Development of a mathematical model to elucidate the cross-linking of GSK1995057 and ADAs and binding to TNFR1 receptor – filling the gap between hypothesis and reality.

Tamara van Steeg[1], Anne Keunecke[1], David Fairman[2]

[1]LAP&P Consultants Leiden, The Netherlands, [2]GSK Medicines Research Centre, Stevenage, Hertfordshire, SG1 2NY, UK

Contact: info@lapp.nl web: www.lapp.nl

Introduction

The variable heavy (VH) chain domain antibody (dAb™) GSK1995057 (GSK-057) was developed to selectively block TNF-α receptor 1 (TNFR1). Recently, pre-existing auto-antibodies (ADAs/human anti-VH (HAVH) autoantibodies) were discovered in approximately 50% of the GSK-057-naïve healthy human subjects which together with the dAb were associated with activation of the TNFR1 membrane bound receptor (mTNFR1), leading to symptoms of cytokine release [1].

Objectives

The aim of the current investigation was to explain the apparent reversible agonistic effects of dAbs in subjects expressing pre-existing ADAs by development of a mathematical framework for the interaction between dAb, ADA and both soluble (sTNFR1) and membrane-bound TNFR1 (mTNFR1).

Methods

A phase I trial investigating the safety and tolerability of GSK-057 in humans provided data on PK, free and total sTNFR1 levels after administration of a wide dose range of GSK-057 (single dose administration, 0.0004 up to 2.0 mg/kg). ADA positive subjects were excluded from this phase of the study. This post hoc analyses of the Phase I data were performed in NONMEM (Version 7.3, method FOCE with interaction). Model extension based on theoretical considerations, simulations and all graphical explorations were performed using R (version 3.3.2) and Rstudio (version 1.0.44). Relevant model parameters were estimated or obtained from theoretical concepts, literature or in-vitro experiments.

Results

- A full TMDD model was able to describe the observed profiles for unbound dAb, free and total sTNFR1 (sTNFR1 complexed with GSK-057) (Table 1)
- Biomarker profiles (free and total sTNFR1) were captured adequately (Figure 1)
- However, analysis revealed that the interaction with the membrane-bound receptor could not be derived from the data. Thus indicating that it's abundance/turnover was likely to be lower than that of sTNFR1

