PharmML & SO – Standards for Encoding Models and Results in Pharmacometrics and Quantitative Systems Pharmacology

Maciej J. Swat (1), Nadia Terranova (2), on behalf of all DDMoRe contributors.
(1) EMBL-European Bioinformatics Institute, Cambridge, UK, (2) Merck Institute for Pharmacometrics, Merck Serono S.A., Switzerland.

Introduction: New formats enabling the efficient exchange and integration of Pharmacometric and Quantitative System Pharmacology models across software tools have been defined and implemented as key elements of the DDMoRe interoperability platform [1]. Specifically, PharmML has been designed as the exchange medium for mathematical and statistical models [2, 3], and the Standard Output (SO) has been developed as a complementary component for storing typical output produced in a pharmacometric workflow. PharmML and SO, as essential elements of the DDMoRe interoperability platform, proved to be capable to handle complex modeling scenarios, and to facilitate model exchange and results storage across various tools.

Role of exchange formats

- Smooth and error-free transmission of models between tools.
- Use of complex workflows via standardised model and output definitions.
- Reproducibility of research.
- Easier reporting and bug tracking.
- Improved interaction with regulatory agencies regarding modeling and simulation.
- Development of new tools and methods.
- Expanding the community developing/applying pharmacometric models.
- Reuse of existing models, e.g. BioModels database of computational models of biological processes (SBML).

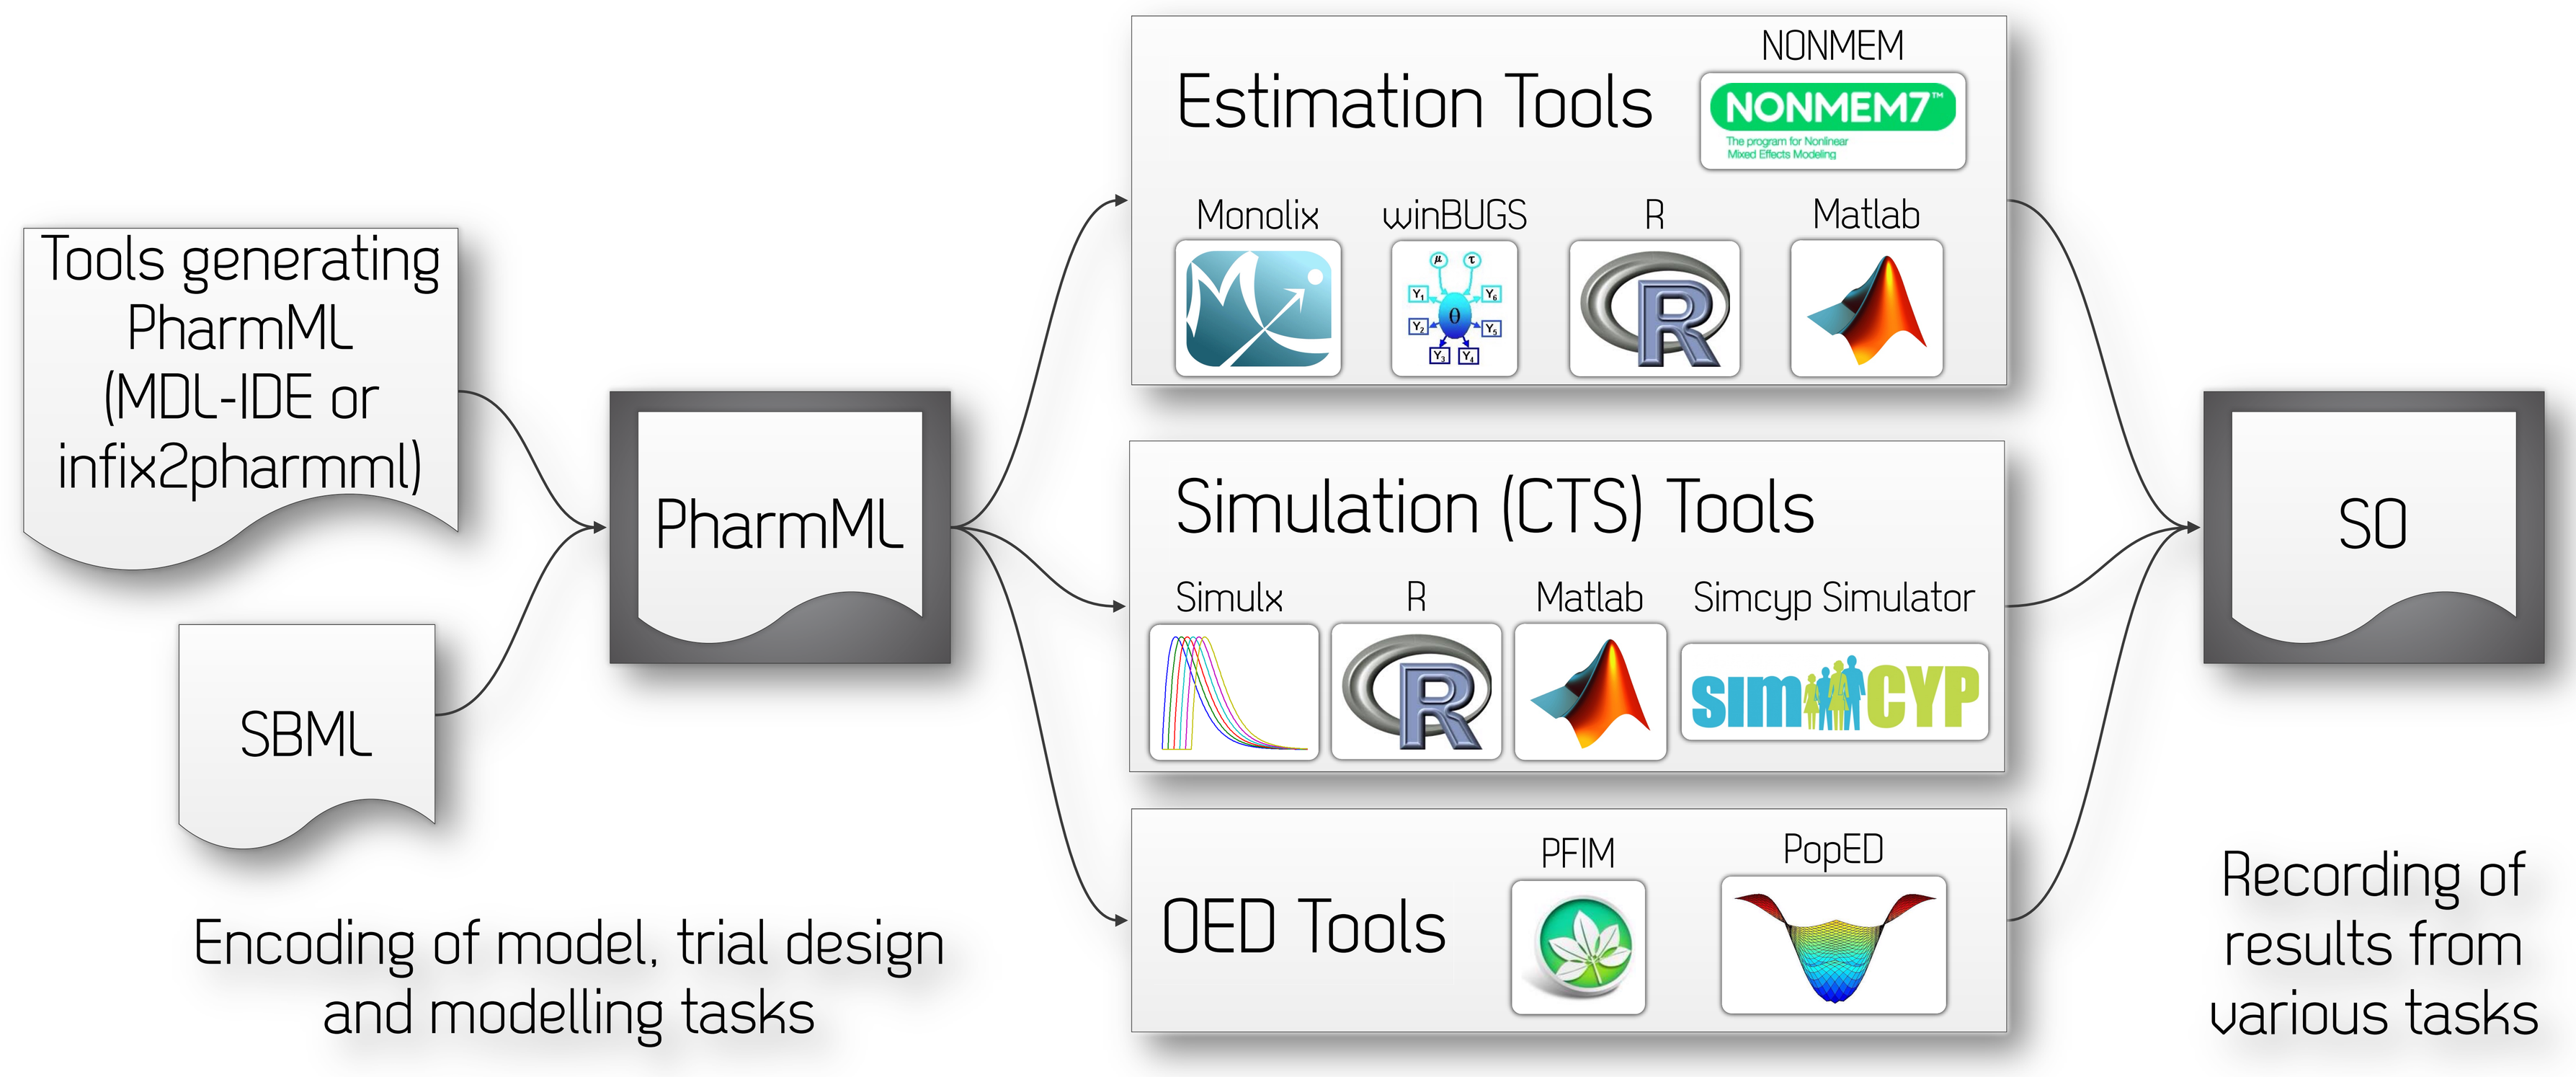
Tools generating PharmML (MDL-IDE or infix2pharmml)
SBML
PharmML
Estimation Tools
NONMEM
Monolix
winBUGS
R
Matlab
Simulation (CTS) Tools
Simulx
R
Matlab
Simcyp Simulator
OED Tools
PFIM
PopED
SO
Encoding of model, trial design and modelling tasks
Recording of results from various tasks

