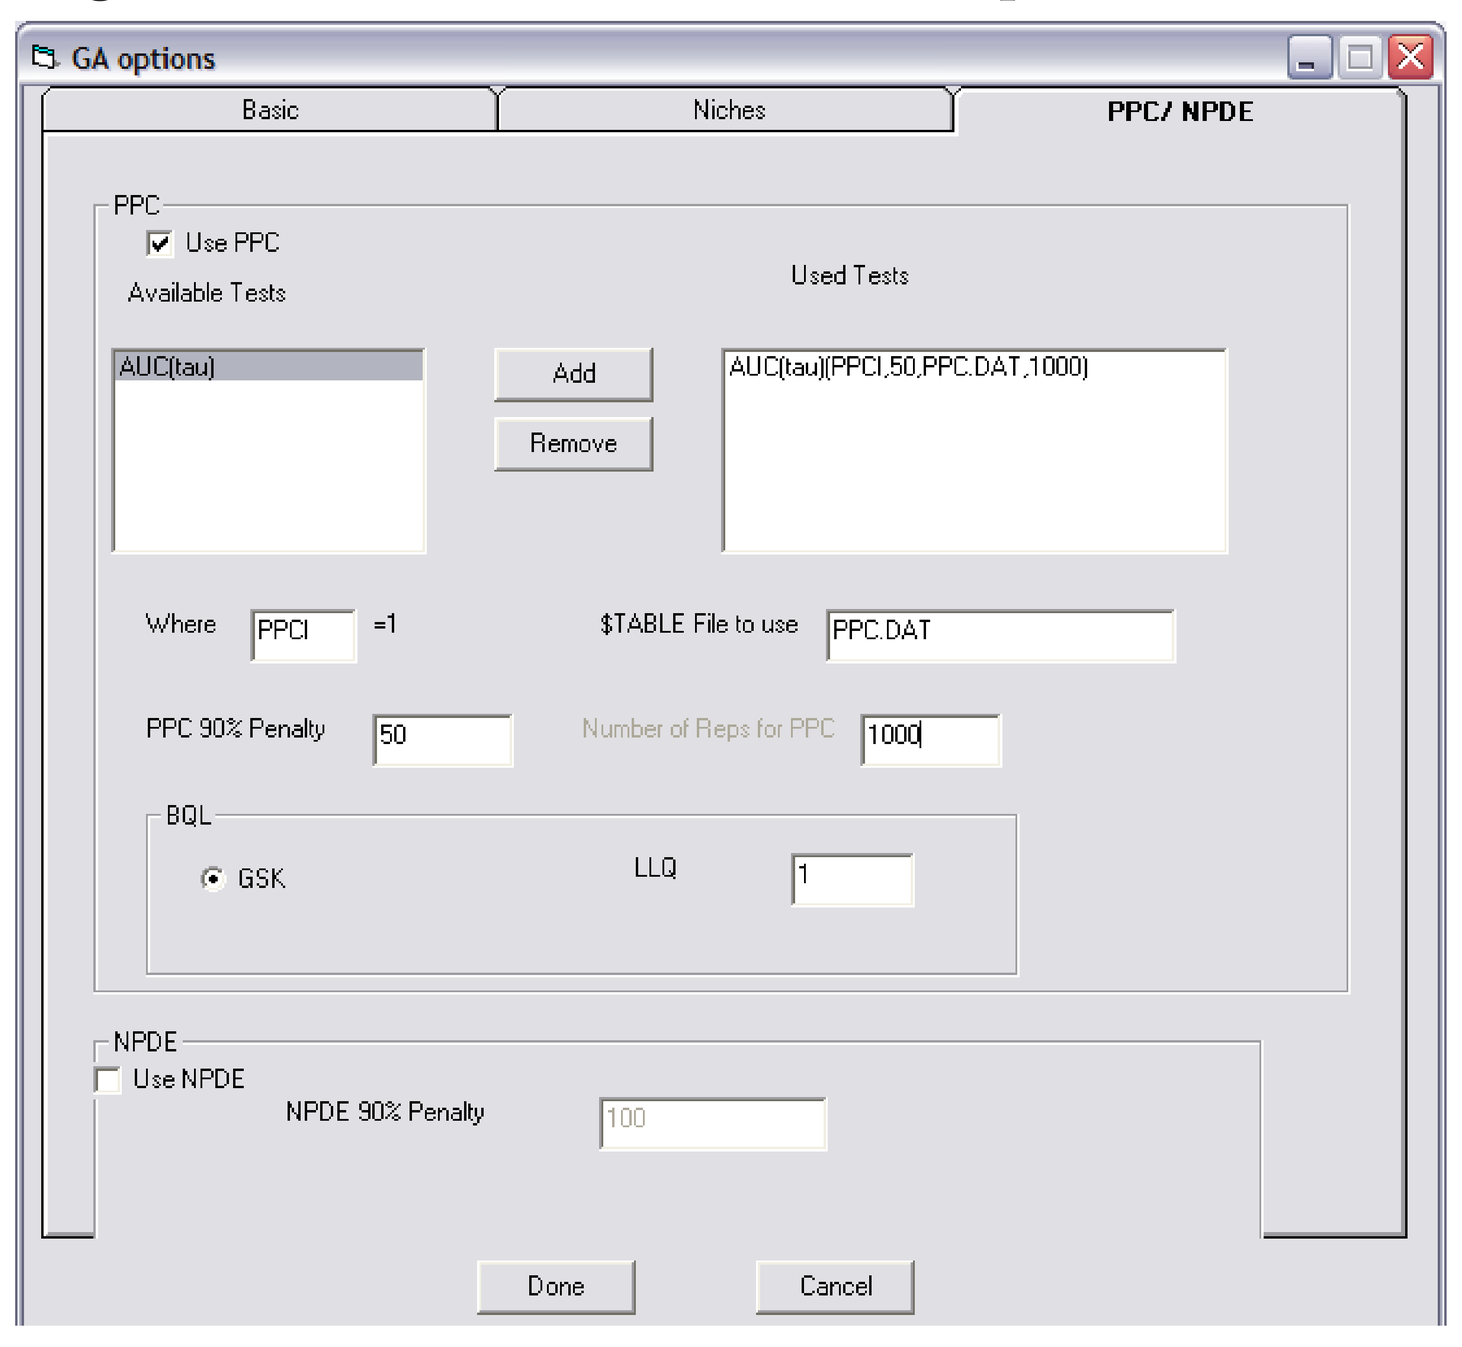
GA options
Basic
Niches
PPC/ NPDE
PPC
Use PPC
Available Tests
Used Tests
AUC(tau)
Add
Remove
AUC(tau)(PPCI,50,PPC.DAT,1000)
Where PPCI =1
$TABLE File to use PPC.DAT
PPC 90% Penalty 50
Number of Reps for PPC 1000
BQL
GSK
LLQ 1
NPDE
Use NPDE
NPDE 90% Penalty 100
Done
Cancel

