A semi-mechanistic population pharmacokinetic model for trastuzumab emtansine (T-DM1) antibody-drug conjugate and total antibody in patients with metastatic breast cancer (mBC)

Franziska Schaedeli Stark[1], Vaishali L. Chudasama[2], Manish Gupta[3], Jay Tibbitts[3], Nicolas Frey[1], Donald E. Mager[2]

[1]Translational Research Sciences, F. Hoffman-La Roche Ltd, Basel, Switzerland, [2]Department of Pharmaceutical Sciences, University at Buffalo, SUNY, Buffalo, NY, [3]Pharmacokinetics and Pharmacodynamics, Genentech Inc., South San Francisco, CA

Introduction

Antibody-drug conjugates (ADC) are a class of targeted drugs with antibodies bearing covalently bound cytotoxic agents designed to target antigen-specific cells to enhance efficacy and reduce the toxicity associated with the cytotoxic agent alone.

Trastuzumab-emtansine (T-DM1) is an ADC with the antimicrotubule agent DM1 conjugated to trastuzumab through a stable thioether linker. The drug-antibody ratio (DAR), i.e. the number of DM1 molecules attached to trastuzumab, can range from 0 to 8 (average DAR = 3.5).

T-DM1 concentrations decrease more rapidly than total trastuzumab (TT), i.e. conjugated and unconjugated T-DM1 (Figure 1). The decrease in T-DM1 concentration is driven by two processes: deconjugation and antibody elimination.

Preclinical T-DM1 PK data demonstrated that high DAR species disappear faster than low DAR species [1].

A semi-mechanistic pharmacokinetic model on monkey data representing 8 measured DAR species with a chain of 8 transit compartments has previously been proposed [1].

Objective

The objective of this study is to develop a semi-mechanistic population pharmacokinetic model to better understand the differences between T-DM1 and total trastuzumab concentrations in patients with HER2+ metastatic breast cancer.

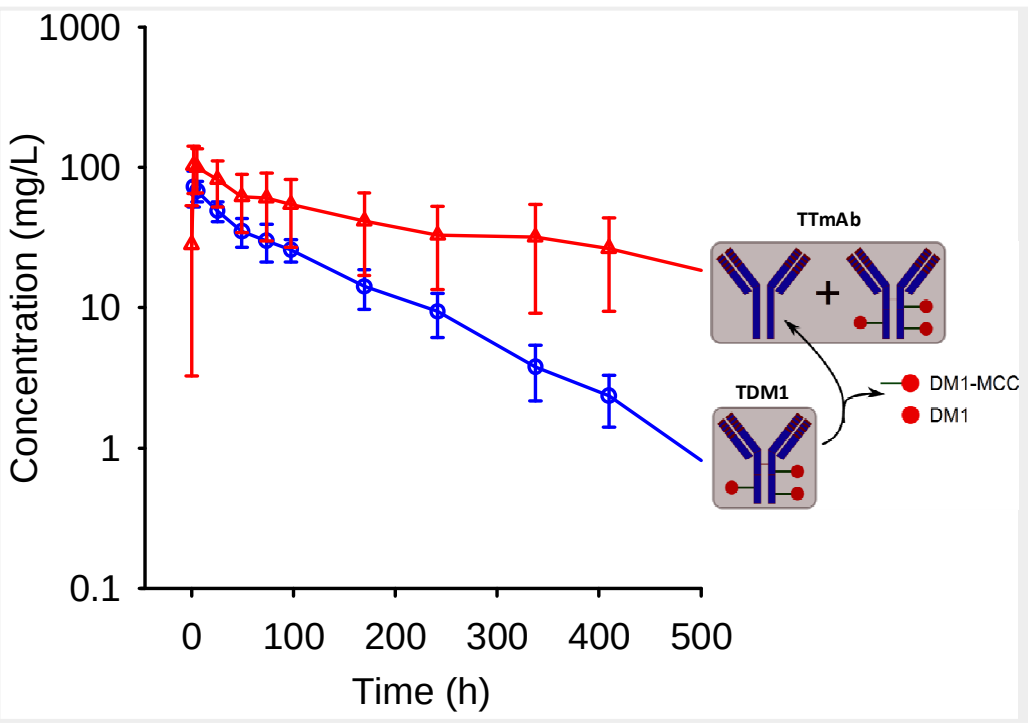
1000
100
10
1
0.1
Concentration (mg/L)
0
100
200
300
400
500
Time (h)
TTmAb
TDM1
DM1-MCC
DM1

Figure 1: Time-course of T-DM1 (blue) and TT (T-DM1 +Tmab, red lines) for 3.6 mg/kg Q3W treatment in study TDM3569g. Symbols are observed mean data and error bars are SD.