Figure 1: The role of PharmML/SO and their connection to the target tools.

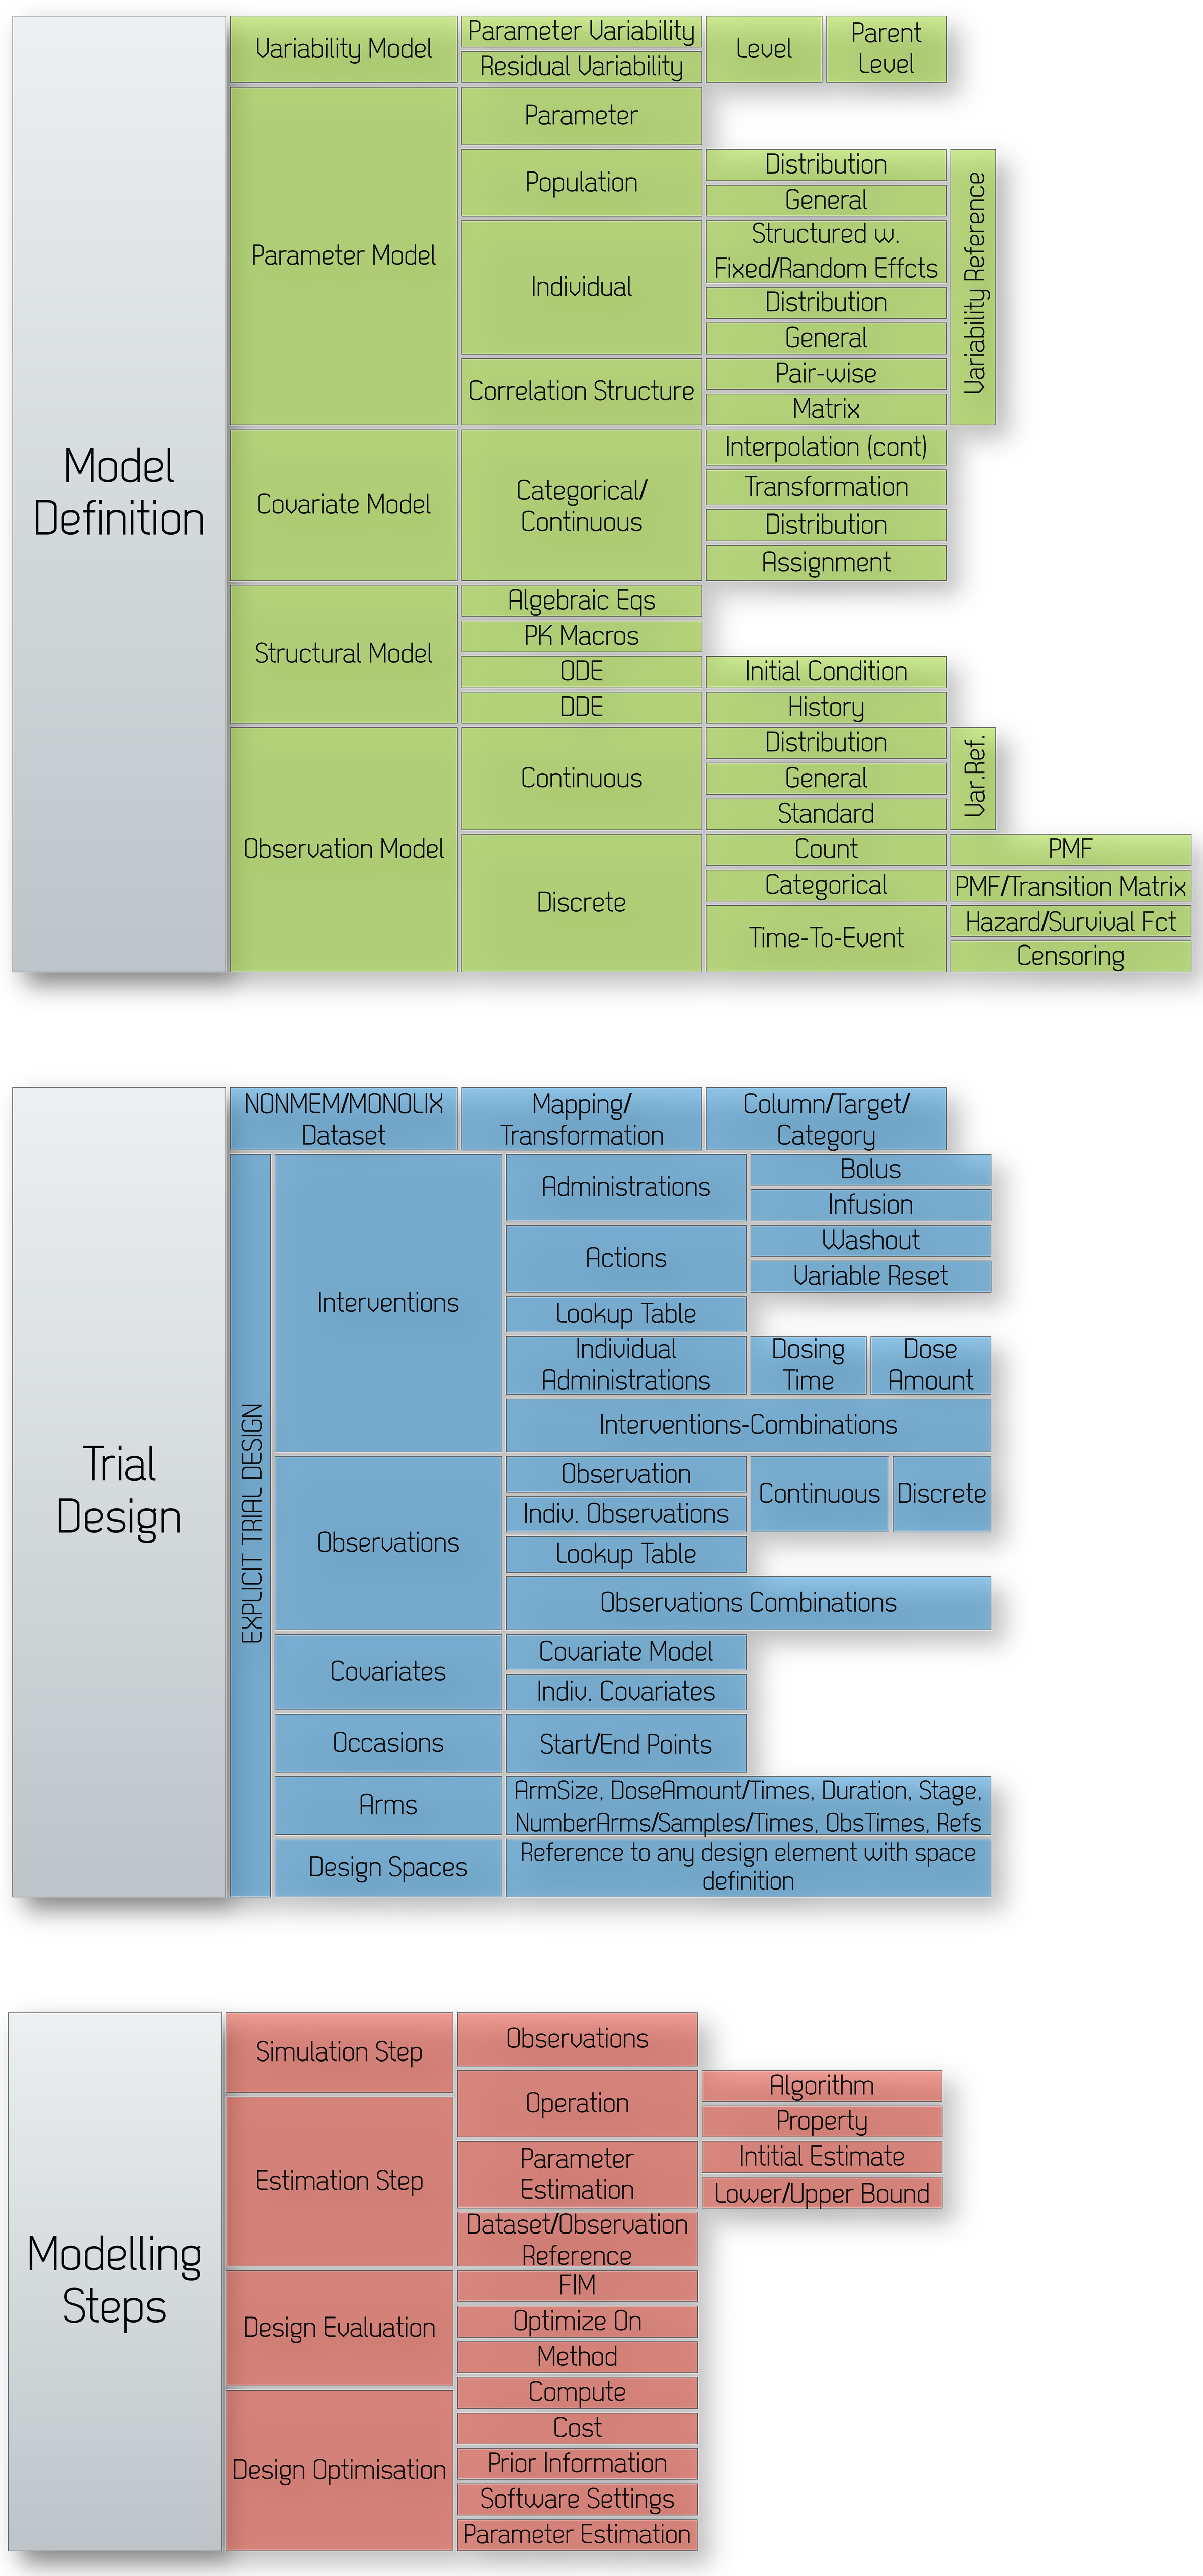
Model Definition
Variability Model
Parameter Variability
Residual Variability
Level
Parent Level
Parameter Model
Parameter
Population
Individual
Correlation Structure
Distribution
General
Structured w. Fixed/Random Effcts
Distribution
General
Pair-wise
Matrix
Variability Reference
Covariate Model
Categorical/ Continuous
Interpolation (cont)
Transformation
Distribution
Assignment
Structural Model
Algebraic Eqs
PK Macros
ODE
DDE
Initial Condition
History
Observation Model
Continuous
Distribution
General
Standard
Var.Ref.
Discrete
Count
Categorical
Time-To-Event
PMF
PMF/Transition Matrix
Hazard/Survival Fct
Censoring
Trial Design
NONMEM/MONOLIX Dataset
Mapping/ Transformation
Column/Target/ Category
EXPLICIT TRIAL DESIGN
Interventions
Administrations
Bolus
Infusion
Actions
Washout
Variable Reset
Lookup Table
Individual Administrations
Dosing Time
Dose Amount
Interventions-Combinations
Observations
Observation
Indiv. Observations
Lookup Table
Continuous
Discrete
Observations Combinations
Covariates
Covariate Model
Indiv. Covariates
Occasions
Start/End Points
Arms
ArmSize, DoseAmount/Times, Duration, Stage, NumberArms/Samples/Times, ObsTimes, Refs
Design Spaces
Reference to any design element with space definition
Modelling Steps
Simulation Step
Observations
Estimation Step
Operation
Algorithm
Property
Parameter Estimation
Intitial Estimate
Lower/Upper Bound
Dataset/Observation Reference
Design Evaluation
FIM
Optimize On
Method
Compute
Design Optimisation
Cost
Prior Information
Software Settings
Parameter Estimation