Figure 2: GA PPC and NPDE options

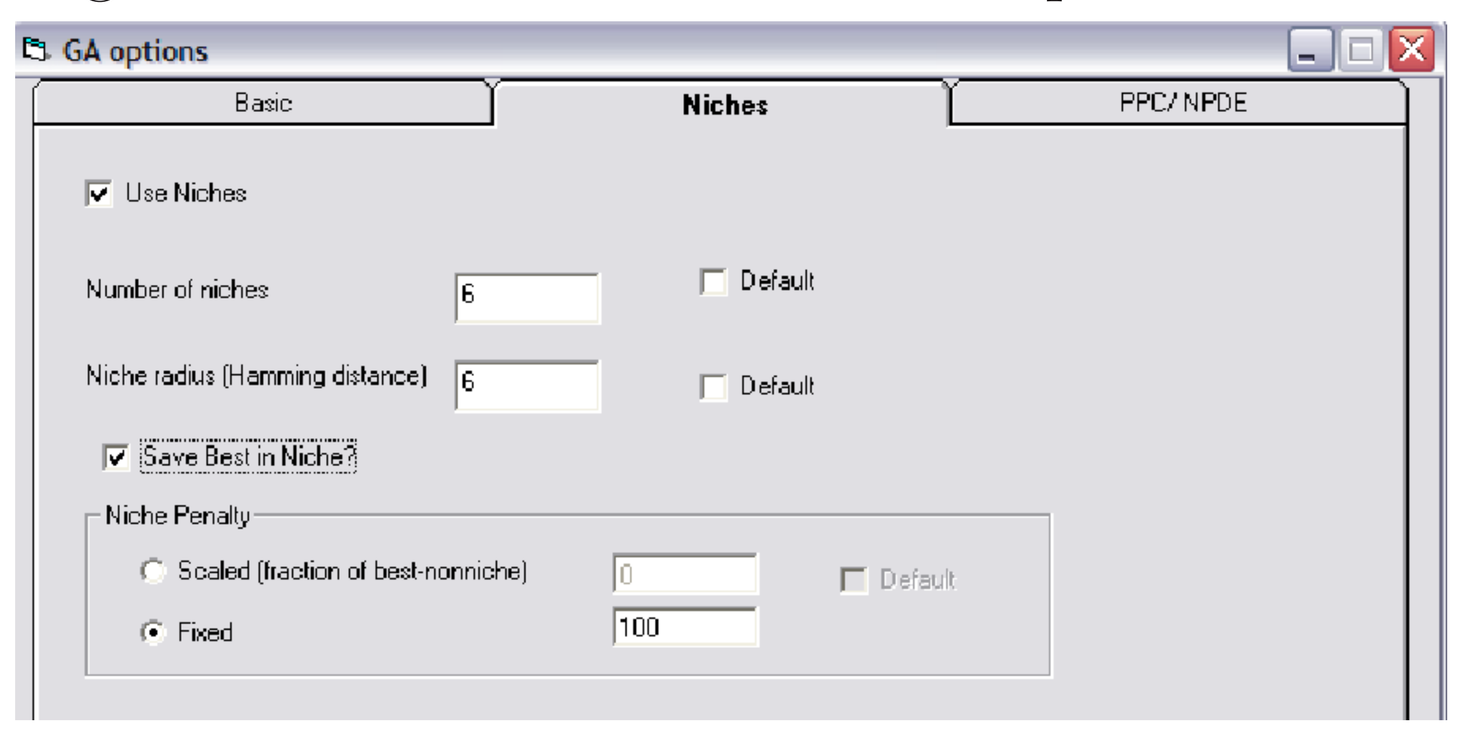
GA options
Basic
Niches
PPC/ NPDE
Use Niches
Number of niches 6
Default
Niche radius (Hamming distance) 6
Default
Save Best in Niche?
Niche Penalty
Scaled (fraction of best-nonniche) 0
Default
Fixed 100

Figure 3: GA niche options

Results

GA versus Standard Approach

Table 2: Summary of model results using both stepwise or GA approaches

Compound	Search	AIC	OBJ	θ	η	ϵ	Covariates	Δ AIC	Comments
citalopram	Step	5713.0	5699.0	8	3	2	CL (weight), Q (fat mass), V_2 (weight, sex)	—	no covariance step
	GA	5345.9	5304.9	17	3	1	CL (sex, weight, body mass index, fat mass) V_1 (sex, body mass index, free fat mass) Q (sex, free fat mass), V_2 (sex, weight, free fat mass)	367.1	covariance step converged
perphenazine	Step	561.9	537.9	12	3	1	CL (race, smoking status, other meds)	—	k_a not estimated
	GA	556.9	531.9	10	1	1	CL (race, # cigarettes, smoking status, other meds) V (age, sex, smoking status)	5.01	covariance step with k_a
olanzapine	Step	10365.8	10347.8	6	2	1	CL (sex, smoking status, race)	—	
	GA	9850.8	9832.8	6	1	2	CL (sex, smoking status, race)	515.0	different error structures
quetiapine	For.	11126	11110.4	4	3	3	V (weight)	—	k_a not well estimated
	GA	10114	10095.7	3	2	2	no significant covariates	1014.7	estimated k_a, no covariates identified
risperidone	Step	5119.1	5103.1	7	5	4	no significant covariates	—	3 CL group mixture model
	GA	4694.9	4662.9	11	3	2	CL (age, sex, race, other meds), V (sex)	424.2	2 CL group mixture model

