PK and PK/PD modeling of CB1 blocker antagonism of THC induced CNS and Heart Rate effects

G Ferron[a], L Klumpers[b], J van Gerven[b], and C Roy[a]
(a) sanofi-aventis and (b) Centre for Human Drug Research

INTRODUCTION

Surinabant (SR) is an orally active selective antagonist for the cannabinoid CB1 receptor which is currently in clinical development.

AIM

Develop a pharmacokinetic / pharmacodynamic (PK/PD) model for the characterization of the inhibition of CNS and heart rate effects of delta9-tetrahydrocannabinol (THC) by surinabant in healthy subjects.

METHODS

A double blind, placebo-controlled, randomized, 6-treatment, 4-period, 6-sequence incomplete balanced cross-over study was performed in 36 healthy young male occasional cannabis users (<1/week). Single oral dose of surinabant (5, 20 or 60 mg) or placebo was administered followed 1.5 h later by 4 increasing doses of THC inhaled at 1 h intervals. PD measurements were body sway, alertness factor from Bond and Lader visual analogue scales (VAS), item “feeling high”, composite factors “internal and external perception” from Bowdle VAS, and heart rate. THC and surinabant PK were obtained in each period. An integrated population PK/PD model was built describing the effect of THC on each PD end-points and the antagonism of these effects by surinabant. NONMEM V (Globomax, LLC, Hanover, MD) was used for the analysis.

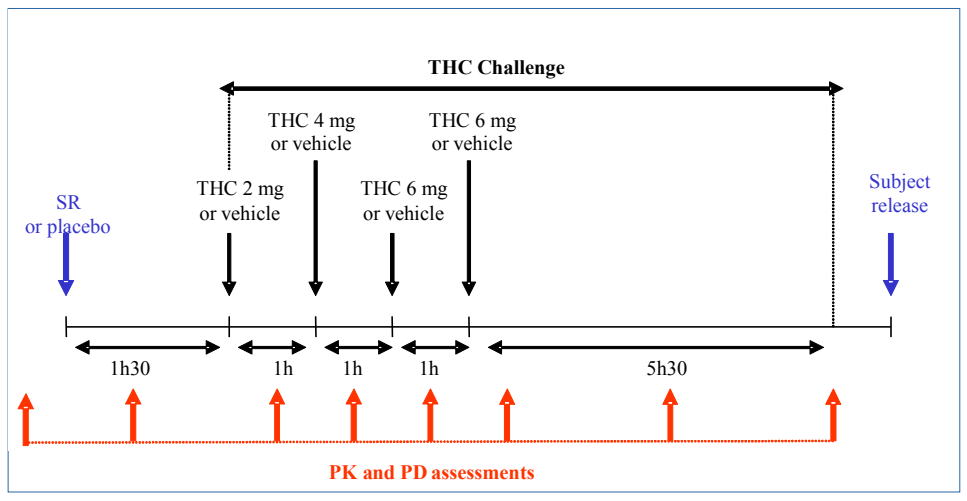
THC Challenge
THC 4 mg or vehicle
THC 6 mg or vehicle
SR or placebo
THC 2 mg or vehicle
THC 6 mg or vehicle
Subject release
1h30
1h
1h
1h
5h30
PK and PD assessments

PK MODEL FOR THC

A two-compartment model with intra-individual variability on absorption and linear elimination adequately described THC PK. Surinabant was not found to impact THC PK.

Table 1 - Final population parameters for THC (n=30, 913 values)

Parameter	Explanation	Estimate	%RSE
TV1CL [θ_1], L/h	Typical clearance	293	7.58%
TVV1 [θ_2], L	Typical central volume	43.6	8.03%
TVQ [θ_3], L/h	Typical intercompartmental clearance	166	8.01%
TVV2 [θ_4], L	Typical peripheral volume of distribution	136	8.97%
Between-subject variability (%CV)			
η_{CL} [η_1]	Inter-individual error on CL	11.8%	25.0%
η_{V2} [η_2]	Inter-individual error on V2	15.2%	36.0%
η_F [η_3]	Interoccasion variability on Bioavailability	55.8%	12.6%
Residual variability (SD): Y = F + F* eps $_1$			
σ_1	Proportional residual error	15.9%	14.6%

