Population Pharmacokinetics of Eltrombopag in Healthy Subjects and Patients with Chronic Idiopathic Thrombocytopenic Purpura

Ekaterina Gibiansky (1), Jianping Zhang (2), Daphne Williams (2,3), Zhao Wang (1), Daniele Ouellet (2)

(1) ICON Development Solutions, Ellicott City, MD, USA; (2) GlaxoSmithKline, Research Triangle Park, NC, USA;
(3) Current Address: Bristol-Myers Squibb, Princeton, NJ, USA

A Symbol of Excellence

BACKGROUND

Eltrombopag is an orally bioavailable small molecule agonist of thrombopoietin receptor that has been recently approved in the USA for treatment of chronic idiopathic (immune) thrombocytopenic purpura (ITP). The aim of this analysis was to develop a population pharmacokinetic (PK) model of eltrombopag, predict steady-state exposure at therapeutic doses, identify demographic/covariate factors influencing eltrombopag exposure and quantify relative impact of these covariates.

DATA

Three Phase 1 studies: Total 111 healthy volunteers, 18-49 years old, weight 49-95 kg.

- 13% females, 28% East Asian (all males), 8% African-Americans, 50% administered capsules.
 - Doses of 5-200 mg, administered as a single dose and/or QD for 5-10 days.
 - Rich PK sampling - 3627 eltrombopag concentrations.

88 ITP patients (83 from Phase 2 study, 5 from Phase 3 study), 18-85 years old, weight 43-122 kg.

 - 65% females, 20% East Asian (of them 72% females), 100% administered tablets, 27% corticosteroid use.
 - Doses of 30-75 mg QD administered for up to 6 weeks.
 - Mix of sparse and serial PK sampling – 466 eltrombopag concentrations.

METHODS

Population PK Modeling

- Modeling was performed using a mixed-effects modeling approach with the first-order conditional method (FOCEI) of NONMEM VI.
- Model selection was driven by the data and was based on evaluation of goodness-of-fit plots, successful convergence, plausibility and precision of parameter estimates, and the minimum objective function value.

Covariate modeling

- Covariates selected based on the representation, range and correlation of covariate values in the dataset, scientific interest, mechanistic plausibility, and exploratory graphics.
- Covariates chosen for modeling: Weight, Population (Patients/healthy volunteers), Race, Gender, estimated creatinine clearance (CRCL), Age, Concomitant use of corticosteroids, Formulation, and Low Dose (< 20 mg, standing for 5 mg capsule formulation).
- Covariates modeled multiplicatively as power function.
 - $TVP_i = \theta_1{*}(COV_i/COV_{Ref})^{\theta 2}$ *for continuous covariates*
 - $TVP_i = \theta_1{*}\ \theta_2^{IND_i}$ *for categorical covariates*
- The full model approach was implemented: all apriori chosen covariate-parameter relationships of interest were entered in the model simultaneously, and parameters were estimated. Insignificant or poorly estimated covariates (based on confidence intervals (CI) of parameter estimates) could then be excluded from the model.
- The full model did not simultaneously include highly correlated covariates; therefore several full models (with one of the competing correlated covariates) were investigated.

Model evaluation

- Visual predictive check simulations (VPC), overall and stratified by dose and study.
- Bootstrap analysis stratified by covariates.

RESULTS

Base model

2 compartment linear model with dual sequential first-order absorption, absorption lag-time, inter-occasion variability in absorption, and higher residual variability during absorption phase (Table 1).

Covariate model

- CL/F and Q/F, and Vc/F and Vp/F increased with Weight as power model (Figure 1).
 - Change from 26% lower to 41% higher than for a 70-kg individual (weight range of 43 to 122 kg).
 - Relationships shallower than allometric, although CI included (for CL/F) or nearly included (for Vc/F) allometric values.
- CL/F was influenced by East Asian Race > Corticosteroid intake > Sex > Population.
 - 33% (CI 26%, 41%) lower in East Asians compared to all other races,
 - 26% (CI 7%, 45%) lower in patients taking corticosteroids concomitantly,
 - 19% (CI 7%, 31%) lower in females,
 - 17% (CI 0%, 34%) higher in healthy volunteers than in ITP patients.
- Due to correlations between covariates, AUCss increased 87% in East Asian patients, 50% in female patients, and 31% in patients taking corticosteroids, compared to all patients, and decreased 35% in healthy volunteers compared to patients.
- CL/F and Vc/F were 68% and 55% higher at low doses (< 20 mg).
 - Possibly due to lower bioavailability with 5 mg capsule formulation.
- Mild renal impairment (CRCL < 80), and age in ITP patients did not influence PK.
 - Point estimates near null values and narrow CIs that include null values.
- African-Americans: No trends, but insufficient information.
 - Point estimates near null values, no decrease in OFV, but wide CIs.