The hypothesis is that deconjugation of DM1 containing entities leads to faster decrease of T-DM1, compared to TT.

Methods

Patient population and study design

Pharmacokinetic data from a phase I dose escalation study (TDM3569g) in 52 patients [2], and from a phase II study (TDM4258g) in 111 patients with heavily pre-treated HER2+ metastatic breast cancer [3] were available for model building.

In the phase I study doses ranged from 1.2 to 2.9 mg/kg (1.2, 1.6, 2.0, 2.4, and 2.9 mg/kg) in the once-weekly study arm (n=28) and from 0.3 to 4.8 mg/kg (0.3, 0.6, 1.2, 2.4, 3.6 and 4.8 mg/kg) in the Q3W regimen (n=24). In the phase II study 3.6 mg/kg of T-DM1 was given Q3W. T-DM1 was administered via intravenous (IV) infusion.

Concentrations of T-DM1 and TT were measured throughout the treatment course, with a frequent sampling schedule in cycles 1 and 4, and pre- and post-infusion in all other cycles (see [2,3] for details).

Pharmacokinetic model

The proposed pharmacokinetic model (Figure 2) is based on the semi-mechanistic model developed for T-DM1 kinetics in monkeys [1].

The number of individual T-DM1 DAR compartments was optimized, from originally eight to five, by removing one compartment at a time and comparing model-fitting criteria.

T-DM1 elimination was described as the net effect of transition from higher to lower DAR species (deconjugation), and elimination from each DAR compartment (antibody degradation). The slower transition kinetics from DAR1 to DAR0 (0.5*ktd) is consistent with pre-clinical observations [1].

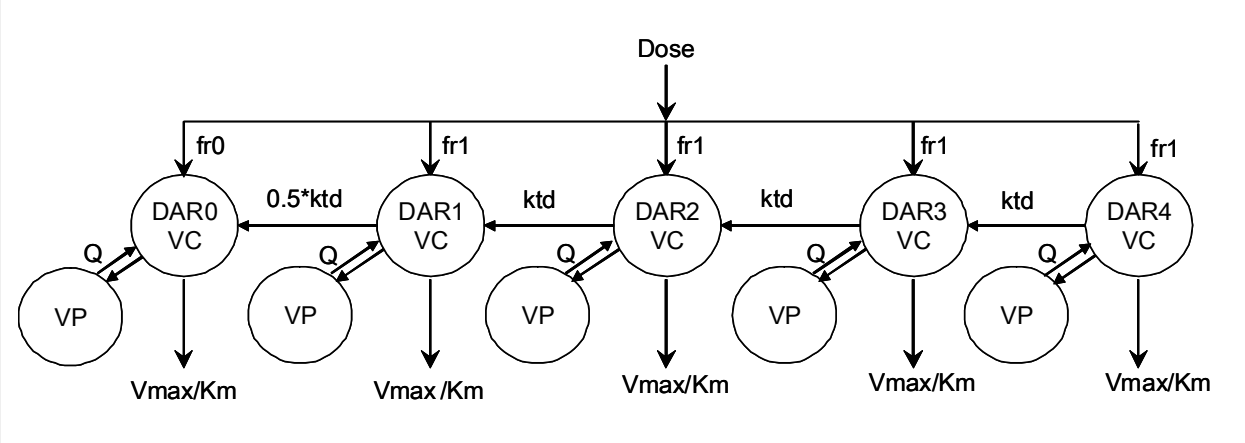
Dose
fr0
fr1
fr1
fr1
fr1
DAR0 VC
0.5*ktd
DAR1 VC
ktd
DAR2 VC
ktd
DAR3 VC
ktd
DAR4 VC
Q
VP
Vmax/Km

Figure 2: Semi-mechanistic pharmacokinetic model of T-DM1 and TT disposition.