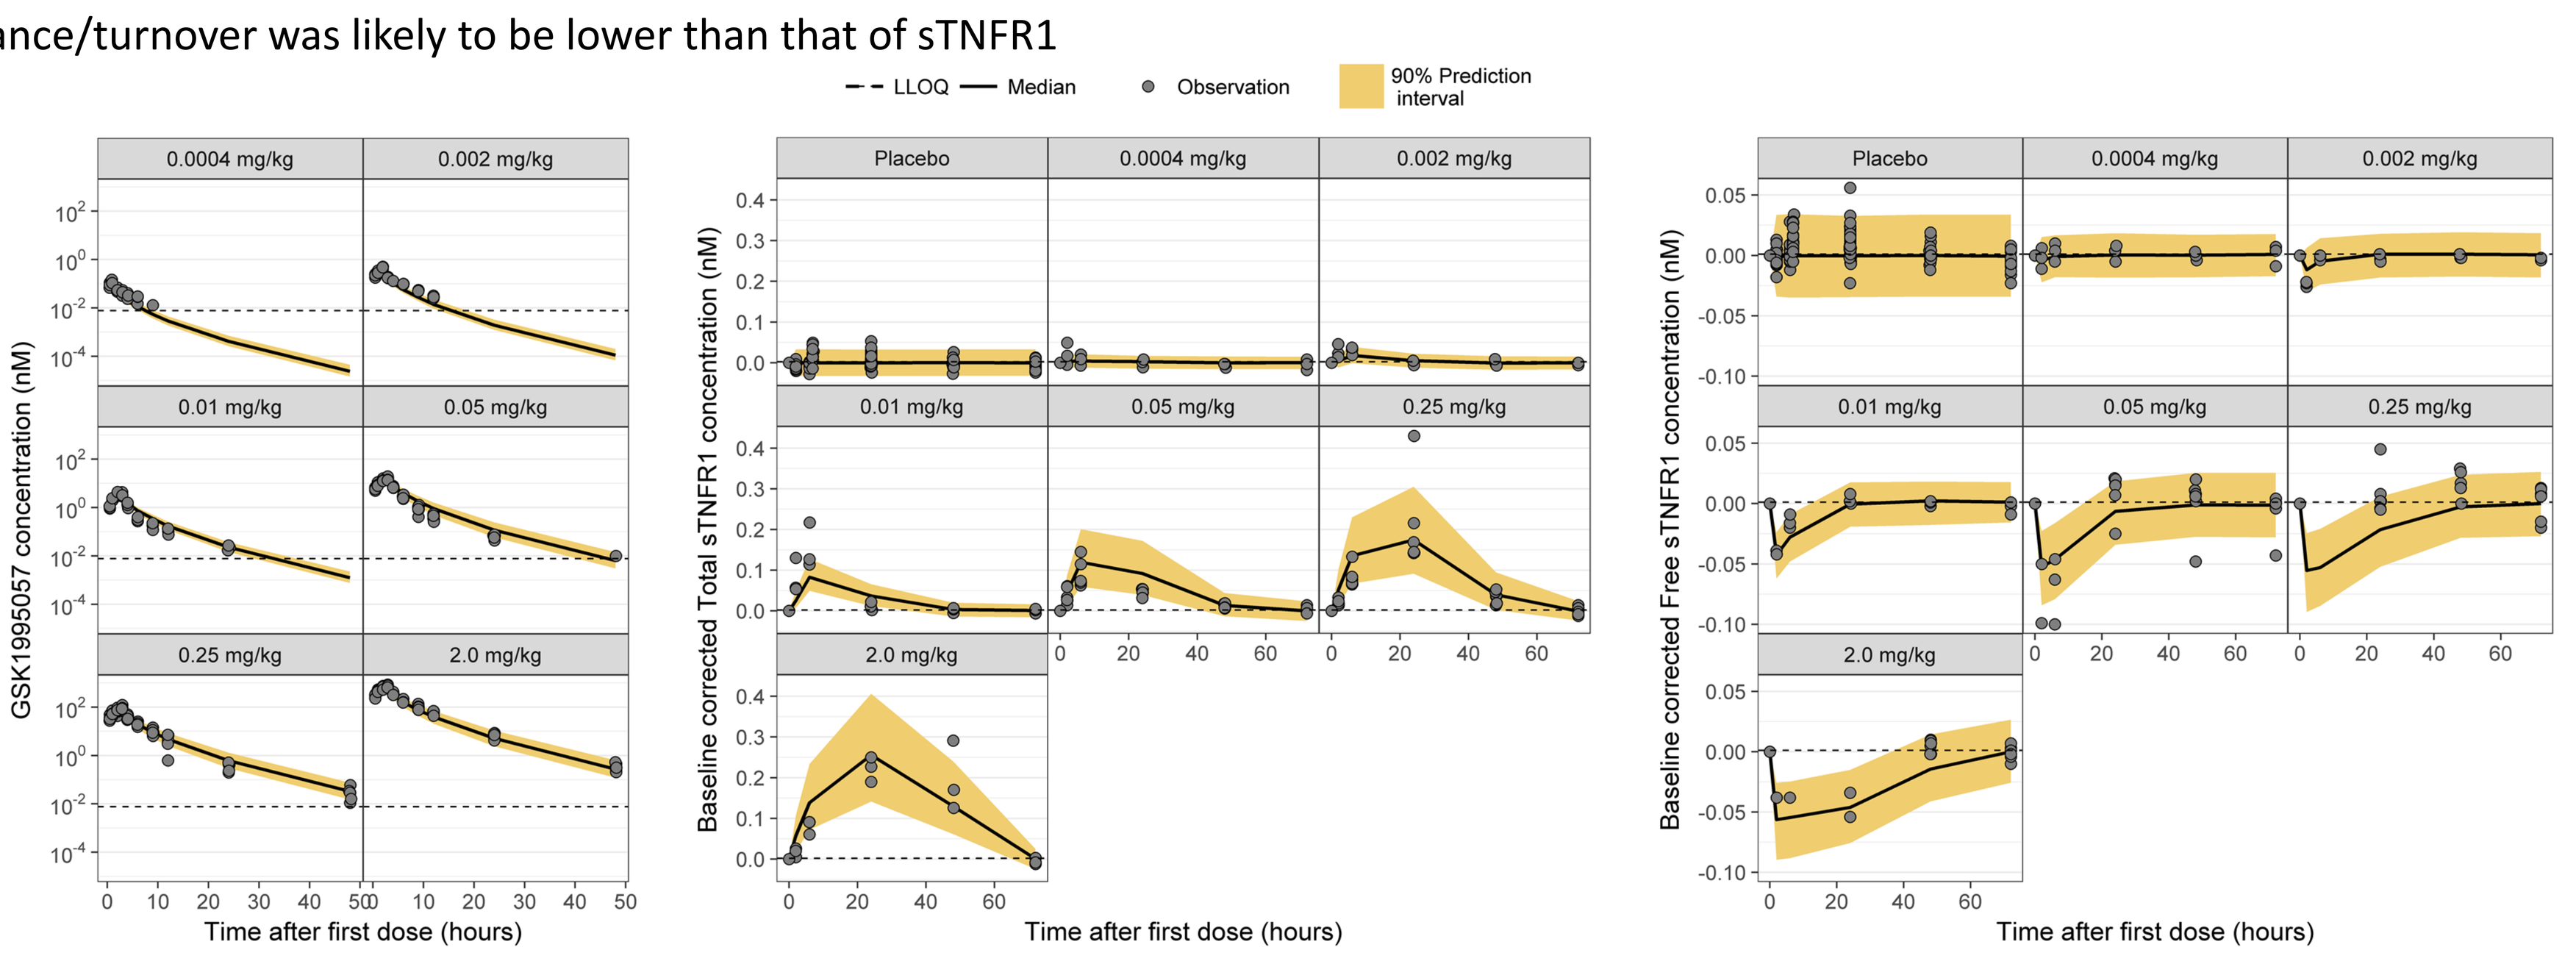
LLOQ
Median
Observation
90% Prediction interval
GSK1995057 concentration (nM)
Baseline corrected Total sTNFR1 concentration (nM)
Baseline corrected Free sTNFR1 concentration (nM)
Placebo
0.0004 mg/kg
0.002 mg/kg
0.01 mg/kg
0.05 mg/kg
0.25 mg/kg
2.0 mg/kg
Time after first dose (hours)

Figure 1 VPC of the full TMDD model fitted to the phase I on PK, free and total sTNFR1 levels

Table 1: Parameter estimates of the full TMDD model fitted to the phase I on PK, free and total sTNFR1 levels

Parameter	Value	SE	CV (%)	Parameter	Value	SE	CV (%)
Fixed Parameters (PK)				*Fixed Parameters (PD) representing binding to sTNFR1*			
CL [L/h/kg]	0.054	0.00185	3.43	B_{max} [nM]	0.0541	0.00234	4.33
V_c [L/kg]	0.0692	0.00169	2.44	K_D [nM]	0.0143 FIXED		
Q (L/h/kg)	0.0036	0.0003	7.48	K_{on} [$nM^{-1}\bullet h^{-1}$]	0.778	1.09	140
V_{p1} [L/kg]	0.0261	0.00106	4.06	K_{deg} [1/h]	0.709	0.127	17.9
Q_2 [L/h/kg]	0.0314	0.00145	4.62	K_{int} [1/h]	0.108	0.0488	45.2
V_{p2} [L/kg]= V_c [L/kg]							
Fixed Parameters (Bioavailability)							
F1 (Dose = 0.0004 mg/kg)	0.534 FIXED						
F1 (Dose = 0.002 mg/kg)	0.477 FIXED						
F1 (Dose > 0.002 mg/kg)	1 FIXED						
Random Parameters (IIV)				*Random Parameters (Residual error)*			
Ω^2 CL	0.0265	0.0038	14.3	Prop. Res. Error (057)	0.23	0.012	5.22
Ω^2 BMax	0.107	0.0503	47	Add. Res. Error (free sTNFR1)	0.0154	0.00203	13.2
				Prop. Res. Error (total sTNFR1)	0.307	0.0298	9.71

- Model was extended to capture mTNFR1 binding (Figure 2).
 (i) the synthesis and degradation of mTNFR1 (Table 2)
 (ii) the production of sTNFR as a result of shedding of mTNFR1 (Table 2) ,
 , (iii) the internalization and shedding of the dAb-mTNFR1 complex,

