Using an Innovative Design in Behavioural Pharmacology Studies Saves Money and Animal Lives

Anders Viberg (1), John Martino (2), Etienne Lessard (2) and Jennifer Laird (2)
(1) AstraZeneca R&D Södertälje, (2) AstraZeneca R&D Montreal

1904

Objectives:
Results from pre-clinical efficacy studies are of importance for many decisions in drug development. They are used to predict active concentrations in the biophase in man, and thus have a direct impact on the evaluation of toxicology and Phase I results and they are also an aid in dose setting in Phase II studies. Typically, pre-clinical studies in analgesia consist of at least three different studies; dose-finding, effect-duration and tolerance development studies. In behavioral studies, the exposure is usually measured in a separate group of animals because stress associated with sampling can affect the PD measures, which may compromise the precision in describing PKPD relationships. The objective of this study was to investigate if pre-clinical analgesic studies in rats could be more effectively performed using sparse PK sampling in the PD tested animals and thereafter evaluate the results using a population approach.

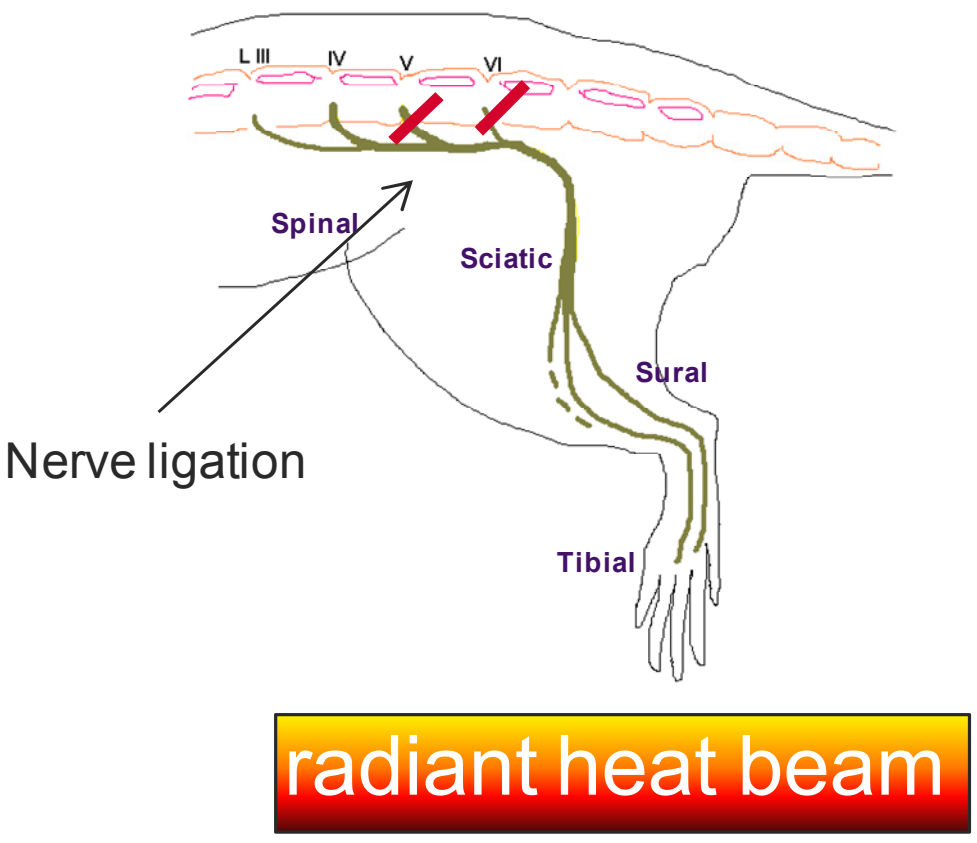
L III
IV
V
VI
Spinal
Sciatic
Sural
Tibial
Nerve ligation
radiant heat beam

Figure 1 Schematic picture of the Chung hyperalgesia model

Methods:
In order to induce hyperalgesia in male Sprague-Dawley rats the model described by Kim and Chung was used [1]. The L5 and L6 spinal nerves were isolated and tightly ligated distal to the dorsal root ganglion under isoflurane anesthesia (Figure 1). Rats were thereafter allowed to recover and all experiments were conducted between postoperative days 7-25. During the experiment, a thermal stimulus in the form of a radiant heat beam was focused onto the plantar surface of the affected paw. In each test session the paw withdrawal latency, measured in seconds, were tested twice for each rat. Naive rats were used as controls. A sampling strategy was developed for drug X where PK and PD were measured in the same animal (Table 1). To minimize the potential effect the PK sampling could have on PD measurement, blood samples of PK were withdrawn on alternate days to the PD measurements. To test if the PK sampling affected the PD measurements a separate experiment using animals without PK sampling was done. Data was analyzed using a population approach in NONMEM VI.

