Optimising Phase 1 oncology dosing schedule of an ATR inhibitor in real time using a model informed approach to predict myelosuppression

S. Y. Amy Cheung[1], Alienor Berges[1], James W. T. Yates[1], Nuria Buil Bruna[1], Graham Ross[1], Simon Smith[1], Brunella Felicetti[1], Musaddiq Khan[1], Christine Stephens[1], Jean-Charles Soria[2], Kevin Harrington[3], Magnus Dillon[3] and Simon J Hollingsworth[1]

Innovative Medicines and Early Development, AstraZeneca UK[1], Institut Gustave Roussy, Paris, France[2], The Institute of Cancer Research/The Royal Marsden Hospital, London[3]

The ROYAL MARSDEN NHS Foundation Trust

GUSTAVE ROUSSY CANCER CAMPUS GRAND PARIS

Abstract

Oncology phase 1 development is crucial to establish the recommended dose and schedule of novel agents and their combinations for phase 2 trials through dose escalation. Toxicities are important endpoints for phase 1 trials involving cytotoxic agents, it is essential to avoid exposing too many patients to sub-therapeutic doses while preserving safety, maintaining rapid accrual and if possible detect early signals of efficacy and toxicity. Here we utilize emerging data from the AZD6738 phase 1 program and model the relationship between drug exposure and patient safety of a novel oncology compound AZD6738. A quantitative understanding of duration and severity of myelosuppression is vital for dose-regimen optimization in this phase [1].

Introduction

- AZD6738 is a potent, highly specific ATR kinase inhibitor being tested in phase 1 clinical trials in patients with solid malignancies as monotherapy and in combination:
 - ➢ An AZ sponsored phase 1 study with a modular protocol:
 - ❖ Module 1, combining AZD6738 with chemotherapy, carboplatin
 - ❖ Module 2, combining with a PARP-1 inhibitor olaparib.
 - ➢ An externally sponsored research (ESR) phase 1 study (PATRIOT) to access AZD6738 as a single agent and in combination with palliative radiation therapy.
- Thrombocytopenia and neutropenia are known adverse events with carboplatin monotherapy, hence the understanding of the relationship between AZD6738, carboplatin dose and exposure to platelet or neutrophil nadir is essential for selection of tolerated dose and schedule in combination.
- As part of a model informed drug development [2], a PK-safety modelling approach was applied by integrating data across phase 1 studies to support dose-regimen selection.

Methods

1. Data

Emerging plasma concentration (PK), absolute platelet count (APC) and absolute neutrophil count (ANC) data from 2 phase 1 studies of AZD6738 dosed alone or in combination intermittent doses with carboplatin and olaparib were used.