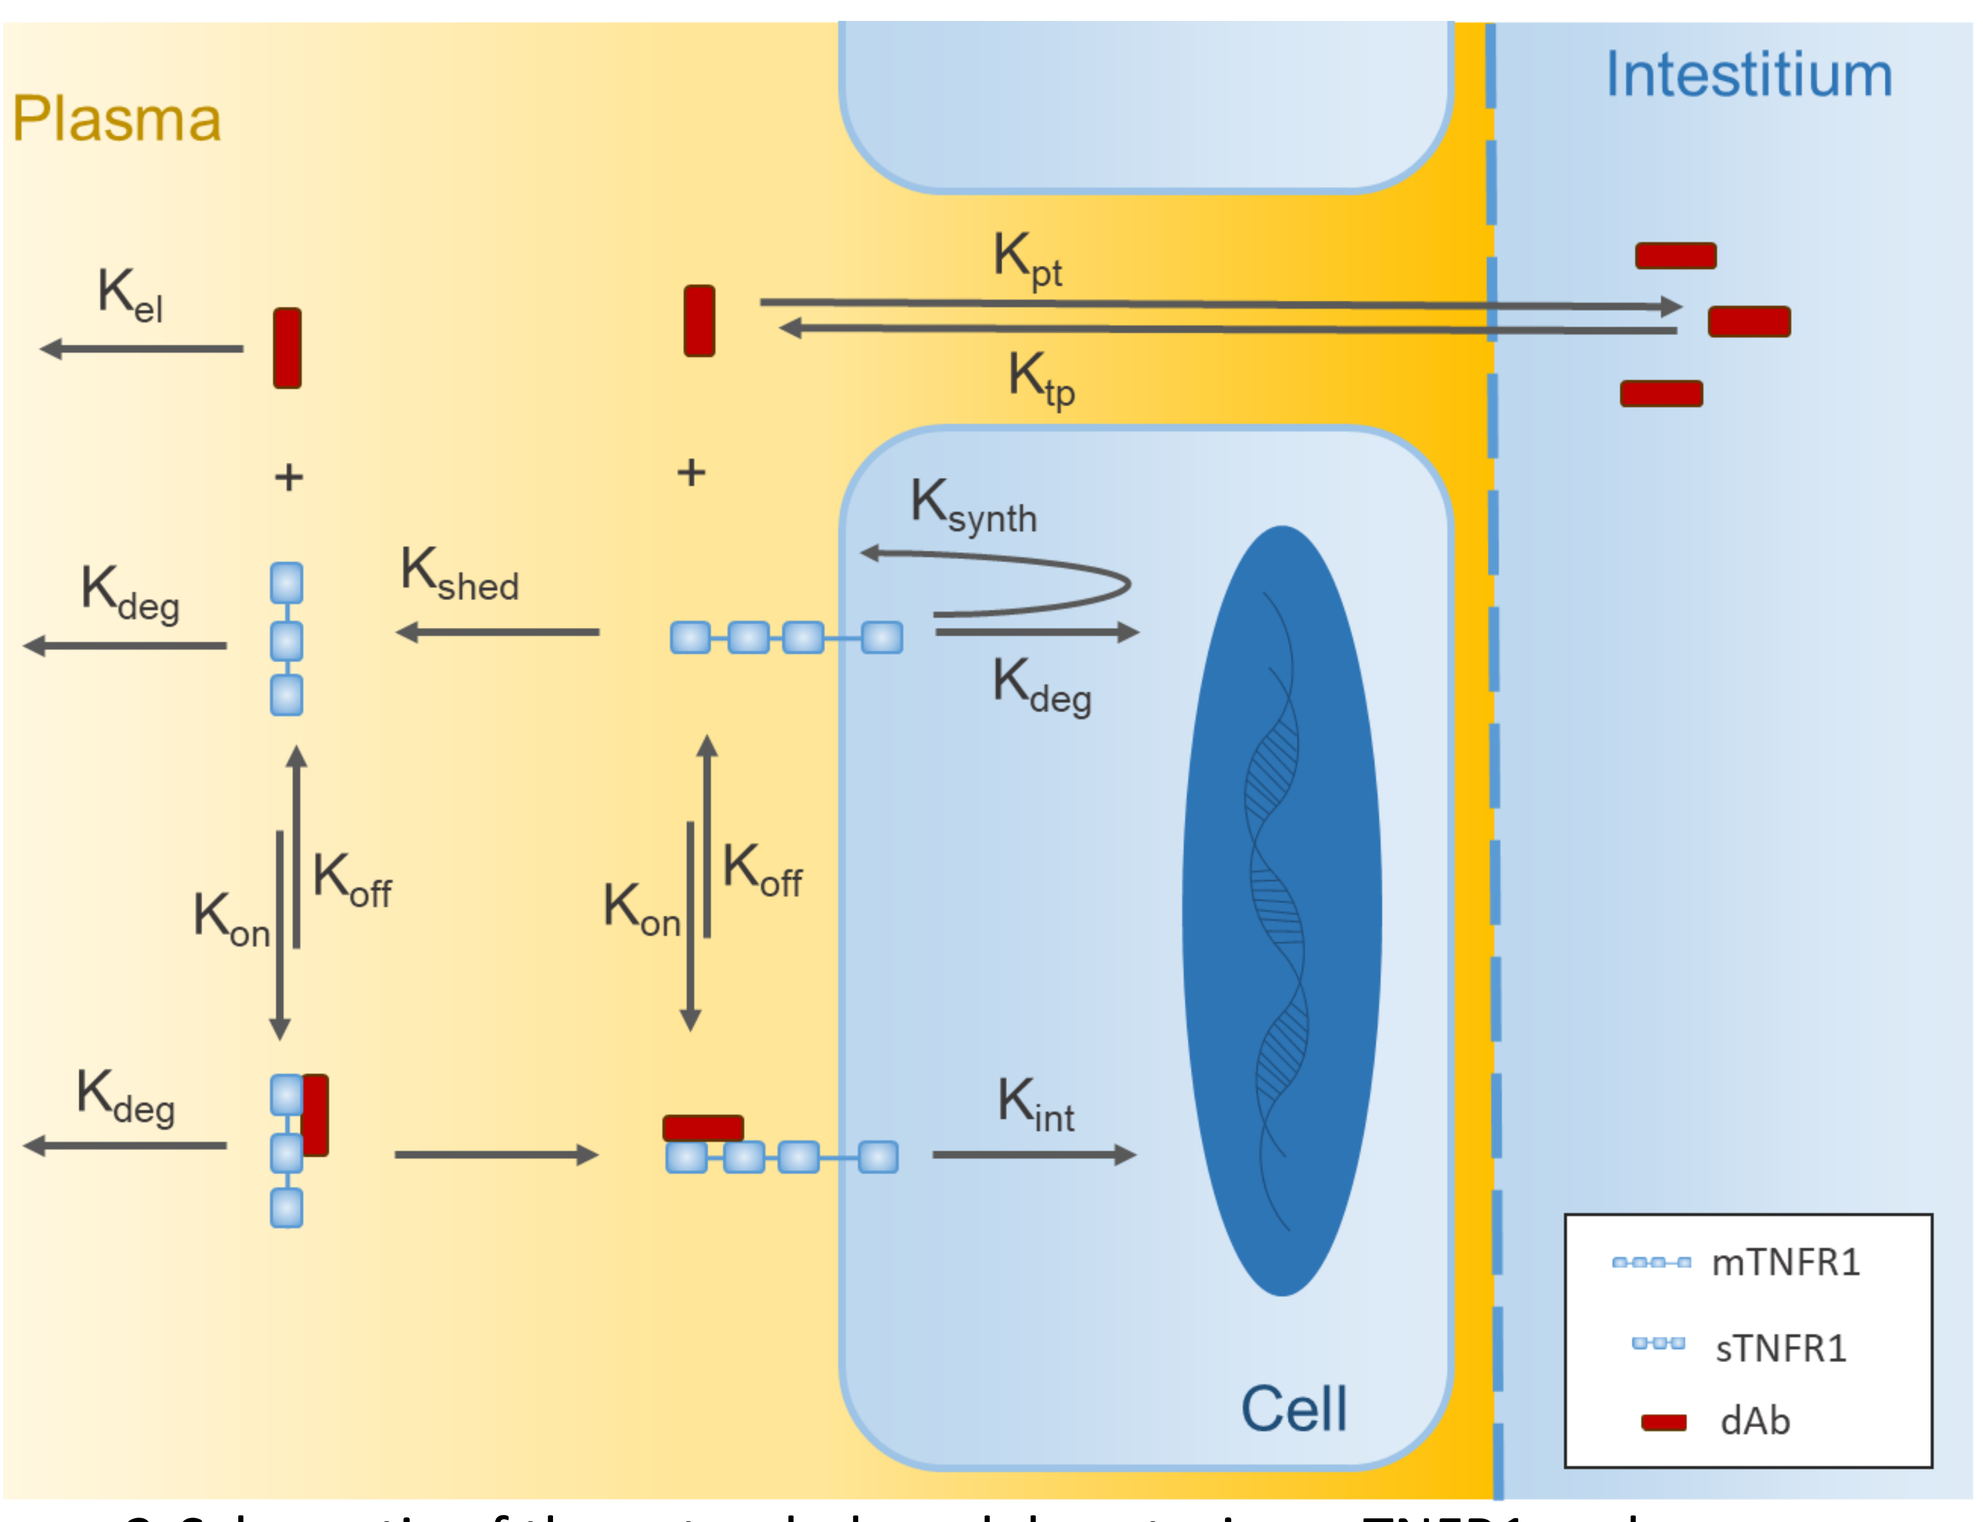
Plasma
Intestitium
Cell
K_{el}
K_{pt}
K_{tp}
K_{synth}
K_{deg}
K_{shed}
K_{on}
K_{off}
K_{int}
mTNFR1
sTNFR1
dAb

