Adaptive Optimal Design in PET Occupancy Studies

Stefano Zamuner (1), Vincenzo L. Di Iorio (1), Joakim Nyberg (2), Roger N. Gunn (3),
Vincent J. Cunningham (3), Andrew Hooker (2) and Roberto Gomeni (1)

1) GlaxoSmithKline, Clinical Pharmacokinetics/Modeling&Simulation, Verona, Italy. (2) Div. of Pharmacokinetics and Drug Therapy, Dept. of Pharmaceutical Biosciences, Uppsala University, Sweden. (3) GlaxoSmithKline, Clinical Imaging Centre, Hammersmith Hospital, UK.

Background

To increase the efficiency of trials in drug development, optimal experimental design has been used to successfully optimize dose allocation and sampling schedules [1,2]. Adaptive optimal design has recently been proposed as a method to improve the assessment of receptor occupancy time-courses in PET experiments [3]. In this work we have further developed this concept to include the optimisation of dose and also improve the adaptation/optimization algorithm. In addition, a kon-koff model using the binding potential (BP) estimates from PET studies [4] has been applied to account for baseline inter-subject variability in these experiments.

Objective

To investigate advantages of adaptive optimal designs vs. traditional designs with fixed or educated selection of PET scan allocations, when optimizing over both sampling schedule and dose.

Methods

Adaptation/optimization algorithm

In the proposed adaptive algorithm, once the model has been selected according to preclinical data or other criteria, an initial group will receive a selected dose and PET scan will be acquired at a pre-specified time-points. This initial information will be then used as a prior to selected doses and PET scan times in the subsequent groups (see Figure 1).

Parameters estimates were performed in NONMEM VI. Optimization was performed on scanning times only and scanning times and doses using a D-optimality criterion as implemented in the PopED software [2,5]. Information about previous cohorts were included in the optimal design program as a prior to the Fisher information matrix.

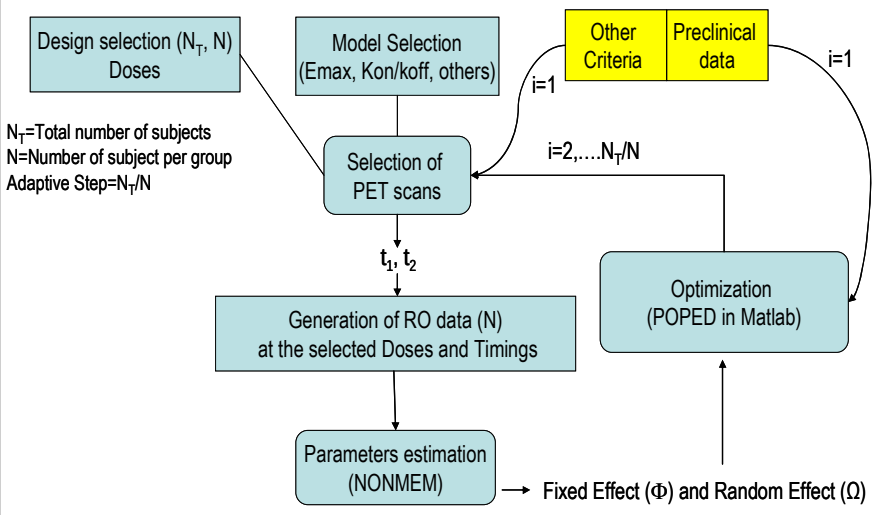
Design selection (N_T, N)
Doses
Model Selection
(Emax, Kon/koff, others)
Other Criteria
Preclinical data
i=1
i=1
N_T=Total number of subjects
N=Number of subject per group
Adaptive Step=N_T/N
Selection of PET scans
i=2,....N_T/N
t_1, t_2
Optimization
(POPED in Matlab)
Generation of RO data (N)
at the selected Doses and Timings
Parameters estimation
(NONMEM)
Fixed Effect (Φ) and Random Effect (Ω)

Figure 1. *Adaptive-optimal design framework. Initial model, dose and scanning times will be selected according prior information. At each adaptive step, parameter estimates from the previous cohorts were determined and used to determine designs for the next cohort.*