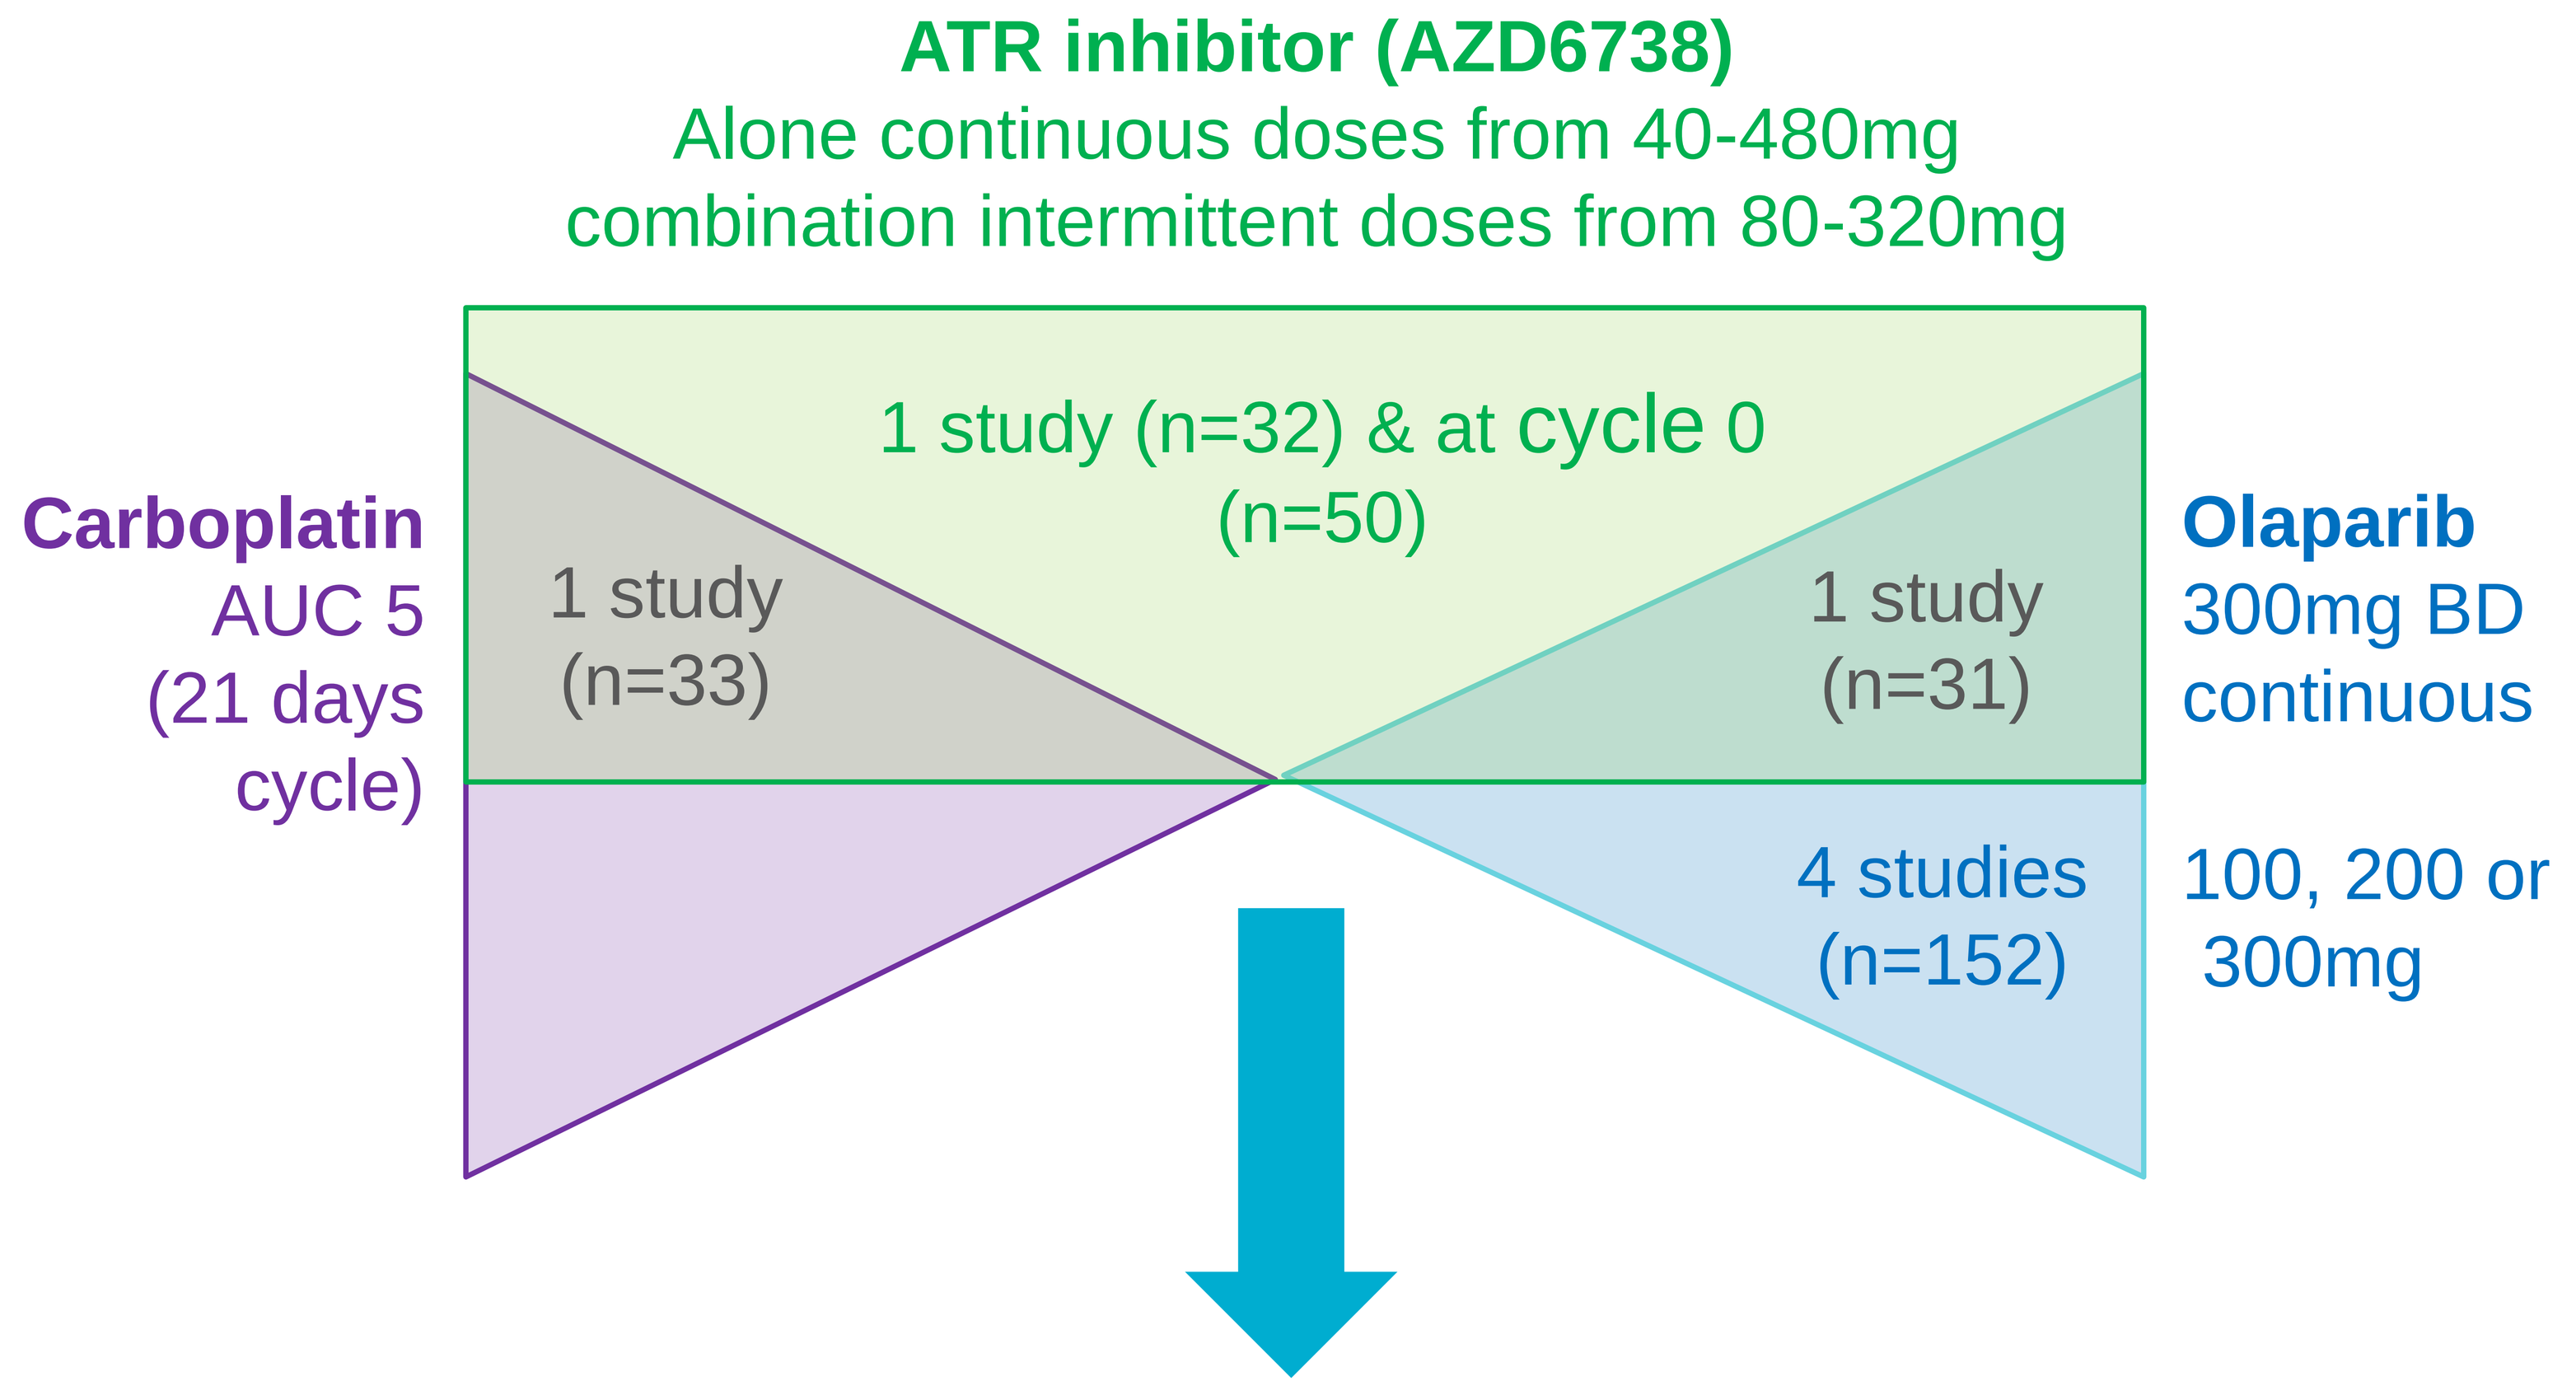
ATR inhibitor (AZD6738)
Alone continuous doses from 40-480mg
combination intermittent doses from 80-320mg
1 study (n=32) & at cycle 0
(n=50)
Carboplatin
AUC 5
(21 days cycle)
1 study
(n=33)
1 study
(n=31)
Olaparib
300mg BD continuous
4 studies
(n=152)
100, 200 or 300mg