Model evaluation

- Graphical diagnostics (DV vs PRED, IPRED; WRES, IWRES vs PRED, TIME, TAD; distributions and correlations of random effects, overall and stratified by study) did not show any deficiencies (Figure 2).
- All covariate dependences observed in the base model were accounted for in the final model (Figure 3). No unaccounted relationships remained.
- VPC: no bias evident; approximately 10% of observations outside 90% prediction intervals for each of doses and studies (11% overall).
- Nearly identical Nonmem and bootstrap point estimates and confidence intervals (Table 1).

CONCLUSIONS

The developed population pharmacokinetic model identified and quantified patient characteristics predictive of eltrombopag exposure, and enabled further analysis to characterize pharmacokinetic-pharmacodynamic relationships.

Table 1 *Parameter Estimates of Final Population Pharmacokinetic Model*

Parameter [Units]	NONMEM Estimates				Bootstrap Estimates [a]	
	Point Estimate	%RSE	95% CI		Median	95% CI
CL/F [L/hr]	0.668	8.19	0.561-0.775		0.663	0.571-0.786
Vc/F [L]	8.76	3.61	8.14-9.38		8.72	8.13-9.43
Vp/F [L]	11.3	5.63	10.1-12.5		11.4	10.1-12.8
Q/F [L/hr]	0.399	4.81	0.361-0.437		0.4	0.364-0.436
Ka1 [hr^{-1}]	0.503	7.38	0.43-0.576		0.501	0.428-0.58
Ka2 [hr^{-1}]	10.4	18.8	6.58-14.2		10.3	6.07-17
ALAG1 [hr]	0.457	1.82	0.441-0.473		0.457	0.438-0.471
MTIME [hr]	1.94	1.22	1.89-1.99		1.94	1.86-1.98
σProp ~ITP	1.42	8.66	1.18-1.66		1.41	1.17-1.64
σProp ~TAD<4hr	1.4	5.40	1.25-1.55		1.4	1.26-1.57
CL/F ~WT, Q/F~WT	0.616	13.9	0.449-0.783		0.605	0.312-0.93
Vc/F ~WT, Vp/F~WT	0.617	30.0	0.254-0.98		0.611	0.24-1
CL/F ~DOSE<20 mg	1.68	6.67	1.46-1.9		1.69	1.47-1.9
Vc/F ~DOSE<20 mg	1.55	6.00	1.37-1.73		1.55	1.38-1.74
CL/F ~Healthy	1.17	7.6	0.996-1.34		1.17	1.01-1.35
CL/F ~Asian	0.665	5.74	0.59-0.74		0.662	0.582-0.754
CL/F ~Female	0.808	7.54	0.689-0.927		0.808	0.69-0.937
CL/F ~CORT	0.742	13.0	0.553-0.931		0.748	0.572-0.953
Inter-individual or inter-occasion variability				**CV% or R [b]**		
ω^2_{CL}	0.165	17.3	0.109-0.221	CV= 40.6%	0.16	0.115-0.211
Covar $\omega_{CL}\ \omega_{Vc}$	0.113	22.7	0.0628-0.163	R= 0.743	0.111	0.0728-0.161
ω^2_{VC}	0.14	22.4	0.0787-0.201	CV= 37.4%	0.139	0.0929-0.203
$\omega^2_{IOV\ Ka}$	1.62	11.0	1.27-1.97	CV= 127%	1.64	1.32-2.04
Residual variability				**CV% or SD**	**Median**	**95% CI**
σ^2_{prop}	0.0433	10.5	0.0344-0.0522	CV= 20.8%	0.0427	0.034-0.0524
σ^2_{add}	899	42.5	148-1630	SD= 29.8	891	353-2100