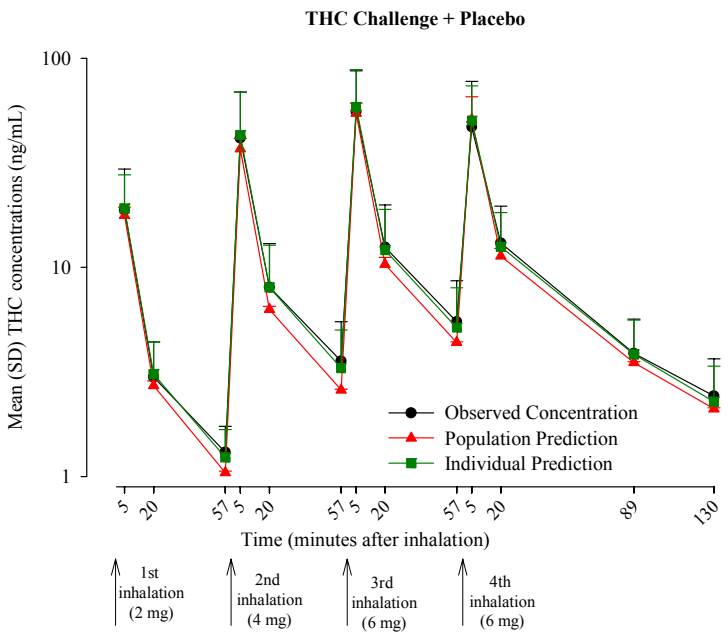
THC Challenge + Placebo
Mean (SD) THC concentrations (ng/mL)
100
10
1
Observed Concentration
Population Prediction
Individual Prediction
5
20
57
5
20
57
5
20
57
5
20
89
130
Time (minutes after inhalation)
1st inhalation (2 mg)
2nd inhalation (4 mg)
3rd inhalation (6 mg)
4th inhalation (6 mg)

Figure 1 – Mean (SD) Observed and Predicted (Population and Individual) of THC concentrations

PK MODEL FOR SURINABANT

A two-compartment model with first order absorption (ka) and elimination, a lag-time, and dose effect on ka was used for surinabant.

Table 2 - Final population parameters for SR147778 (n=30, 731 values)

Parameter	Explanation	Estimate	%RSE
TVCL [θ_1], L/h	Typical clearance	4.69	13.0%
TVV2 [θ_2], L	Typical central volume	3.74	22.0%
TVQ [θ_3], L/h	Typical intercompartmental clearance	15.3	3.70%
TVV3 [θ_4], L	Typical peripheral volume of distribution	491	6.27%
TVKA [θ_5], h^{-1}	Typical absorption rate constant	0.406	3.18%
TVLAG [θ_6], h	Typical lag time	0.591	5.91%
Dose effect on KA [θ_7]	Dose effect on ka	-0.00164	16.4%
Between-subject variability (%CV)			
η_{CL} [η_1]	Inter-individual error on CL	72.1%	27.7%
η_{V2} [η_2]	Inter-individual error on V2	74.8%	34.9%
η_Q [η_3]	Inter-individual error on Q	16.3%	30.8%
η_{V3} [η_4]	Inter-individual error on V3	30.6%	23.9%
η_{ka} [η_5]	Inter-individual error on KA	6.40%	115%
Residual variability (SD): Y = F + F* eps $_1$			
σ_1	Proportional residual error	18.2%	10.0%

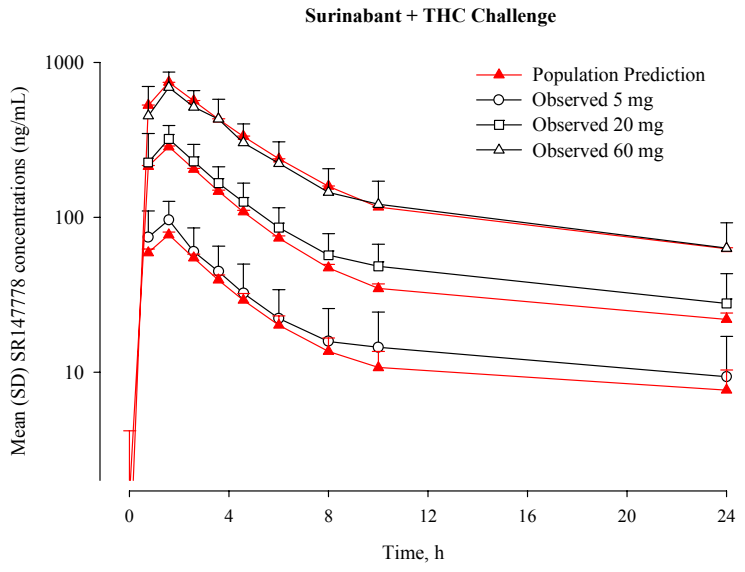
Surinabant + THC Challenge
Mean (SD) SR147778 concentrations (ng/mL)
1000
100
10
Population Prediction
Observed 5 mg
Observed 20 mg
Observed 60 mg
0
4
8
12
16
20
24
Time, h