2. Models

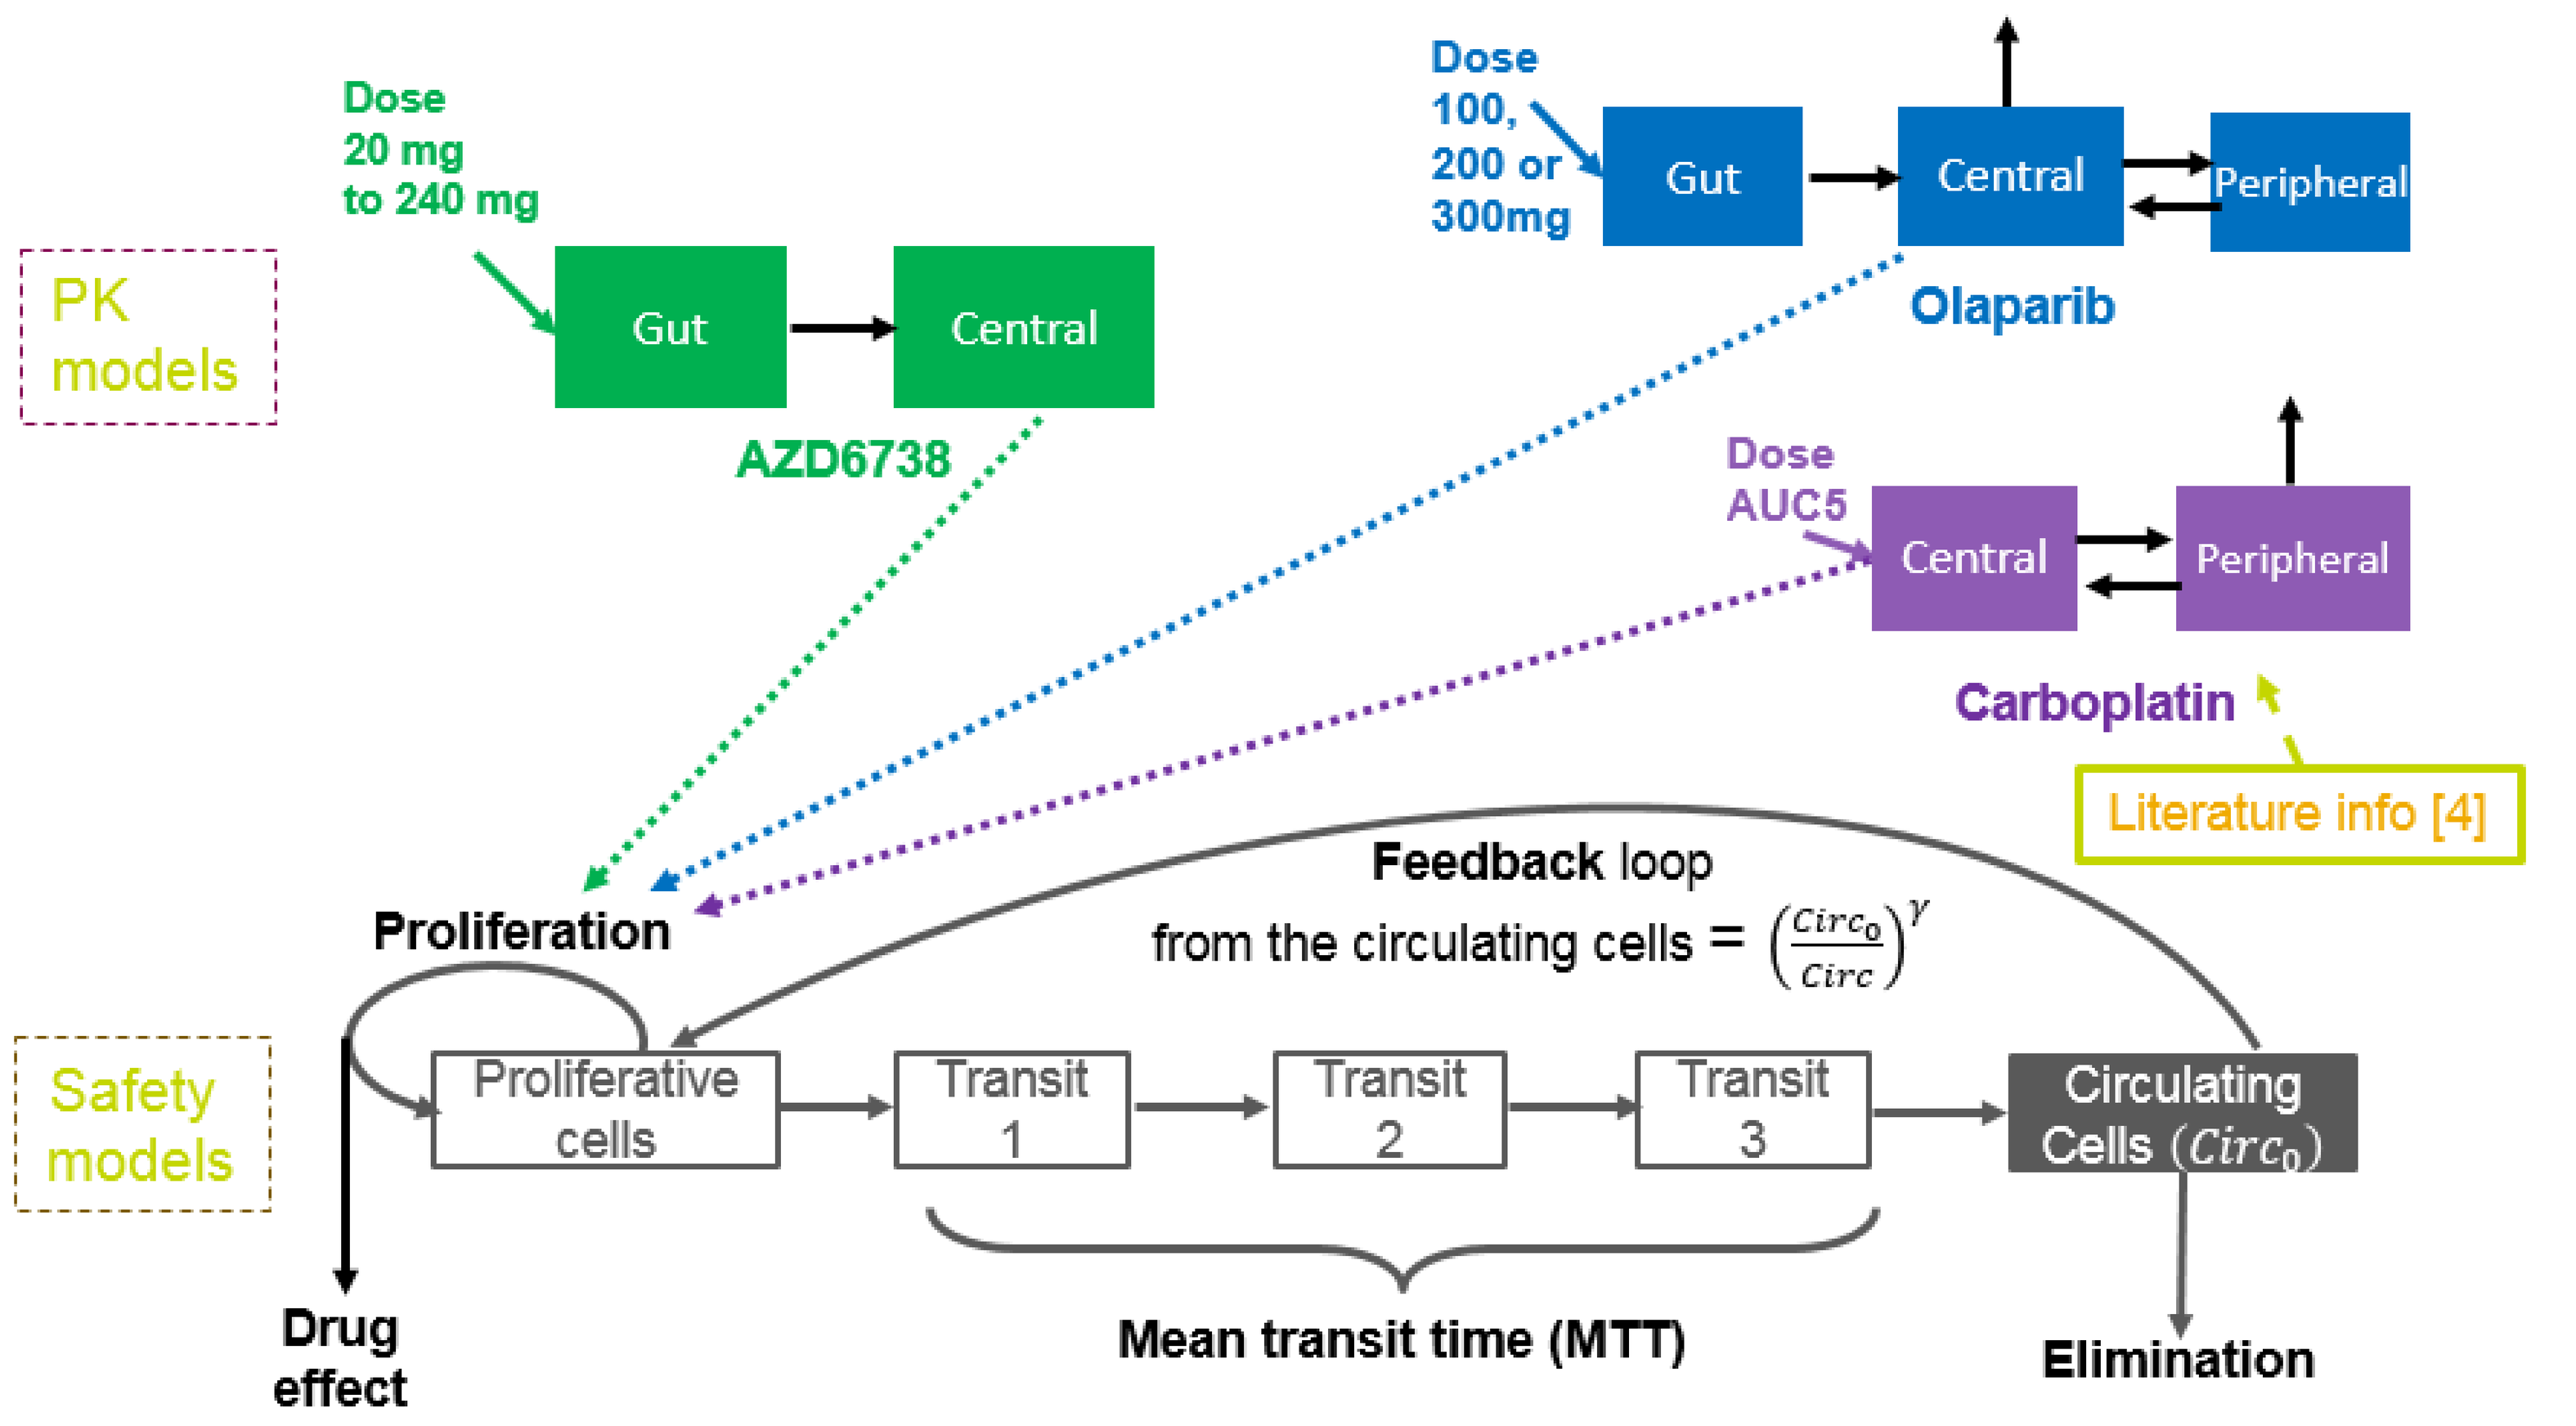
PK models
Dose 20 mg to 240 mg
Gut
Central
AZD6738
Dose 100, 200 or 300mg
Gut
Central
Peripheral
Olaparib
Dose AUC5
Central
Peripheral
Carboplatin
Literature info [4]
Feedback loop
from the circulating cells = $\left(\frac{Circ_0}{Circ}\right)^{\gamma}$
Proliferation
Safety models
Proliferative cells
Transit 1
Transit 2
Transit 3
Circulating Cells ($Circ_0$)
Drug effect
Mean transit time (MTT)
Elimination

