Optimal two-stage design for a population pharmacokinetic study in children

Cyrielle Dumont (1,2), Marylore Chenel (2) and France Mentré (1)

(1) UMR 738 INSERM and University Paris Diderot, Paris, France

(2) Department of Clinical Pharmacokinetics, Institut de Recherches Internationales Servier, Paris, France.

Introduction

Pharmacokinetic (PK) studies in children

✦ Are mainly analysed by nonlinear mixed-effect models (NLMEM) [1,2] as recommended in guideline [3]
✦ Approaches based on the Fisher information matrix (M_F) [4] can be used to optimize their designs and are based on *a priori* information
✦ PK data in children are often not available and methods as allometry or PBPK are used to predict 'initial' PK parameters

➡ Adaptive designs [5,6], among which two-stage designs, are useful to provide flexibility and two-stage designs are easier to conduct

Objectives

1) To analyse concentration-time data obtained from PBPK simulations in children after oral absorption of a drug X in development
2) To develop and evaluate the impact of two-stage designs when children 'true' parameters are different from initial ones

Methods

1) Data and modelling

✦ Parent PK profiles obtained by simulation in scaling the existing PBPK model in adults to children using the software SIMCYP (version 9) [7]
✦ 100 children between 6 months and 18 years old
✦ Oral absorption of a dose equal to 0.1 mg/kg
✦ Simulated data analysed by NLMEM (FOCEI algorithm in NONMEM 7.2 [8])
✦ Dose and parameters per kg

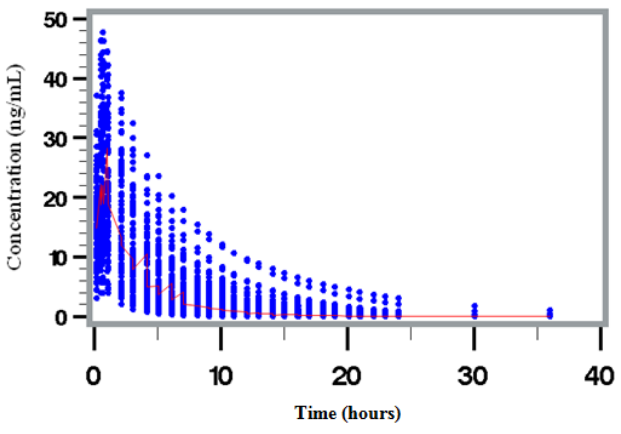
Concentration (ng/mL)
Time (hours)
0
10
20
30
40
50

Figure 1: Simulated pharmacokinetic profiles

2) Two-stage design

✦ Assumption here: same elementary design (ξ) for all subjects

✦ Notations

- Ψ_0: initial parameters
- Ψ^*: true parameters
- ξ_1: optimized design obtained with parameters Ψ_0 for N_1 subjects
- Ψ_1: estimated parameters from data Y_1 with design ξ_1 and N_1 subjects
- ξ_2: optimized design obtained with estimated parameters Ψ_1 for N_2 subjects
- Ψ_2: estimated parameters from data Y_2, obtained with design ξ_2 for N_2 subjects, and Y_1

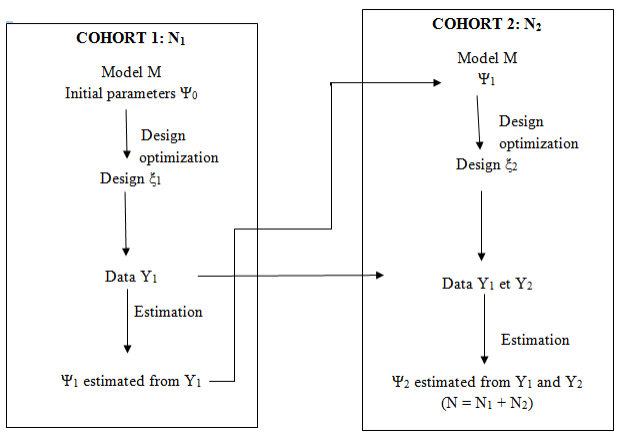
COHORT 1: N₁
Model M
Initial parameters Ψ₀
Design optimization
Design ξ₁
Data Y₁
Estimation
Ψ₁ estimated from Y₁
COHORT 2: N₂
Model M
Ψ₁
Design optimization
Design ξ₂
Data Y₁ et Y₂
Estimation
Ψ₂ estimated from Y₁ and Y₂
(N = N₁ + N₂)