Receptor-Time Course Model using Binding Potential

A general representation of PK-occupancy time-course model is described in Figure 2. However, in PET studies where only few PET scans per subjects can be acquired, this model can not be applied and a simplified version needs to be considered. In our study, a kon-koff model using the binding potential data derived from PET study was considered. Assuming RO derived from the binding potential measurement as follow:

$$RO = \frac{BP_0 - BP}{BP_0}$$

where BP_0 is the baseline binding potential and BP that after dosing. The equation for the receptor-time course model can be described as:

$$d\left(\frac{BP_0 - BP}{BP_0}\right) / dt = k_{on} \cdot C_P \cdot \left(1 - \frac{BP_0 - BP}{BP_0}\right) - k_{off} \cdot \frac{BP_0 - BP}{BP_0}$$

$$\frac{dBP}{dt} = k_{off} \cdot BP_0 - (C_P \cdot k_{on} + k_{off})BP$$

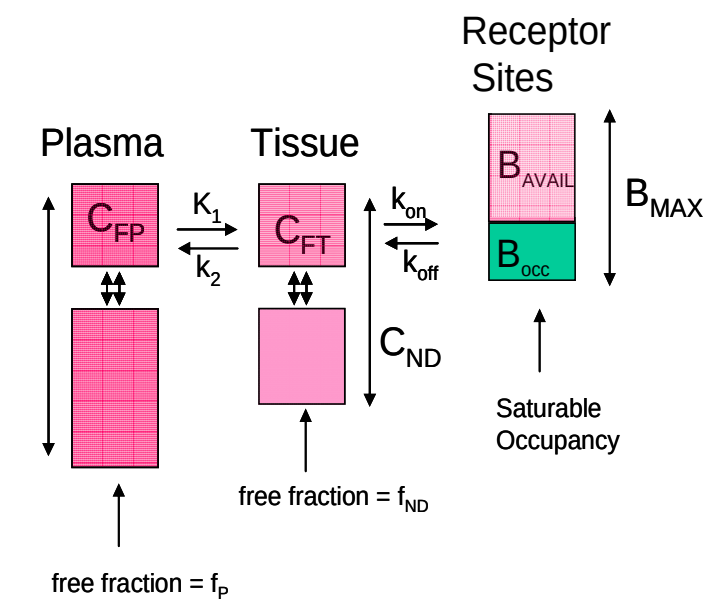
Receptor Sites
Plasma
Tissue
C_{FP}
K_1
k_2
C_{FT}
k_{on}
k_{off}
B_{AVAIL}
B_{occ}
B_{MAX}
C_{ND}
Saturable Occupancy
free fraction = f_{ND}
free fraction = f_P

Figure 2*. Schematic representations of a PK-receptor binding model*

Simulation Study

Adaptive optimization was performed on the PK-BP model with the following parameters: kon=0.088 hrs^{-1} and koff=0.221 hrs^{-1}, BP_0=3, inter-subject variability=30%, proportional error model.

A total of 12 subjects was considered with 5 possible doses (1.5, 3, 4, 6 and 8mg) and designs with 3, 4, and 6 adaptive steps were investigated. At each adaptive step, parameter estimates from the previous cohorts were determined and used to determine designs for the next cohort. The BP time-courses from these designs (empirical dummy using samples at Tmax and trough, educated – samples selected from an independent expert - and optimal) were then simulated under the true model. One hundred studies per each design were simulated to test the performance of the adaptive/optimal algorithm.

Results

A binding potential time-course for a typical simulated study with optimal PET scan allocation is illustrated below.

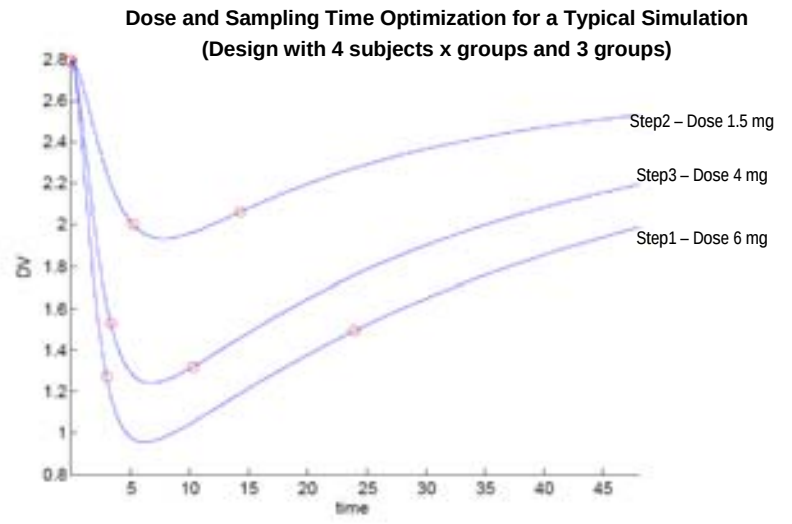
Dose and Sampling Time Optimization for a Typical Simulation
(Design with 4 subjects x groups and 3 groups)
Step2 – Dose 1.5 mg
Step3 – Dose 4 mg
Step1 – Dose 6 mg
time

Figure 3. *Binding potential time-course at three different dose levels. Red circles represents the optimal PET scan allocation per each group.*

A clear improvement in terms of bias (SME), precision (CV) and accuracy (RMSE) of the population estimates (Kon and koff) was found when comparing dummy vs. educated vs. optimal. Unbiased mean estimates were found for the optimal designs; a great improvement in accuracy was found when comparing optimal vs. dummy designs (25-30 fold) and still a significant improvement was found when comparing optimal vs. educated designs (2-3 fold).

