Population pharmacokinetic modeling and simulation of imeglimin in type 2 diabetes patients to support dose recommendations to patients with renal impairment

Enrica Mezzalana (2), Yoshiko Tomita (1), Emma Hansson (2), Florent Mazuir (3), Gustaf Wellhagen (2), Qing Xi Ooi (2), Atsushi Kitamura (1), Daisuke Nemoto (1), Sebastien Bolze (3)

(1) Sumitomo Dainippon Pharma Co., Ltd., Osaka, Japan, (2) Pharmetheus AB, Uppsala, Sweden, (3) Poxel SA, Lyon, France

Objectives

To describe the PK characteristics of imeglimin following single and repeated oral administration in healthy volunteers and type 2 diabetes (T2DM) patients with different degrees of chronic kidney disease (CKD) and to identify covariates of clinical relevance.

Background

Imeglimin is a novel first-in class oral antidiabetic drug to treat T2DM [1]. Imeglimin is not metabolized but it is eliminated unchanged into urine [2]. Thus, a dosage reduction may be required in renally impaired patients.

Phase 2b studies showed that the optimal dose was 1000 mg BID in Japanese T2DM patients [3] and 1500 mg twice daily (BID) in Western T2DM patients [4] and slightly higher imeglimin exposures were observed in Japanese vs Western T2DM patients.

Methods

Data from four Phase I studies, four Phase II studies and one Phase III study were used for the PK model development. Moreover, two additional Phase III studies were used for external evaluation of the PK model.

Nonlinear mixed effects modelling was conducted using NONMEM v 7.3.0 [5].

Based on the final population PK model, imeglimin AUCs were simulated in 1000 Japanese reference patients sampled randomly with replacement from P2b and P3 studies in Japanese T2DM patients. The reference patients were duplicated with estimated glomerular filtration rate (eGFR) sampled from uniform distributions in the eGFR ranges of CKD stages:

- **G3a**: 45 to < 60
- **G3b**: 30 to < 45
- **G4**: 15 to < 30 mL/min/1.73 m^2

and the mean age was increased based on literature data in Japanese patients with CKD [6][7][8] to adjust for the known increase in age with CKD stage.

Simulations of AUC in Japanese and Western patients were also performed by sampling patients' covariates from relevant studies in Japanese and Western T2DM patients. The Western reference subjects were duplicated:

- (i) with similar eGFR distribution
- (ii) with similar eGFR and body weight distributions

as in the Japanese reference patients.

Results

- The analysis dataset for PK model development contained 8256 PK observations from 867 individuals with a median age of 59.0 (range 20.0 - 80.0) years, body weight of 77.0 (range 35.6 - 148) kg and eGFR of 81.4 (14.1 - 152) mL/min/1.73 m^2. The subjects were mainly Western (55.6%), and male (56.9%).

