Using Modelling & Simulation Techniques to Optimise the Design of a Paediatric Simulation Study

(Clare Gaynor, Karl Brendel), Cyrielle Dumont, Alexia Blesius and Marylore Chenel

Department of Clinical Pharmacokinetics, Institut de Recherches Internationales Servier, Courbevoie, France.

Introduction

European Paediatric Regulations in force since January 2007 specify that, when applying for a new indication or new route of administration in adults, a corresponding Paediatric Investigation Plan (PIP) must be submitted to the European Medicines Agency Paediatric Committee (EMEA PdCO). A development plan for use of Servier drug X in the paediatric population is, therefore, underway. The study being planned is a dose-finding PK/PD study which involves a single i.v. bolus administration of the compound to children in the target population.

Modelling & Simulation techniques are playing a key role in designing this study to ensure that the ideal dose is identified as early as possible and that blood samples will be taken at optimum times.

Methods

Data

A physiologically-based (PB) PK model has previously been developed for this compound in adults; this existing model was adapted and used to simulate the plasma concentration-time profiles which would result from an intravenous bolus dose of 0.1 mg/kg in children from 2 to18 years (Figure 1).

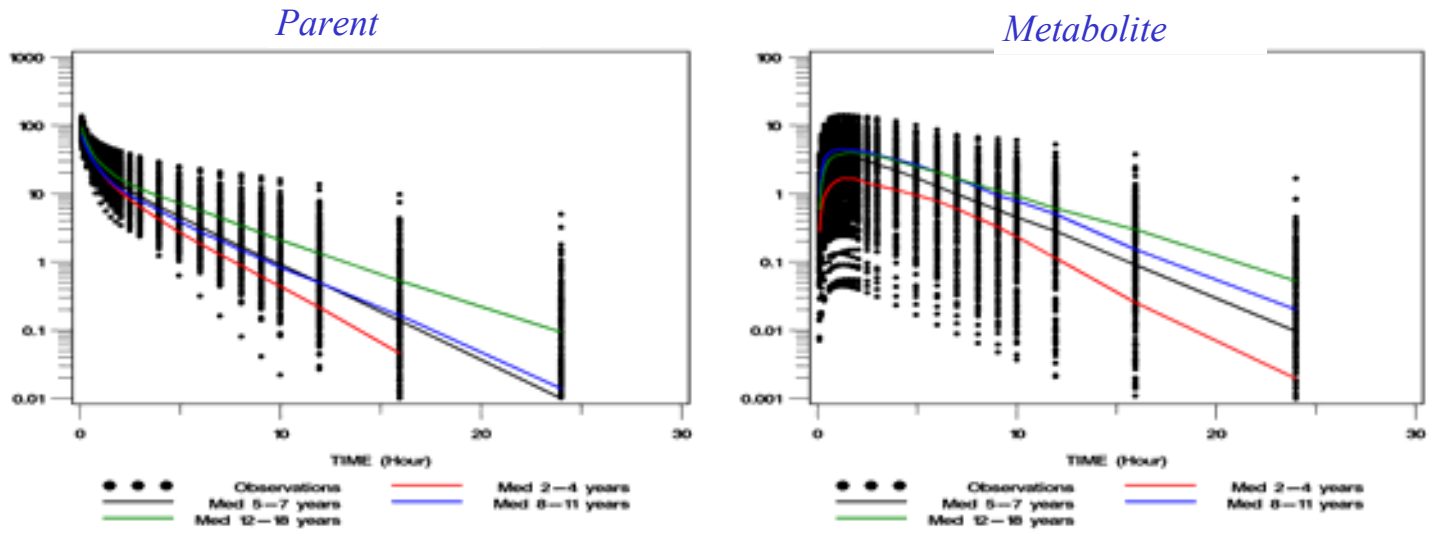
Parent
Metabolite
TIME (Hour)
Observations
Med 2–4 years
Med 5–7 years
Med 8–11 years
Med 12–18 years

Fig.1: Simulated PK profiles of Parent and Metabolite in children (by age class) after iv admin 0.1 mg/Kg

Population PK model

A population PK model for both the parent drug and its active metabolite, was built on these plasma concentration-time. The first step of the modelling was to build the basic population model, made of the structural model (i.e. the number of compartment…), the random effect model (i.e. the inter-individual variability, inter-occasion variability) and the residual error model. Model selection was based on the comparison of the objective function (OF) given by NONMEM. For nested models, a likelihood ratio test (LRT) was performed with a p-value of 0.05.