Figure 2 – Mean (SD) Observed and Population Predicted Surinabant Concentrations

PK/PD MODELS

The PK/PD model describing THC effect on PD measures was comprised of an effect compartment, an E_{max} (body sway, feeling high, alertness and heart rate) or linear (internal and external perception) model and intra-individual variability on baseline PD. CB1 antagonism effect was included using a competitive binding equation. In addition, surinabant only partially inhibited THC effect on feeling high. Individual PK values were fixed.

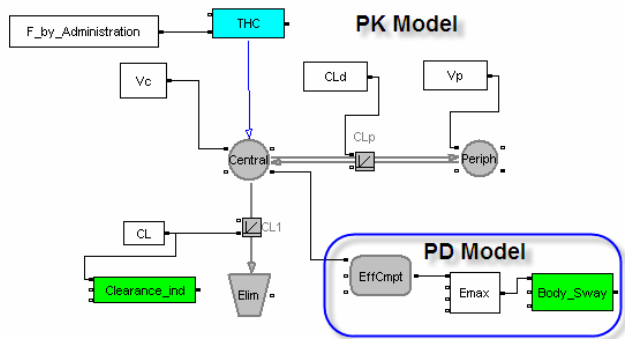
F_by_Administration
THC
PK Model
Vc
CLd
Vp
CLp
Central
Periph
CL
CL1
Clearance_ind
Elim
PD Model
EffCmpt
Emax
Body_Sway

$$Effect_{THC+SR} = \frac{E_{\max} \bullet \dfrac{THC_{Conc}}{EC_{50,THC}}}{1 + \dfrac{SR_{Conc}}{IC_{50,SR}} + \dfrac{THC_{Conc}}{EC_{50,THC}}}$$

$$Effect_t = E_{0,occasion} + Effect_{THC+SR,t}$$

Table 3 - Final population PK/PD parameters (n=30, 2510 to 2538 values)

		Body Sway		Alertness		Feeling High		External Perception		Internal Perception		Heart Rate	
Parameter	**Explanation**	**Estimate**	**%RSE**	**Estimate**	**%RSE**	**Estimate**	**%RSE**	**Estimate**	**%RSE**	**Estimate**	**%RSE**	**Estimate**	**%RSE**
TVEo [θ_1]	Baseline	5.46	1.26%	49.4	1.10%	0.321	3.96%	0.367	0.529%	0.366	0.508%	64.2	1.25%
TVE$_{max}$ [θ_2]	E$_{max}$ value	0.829	24.5%	NA	NA	0.713	31.6%	NA	NA	NA	NA	37.8	31.2%
TVEC50 or slope [θ_3]	EC50 or slope of THC effect	7.24	42.8%	0.547	45.2%	6.98	33.5%	0.00258	41.9%	8.69 .10^{-4}	38.2%	20.3	45.0%
TVkeo [θ_4], h^{-1}	Bffect compartment rate constant	0.466	17.9%	0.347	33.7%	1.04	17.4%	0.868	16.9%	0.955	20.1%	0.945	27.7%
TVICsr [θ_5], ng/mL	IC50 of SR147778 effect	22.0	45.2%	33.6	45.8%	30.5	61.6%	37.1	59.6%	58.8	44.2%	25.6	32.5%
TVINH [θ_6]	% Inhibition of THC effect by Surinabant	1 FIXED		1 FIXED		0.751	20.6%	1 FIXED		1 FIXED		1 FIXED	
Between-subject variability (%CV)													
η_{Eo} [η_1]	Inter-subject on Eo	6.66%	24.3%	5.13%	47.9%	21.6%	38.5%	NA	NA	2.68%	68.2%	6.46%	33.3%
η_{Eo} [η_4]	Intra-subject on Eo	3.00%	32.2%	180%	37.0%	NA	NA	3.86%	46.1%	1.46%	36.9%	5.68%	16.7%
η_{Emax} [η_2]	Inter-subject on E$_{max}$	68.6%	40.2%	98.1%	53.5%	124%	39.6%	154%	29.4%	151%	35.1%	70.5%	42.1%
η_{keo} [η_3]	Inter-subject on keo	73.4%	33.6%	4.64%	26.0%	71.6%	32.4%	69.9%	30.1%	71.4%	45.5%	124%	33.8%
Residual variability (SD): Y = F + eps $_1$													
σ_1	Additive residual error	0.212	10.5%	3.30%	18.3%	0.254	19.1%	0.0182	19.1%	0.0123	22.8%	6.53	5.66%