Figure 1. *Imeglimin PK model.*

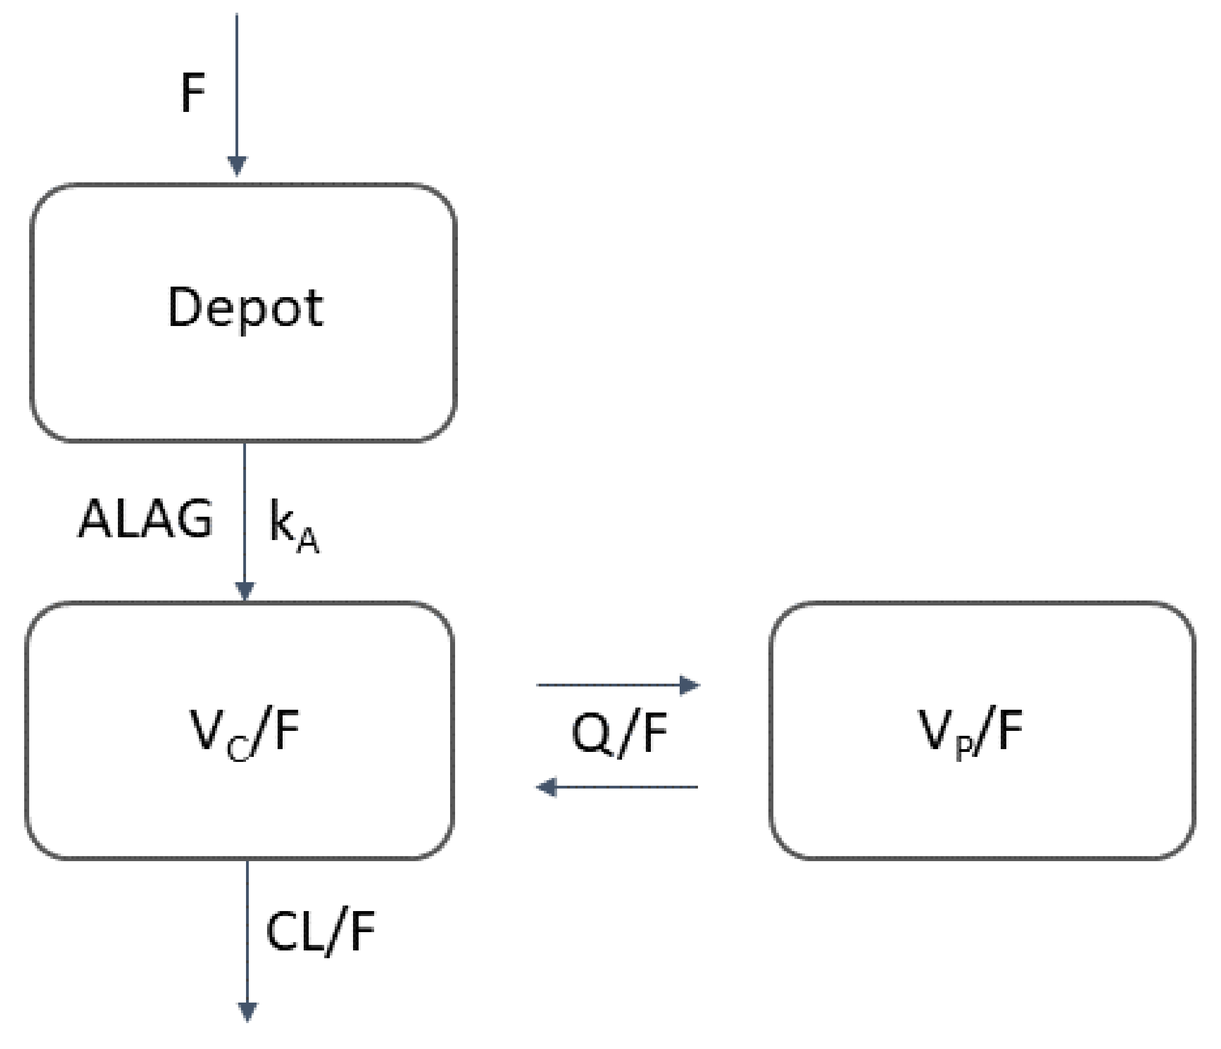
F
Depot
ALAG
k_A
V_C/F
Q/F
V_P/F
CL/F

- Imeglimin population PK was described by a 2-compartment model with first-order absorption (ka) with a lag time (ALAG) and a first-order elimination from the central compartment. Inter-individual variability was supported on k_A, relative bioavailability (F), volumes of distribution (Vc, Vp) and clearance (CL) with a correlation between CL and F (**Figure 1**).

- The population PK model accurately described imeglimin PK characteristics across CKD stages (**Figure 2**).