The PK-safety modelling [3] was performed sequentially using FOCEI in NONMEM 7.3 and PSN. The myelosuppression model [3] describes the baseline circulating count, a linear relationship between drug concentration and reduced proliferation in the bone marrow precursor cell population, a mean transit time (MTT) for the delay before reduction is seen in circulating cell counts and homeostasis increasing precursor proliferation to return cell counts to baseline combination effect was tested by an additional effect when both drugs were present. Simulations of the model in the software R were used to explore dose and schedule options.

Results

A

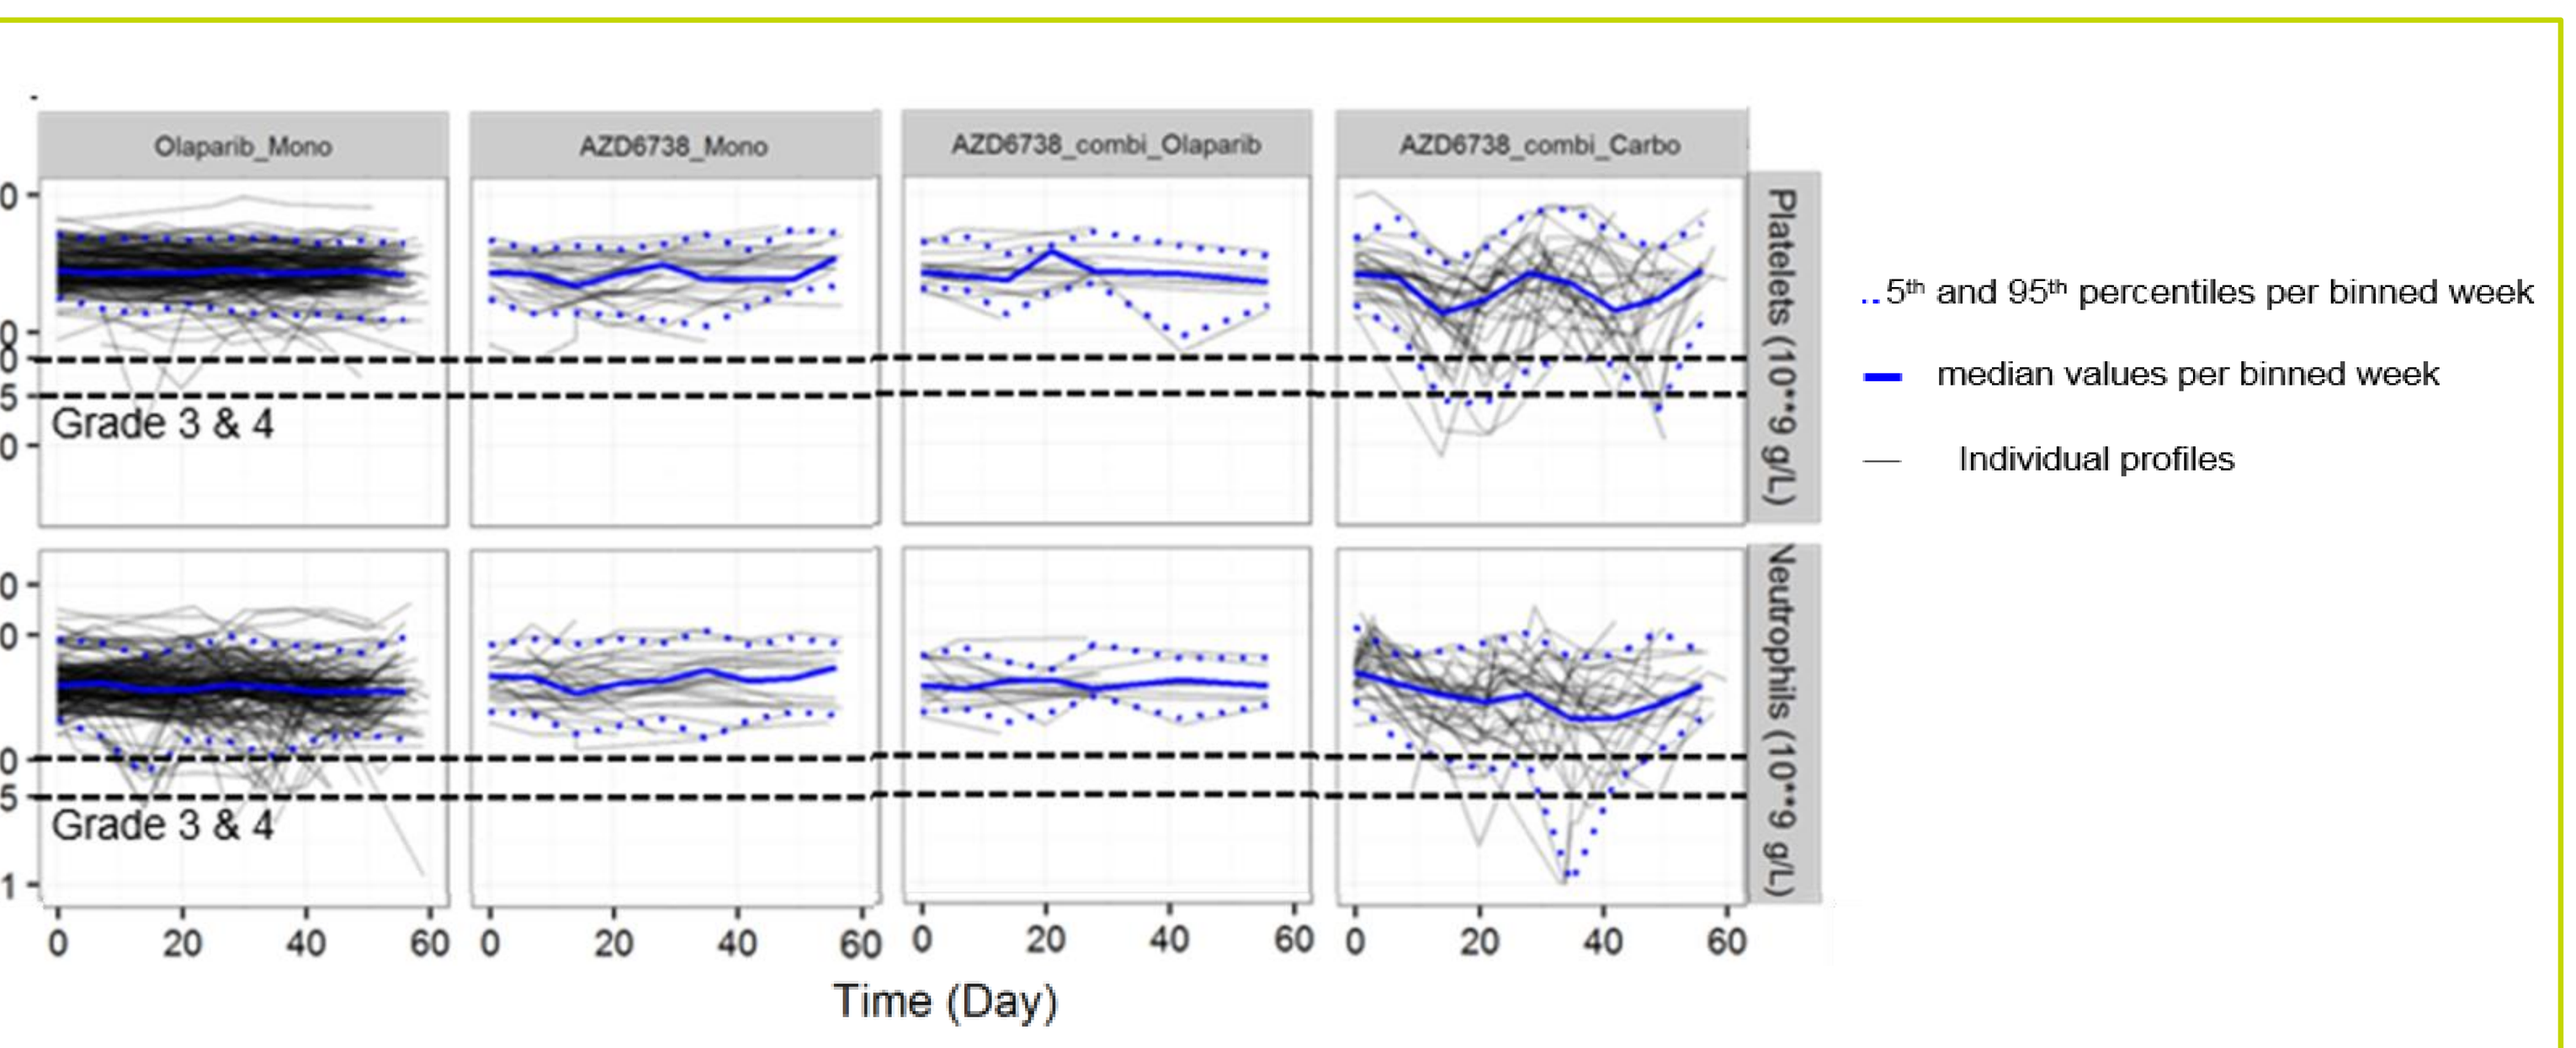
Olaparib_Mono
AZD6738_Mono
AZD6738_combi_Olaparib
AZD6738_combi_Carbo
Platelets (10**9 g/L)
Neutrophils (10**9 g/L)
Grade 3 & 4
Time (Day)
5th and 95th percentiles per binned week
median values per binned week
Individual profiles