Design	Performance	Dummy		Educated		Optimal (time)	
		Kon	Koff	Kon	Koff	Kon	Koff
3 groups 4 subj x group	Bias SME	0.87	1.00	0.13	0.12	-0.0095	-0.029
	Precision (CV)	256	282	38.3	43.7	18.5	20.5
	Accuracy (RMSE)	0.43	1.27	0.040	0.11	0.016	0.044
4 groups 3 subj x group	Bias (SME)	0.83	0.93	0.096	0.076	-0.0010	-0.012
	Precision (CV)	300	319	43.4	48.7	19.9	23.8
	Accuracy (RMSE)	0.49	1.37	0.043	0.12	0.017	0.052
6 groups 2 subj x group	Bias (SME)	0.87	1.00	0.13	0.12	0.013	0.0022
	Precision (CV)	256	282	38.3	43.7	19.6	22.7
	Accuracy (RMSE)	0.43	1.27	0.040	0.11	0.017	0.050

No clear advantages were found when optimizing both time and dose. The number of adaptive steps was less influential on design performance than the method of designing the next step. No improvement was obtained for inter-subject variability estimates when comparing optimal vs. non optimal designs.

Design	Performance	Optimal (time)		Optimal (time + dose)	
		Kon	Koff	Kon	Koff
3 groups 4 subj x group	Bias SME	-0.0095	-0.029	0.0086	-0.0088
	Precision (CV)	18.5	20.5	19.5	19.6
	Accuracy (RMSE)	0.016	0.044	0.017	0.043
4 groups 3 subj x group	Bias (SME)	-0.0010	-0.012	-0.0077	-0.021
	Precision (CV)	19.9	23.8	20.1	21.4
	Accuracy (RMSE)	0.017	0.052	0.018	0.046
6 groups 2 subj x group	Bias (SME)	0.013	0.0022	0.011	0.0004
	Precision (CV)	19.6	22.7	18.2	20.0
	Accuracy (RMSE)	0.017	0.050	0.016	0.044

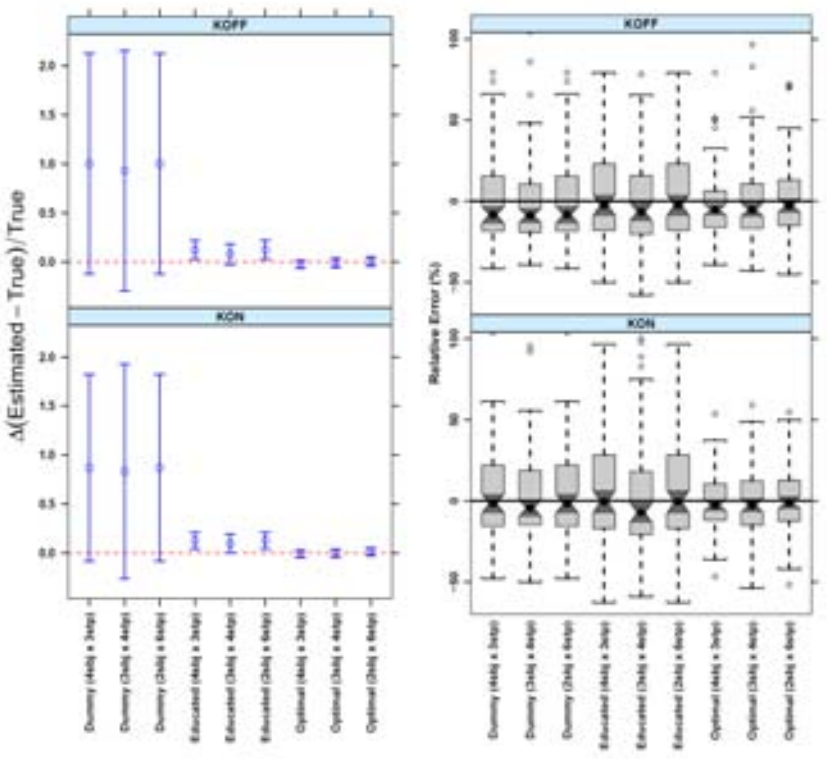
KOFF
KON
Δ(Estimated – True) / True
Relative Error (%)