Figure 2: Two-stage design

✦ M_F for a two-stage design
First stage: ξ_1 is the design which optimizes the following M_F

$$M_F(\Psi_0, N_1\xi) = N_1 M_F(\Psi_0, \xi)$$

Second stage: ξ_2 is the design which optimizes the following M_F using estimated Ψ_1

$$M_F(\Psi_1, N_1\xi_1 + N_2\xi) = N_1 M_F(\Psi_1, \xi_1) + N_2 M_F(\Psi_1, \xi)$$

3) Simulation study

✦ Application to drug X for a trial with N = 60 children
✦ 'Initial' parameters Ψ_0 are different from the 'true' parameters Ψ^*
✦ Steady-state bid and dose equal to 0.1 mg/kg
✦ Optimization
- according to the D-optimality criterion with PFIM [9,10] in R
- 5 sampling times among the possible sampling times 0.25, 0.5, 0.75, 1, 2, 3, 4, 5, 6, 7, 8, 9, 10, 11, 12

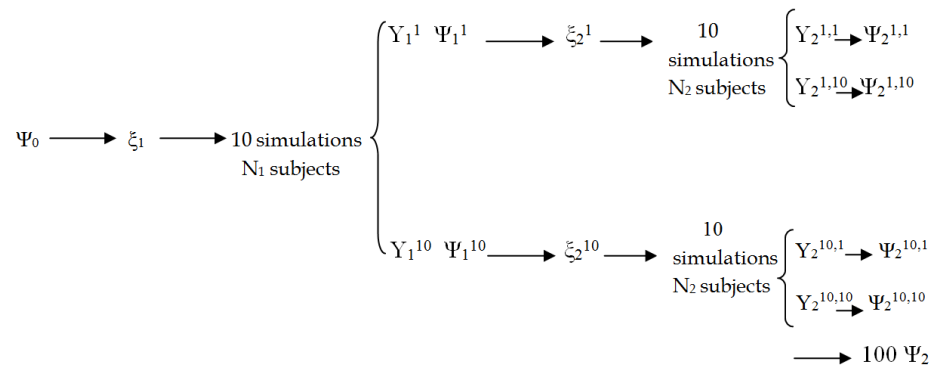
Ψ₀
ξ₁
10 simulations
N₁ subjects
Y₁¹ Ψ₁¹
ξ₂¹
10 simulations
N₂ subjects
Y₂¹·¹ Ψ₂¹·¹
Y₂¹·¹⁰ Ψ₂¹·¹⁰
Y₁¹⁰ Ψ₁¹⁰
ξ₂¹⁰
10 simulations
N₂ subjects
Y₂¹⁰·¹ Ψ₂¹⁰·¹
Y₂¹⁰·¹⁰ Ψ₂¹⁰·¹⁰
100 Ψ₂

Figure 3: Simulation plan

✦ Estimation of Ψ_1 and Ψ_2 with saemix [11] in R
✦ Comparison of the relative bias and relative root mean square error (RMSE) for the estimated Ψ_1 (first stage) and the estimated Ψ_2 (second stage)

Results

1) PK model and parameters estimated for the parent drug

✦ Two-compartment model (five parameters)