Figure 2 Schematic of the extended model capturing mTNFR1 and sTNFr1 binding

Table 2: Parameter values of the extended model to capture mTNFR1 and sTNFR1 binding based on discussions with the clinical/preclinical project teams and literature.

Parameter	Description	Value (simulation range)	Reference
sTNFR1			
T_0S [nM]	Baseline sTNFr concentration	0.04	GSK Team
KD_S [nM]	Affinity of dAb binding sTNFR	0.0143	*In-vitro* experiments
K_{on} [nM^{-1} h^{-1}]	Association rate constant of sTNFR-dAb complex formation	0.405	Estimated from clinical data
K_{deg_S} [h^{-1}]	Degradation constant of sTNFR	0.671	Estimated from clinical data
mTNFR1			
T_0M [nM]	Baseline mTNFr concentration	$K_{deg_S}/(K_{shed} \bullet T_0S)$	
KD_M [nM]	Affinity of dAb binding mTNFR	0.0143	*In-vitro* experiments
K_{on} [nM^{-1} h^{-1}]	Association rate constant of mTNFR-dAb complex formation	35.78	*In-vitro* experiments
K_{deg} [h^{-1}]	Degradation constant of mTNFR	8.3	Estimated from clinical data
K_{int} [h^{-1}]	Internalization constant of mTNFR-dAb complex	0.0873	Discussion with GSK Team
K_{shed} [h^{-1}]	Shedding constant of mTNFr	0.231	Discussion with GSK Team
K_{syn_M} [nM h^{-1}]	Synthesis rate of mTNFR	$(K_{int_M} + K_{shed}) \bullet T_0M$	

Table 3: Parameter values of the extended model to capture GSK-057-ADA complex formation and subsequent mTNFr1 crosslinking based on discussions with the clinical/preclinical project teams and literature.