Figure 4. *Comparison of performances (Bias and relative error) between adaptive-optimal vs non optimal approaches on different designs.*

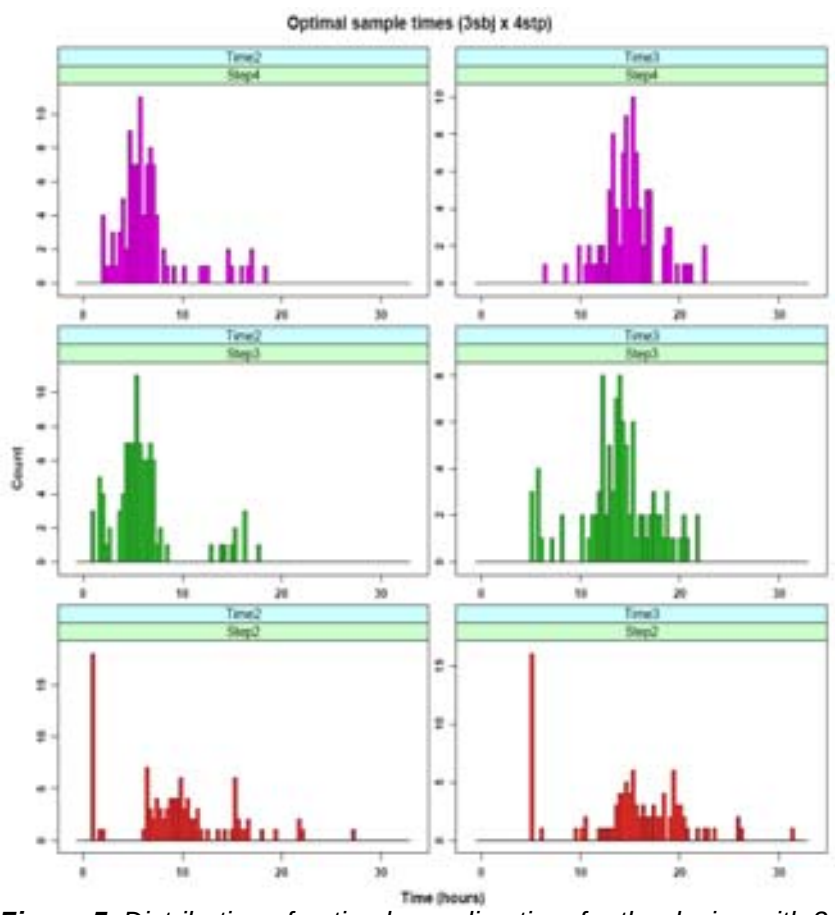
Optimal sample times (3sbj x 4stp)
Time2
Step4
Time3
Step3
Step2
Count
Time (hours)

Figure 5. *Distribution of optimal sampling time for the design with 3 subjects x group and 4 group (100 simulated studies).*

Discussion

These results indicate that adaptive optimal design of PET occupancy studies provides more accurate information on the PK-occupancy relationship. In this work, doses were initially selected at high, medium and low occupancy levels based on previous knowledge of the system. Consequently, optimization of dose was not found to influence the results. In experiments where initial dose selection is misleading it is expected that dose optimization will have a greater impact.

References

[1] Mentré, F., Mallet, A. and Baccar, D., Optimal design in random-effects regression models. Biometrika, 1997. 84(2): p. 429-442.
[2] Foracchia, M., Hooker, A., Vicini, P. and Ruggeri, A., POPED, a software for optimal experiment design in population kinetics. Comput Methods Programs Biomed, 2004. 74(1): p. 29-46.
[3] Petrone, M., Zamuner, S., Di Iorio, V., Gunn, R., Laruelle, M., Gomeni, R. Model-based sequential human PET study design for Optimal PK/RO assessment. PAGE 16 (2007) Abstr. 1183
[4] Mintun MA, Raichle ME, Kilbourn MR, Wooten GF, Welch MJ. A quantitative model for the in vivo assessment of drug binding sites with positron emission tomography. Ann Neurol, 1984. 15(3): p. 217-27.
[5] Nyberg, J., Karlsson, MO, Hooker, A., Sequential versus simultaneous optimal experimental design on dose and sample times. PAGE 16 (2007) Abstr 1160

Created by Web Broadcasting Service
live | video | interactive | design