FACULTY OF HEALTH AND MEDICAL SCIENCES

Eva Sverrisdóttir[1], Mads Kreilgaard[2], Anne Brokjær[3], Katja Kjeldgaard Miltersen[4], Anne Estrup Olesen[1,3], Lona Louring Christrup[1], Asbjørn Drewes[3,5]
[1]Department of Drug Design and Pharmacology, Faculty of Health Sciences, University of Copenhagen, Universitetsparken 2, DK-2100 Copenhagen, Denmark; [2]Novo Nordisk, Måløv, Denmark; [3]Mech-Sense, Department of Gastroenterology & Hepatology, Aalborg University Hospital, Mølleparkvej 4, DK-9000 Aalborg, Denmark; [4]Klinisk Biokemisk Afdeling, Aarhus Universitetshospital, Palle Juul-Jensens Boulevard 99, DK-8200 Aarhus N, Denmark, [5]Department of Clinical Medicine, Aalborg University, Sdr. Skovvej 11, DK-9000 Aalborg, Denmark

Modelling drug-drug interactions between morphine and methylnaltrexone

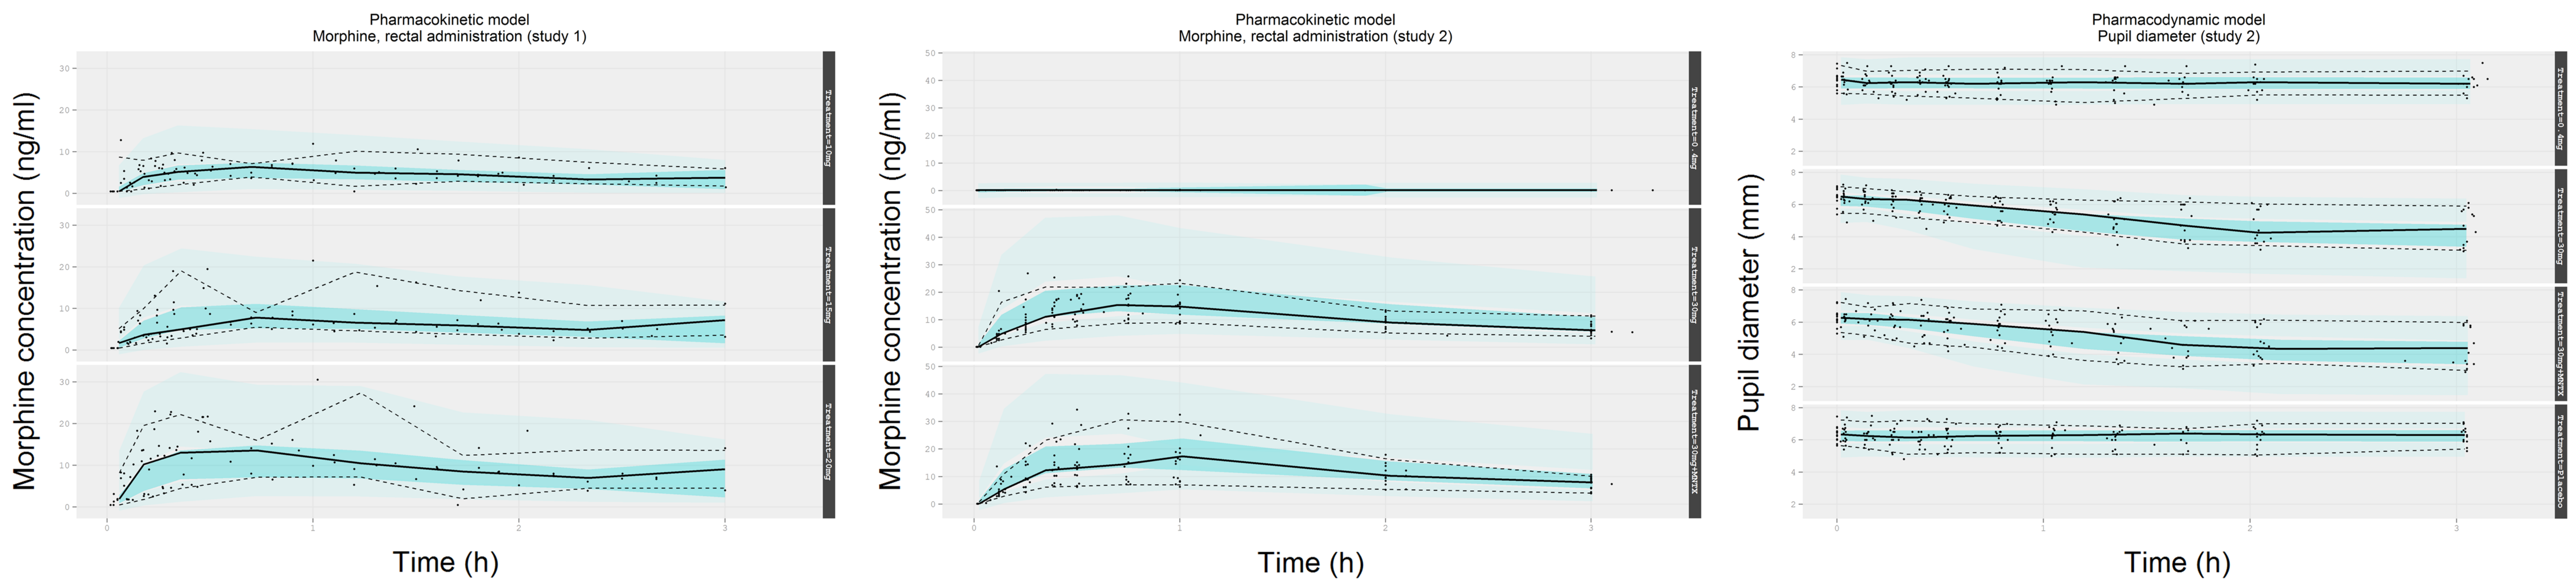
Pharmacokinetic model
Morphine, rectal administration (study 1)
Morphine concentration (ng/ml)
Time (h)
Pharmacokinetic model
Morphine, rectal administration (study 2)
Morphine concentration (ng/ml)
Time (h)
Pharmacodynamic model
Pupil diameter (study 2)
Pupil diameter (mm)
Time (h)