Parameters (units)	Estimates (RSE %)	Ψ_0	Ψ^*
ka (h^{-1})	3.0 (20.0)	3.0	3.0
CL ($L.h^{-1}.kg^{-1}$)	1.3 (7.5)	1.3	3.4
V1 ($L.kg^{-1}$)	2.1 (13.0)	2.1	2.1
Q ($L.h^{-1}.kg^{-1}$)	0.91 (8.5)	0.91	0.91
V2 ($L.kg^{-1}$)	1.3 (7.3)	1.3	1.3
ω^2_{ka}	1.4 (14.5)	1.4	1.4
ω^2_{CL}	0.53 (11.4)	0.53	0.53
ω^2_{V1}	1.1 (13.9)	1.1	1.1
COV/CORR(CL ;V$_1$)	0.55 (14.7) / 0.73 (7.1)	0.00	0.00
σ_{inter} ($\mu g.L^{-1}$)	0.00060 (17.5)	0.13	0.13
σ_{slope}	0.12 (8.1)	0.12	0.12

Table 1: Population PK parameter values

2) Two-stage design

Ξ_0: N = 60 and ξ_0 = 0.25, 0.75, 2, 5, 12 (optimal for Ψ_0)
Ξ^*: N = 60 and ξ^* = 0.25, 0.75, 2, 4, 7 (optimal for Ψ^*)
Ξ_{2S}: N_1 = 30 with ξ_0 and N_2 = 30 with ξ^*

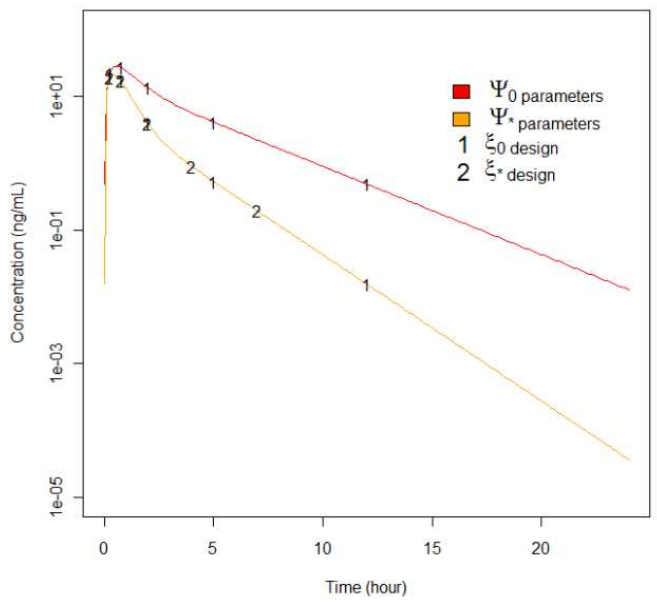
Ψ₀ parameters
Ψ* parameters
1 ξ₀ design
2 ξ* design
Concentration (ng/mL)
1e+01
1e-01
1e-03
1e-05
0
5
10
15
20
Time (hour)

Figure 4: Mean profiles in semi-log scale

Parameters / Design	Ψ_0 / Ξ_0	Ψ^* / Ξ_0	Ψ^* / Ξ^*	Ψ^* / Ξ_{2S}
Criterion (Determinant)$^{1/10}$	78.8	47.0	61.7	58.6
Predicted RSE (%)				
ka	20.8	**63.2**	27.9	**30.8**
CL	9.5	9.5	9.5	9.5
V1	17.5	**60.1**	24.8	27.9
Q	19.9	**74.1**	19.8	25.8
V2	13.0	27.0	11.2	12.6
ω^2_{ka}	20.0	24.3	23.4	23.8
ω^2_{CL}	18.4	18.5	18.5	18.5
ω^2_{V1}	19.2	23.2	22.5	22.8
σ_{inter}	24.1	8.8	12.1	10.1
σ_{slope}	10.4	13.5	12.9	13.0

Table 2: Design influence on parameter estimation (PFIM predictions)

➡ One-stage design Ξ_0 when parameters are different (Ψ^*) shows a loss of efficiency. The 'ideal' two-stage design, with $N_1 = N_2 = 30$, allows to partly compensate this loss of information