The dose is fractionated in fr0 (DAR0 = unconjugated T-DM1), and four equal fractions fr1 of conjugated T-DM1 (DAR1-4). DAR deconjugation is represented by a first order transition from higher to lower DAR levels. All compartments share the same volumes (VC, VP) and Michaelis-Menten elimination kinetics.

Population data analysis

All measurements of total T-DM1 and TT concentrations from the pooled phase I and II study were modeled simultaneously using a nonlinear mixed effects modeling approach with the MCPEM algorithm implemented in S-ADAPT 4.

Inter- and intra-individual variability was characterized along with a full variance-covariance matrix. Residual variability was modeled using an additive plus proportional variance model.

Model development and qualification were guided by goodness of fit plots and the precision of parameter estimates. A visual predictive check (VPC) was done for further model qualification.

Conclusions

Preclinical pharmacokinetic modeling efforts in monkeys were utilized as an initial approach to developing a clinical semi-mechanistic model.

The proposed model successfully described the time-courses of T-DM1 and TT concentrations simultaneously using transit compartments to emulate the deconjugation of higher DAR levels to low DAR levels and unconjugated trastuzumab, combined with a common elimination process for all DAR species.

This model supports the hypothesis that the extended terminal half-life of TT relative to T-DM1 is a consequence of the T-DM1 deconjugation process from higher to lower DAR species (Figure 5).

This modeling framework may be useful to further investigate the release kinetics of DM1, and to characterize the PK of other ADC systems.

Results

The final structural model includes a chain of 5 transit compartments representing 5 DAR levels of T-DM1, with shared volumes of distribution and elimination kinetics for each compartment.

The final population PK parameters are presented in Table 1. All parameters were estimated with good precision.