Parameter	Description	Value (simulation range)	Reference
ADA			
T_0ADA [nM]	Baseline ADA concentration	1 (10^{-4} - 10^4)	Discussion with GSK Team
K_{synth_ADA} [h^{-1}]	Synthesis rate of ADA	$T_0ADA \bullet K_{deg_ADA}$	
K_{deg_ADA} [h^{-1}]	Degradation rate constant of ADA	0.00138	IgG1 degradation rate constant
KD_{ADA} [nM]	Affinity of ADA binding dAb	0.165 (0.000165 - 16.5)	Discussion with GSK Team
K_{on_ADA} [nM^{-1} h^{-1}]	Association rate constant ADA-dAb and ADA-$(dAb)_2$ complex	36	Diffusion limit
K_{off_ADA} [h^{-1}]	Association rate constant ADA-dAb and ADA-$(dAb)_2$ complex	$KD_{ADA} \bullet K_{on_ADA}$	
KD_{MA} [nM]	Affinity of ADA-dAb and ADA-$(dAb)_2$ complex to bind to mTNFR	0.165 (0.000165 - 16.5)	Discussion with GSK Team
K_{on_MA} [nM^{-1} h^{-1}]	Association rate constant of ADA-dAb and ADA-$(dAb)_2$ complex to bind to mTNFR	K_{on}	Discussion with GSK Team
K_{off_MA} [h^{-1}]	Dissociation rate constant of ADA-dAb-mTNFR and ADA-$(dAb\text{-}mTNFR)_2$ complex	$KD_{MA} \bullet K_{on_MA}$	
KD_{SA} [nM]	Dissociation rate constant of ADA-dAb-sTNFR and ADA-$(dAb\text{-}mTNFR)_2$ complex	KD_{MA}	Discussion with GSK Team
K_{on_SA} [nM^{-1} h^{-1}]	Association rate constant of ADA-dAb-sTNFR and ADA-$(dAb\text{-}sTNFR)_2$ complex	K_{on}	Discussion with GSK Team
K_{off_SA} [h^{-1}]	Dissociation rate constant of ADA-dAb-sTNFR and ADA-$(dAb\text{-}sTNFR)_2$ complex	$KD_{SA} \bullet K_{on_SA}$	

- Model was extended to GSK-057-ADA complex formation and subsequent mTNFr1 crosslinking (Figure 3) including:
 (iv) dAb-ADA binding (Figure 3) and
 (v) binding of the dAb-ADA complex to mTNFR1 (Figure 3).

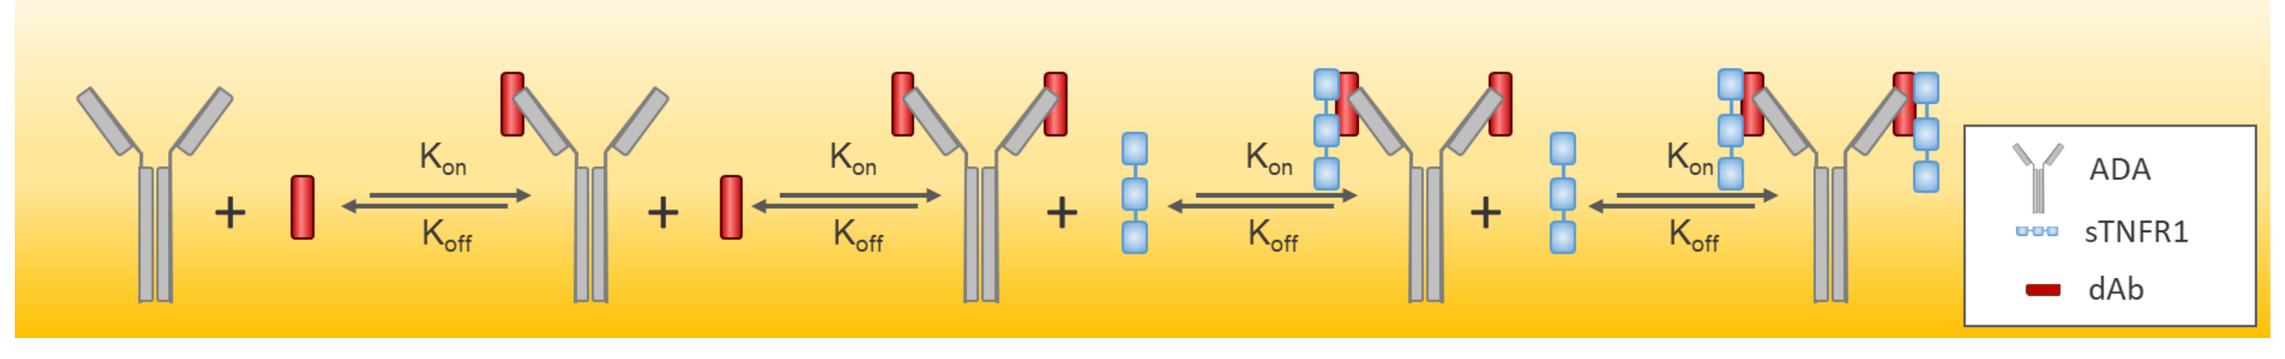
K_{on}
K_{off}
ADA
sTNFR1
dAb

Figure 3 GSK-057-ADA complex formation and subsequent sTNFR1 /mTNFr1 crosslinking. (Examplified through sTNFR1)

- Thus the model reflected all relevant binding processes involved in the activation of the mTNFR1 by dAbs in individuals with pre-existing ADAs (Figure 4) [1].