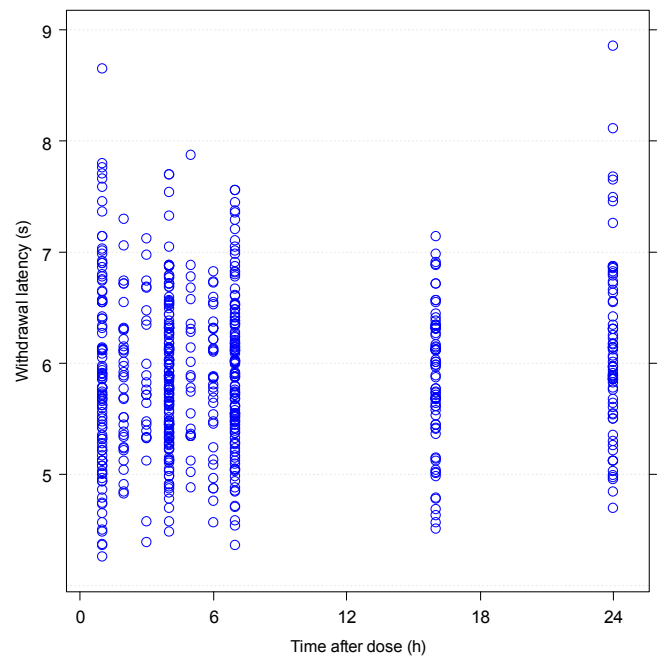
Withdrawal latency (s)
Time after dose (h)
0
6
12
18
24
5
6
7
8
9

Figure 2 Withdrawal latency of vehicle treated rats vs time after vehicle administration

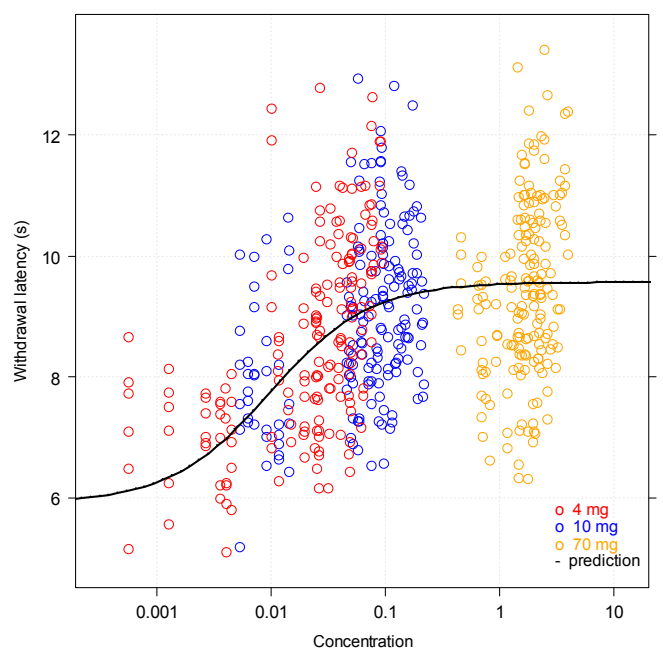
Withdrawal latency (s)
Concentration
0.001
0.01
0.1
1
10
6
8
10
12
4 mg
10 mg
70 mg
- prediction

Figure 3 Withdrawal latency vs concentration of drug treated rats

Results:
The animals with PK sampling had the same therapeutic response as the animals without PK sampling. The PD response in vehicle treated animals was stable over the entire day (Figure 2). The PK was described using a one-compartment model with first order absorption and PD was described using a direct concentration-effect relationship (Figure 3). The parameters were estimated with good precision and no tolerance development was observed. When comparing the new design with to the traditional method, substantial savings were realized. The number of animals was reduced by 44% and the number of working hours in the lab was reduced by 63% compared with the previous approach with three separate studies.

	n	Day 1 (h) 1	2	4	6	7	24	Day 2 (h) 2	4	6	7	24
4 umol/kg	6	X		X	X		X	O		O		
10 umol/kg	6		X	X		X	X		O		O	
70 umol/kg	6	X		X	X		X	O			O	
Vehicle	6		X	X		X	X					
Vehicle	6	X		X	X		X					

	Day 3 (h) 1	2	4	6	7	24	Day 4 (h) 2	4	6	7	24
4 umol/kg		X	X		X	X		O		O	
10 umol/kg	X		X	X		X	O		O		
70 umol/kg		X	X		X	X		O	O		
Vehicle	X		X	X		X					
Vehicle		X	X		X	X					

	Day 5 (h) 1	2	4	6	7	24	Day 8 (h) 3	5	7
4 umol/kg	X		X	X		X	X	X	X/O
10 umol/kg		X	X		X	X	X	X	X/O
70 umol/kg	X		X	X		X	X	X	X/O
Vehicle		X	X		X	X	X	X	X/O
Vehicle	X		X	X		X	X	X	X/O

Satellite	n	Day 1 (h) 1	2	4	6	7	24
4 umol/kg	3	O	O	O	O	O	O
10 umol/kg	3	O	O	O	O	O	O
50 umol/kg	3	O	O	O	O	O	O

Table 1 Sampling schedule where X is PD measurement and O is PK measurement

Conclusions:
The new suggested design results in substantial savings in both animal lives and money. Moreover, the exposure response relationship was described with higher statistical precision compared to using the old design.

Reference:
[1] Kim SH, Chung JM. Pain. 1992 Sep;50(3):355-63.

AstraZeneca