Summary of observed blood count changes observed in patients.

B

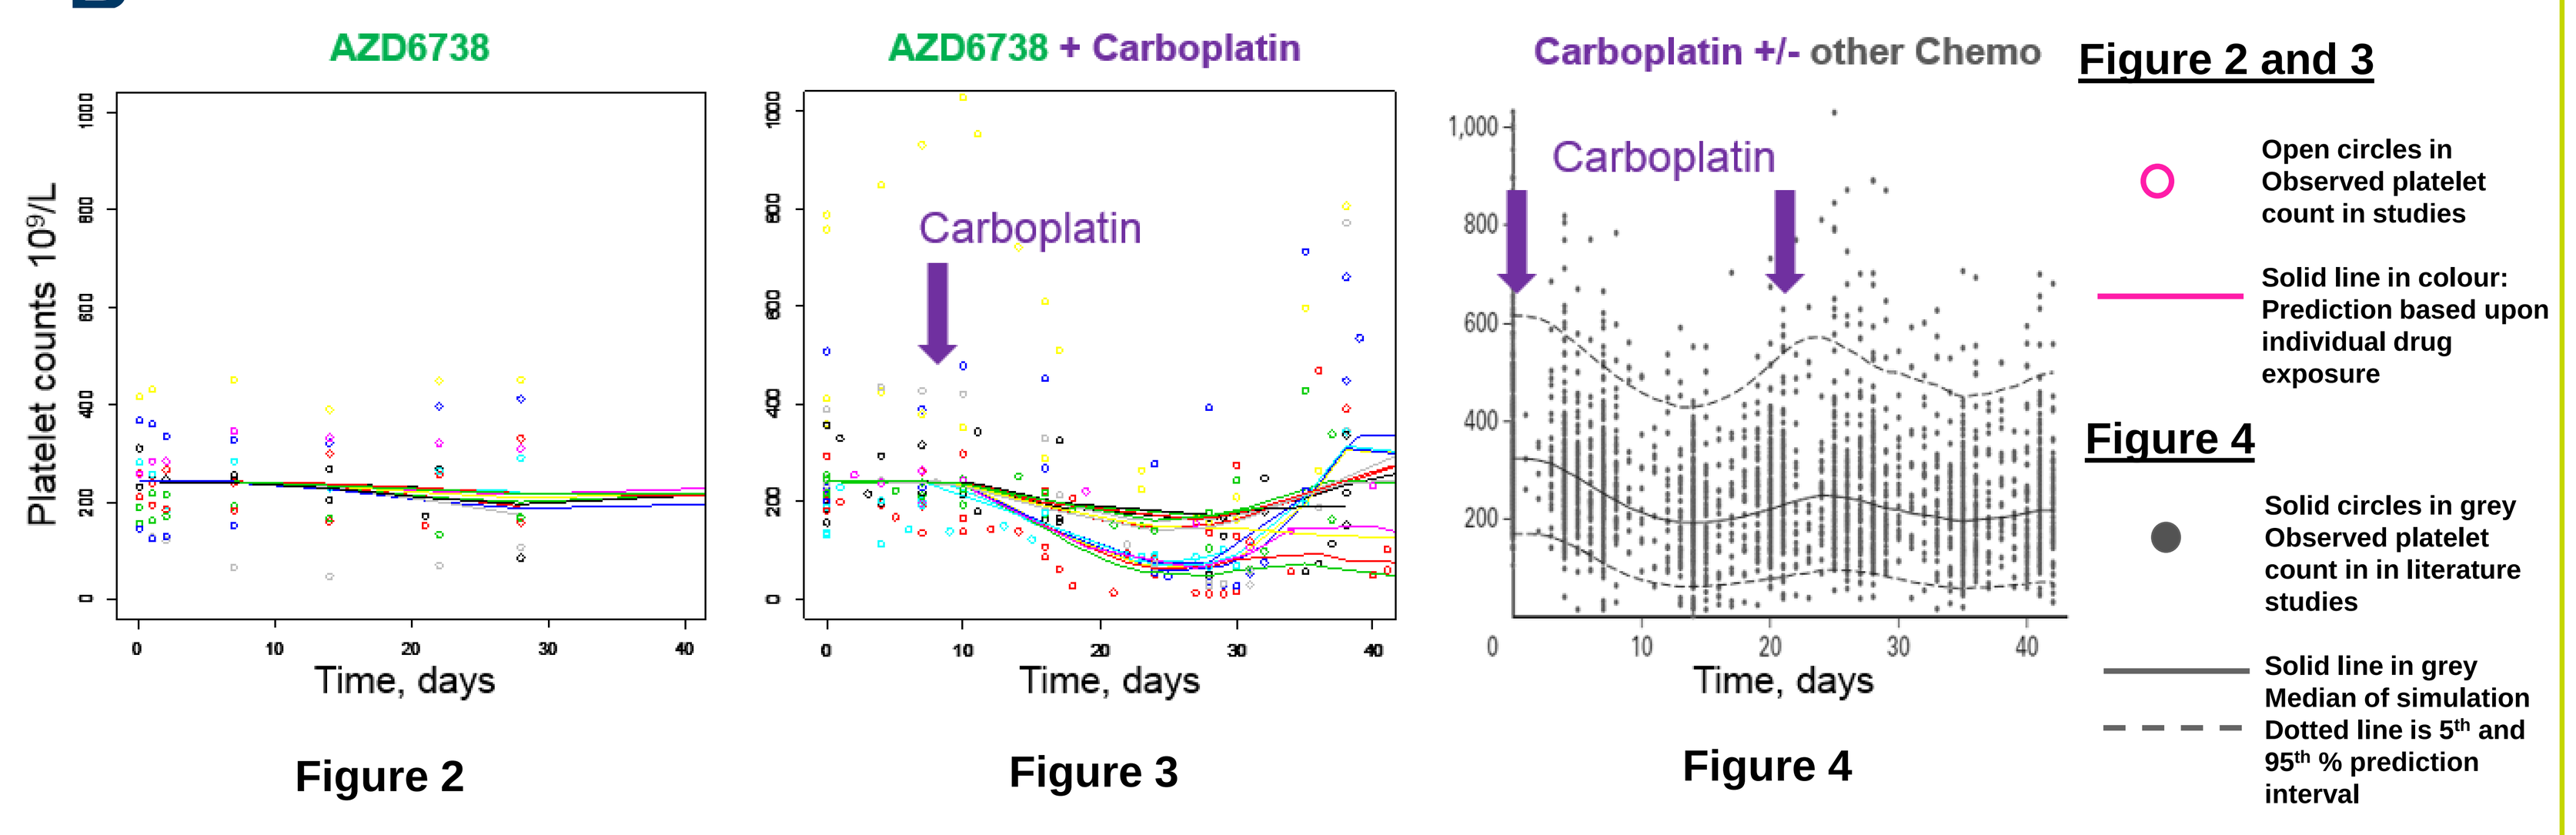
AZD6738
AZD6738 + Carboplatin
Carboplatin +/- other Chemo
Platelet counts 10^9/L
Carboplatin
Time, days
Figure 2
Figure 3
Figure 4
Figure 2 and 3
Open circles in Observed platelet count in studies
Solid line in colour: Prediction based upon individual drug exposure
Figure 4
Solid circles in grey Observed platelet count in in literature studies
Solid line in grey Median of simulation Dotted line is 5th and 95th % prediction interval