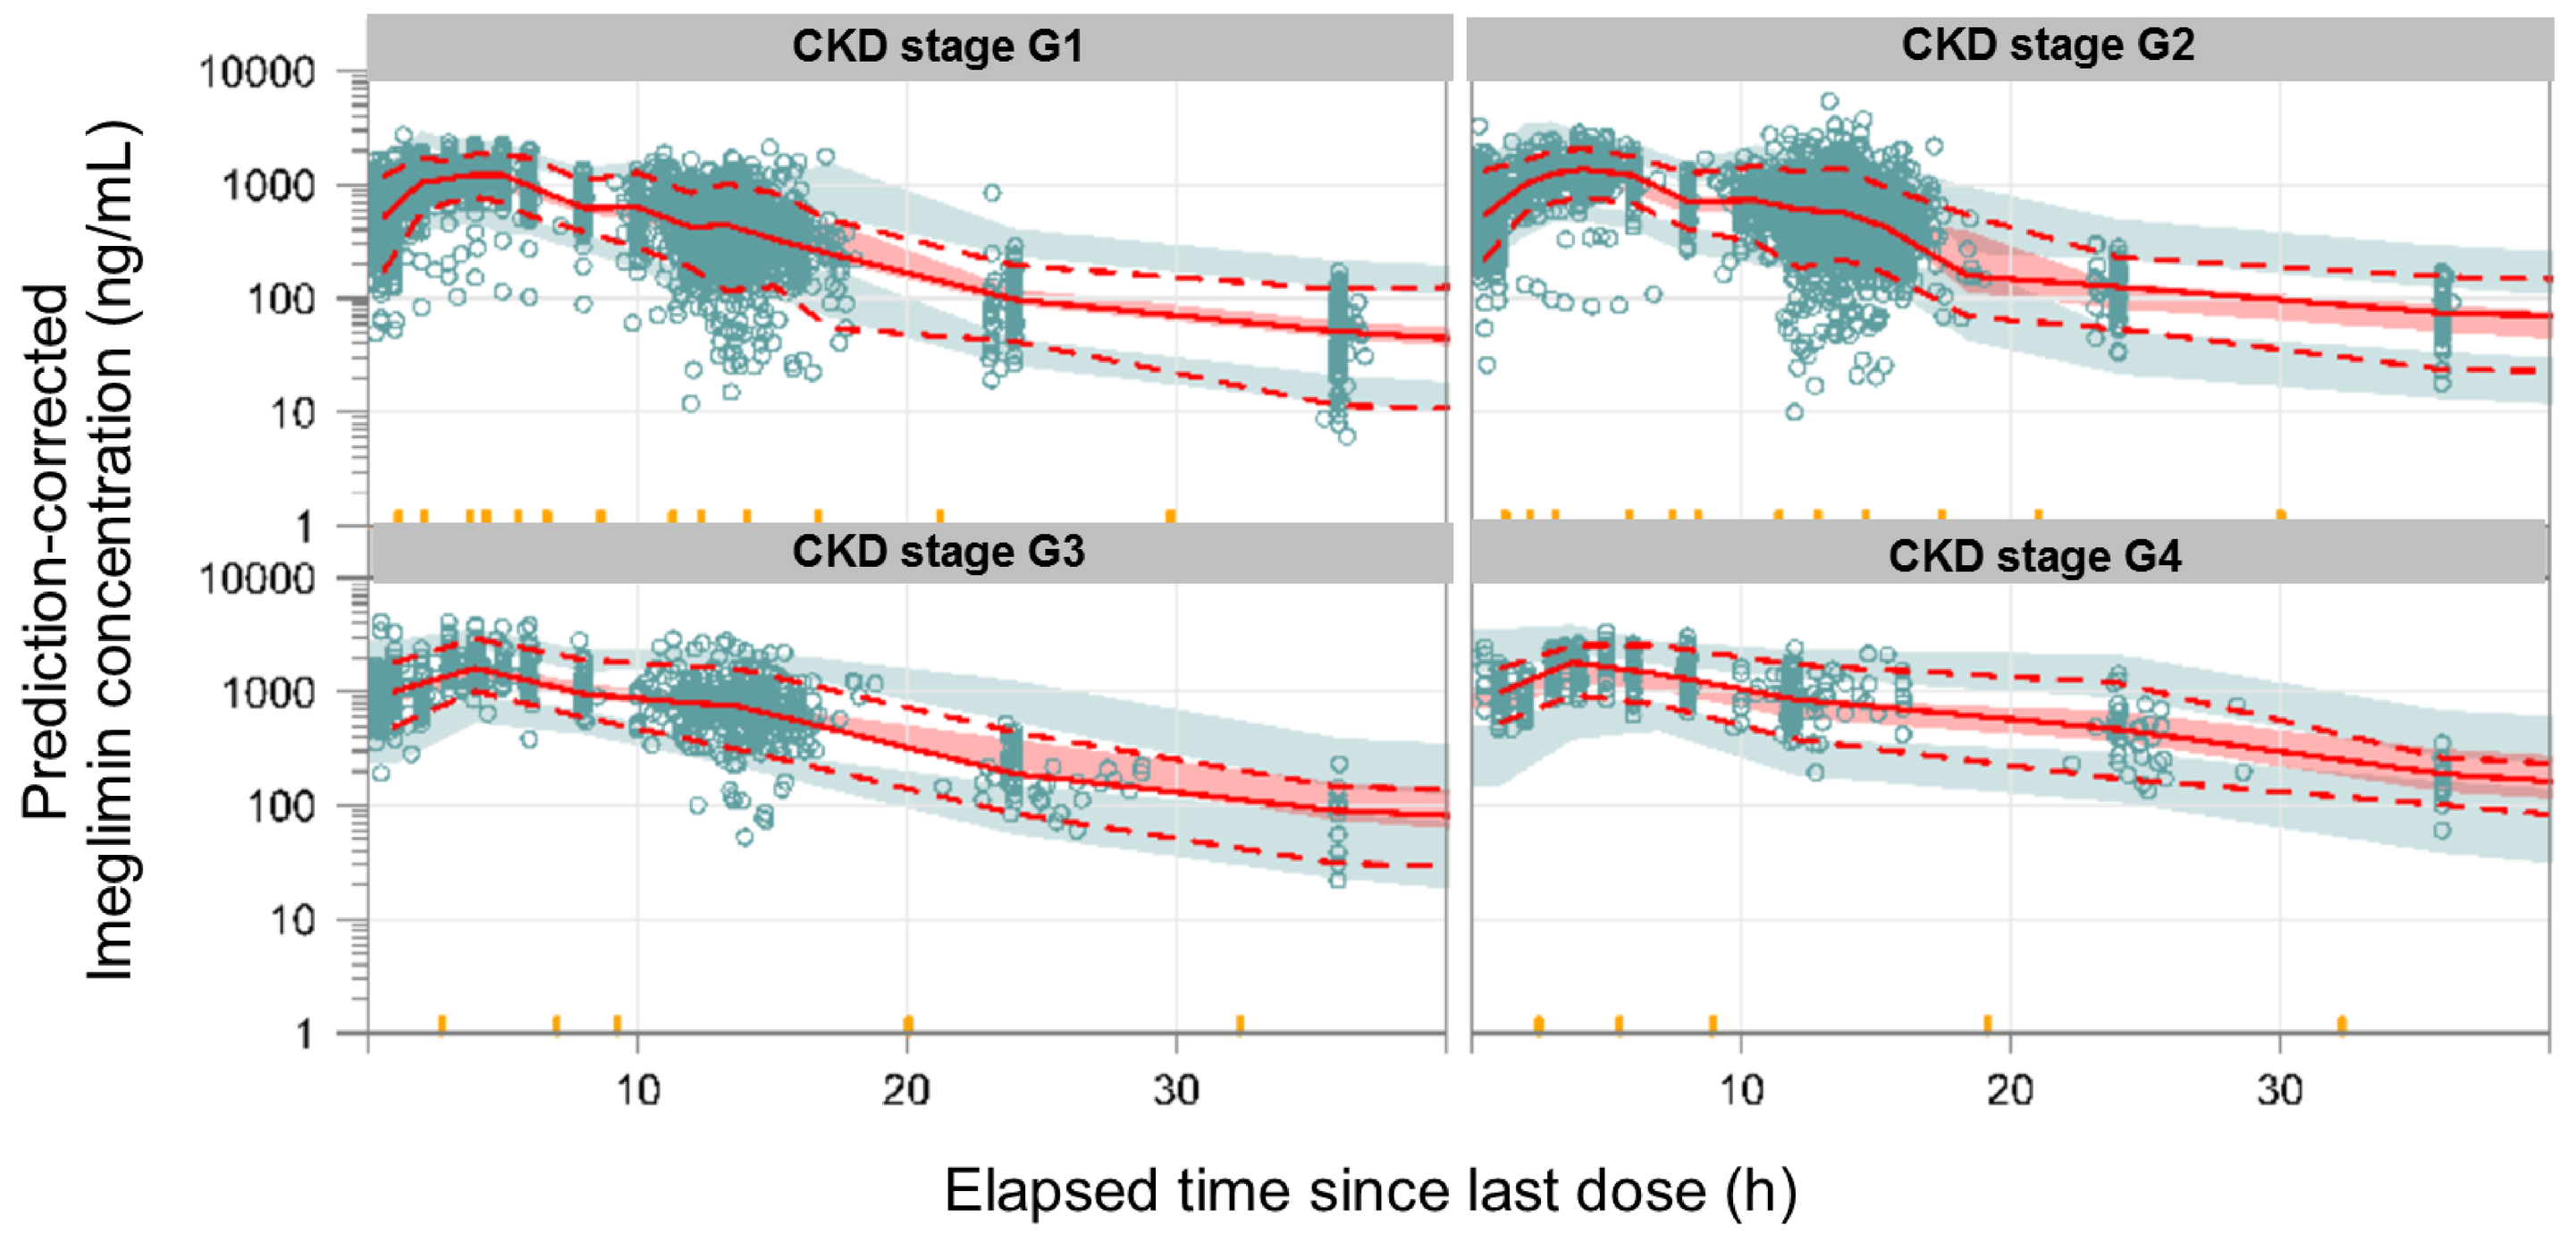
CKD stage G1
CKD stage G2
CKD stage G3
CKD stage G4
Prediction-corrected
Imeglimin concentration (ng/mL)
10000
1000
100
10
1
10
20
30
Elapsed time since last dose (h)