Risperidone

- GA search included the possibility of one, two, or three groups for parent compound CL
 - ⋆ only two CL groups were identified
 - ⋆ improvement over the previously developed model
- GA identified significant covariates on CL and V
 - ⋆ age, sex, race, and other medications
- Sample code shown below

```
$MIX
P(1)=THETA(3)/(1+THETA(3))
P(2)=1-P(1)
NSPOP=2
$PK
  CALLFL =1
  AAGE = (AGE-46.6)/16.8
  AWT = (WGTB-188)/50
  ASEX = SEX-1
  EST=MIXEST
IF(MIXNUM.EQ.1)THEN
BCL=THETA(4)
ELSE
BCL=THETA(5)
ENDIF
  TVCL1 = BCL *EXP(AAGE*THETA(6))
  TVCL2 = TVCL1 *EXP(ASEX*THETA(7)) *EXP(RACE*THETA(8))
  TVCL3 = TVCL2 *EXP(PARX*THETA(9)) *EXP(FLUX*THETA(10))
  CL = TVCL3 *EXP(ETA(1))
```

```
  TVV1 = THETA(1)
  TVV2 = TVV1 *EXP(ASEX*THETA(11))
  TVV3 = TVV2
  V =  TVV3 *EXP(ETA(2))
  KA=THETA(2)
  ;ADVAN2
  S2=V

  IF(MDV.EQ.0.AND.TIME.GE.0.AND.TIME.LE.24) THEN
    PPCI = 1
  ELSE
   PPCI = 0
  END IF
  RP = IREP
$ERROR
   Y=F *EXP(EPS(1))+EPS(2)
   IPRED=F
   OB = DV ; FOR NPDE PROBLEM 1
   FSIM = Y ; FOR NPDE PROBLEM 2
```

Table 3: Summary of GA population size, generations evaluated, and total number of identified models

Compound	# individuals	# generations	unique models	% total
citalopram	300	49	21431	0.05
perphenazine	300	50	13903	0.35
olanzapine	200	50	8033	0.81
quetiapine	200	50	7231	0.73
risperidone	400	30	7948	0.09

Olanzapine: 3 residual unknown variability, 3 parameters with 3 interindividual variability structures, 5 discrete covariates (4 structures), and 2 continuous covariates (6 structures).
Total # $= 3 * 3 * 3 * 4^5 * 6^2$

Olanzapine

- Same number of parameters and significant covariates
- Differences between the models included
 - ⋆ interindividual variability (k_a and CL vs. CL)
 - ⋆ output error (additive vs. proportional and additive)
- Estimate of k_a possible with GA

Summary

- GA search resulted in a lower AIC than stepwise for all five drug models
 - ⋆ geometric and arithmetic means of 210 and 465
- Different models predicted by the GA for all cases
 - ⋆ alternate output error structure (4/5)
 - ⋆ different set of parameters with interindividual variability (4/5)
 - ⋆ significant covariates/covariate relationships (4/5)
 - ⋆ fewer CL categories were necessary in the risperidone mixture model

Acknowledgements

- National Institute of Mental Health Research Grants R01 MH64173, P30 MH30915, R01 MH65376, K24 MH65416, MH52247, N01 MH90001, and MH76420
- CATIE investigators (P.I. Jeffrey Lieberman, MD)