Then, the covariate influence upon the model parameters was studied. All volume and clearance parameters were scaled for body weight (WT). This is illustrated for clearance using WT:

$$CLi = CL.(WT/WTstd)^{\theta cov}$$

Where *WTstd* is a nominal standard weight (median value) of the population and *θcov* the covariate effect, which can be fixed to the allometric values (0.75 for clearance and 1 for volume) or estimated. Age effect was also tested.

Lastly, the final population mixed effect model was evaluated using internal evaluation procedures as Visual Predictive Check (VPC).

PD simulations

PK model is linked to a population PD model previously developed in adults, in order to simulate response-time profiles at different doses and in several age classes over the range being considered. This PD model is an agonist Emax model with an effect compartment taking into account the activity of both the parent drug and its active metabolite.

Four age classes (Figure 1) were determined on the basis of biomarker baseline values as well as response and safety targets. For each envisaged dose and each age class, the proportion of subjects predicted to have a PD response regarding the efficacy target value (% efficacy Responder) were calculated from simulated profiles between time 0 to 1.5h (expected:>50%). Concerning the safety threshold value (% safety Responder) , the proportion of subjects were calculated between time 0 and 5h (expected:<5%).

Results

A model incorporating the PK of the parent drug and its active metabolite with three compartments for the parent and a one compartment for the metabolite (with formation from the central compartment of the parent) were used to fit the simulated concentration-time data. A combined error model best described the residual variability. Weight effects were applied and estimated (i.e. not fixed to the allometric values) on clearances and volumes of distribution.

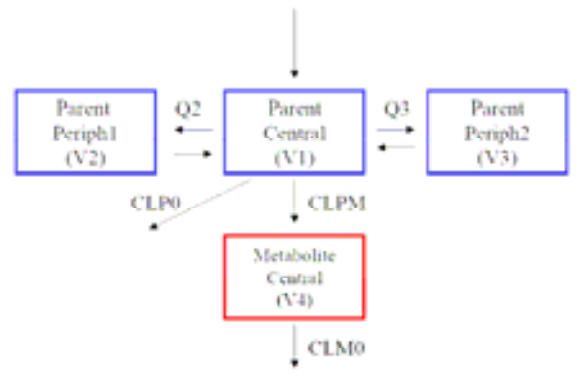
Parent
Periph1
(V2)
Q2
Parent
Central
(V1)
Q3
Parent
Periph2
(V3)
CLP0
CLPM
Metabolite
Central
(V4)
CLM0

Fig.2: Population PK model for parent and metabolite

The estimated population parameters and the precision of their estimation are given in Table 1 for the final model.

Table 1: Population PK parameters estimates and relative standard errors (RSE) for the final model

Parameters	Estimates (RSE%)	IIV (%)
CLPO (L/h)	22.3 (2)	38.1
θWT_CLPO	0.519 (5)	-
Q2	7.22 (17)	-
θWT_Q2	1.35 (20)	-
Q3	24.1 (6)	-
θWT_Q3	0.957 (7)	-
CLPM	4.28 (6)	115
θWT_CLPM	0.977 (10)	-
CLM0	33.6 (3)	44.3
θWT_CLM0	0.726 (5)	-
V1	36.5 (1)	18.7
θWT_V1	0.781 (2)	-
V2	20.1 (12)	-
θWT_V2	1.34 (15)	-
V3	28.8 (9)	-
θWT_V3	1.04 (12)	-
$\sigma^2_{prop_S16257}$ (%)	15.6 (8)	-
$\sigma^2_{add_S16257}$	0.0575 (19)	-
$\sigma^2_{prop_S18982}$ (%)	5.2 (6)	-
$\sigma^2_{add_S18982}$	0.00998 (24)	-

The Visual Predictive Checks were satisfactory for both parent and metabolite (Figure 3).