Figure 3: PharmML sections.

PharmML

- **Model definition, trial design and modelling steps** storing tool-agnostic exchange format.
- **Declarative** structure allows it to play a role as an interoperability hub, a *lingua franca*, between tools used in pharmacometrics.

SCOPE

- **Structural model** defined as a system of ordinary differential equation (ODE), algebraic equations, PK macros & delay differential equations (DDE).
- **Parameter model** allowing for implementation of a wide variety of parameter models.
- Discrete/continuous **covariates**, and if required their transformation, distribution, interpolation features.
- **Variability model** (nested hierarchy) capable of expressing complex random error structures.
- **Observation model** with flexible residual error model supporting untransformed or transformed continuous data.
- **Discrete data models** – supports count, categorical, time-to-event data models with Markov dependencies.
- **Trial design model** with support for common design patterns, drug administration types and encoding of experimental data needed for typical simulation or estimation tasks, such as dosing, observations and covariates. Design spaces for Optimal Design.
- **Hierarchical models/Bayesian inference** definition via assignment of distributions to any model parameter.
- Typical **modeling steps** such as estimation, simulation, design optimization/evaluation.

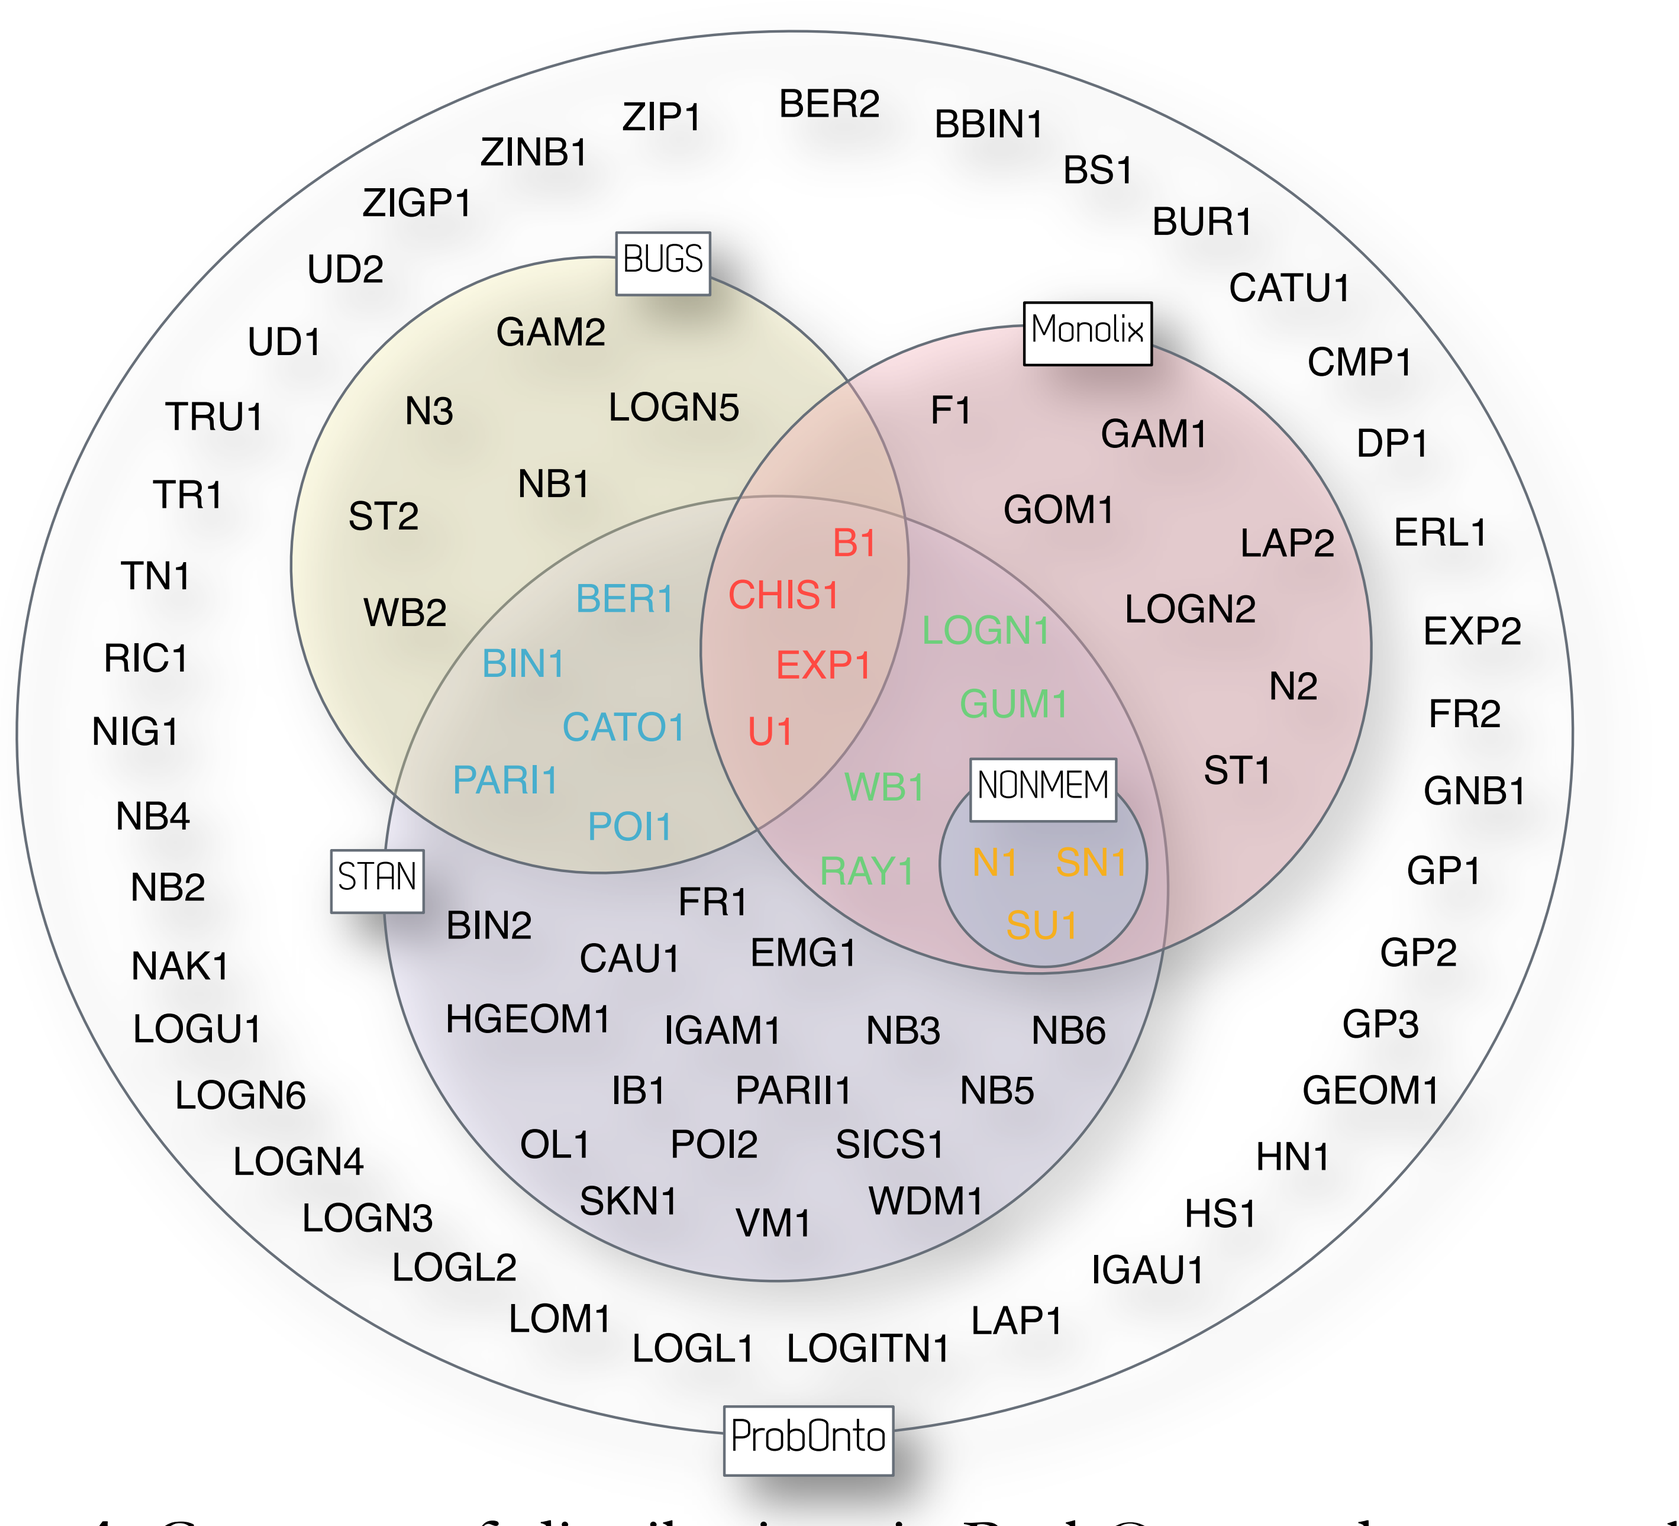
BUGS
Monolix
NONMEM
STAN
ProbOnto
ZIP1 BER2 BBIN1 ZINB1 BS1 ZIGP1 BUR1 UD2 CATU1 UD1 CMP1 TRU1 DP1 TR1 ERL1 TN1 EXP2 RIC1 FR2 NIG1 GNB1 NB4 GP1 NB2 GP2 NAK1 GP3 LOGU1 GEOM1 LOGN6 HN1 LOGN4 HS1 LOGN3 IGAU1 LOGL2 LAP1 LOM1 LOGL1 LOGITN1
GAM2 N3 LOGN5 NB1 ST2 WB2
F1 GAM1 GOM1 LAP2 LOGN2 N2 ST1
B1 CHIS1 EXP1 U1 BER1 BIN1 CATO1 PARI1 POI1
LOGN1 GUM1 WB1 RAY1
N1 SN1 SU1
BIN2 FR1 CAU1 EMG1 HGEOM1 IGAM1 NB3 NB6 IB1 PARII1 NB5 OL1 POI2 SICS1 SKN1 VM1 WDM1