Predictions, platelet model & carboplatin combination example.
Good data description by the model, following AZD6738 alone and in combination with carboplatin AUC5 every 3 weeks and olaparib 300mg twice daily continuous dosing. A comparison with the literature is shown [4].

C

PD variable	Mean values	Historical data [4]
Baseline level (10^9/L, $Cric_0$)	235.6	332 to 358
Mean transit time (h, MTT)	208.5	203 to 245
Feedback (γ)	0.259	0.5 to 0.6
$slope_{Carboplatin}$ (mL/ng)	6.61×10^{-4}	5×10^{-4} to 6.58×10^{-4}
$slope_{AZD6738}$ (mL/ng)	4.35×10^{-6}	NA

Parameter estimation, platelet model & carboplatin combination example:
AZD6738 alone showed a minimal impact on blood cell count (platelet) compared to carboplatin. No synergy with carboplatin or olaparib could be estimated.

D

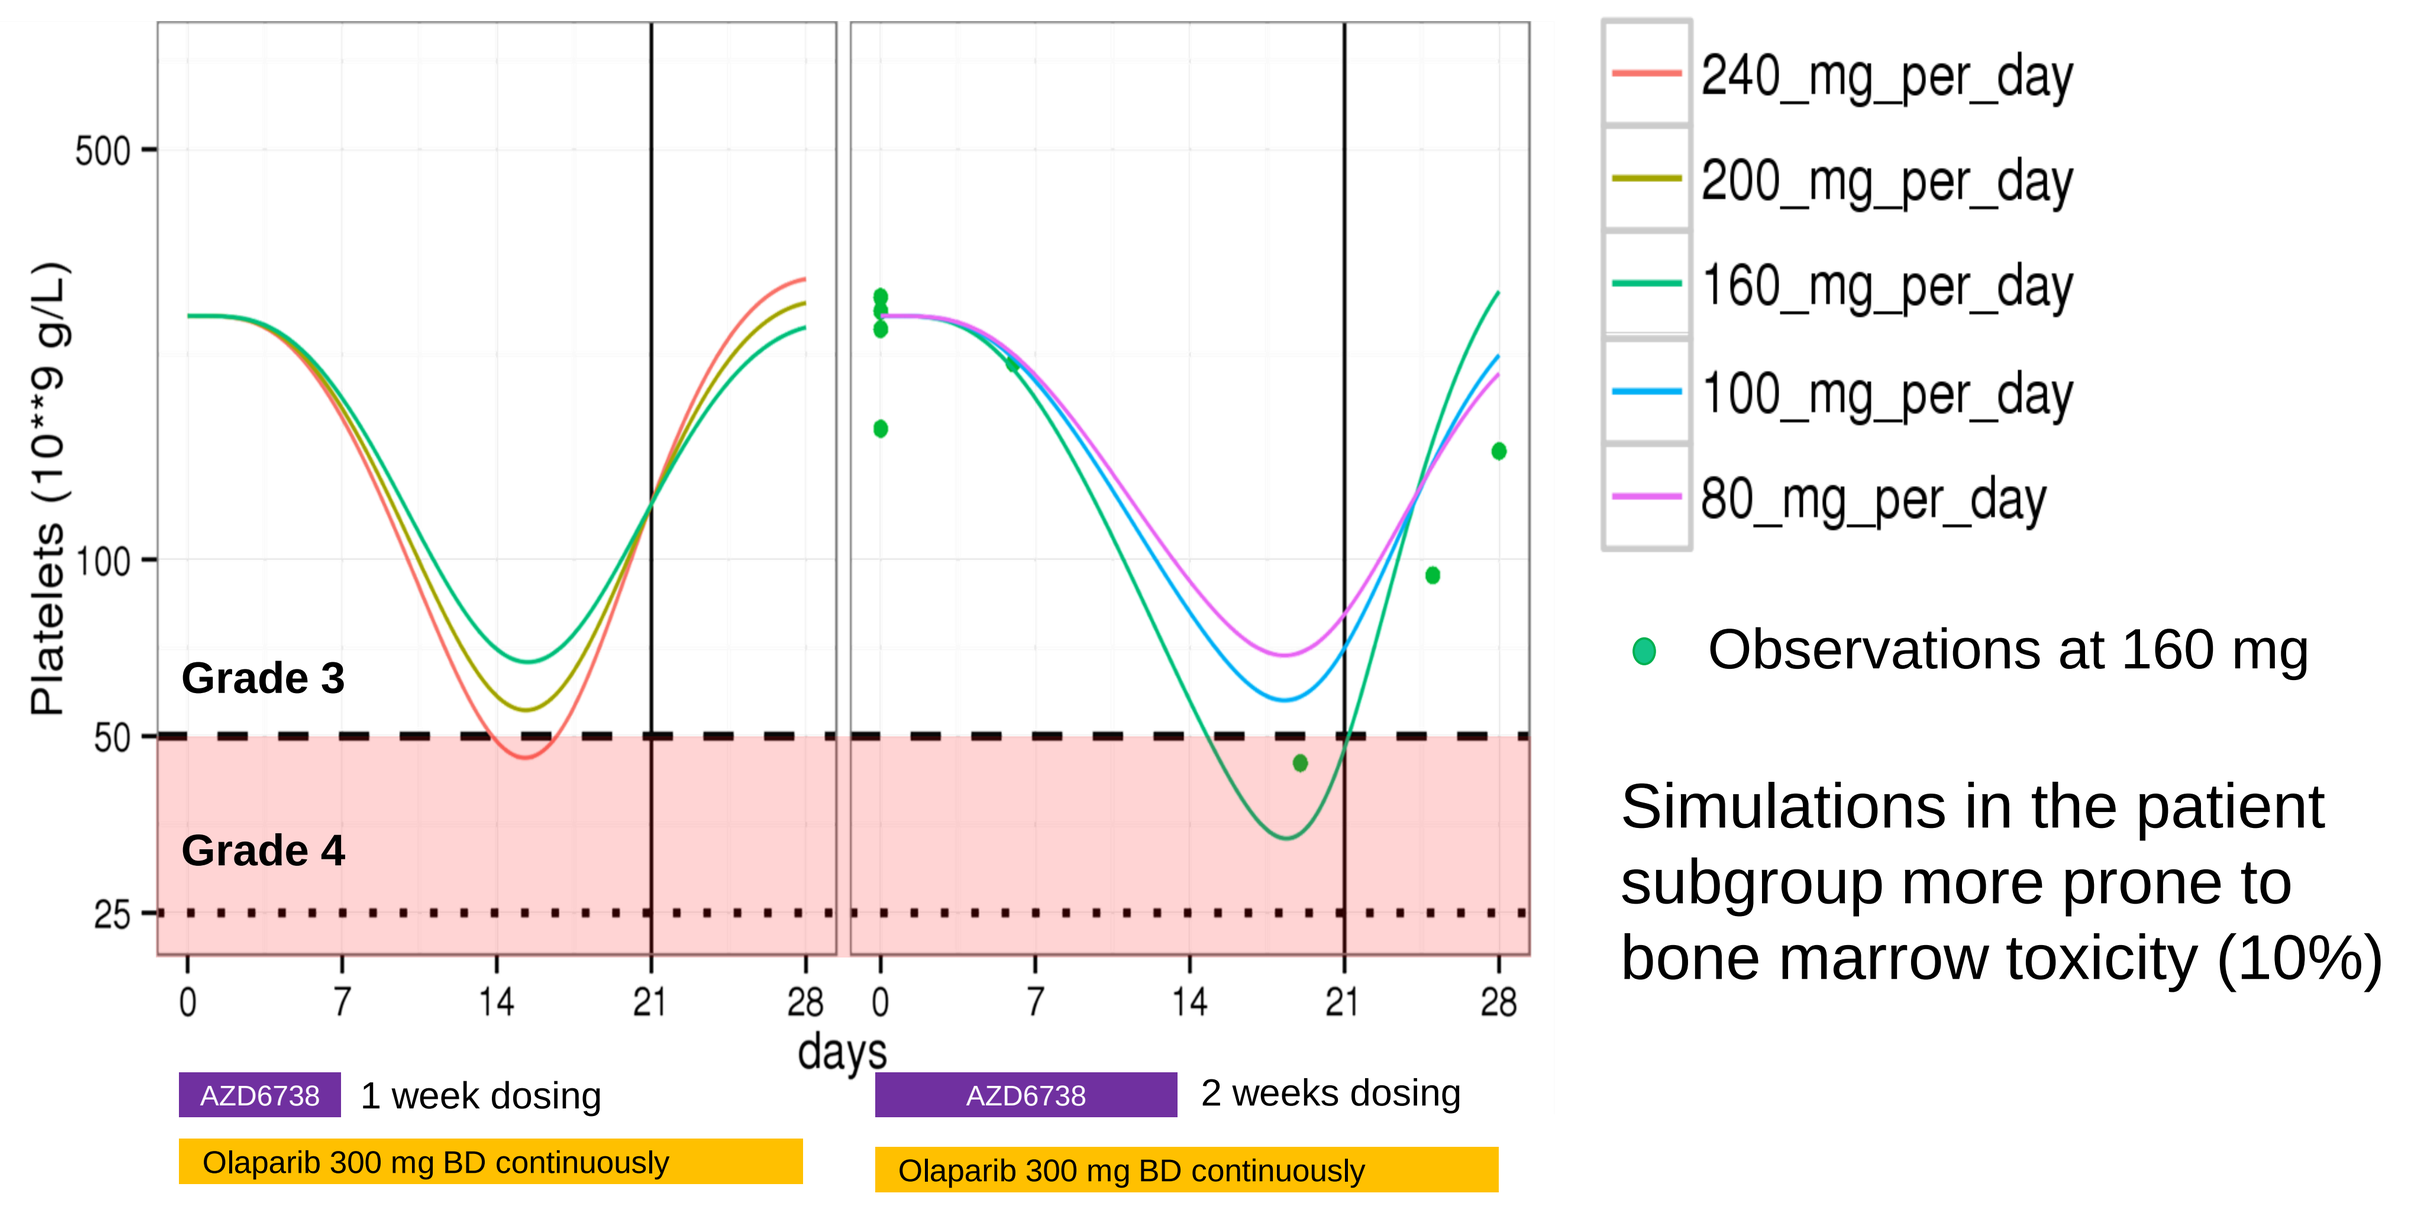
Platelets (10**9 g/L)
Grade 3
Grade 4
days
AZD6738 1 week dosing
Olaparib 300 mg BD continuously
AZD6738 2 weeks dosing
Olaparib 300 mg BD continuously
240_mg_per_day
200_mg_per_day
160_mg_per_day
100_mg_per_day
80_mg_per_day
Observations at 160 mg
Simulations in the patient subgroup more prone to bone marrow toxicity (10%)

Simulations, platelet model and olaparib combination example:
- Simulations for olaparib combination indicates total platelet counts recover to 90% baseline 21 days after AZD6738 first dose.
- Continued reductions in cell counts are not predicted by the model whereas some patients with grade 2 reductions on cycle 1 experienced grade 4 on repeated cycles: Evaluation of alternative mathematical myelosuppression models [5] to describe potential cumulative toxicity resulted from targeted treatment is ongoing.

Conclusions

- The model simulations supported team decisions on dose and schedule and will minimize dose adjustments in future trials.
- Understanding single agent and combination safety profiles provided confidence for combination choices and differentiation strategy.

References
1. Berges et al PAGE 2017
2. Marshall et al CPT: PSP 2017
3. Friberg et al J Clin Oncol 2002
4. Schmitt et al. J Clin Oncol 2010
5. Buil Bruna et al PAGE 2017

Acknowledgements
PATRIOT (Study 2) Clinical data from the Royal Marsden Hospital.
Many thanks all the PATRIOT, Study 4 investigators and AZ Project team.
All the patients participant in the studies and their families.

Supported by AstraZeneca IMED Biotech Unit