Figure 2. *Prediction-corrected visual predictive check (pcVPC) of imeglimin plasma concentrations vs. elapsed time since last dose stratified by estimated glomerular filtration rate (eGFR) range, CKD stages G1, ≥ 90; G2, 60 to < 90; G3, 30 to < 60; G4, 15 to < 30 mL/min/1.73 m^2 .*

- Several covariates were identified as significant predictors for imeglimin PK. Lower body weight, higher age, and lower eGFR all lead to lower CL while higher dose leads to lower F. Among these covariates, eGFR had the largest impact on imeglimin exposure (**Figure 3**).

- Simulations for Japanese and Western T2DM patients showed that the difference in plasma AUC between these populations at the same dose was mainly driven by eGFR differences (+15.24 mL/min/1.73 m^2 in Western) (**Table 1**).

Table 1 *Simulated imeglimin $AUC_{24,ss}$ and its ratio to Japanese reference patients in Western T2DM patients with changes in eGFR and body weight (median [min, max] for covariates and median [2.5th percentile, 97.5th percentile] for $AUC_{24,ss}$ and ratio).*

		Japanese reference	Westerners reference	Westerners with eGFR similar to Japanese	Westerners with eGFR & weight similar to Japanese
Age		62 [32, 83]	59 [20, 75]	59 [20, 75]	59 [20, 75]
Weight		68.8 [35.6, 124]	85.8 [54.0, 135]	85.8 [54.0, 135]	68.5 [36.7, 118]
eGFR		72.6 [47.2, 138]	92.8 [47.3, 125]	77.6 [32.2. 110]	77.6 [32.2, 110]
2000 mg BID	$AUC_{24,ss}$	46 [23, 96]	37 [18, 75]	42 [20, 88]	45 [22, 95]
	$AUC_{24,ss}$ ratio	1.6 [0.81, 3.4]	1.3 [0.65, 2.7]	1.5 [0.72, 3.2]	1.6 [0.79, 3.4]
1500 mg BID	$AUC_{24,ss}$	37 [17, 75]	29 [14, 59]	34 [16, 71]	37 [18, 76]
	$AUC_{24,ss}$ ratio	1.3 [0.62, 2.7]	1.0 [0.51, 2.1]	1.2 [0.57, 2.5]	1.3 [0.64, 2.7]
1000 mg BID	$AUC_{24,ss}$	28 [13, 55]	22 [11, 43]	25 [12, 50]	27 [13, 55]
	$AUC_{24,ss}$ ratio	1 [0.47, 2.0]	0.77 [0.38, 1.6]	0.9 [0.43, 1.8]	0.98 [0.47, 2.0]

- Simulations of AUC in T2DM Japanese patients with moderate to severe renal impairment suggested that imeglimin dose should be reduced from 1000 to 500 mg BID for patients with eGFR ranging from 15 to 45 mL/min/1.73 m^2 (**Table 2**).

Table 2. *Simulated imeglimin $AUC_{24,ss}$ and its ratio to reference in patients with renal impairment (median [min, max] for covariates and median [2.5th percentile, 97.5th percentile] for $AUC_{24,ss}$ and ratio) .*

		Japanese reference	CKD stage G3a	CKD stage G3b	CKD stage G4
Age		62.0 [32, 83]	68 [38, 89]	70 [40, 91]	70 [40, 91]
Weight		68.8 [35.6, 124]	68.8 [35.6, 124]	68.8 [35.6, 124]	68.8 [35.6, 124]
eGFR		72.6 [47.2, 138]	52.5 [45.0, 60.0]	37.6 [30.0, 45.0]	22.4 [15.0, 30.0]
1000 mg BID	$AUC_{24,ss}$	27 [13, 55]	36 [19, 71]	45 [23, 92]	60 [32, 120]
	$AUC_{24,ss}$ ratio	1 [0.48, 2.0]	1.3 [0.71, 2.6]	1.7 [0.85, 3.4]	2.2 [1.2, 4.4]
500 mg BID	$AUC_{24,ss}$	15 [7.3, 30]	20 [11, 39]	26 [14, 47]	33 [18, 69]
	$AUC_{24,ss}$ ratio	0.56 [0.27, 1.1]	0.74 [0.39, 1.4]	0.95 [0.51, 1.7]	1.2 [0.66, 2.5]

Conclusions

- A model characterizing imeglimin PK in T2DM patients was developed and used to derive expected exposures in moderate/severe renal impaired patients.
- Simulations helped to recommend dosing adjustment in renally impaired patients (eGFR between 15 and 45 mL/min/1.73 m^2)
- This work revealed that plasma exposure differences observed between Japanese and Western populations were mainly driven by difference in eGFR.