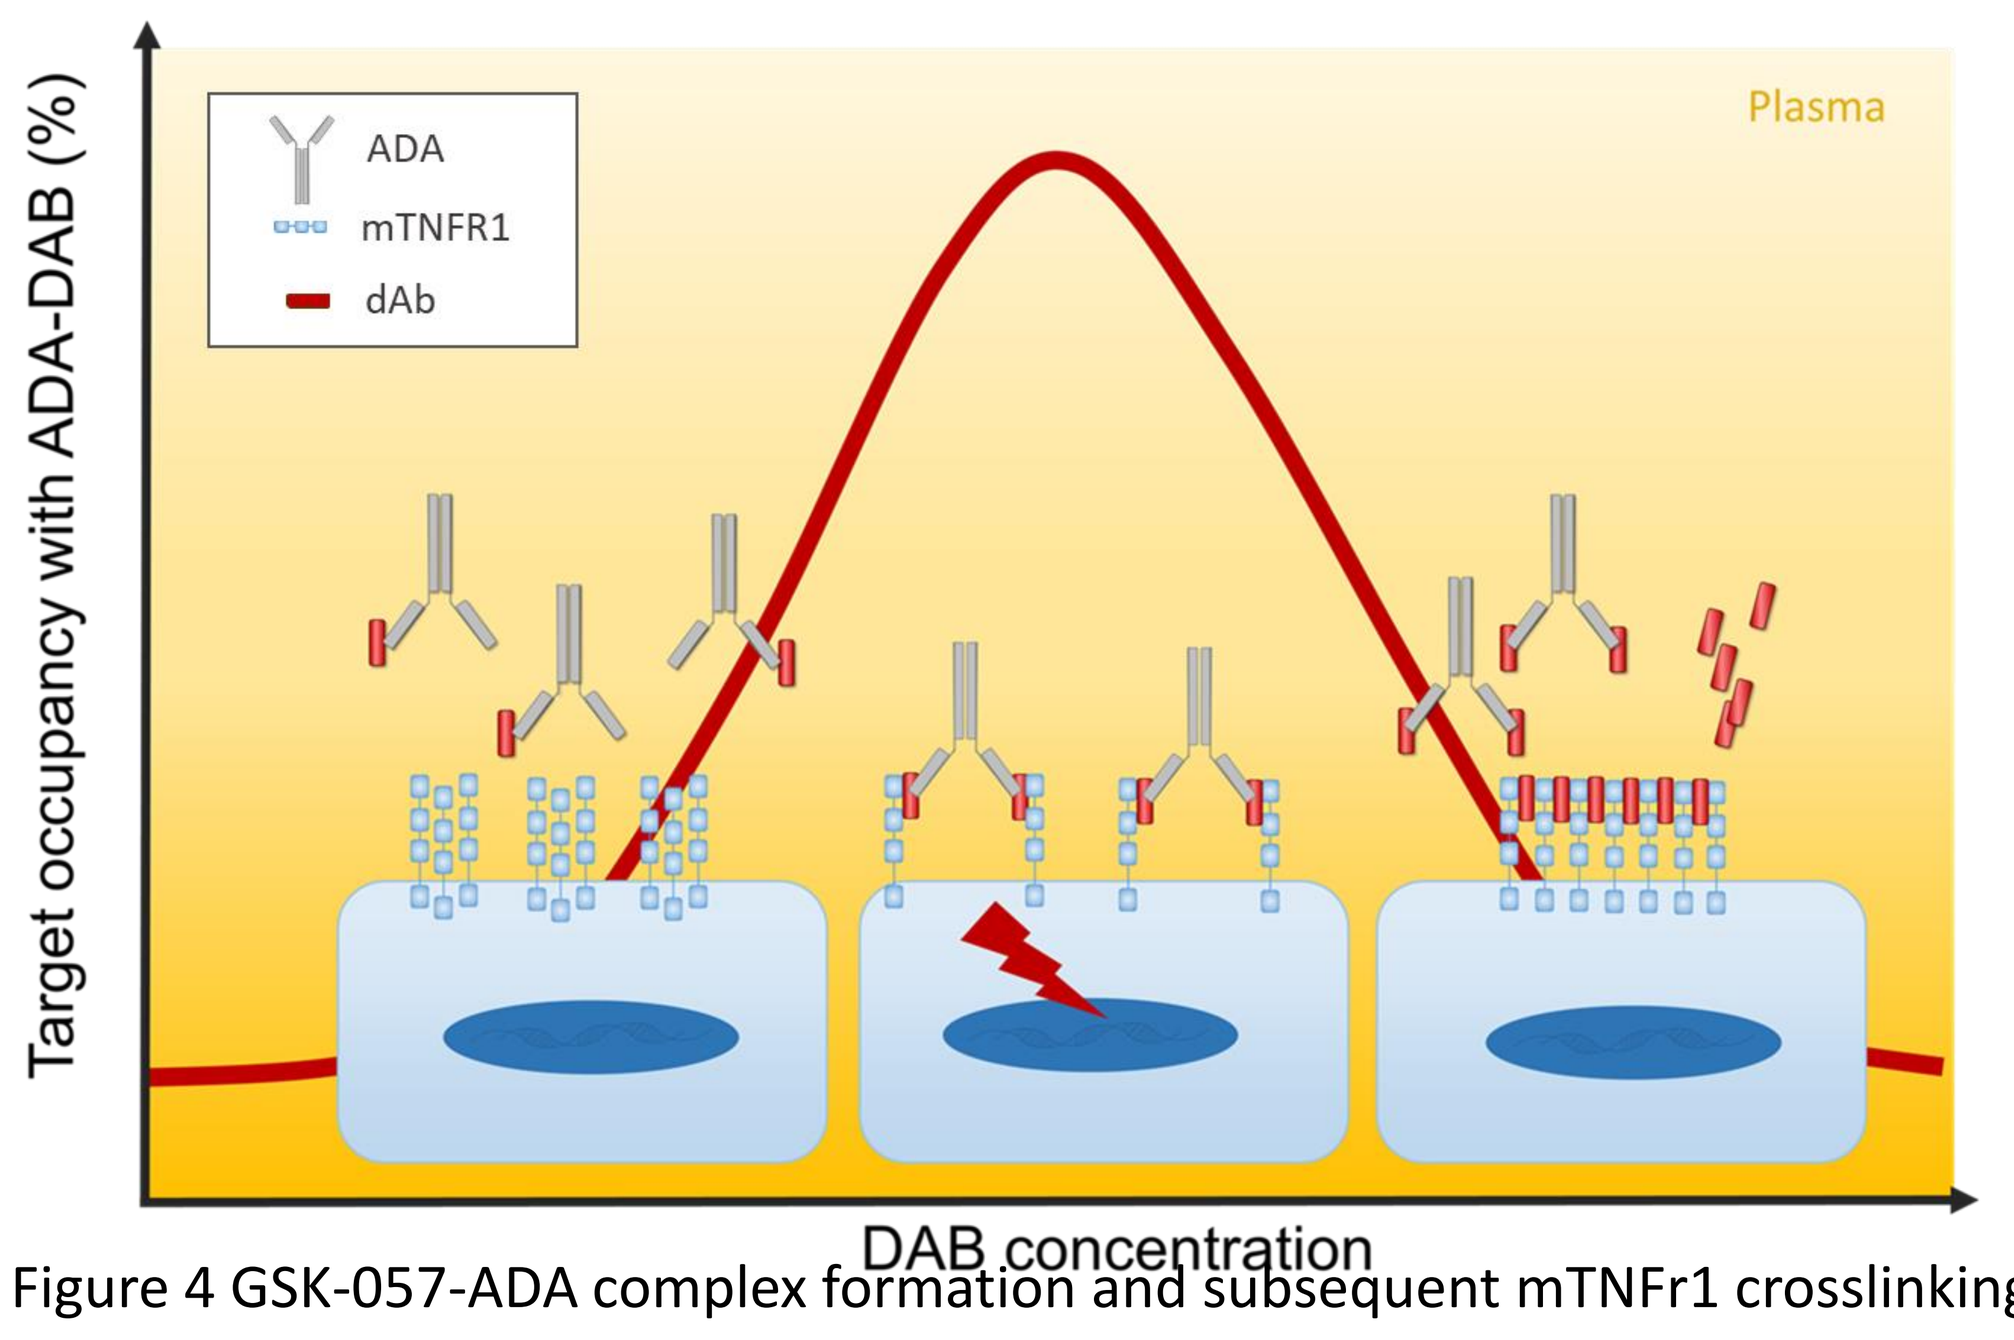
ADA
mTNFR1
dAb
Plasma
Target occupancy with ADA-DAB (%)
DAB concentration

Figure 4 GSK-057-ADA complex formation and subsequent mTNFr1 crosslinking.