Figure 4: Coverage of distributions in ProbOnto and target tools.

ProbOnto – knowledge base of probability distributions, featuring more than 100 uni- and multivariate distributions with their defining functions, characteristics, relationships and re-parameterization formulas [3]. Facilitates the encoding of distribution-based models, related functions and can be used for model annotation.

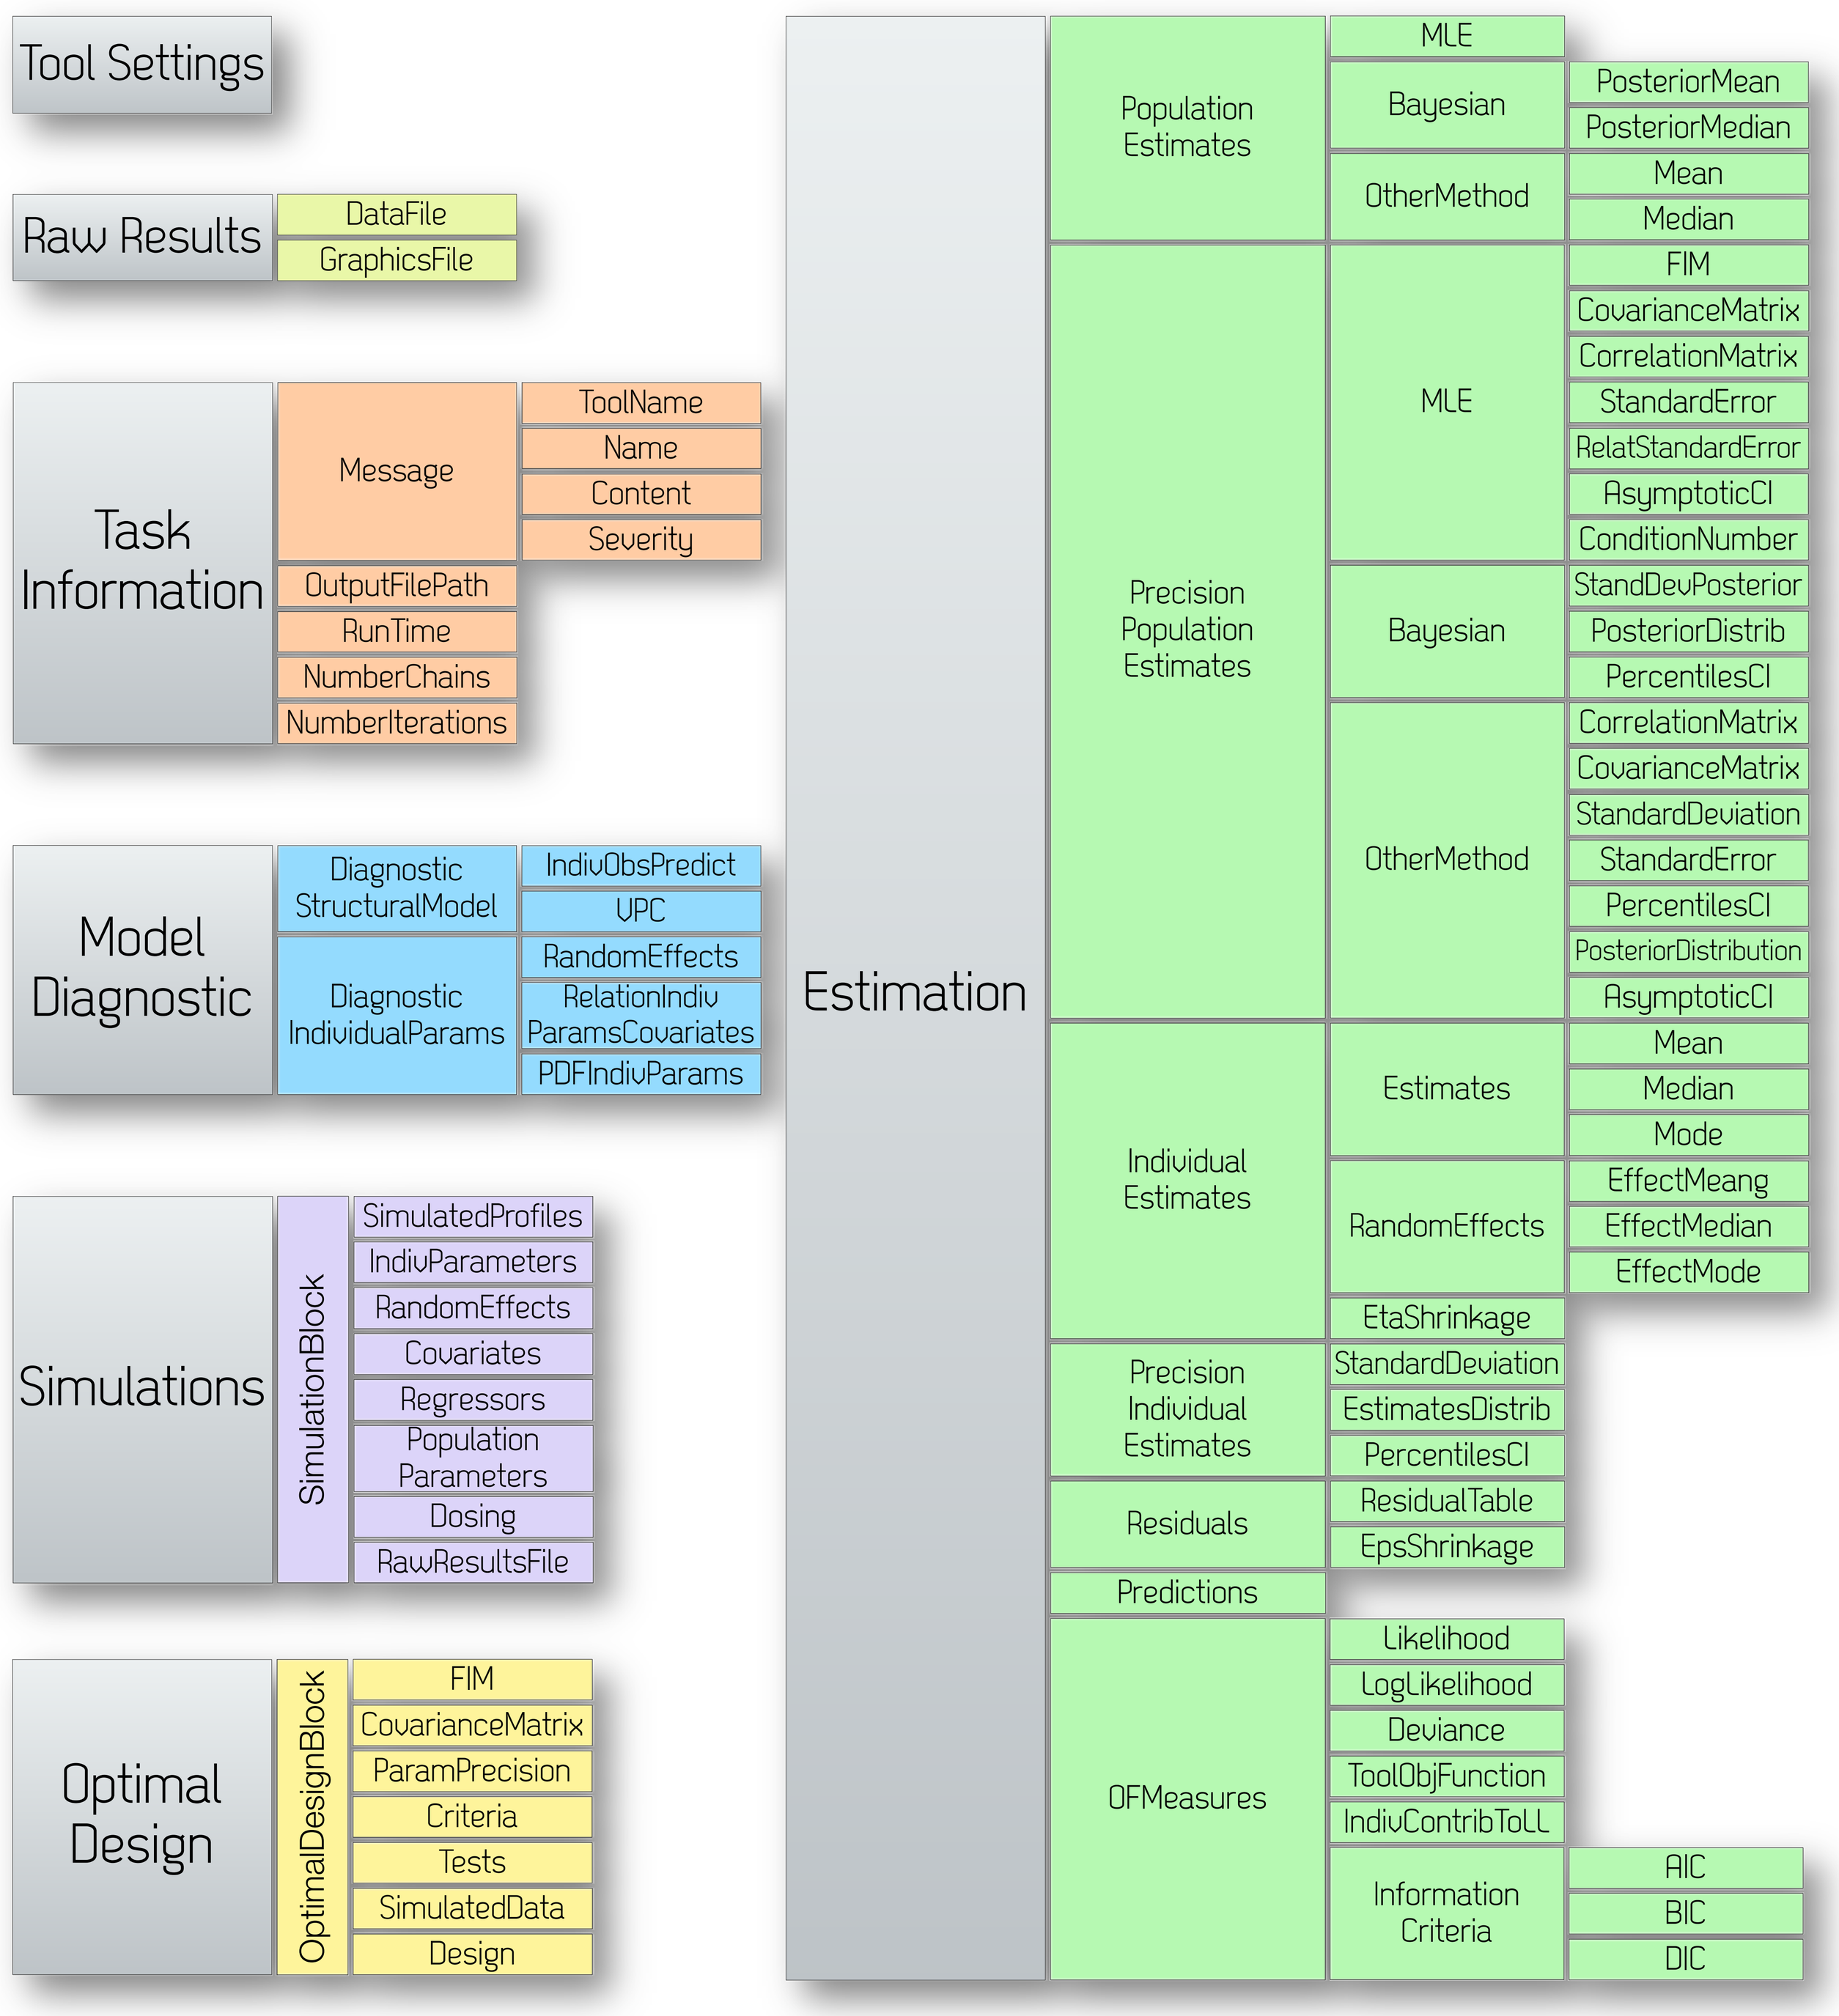
Tool Settings
Raw Results
DataFile
GraphicsFile
Task Information
Message
ToolName
Name
Content
Severity
OutputFilePath
RunTime
NumberChains
NumberIterations
Model Diagnostic
Diagnostic StructuralModel
Diagnostic IndividualParams
IndivObsPredict
VPC
RandomEffects
RelationIndiv ParamsCovariates
PDFIndivParams
Simulations
SimulationBlock
SimulatedProfiles
IndivParameters
RandomEffects
Covariates
Regressors
Population Parameters
Dosing
RawResultsFile
Optimal Design
OptimalDesignBlock
FIM
CovarianceMatrix
ParamPrecision
Criteria
Tests
SimulatedData
Design
Estimation
Population Estimates
MLE
Bayesian
PosteriorMean
PosteriorMedian
OtherMethod
Mean
Median
Precision Population Estimates
MLE
FIM
CovarianceMatrix
CorrelationMatrix
StandardError
RelatStandardError
AsymptoticCI
ConditionNumber
Bayesian
StandDevPosterior
PosteriorDistrib
PercentilesCI
OtherMethod
CorrelationMatrix
CovarianceMatrix
StandardDeviation
StandardError
PercentilesCI
PosteriorDistribution
AsymptoticCI
Individual Estimates
Estimates
Mean
Median
Mode
RandomEffects
EffectMeang
EffectMedian
EffectMode
EtaShrinkage
Precision Individual Estimates
StandardDeviation
EstimatesDistrib
PercentilesCI
Residuals
ResidualTable
EpsShrinkage
Predictions
OFMeasures
Likelihood
LogLikelihood
Deviance
ToolObjFunction
IndivContribToLL
Information Criteria
AIC
BIC
DIC