3) Simulation study

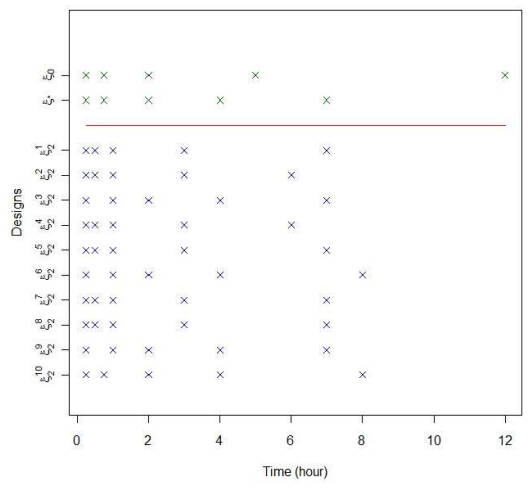
Designs
Time (hour)
0
2
4
6
8
10
12

Figure 5: The ten second-stage designs (ξ_2) optimized from the ten estimated Ψ_1

➡ Seven designs are different and the other are identical. None of them are identical to ξ^* nor ξ_0

Parameters	First stage (Ψ_1) Relative bias (%)	First stage (Ψ_1) Relative RMSE (%)	Second stage (Ψ_2) Relative bias (%)	Second stage (Ψ_2) Relative RMSE (%)
ka	24.2	**94.0**	1.87	**43.6**
CL	-4.15	9.31	-3.73	6.52
V1	-26.2	**40.9**	-24.1	**32.1**
Q	-3.79	12.2	-2.38	10.0
V2	-1.15	13.3	0.627	9.07
ω^2_{ka}	21.7	**131**	-6.44	**55.0**
ω^2_{CL}	-7.49	18.8	-9.60	16.8
ω^2_{V1}	18.1	49.5	32.6	**66.7**
σ_{inter}	0.420	13.0	0.420	8.90
σ_{slope}	-2.07	14.9	-2.07	10.2

Table 3: Relative bias (%) and RMSE (%) for estimated Ψ_1 and Ψ_2

➡ Relative bias and RMSE are lower for Ψ_2 than for Ψ_1. The two-stage design improves the estimation

Conclusion

✦ Two articles in other contexts [12,13] discussed that two-stage designs could be more efficient than fully adaptive designs
✦ Two-stage designs are a good alternative for designing PK studies in children

✦ Perspectives
- To study the impact of the two-stage design with the metabolite
- To investigate the choice of the ratio of the sample sizes between the two stages

References

[1] Mentré F, Dubruc C, Thénot JP (2001). *J Pharmacokinet Pharmacodyn*, 28: 299-319.
[2] Tod M, Jullien V, Pons G (2008). *ClinPharmacokinet*, 47: 231-243.
[3] EMEA (2006). Guideline on the role of pharmacokinetics in the development of medicinal products in the paediatric population. *Scientific guideline.*
[4] Mentré F, Mallet A, Baccar D (1997). *Biometrika*, 84: 429-442.
[5] Foo LK, Duffull S (2012). *Pharmaceutical Research*, 29: 1530-1543.
[6] Zamuner S, Di Iorio VL, Nyberg J, Gunn RN , Cunningham VJ, Gomeni R and Hooker AC (2010). *ClinPharmacol & Ther*, 87: 563–571.
[7] Jamei M , Marciniak S, Feng K, Barnett A, Tucker G, Rostami-Hodjegan A (2009). *Expert Opin Drug Metab Toxicol*, 5: 211-223.
[8] Dumont C, Chenel M, Mentré F (2011). *Population Approach Group in Europe*, Abstr 2160 [www.page-meeting.org/?abstract=2160].
[9] Bazzoli C, Retout S, Mentré F (2010). *Comput Methods Programs Biomed*, 98: 55-65.
[10] www.pfim.biostat.fr.
[11] Comets E, Lavenu A, Lavielle M (2011). *Population Approach Group in Europe*, Abstr 1695 [www.page-meeting.org/default.asp?abstract=2173].
[12] Federov V, Wu Y, Zhang R (2012). *Stat Med*, 31: 217-234.
[13] Chen TT (1997). *Stat Med*, 16: 2701-2711.