Visual predictive checks (VPCs) of the morphine pharmacokinetic (PK) model for rectal administration of morphine in study 1 and study 2, and the pharmacodynamic (PD) model for morphine effect on pupil diameter. The VPCs are stratified on treatment arm. The black solid line is the observed population median, the dashed lines are the 5th and the 95th percentiles of the observed data. The dark coloured area is the 95% confidence intervals for the predicted population median and the light areas are the 95% confidence intervals of the predicted 5th and 95th percentiles. The VPCs are based on 2000 simulations. MNTX: methylnaltrexone.

OBJECTIVES

- **Establish the population PK-PD relationship of morphine after rectal administration**
- **Use pupil diameter as a central biomarker for morphine response**
- **Investigate if methylnaltrexone (peripheral opioid receptor antagonist) interacts with central morphine response**

BACKGROUND

- Morphine is the mainstay of treatment of moderate to severe pain
- Rectal administration of morphine is a cheap and non-invasive alternative to oral administration of morphine
- Many morphine-induced adverse effects occur in the gastrointestinal tract
- Methylnaltrexone is a peripheral μ opioid receptor antagonist
- Pupil diameter is thought to be a useful central biomarker for morphine response [1]

Parameter	Estimate	IIV	IOV	Description
		Morphine pharmacokinetic parameters		
$CL \cdot \left(\frac{WT}{70kg}\right)^{\frac{3}{4}} (L/h)$	74.2	42.2		Clearance
$V2 \cdot \left(\frac{WT}{70kg}\right)^{\frac{4}{4}} (L)$	8.56			Volume of distribution in central compartment
$Q \cdot \left(\frac{WT}{70kg}\right)^{\frac{3}{4}} (L/h)$	161			Intercompartmental clearance
$V3 \cdot \left(\frac{WT}{70kg}\right)^{\frac{4}{4}} (L)$	121			Volume of distribution in peripheral compartment
$LGT\ (\%)$	-1.32 (F=21%)	37.4	30.4[a]	Logit transformed bioavailability
$MTT(h)$	0.496	64.6	58.7	Transit absorption rate constant
$k_a\ (h^{-1})$	1.48			First-order absorption rate constant
$\sigma_{prop1}\ (\%)$	23			Proportional error, study 1
$\sigma_{add1}\ (ng/ml)$	0.767			Additive error, study 1
$\sigma_{prop2}\ (\%)$	12.5			Proportional error, study 2
$\sigma_{add2}\ (ng/ml)$	-1.25			Additive error, study 2
		Morphine pharmacodynamic parameters		
$Baseline\ (mm)$	6.25	9.1	3	Pupil diameter baseline
$EC_{50}\ (ng/ml)$	12.3	47.6	13.4	Morphine concentration for 50% effect
$k_{e0}(h^{-1})$	0.22			Equilibration rate constant
$Hill$	1.28			Shape coefficient
$\sigma_{add}\ (mm)$	-0.261			Additive error for morphine response

[a]Inter-study variability, IIV: inter-individual variability, IOV: inter-occasion variability