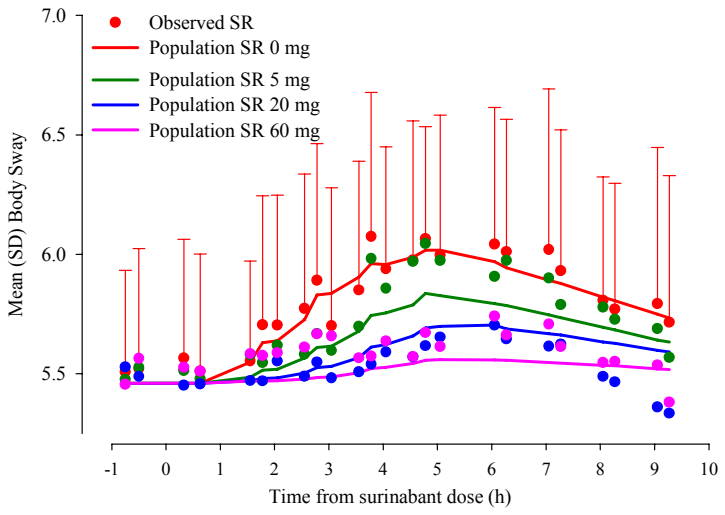
Observed SR
Population SR 0 mg
Population SR 5 mg
Population SR 20 mg
Population SR 60 mg
Mean (SD) Body Sway
7.0
6.5
6.0
5.5
-1
0
1
2
3
4
5
6
7
8
9
10
Time from surinabant dose (h)

Figure 3 – Mean (SD) Observed and Population Prediction of Body Sway with Increased Surinabant Doses

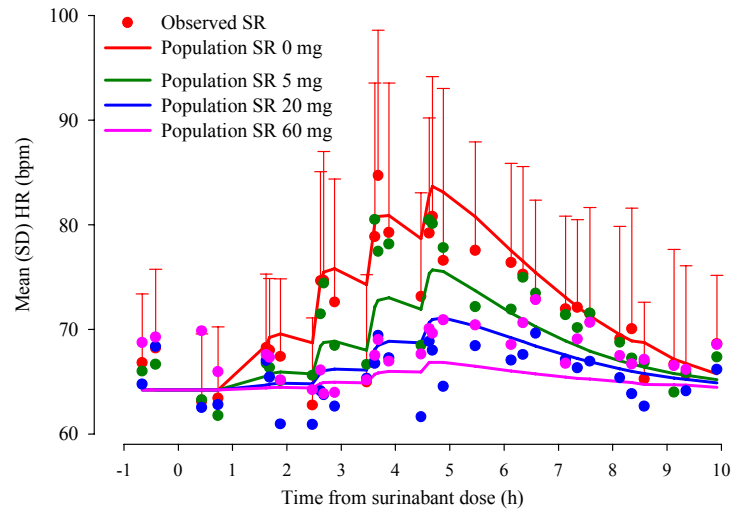
Observed SR
Population SR 0 mg
Population SR 5 mg
Population SR 20 mg
Population SR 60 mg
Mean (SD) HR (bpm)
100
90
80
70
60
-1
0
1
2
3
4
5
6
7
8
9
10
Time from surinabant dose (h)

Figure 4 – Mean (SD) Observed and Population Prediction of Heart Rate with Increased Surinabant Doses

CONCLUSION

- PK/PD models adequately described the time-course of PK and PD effects of THC and surinabant. Surinabant competitively binds to CB1 receptor with concentrations producing 50% reduction of THC effect between 22.0 and 58.8 ng/mL.
- This model could be of value to differentiate CB1 blockers.

sanofi aventis

L’essentiel c’est la santé.