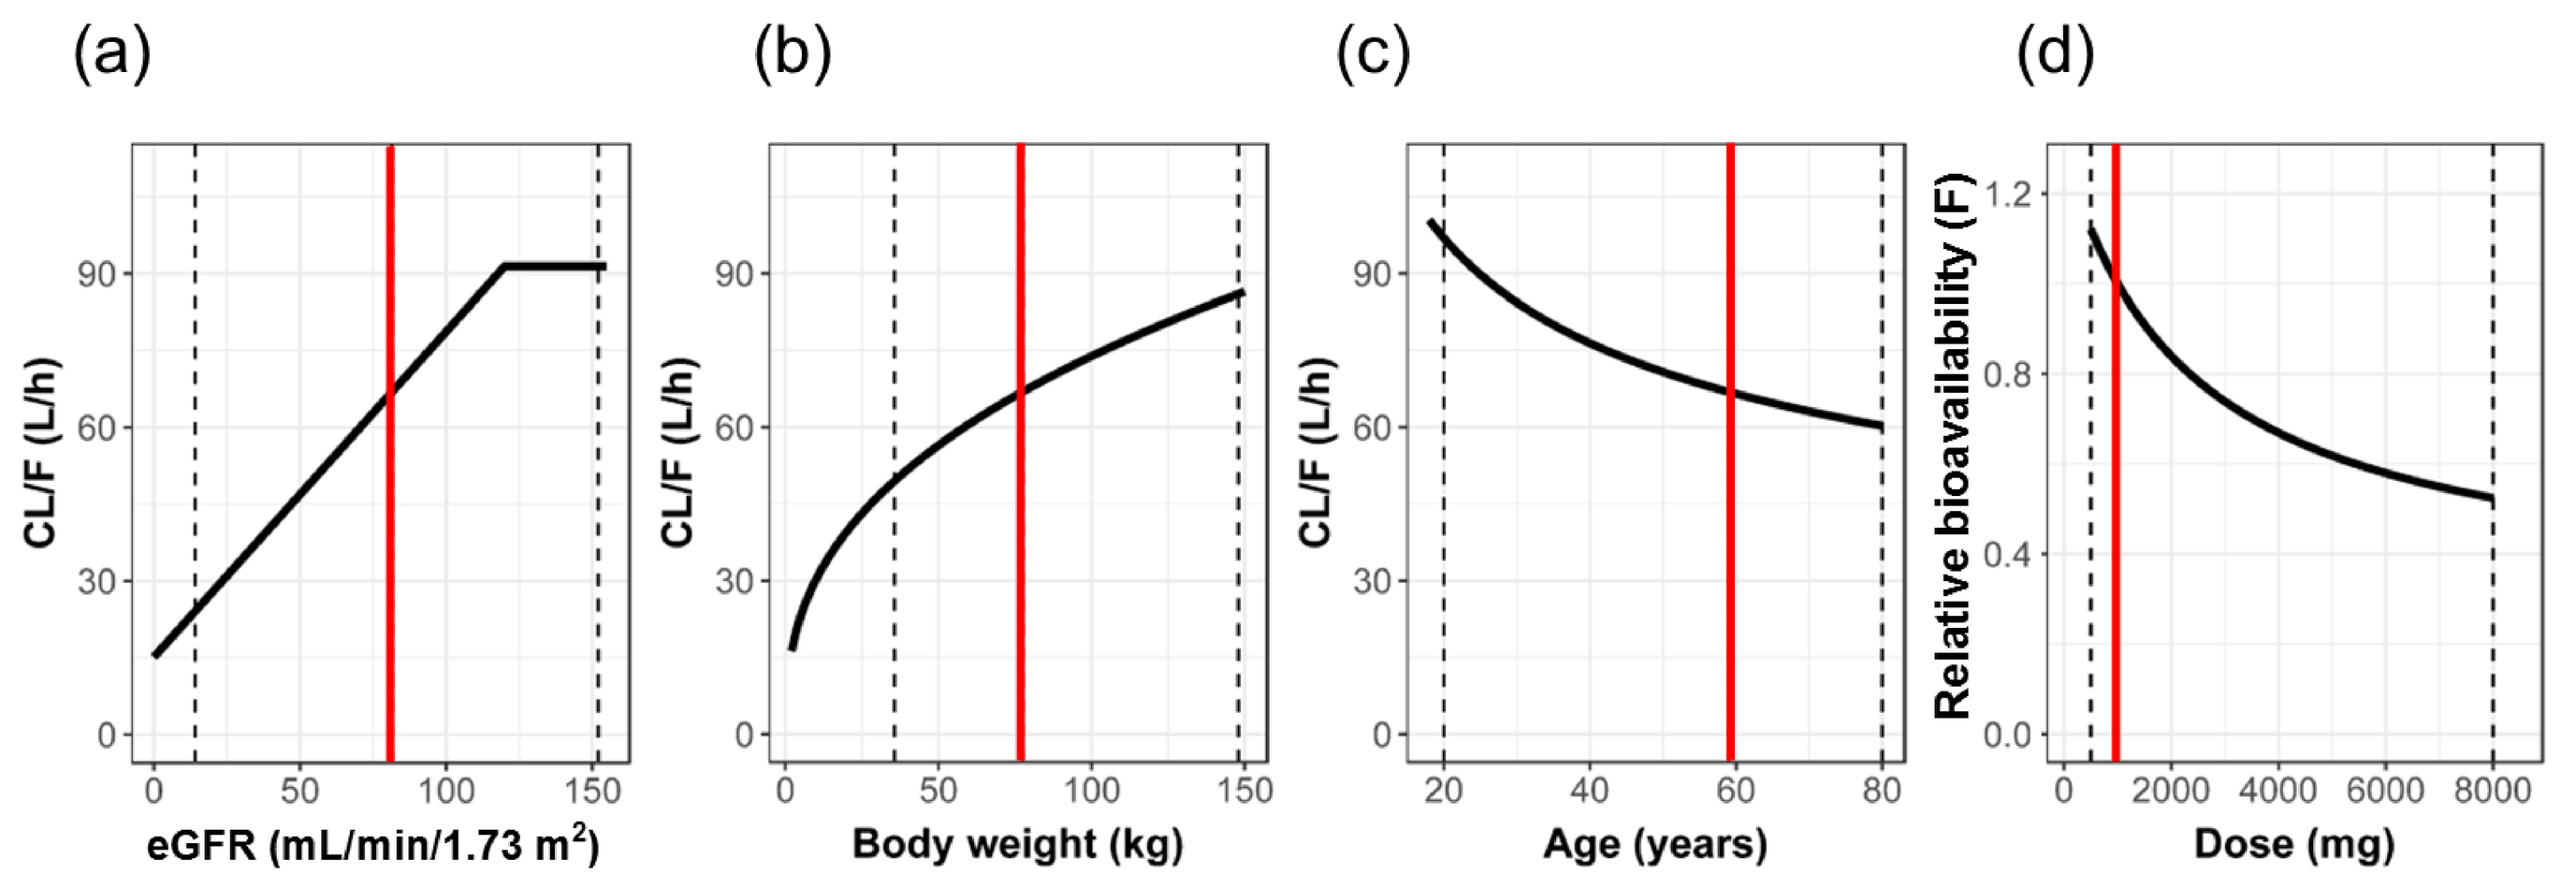
(a)
(b)
(c)
(d)
CL/F (L/h)
Relative bioavailability (F)
eGFR (mL/min/1.73 m²)
Body weight (kg)
Age (years)
Dose (mg)