MATERIALS AND METHODS

- Randomized, placebo controlled, double-blinded, four-way cross-over study
- 30 mg morphine hydrochloride administered rectally
- 15 healthy male volunteers
- 3 h sampling of plasma and pupil diameter
- PK samples from a previous study included [2]
- Sequential approach:
 - PK data fitted alone
 - PK populations parameters fixed in PD modelling
- Variability tested:
 - Inter-individual
 - Inter-occasion
 - Inter-study
- Covariates tested: age, weight, height, co-administration of methylnaltrexone
- Software:
 - NONMEM 7.3
 - PsN 3.7.6
 - R
 - Pirana

RESULTS AND CONCLUSION

- The effect of morphine on pupil diameter was described with an inhibitory sigmoid E_{max} model
- Baseline and EC_{50} varied between individuals and occasions
- Equilibration half-life was 3.15 h
- No covariate relationships identified for morphine PD
- Co-administration of methylnaltrexone did not affect morphine PK or central PD effect

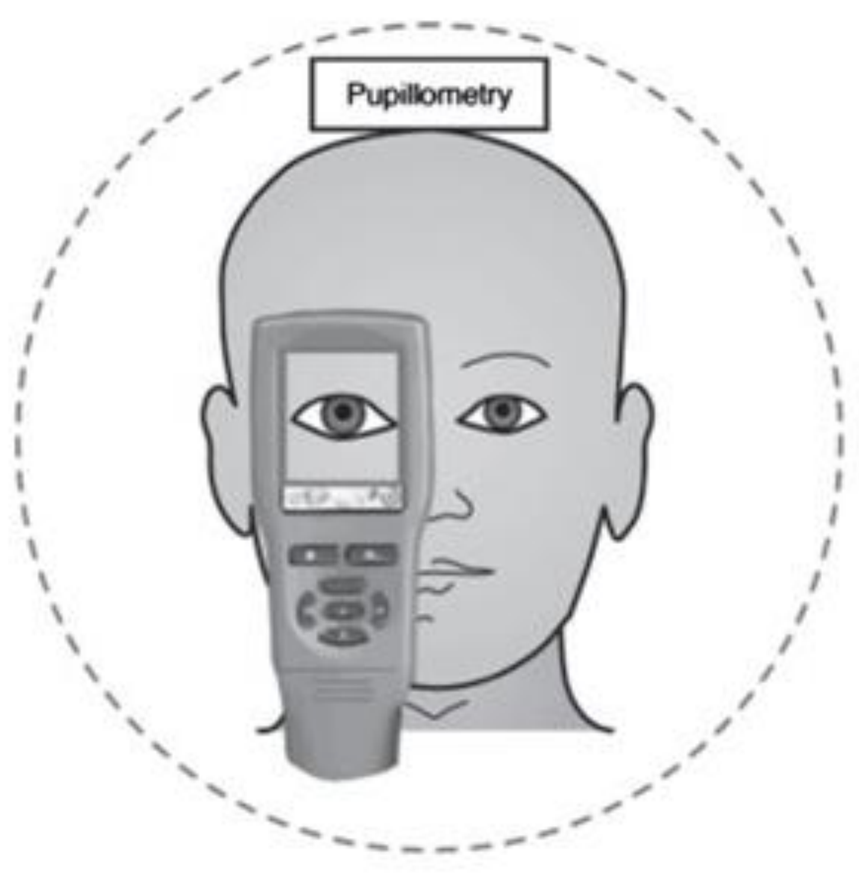
Pupillometry

Pupil diameter was measured at baseline and repeatedly until 3 h after morphine administration. A sampling matrix was used with 12 samples per individual. Figure adapted from Brokjaer et al. 2015 [3].

REFERENCES

[1] Skarke, C., Darimont, J., Schmidt, H., Geisslinger, G., Lötsch, J., 2003. Analgesic effects of morphine and morphine-6-glucuronide in a transcutaneous electrical pain model in healthy volunteers. Clin. Pharmacol. Ther. 73, 107-121. 10.1067/mcp.2003.5
[2] Brokjaer A, Kreilgaard M, Olesen AE, et al. Population pharmacokinetics of morphine and morphine-6-glucuronide following rectal administration--a dose escalation study. *Eur J Pharm Sci*. 2015;68:78-86. doi: 10.1016/j.ejps.2014.12.004
[3] Brokjaer, A., Olesen, A.E., Christrup, L.L., Dahan, A., Drewes, A.M., 2015. The effects of morphine and methylnaltrexone on gastrointestinal pain in healthy male participants. Neurogastroenterol. Motil. 27, 693-704. 10.1111/nmo.12545

ACKNOWLEDGEMENTS

The experimental work was carried out at Mech-Sense, Aalborg University Hospital. The study was supoorted by The Innovation Fund Denmark.