Figure 2: SO sections.

SO

- **Tool-independent** storage format for typical results of M&S analyses performed in DDMoRe target tools.
- Enables effective **data flow** across tasks to ensure optimal interactions among software tools and extends the modeling capabilities of the workflow.
- Facilitates **information retrieval** for post-processing and reporting, by allowing access to M&S results.

SCOPE

- **Tool settings, raw results and task information** sections carry the according information.
- **Estimation** section holds the results from MLE and Bayesian inference analyses for population and individual parameters and their precision.
- **Model Diagnostic** section holds information required for model diagnostic plots - related to structural model, residual error specification and individual parameters.
- **Simulation** section stores results from simulation tasks, such as time course of observations, population and individual parameters, random effects, covariates and dosing records for each subject.
- **Optimal Design** section holds data following design evaluation or optimization steps, e.g. FIM, covariance matrix, parameter values, their precision, information about the adopted criteria and performed tests.

References

[1] Swat, MJ., Moodie S., Wimalaratne S., et al. (2015). Pharmacometrics Markup Language (PharmML) – Opening New Perspectives for Model Exchange in Drug Development, Vol. 4, Issue 6, 316–319.
[2] Terranova, N. et al. (2016) Standard Output (SO) – Format Specification for Version 0.3.1. URL: ddmore.eu/projects/so-standard-output
[3] Swat, MJ., Grenon P., Wimalaratne S. (2016). ProbOnto: ontology and knowledge base of probability distributions. Bioinformatics, 1–3, doi: 10.1093/bioinformatics/btw170.

Acknowledgement: The research leading to these results has received support from the Innovative Medicines Initiative Joint Undertaking under grant agreement n° 115156, resources of which are composed of financial contribution from the European Union's Seventh Framework Programme (FP7/2007-2013) and EFPIA companies' in kind contribution. The DDMoRe project is also financially supported by contributions from Academic and SME partners.