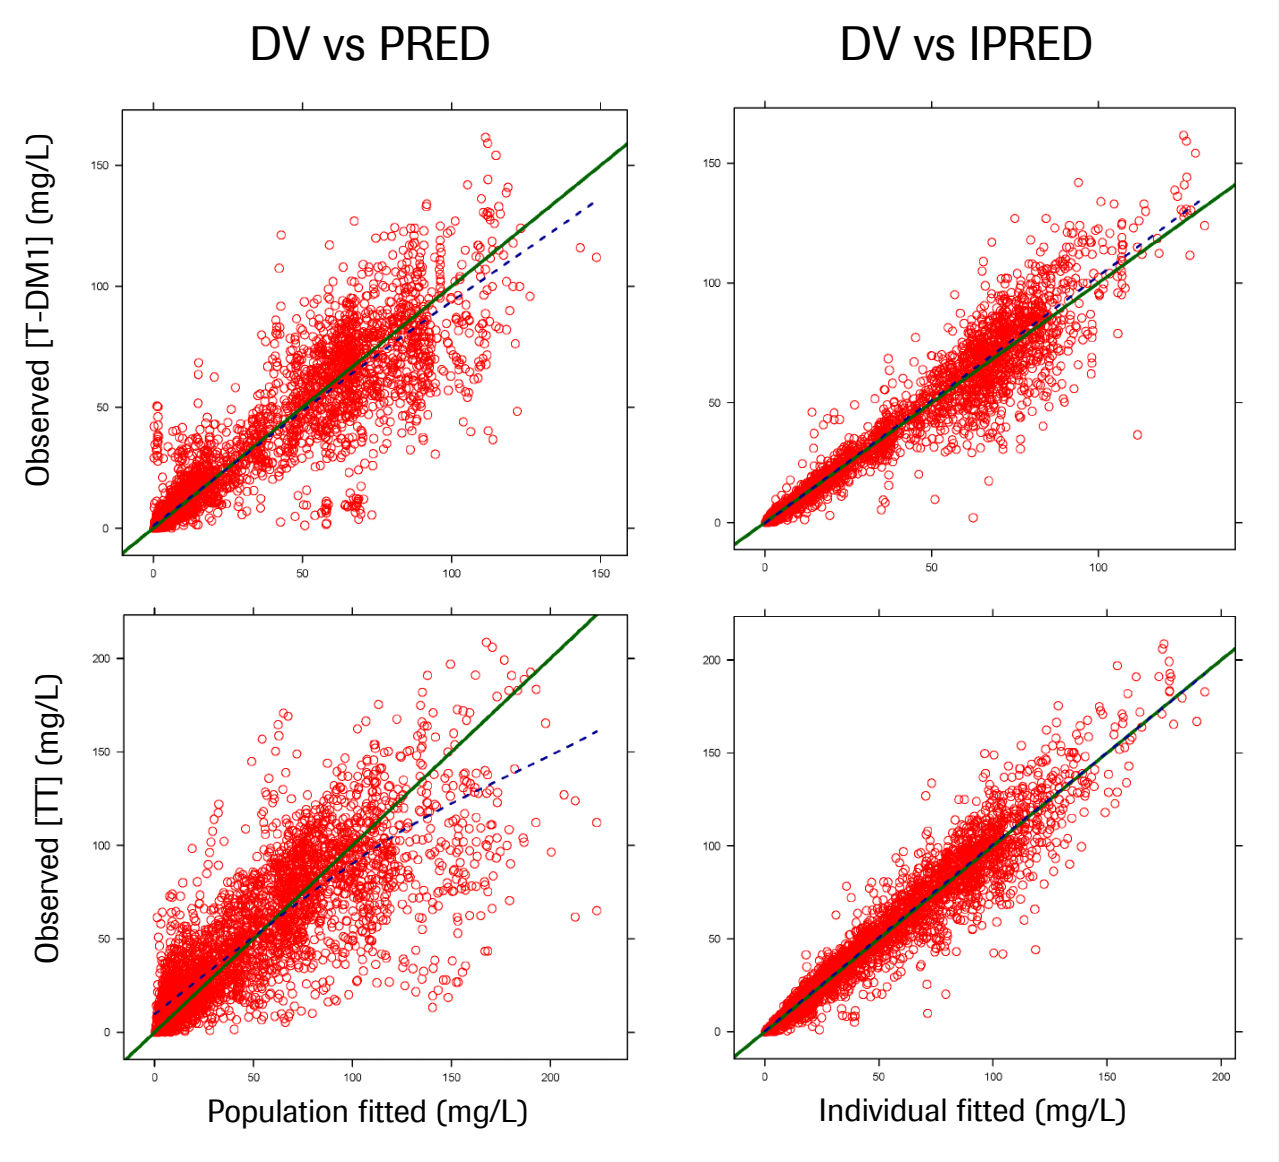
DV vs PRED
DV vs IPRED
Observed [T-DM1] (mg/L)
Observed [TT] (mg/L)
Population fitted (mg/L)
Individual fitted (mg/L)

Figure 3: Goodness of fit plots for T-DM1 (upper panels) and TT (lower panesl). Solid lines: line of identity, broken lines: loess smoothers.