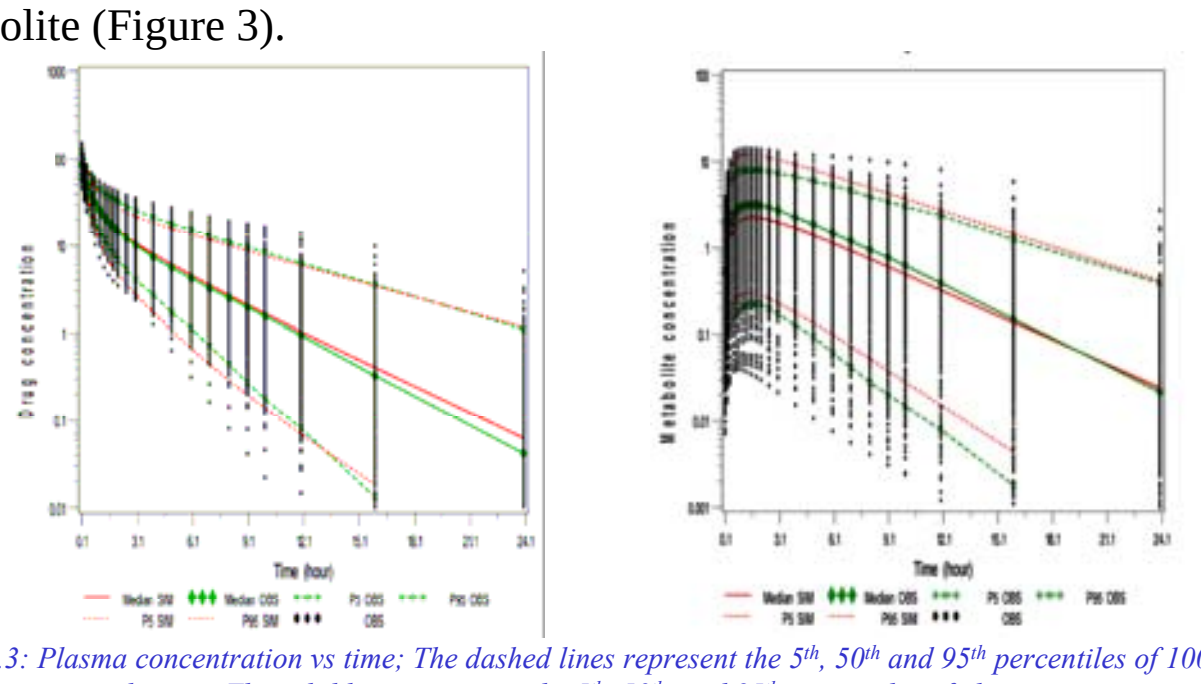
Drug concentration
Metabolite concentration
Time (hour)
Median SIM
Median OBS
P5 OBS
P95 OBS
P5 SIM
P95 SIM
OBS

Fig.3: Plasma concentration vs time; The dashed lines represent the 5th, 50th and 95th percentiles of 1000 simulations; The solid lines represent the 5th, 50th, and 95th percentiles of observations

The % of responders regarding efficacy and safety criteria simulated using PKPD models are given in Table 2 .

Table 2: Simulation of percentage of responders for efficacy and safety criteria, per dose and per age class

AGE CLASS	DOSE (mg/Kg)	%EFFICACY RESPONDERS TIME 0-1.5H	%SAFETY RESPONDERS TIME 0-5H
2-4 years	0.2	23.2	3
5-7 years	0.2	12.7	0.7
8-11 years	0.2	10.7	1.7
12-18 years	0.2	9.8	0.08
2-4 years	0.3	36.9	3
5-7 years	0.3	21.9	0.7
8-11 years	0.3	18.2	1.7
12-18 years	0.3	18.8	0.08
2-4 years	0.5	**59.6**	7.6
5-7 years	0.5	38.1	2.8
8-11 years	0.5	32.1	3.6
12-18 years	0.5	33.3	0.9
2-4 years	0.8	76.2	22.4
5-7 years	0.8	**53.9**	10.4
8-11 years	0.8	**49.3**	7.3
12-18 years	0.8	**49.8**	3.6

Conclusions

Based on this Modelling & Simulation approach a starting dose and a range of doses to be tested, which comply with requirements both in terms of efficacy and safety, can be chosen for each age class.

During the course of the PK/PD study, a continual reassessment method (CRM) design will be used to reassess the distribution of response rates after each cohort of patients (possibly reassessed singly or pairwise), using Bayesian statistical methods and consequently refine the appropriate paediatric doses. As the dose is periodically reassessed, this approach will determine the optimal dose in terms of efficacy and safety as quickly as possible.