- A sensitivity analysis further strengthened the parameter space (Table 3).
- The pre-existing ADA were shown to be approximately 10-fold lower in affinity to GSK-057 than it is to TNFr1 and the maximum ADA baseline estimated to be approximately 0.1 nM (Figure 5).

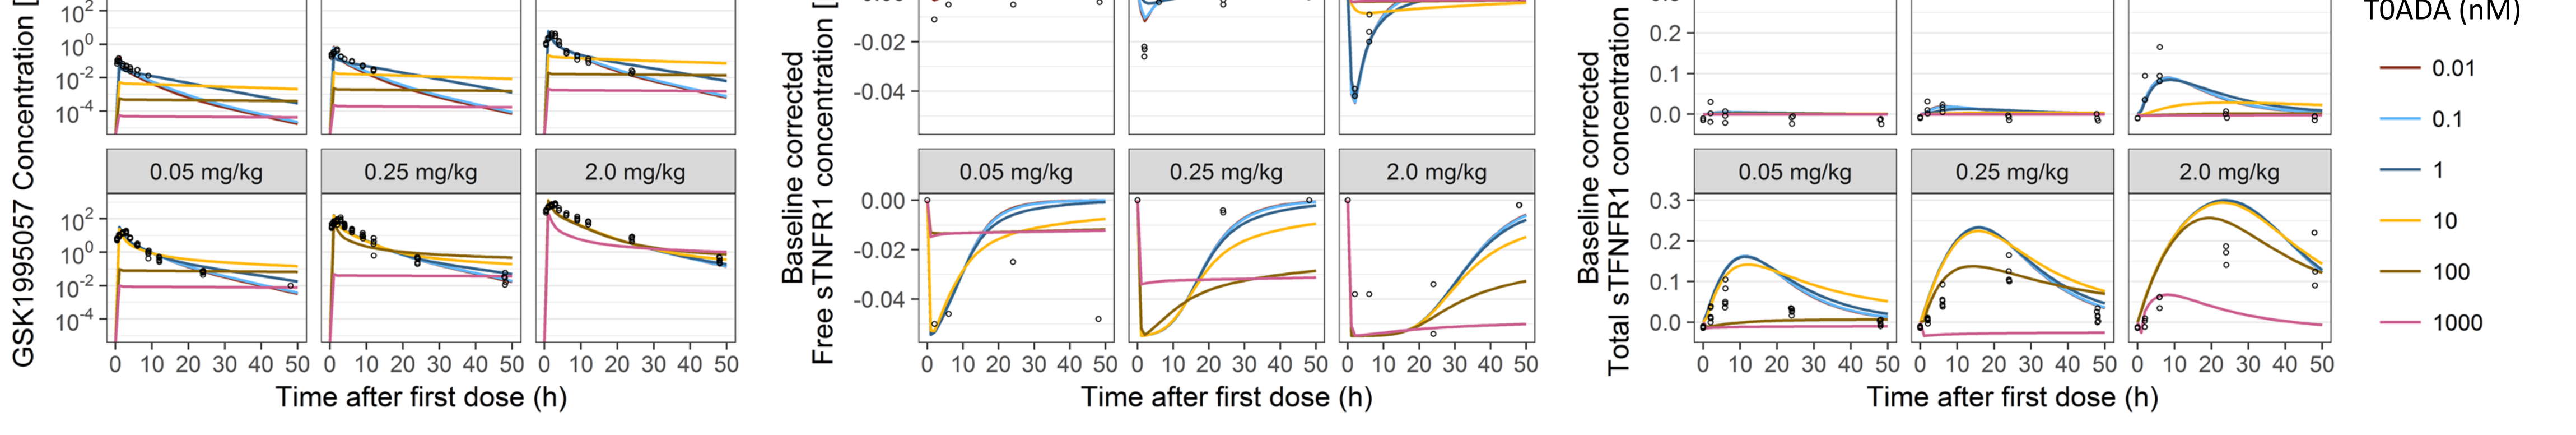
GSK1995057 Concentration [nM]
Baseline corrected Free sTNFR1 concentration [nM]
Baseline corrected Total sTFNFR1 concentration [nM]
0.0004 mg/kg
0.002 mg/kg
0.01 mg/kg
0.05 mg/kg
0.25 mg/kg
2.0 mg/kg
Time after first dose (h)
T0ADA (nM)
0.01
0.1
1
10
100
1000

Figure 5 Sensitivity analysis to evaluate maximum ADA baseline.

- Simulation results were discussed with the clinical/preclinical project teams regularly, thereby challenging the model against the available knowledge on the system.
- Overall, the simulations revealed a bell-shaped binding curve for the dAb-ADA-mTNFR1 complex, thereby demonstrating the reversible nature of the agonistic effects when increasing the dose of the dAb (Figure 6). As expected, the target coverage reduced with a deterioration of affinity. Furthermore, the simulations provided clear insights into the parameters, which governed the reversible nature of the agonist effects.