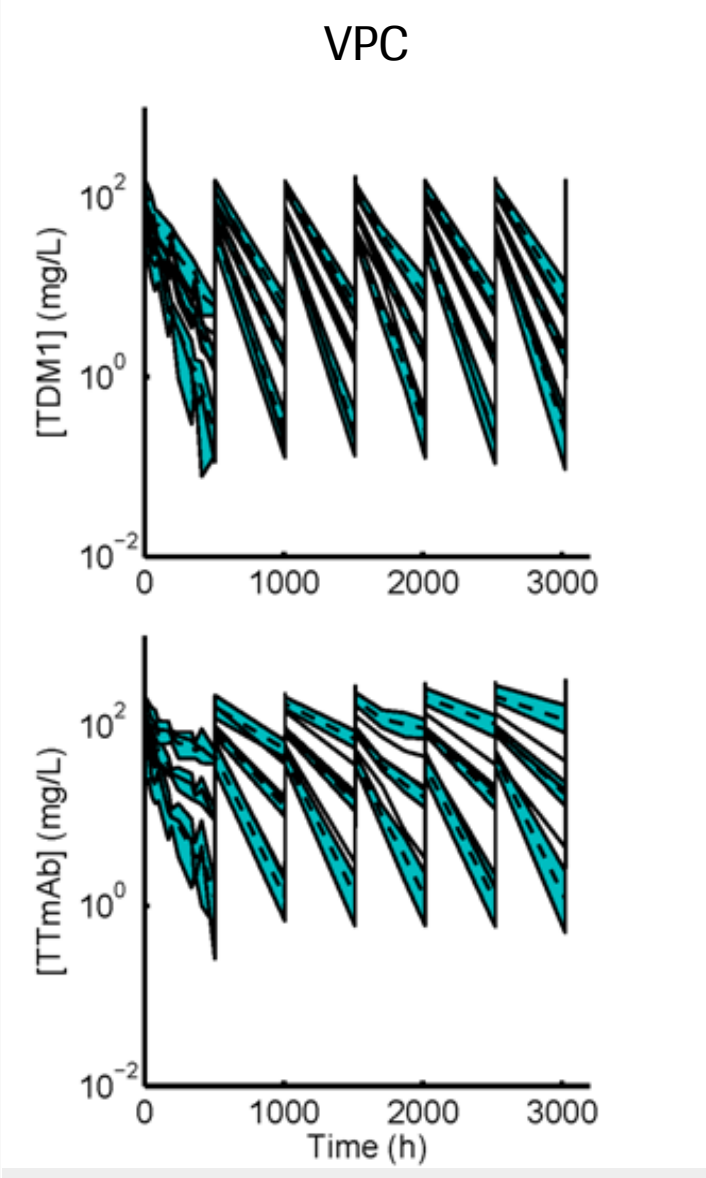
VPC
[TDM1] (mg/L)
[TTmAb] (mg/L)
0
1000
2000
3000
Time (h)

Figure 4: VPC for T-DM1 (upper panel) and TT (lower panel).

Table 1: Final population PK parameter estimates

Parameter	Unit	Definition	Estimate	RSE (%)	IIV (% CV)	RSE (%)
Fr0		Fractional dose input into Tmab compartment	0.045	3.71	63.6	23.5
VC	L	Central Volume of distribution for TDM1 and TT	3.21	1.54	17.5	13.4
ktd	1/day	Transfer rate constant from higher to lower DAR	0.35	1.84	15.5	20.5
Km	mg/L	Michaelis-Menten affinit constant	11.1	11.3	129	14.2
Clint	L/day	Intrinsic clearance	0.751	6.56	75.8	14.3
Q	L/day	Distributional clearance	0.576	9.61	63.1	24.4
VP	L	Peripheral volume of distribution	1.7	5.81	62.3	15.1
Vmax	mg/day	Michaelis-Menten capacity constant	8.338	Clint*Km		