Figure 3*. Impact of key covariates on imeglimin pharmacokinetics. Impact of (a) estimated glomerular filtration rate (eGFR) on apparent clearance (CL/F), (b) body weight on CL/F, (c) age on CL/F, and (d) dose on relative bioavailability (F). Other covariates set to the reference values in the model were age 59 years, body weight 77.35 kg, eGFR 81.4 mL/min/1.73 m^2 , and imeglimin 1,000 mg.*

References

1. Dubourg, J. et al. Diabetes Care 44(4), 952-959 (2021).
2. Clémence, C. et al. Drug Metabolism and Disposition. 48(12), 1330-46 (2020).
3. Dubourg, J. et al. Diabetes Obesity and Metabolism 23(3):800-810 (2021).
4. Johansson, K.S. et al. Expert Opinion on Pharmacotherapy, 21(8), pp.871-882 (2020).
5. Beal SL, NONMEM User's Guides. (1989-2014) Icon Development Solutions, Ellicott City, MD, USA
6. Haneda, M. et al. Clin. Ther. 38, 66-88 (2016).
7. Haneda, M. et al. Diabetes Ther. 9, 1083-1097 (2018).
8. Kobayashi, K. et al. Diab. Vasc. Dis. Res. 16, 103–107 (2019).

Contact

enrica.mezzalana@pharmetheus.com
www.Pharmetheus.com