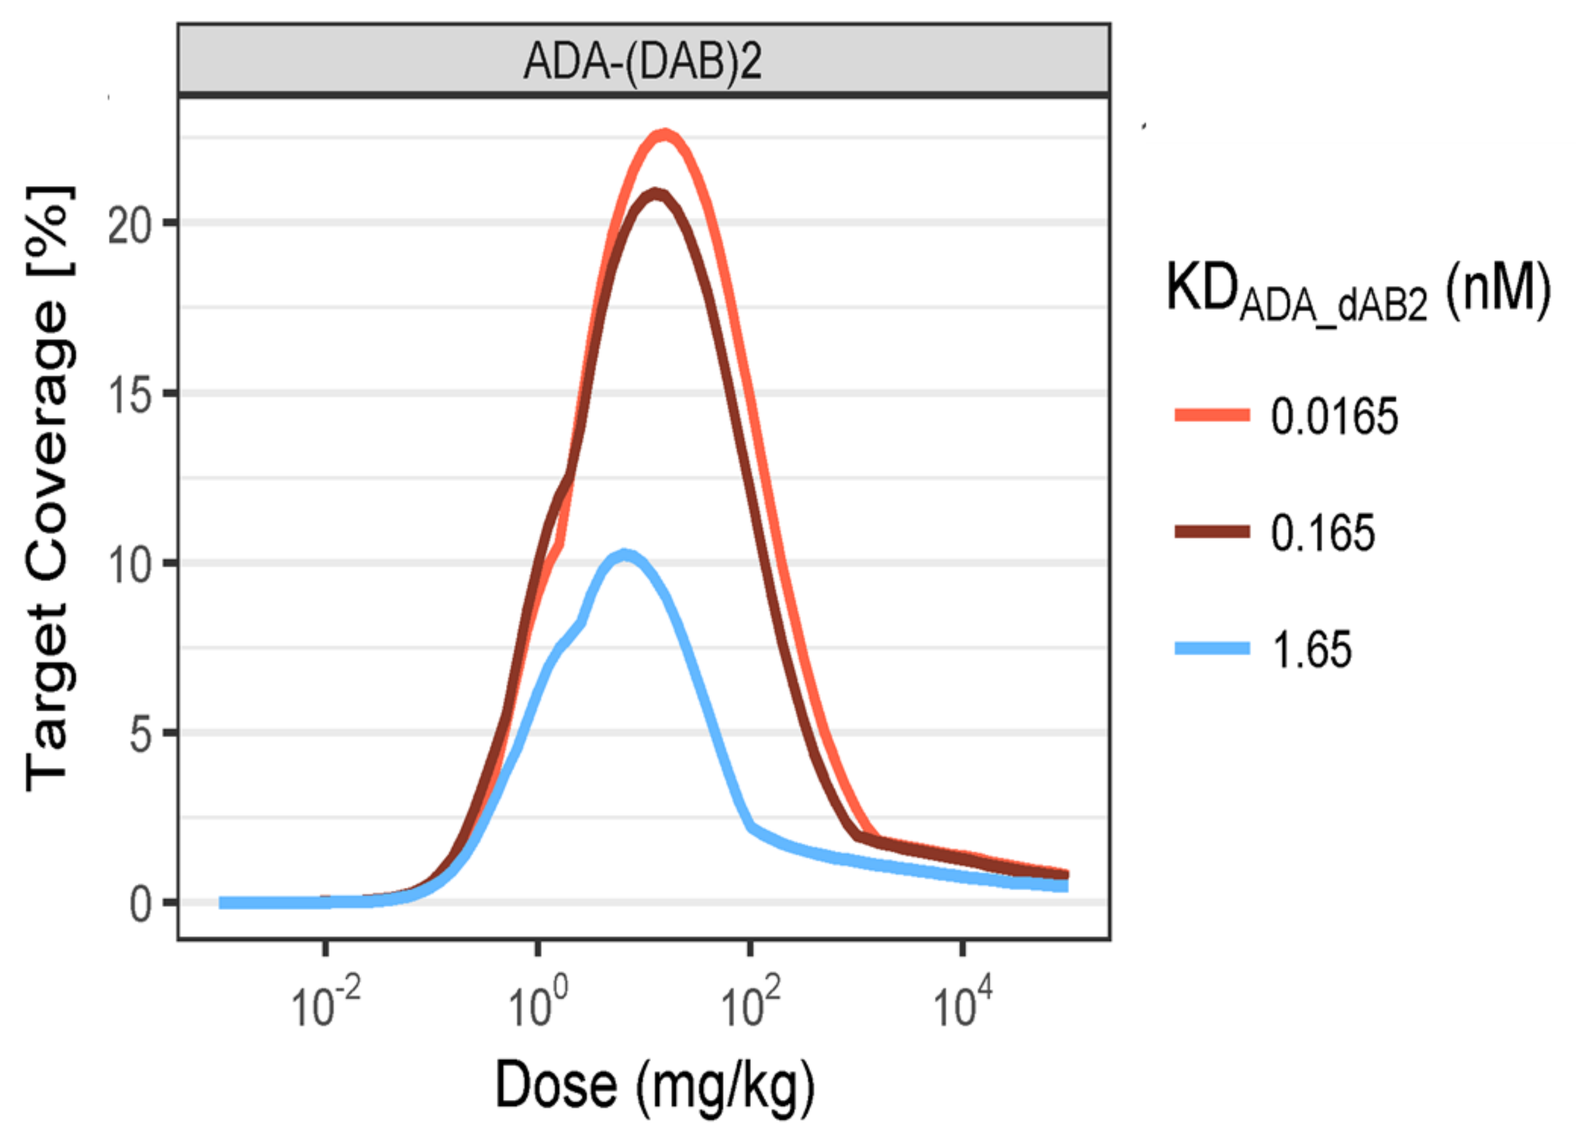
ADA-(DAB)2
Target Coverage [%]
Dose (mg/kg)
KD_{ADA_dAB2} (nM)
0.0165
0.165
1.65

Figure 6 Bell-shaped binding curve for the dAb-ADA-mTNFR1 complex.

Conclusion

Simulations with the developed mathematical model showed that a reversible agonist effect is indeed to be expected based the underlying biological binding principles and the parameters, thereby filling the gap between hypothesis and reality.

[1] Holland MC, Wurthner JU, Morley PJ, Birchler MA, Lambert J, Albayaty M, et al. Autoantibodies to variable heavy (VH) chain Ig sequences in humans impact the safety and clinical pharmacology of a VH domain antibody antagonist of TNF-α receptor 1. Journal of Clinical Immunology. 2013; 33(7):1192–1203.

[2] Wilson MR, Wakabayashi K, Bertok S, Oakley CM, Patel BV, O'Dea KP, et al. Inhibition of TNF receptor p55 by a domain antibody attenuates the initial phase of acid-induced lung injury in mice. Frontiers in Immunology. 2017; 8: 1–12.

GSK funded and was sponsor of study 110951 "Safety Study of GSK1995057 Given as Single and Repeat Intravenous Doses in Healthy Subjects" (NCT01476046)).
DF is a GSK employee and shareholder. TS and AK are employees of LAP&P BV.