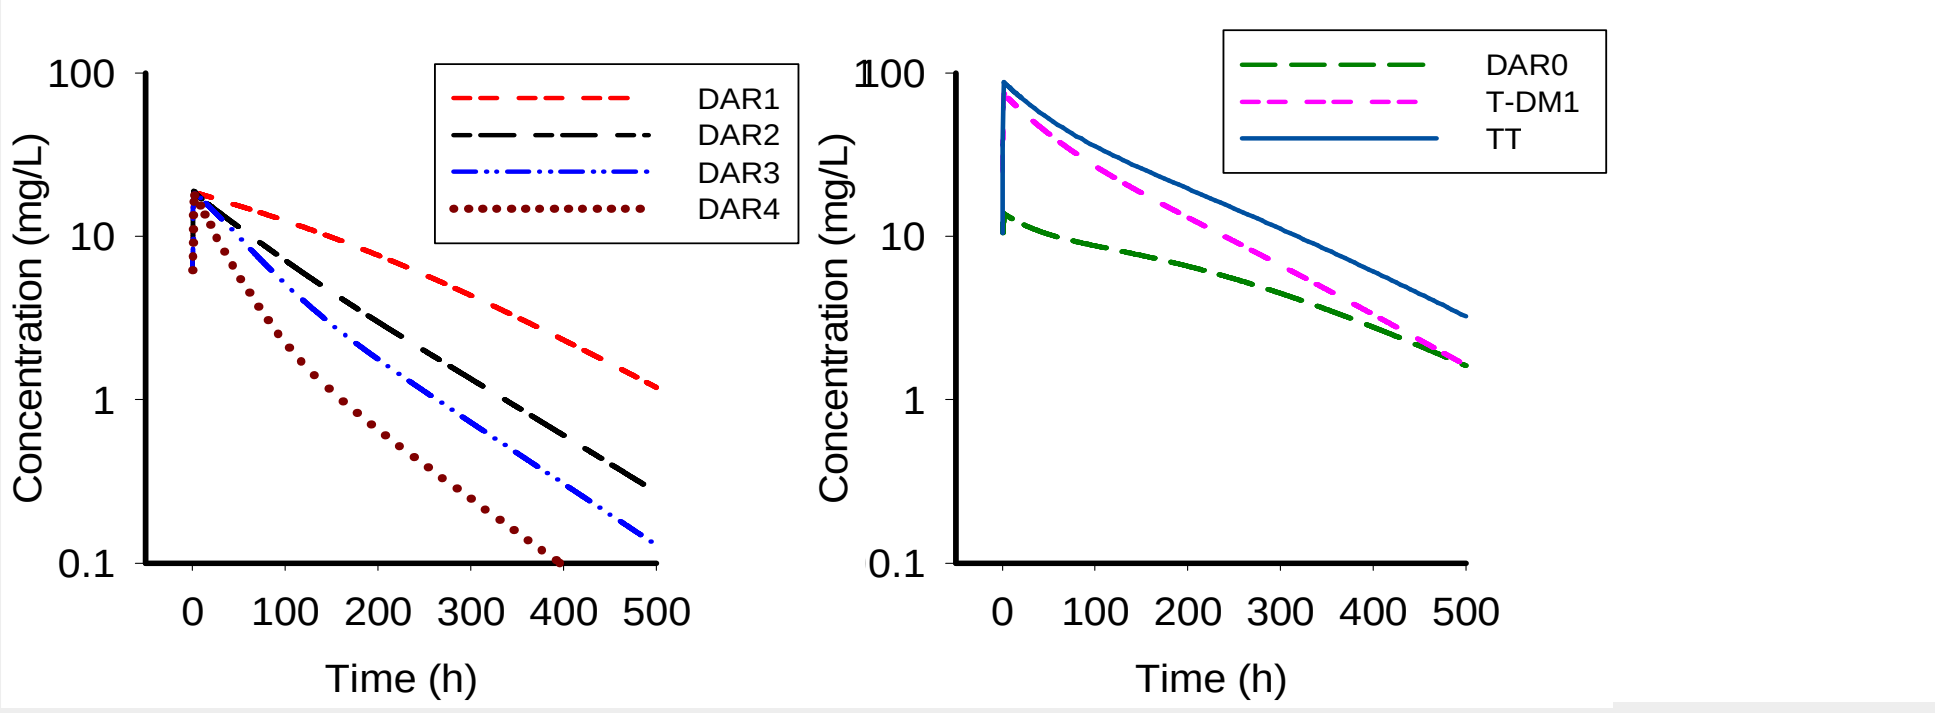
DAR1
DAR2
DAR3
DAR4
DAR0
T-DM1
TT
Concentration (mg/L)
Time (h)

Figure 5: Simulations showing the time-courses of different model DAR compartments (not observed; left panel). Simulated T-DM1 (sum of all DAR>0) and TT (sum of all DAR compartments) are consistent with the observed data. The shape of simulated DAR0 (initial decrease, followed by transient increase) supports the hypothesis that the DAR transition causes the apparently different elimination patterns (right panel).

References

1. Leipold D et al. AACR Meeting. 100, 2914 (2009).
2. Krop IE et al. J Clin Oncol. 28:2698-2704. (2010).
3. Burris HA et al. J Clin Oncol. E-publication ahead of print. Dec 20. 2010.
4. Bulitta JB et al. AAPS J. In press.