a. From 1345 successfully completed bootstrap runs; b. Correlation coefficient

Abbreviations: %RSE: percent relative standard error of the estimate; CL/F = apparent clearance; Vc/F = volume of central compartment; Ka1 and Ka2 = absorption rate constants prior to and after MTIME; Q/F = inter-compartmental clearance; Vp/F = volume of peripheral compartment; ALAG1 = lag-time; σ^2_{prop} and σ^2_{add} = proportional and additive components of the residual error model; σProp ~ITP and σProp ~TAD<4hr = factors of proportional error for ITP subjects and for TAD < 4 hours (absorption time); CI= confidence interval; R= correlation coefficient; ω^2_{CL} and ω^2_{Vc} = variances of random effect on CL/F and Vc/F; Covar ωCL, ωVc = correlation between covariances of CL/F and Vc/F random effects; $\omega^2_{IOV\ Ka}$ = variance of random IOV effect on Ka1; CV= coefficient of variation; SD = standard deviation

The reference population for PK parameters CL/F and Vc/F are 70 kg Caucasian male ITP patients not taking corticosteroids.

Figure 1 *Dependence of Oral Clearance on Weight for Different Subpopulations*

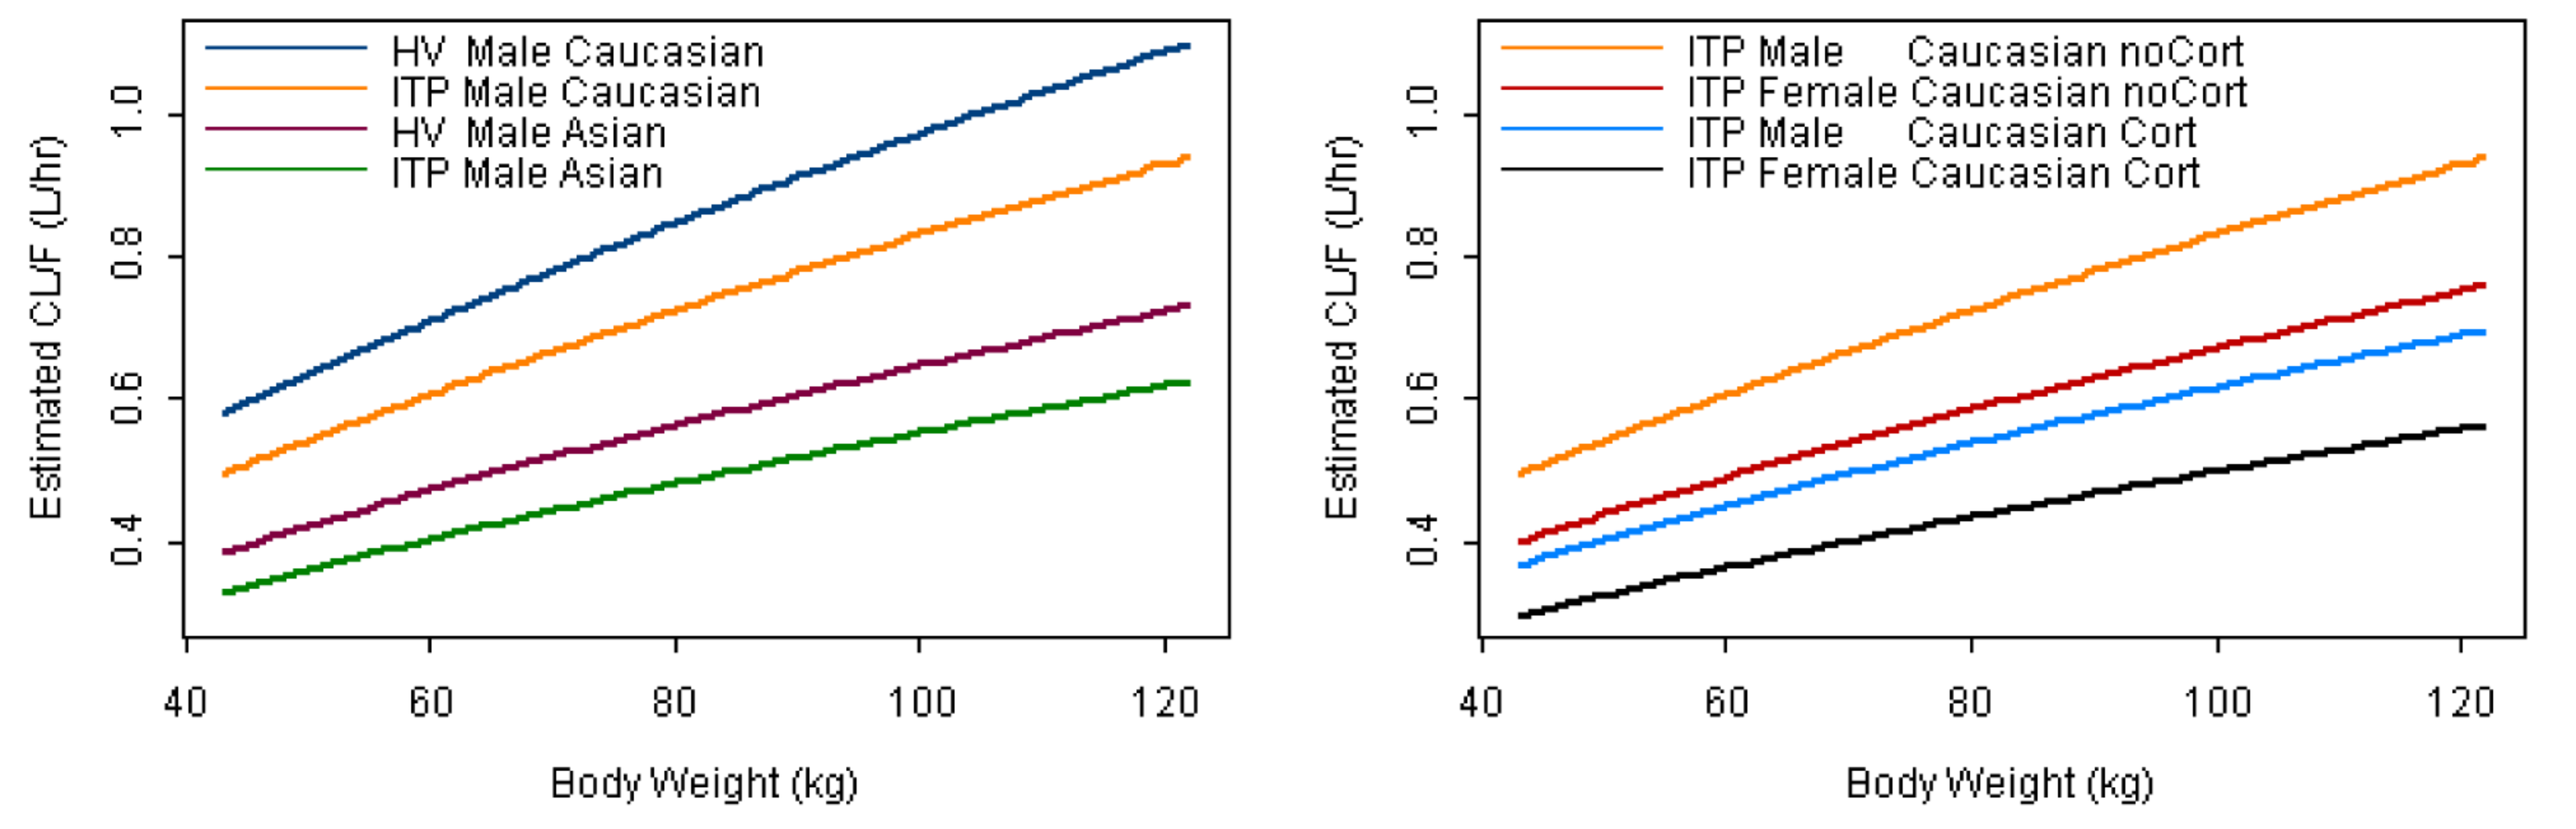
HV Male Caucasian
ITP Male Caucasian
HV Male Asian
ITP Male Asian
ITP Male Caucasian noCort
ITP Female Caucasian noCort
ITP Male Caucasian Cort
ITP Female Caucasian Cort
Estimated CL/F (L/hr)
Body Weight (kg)

Figure 2 *Goodness-of-fit plots for the Final Model*

TAD= time after last dose

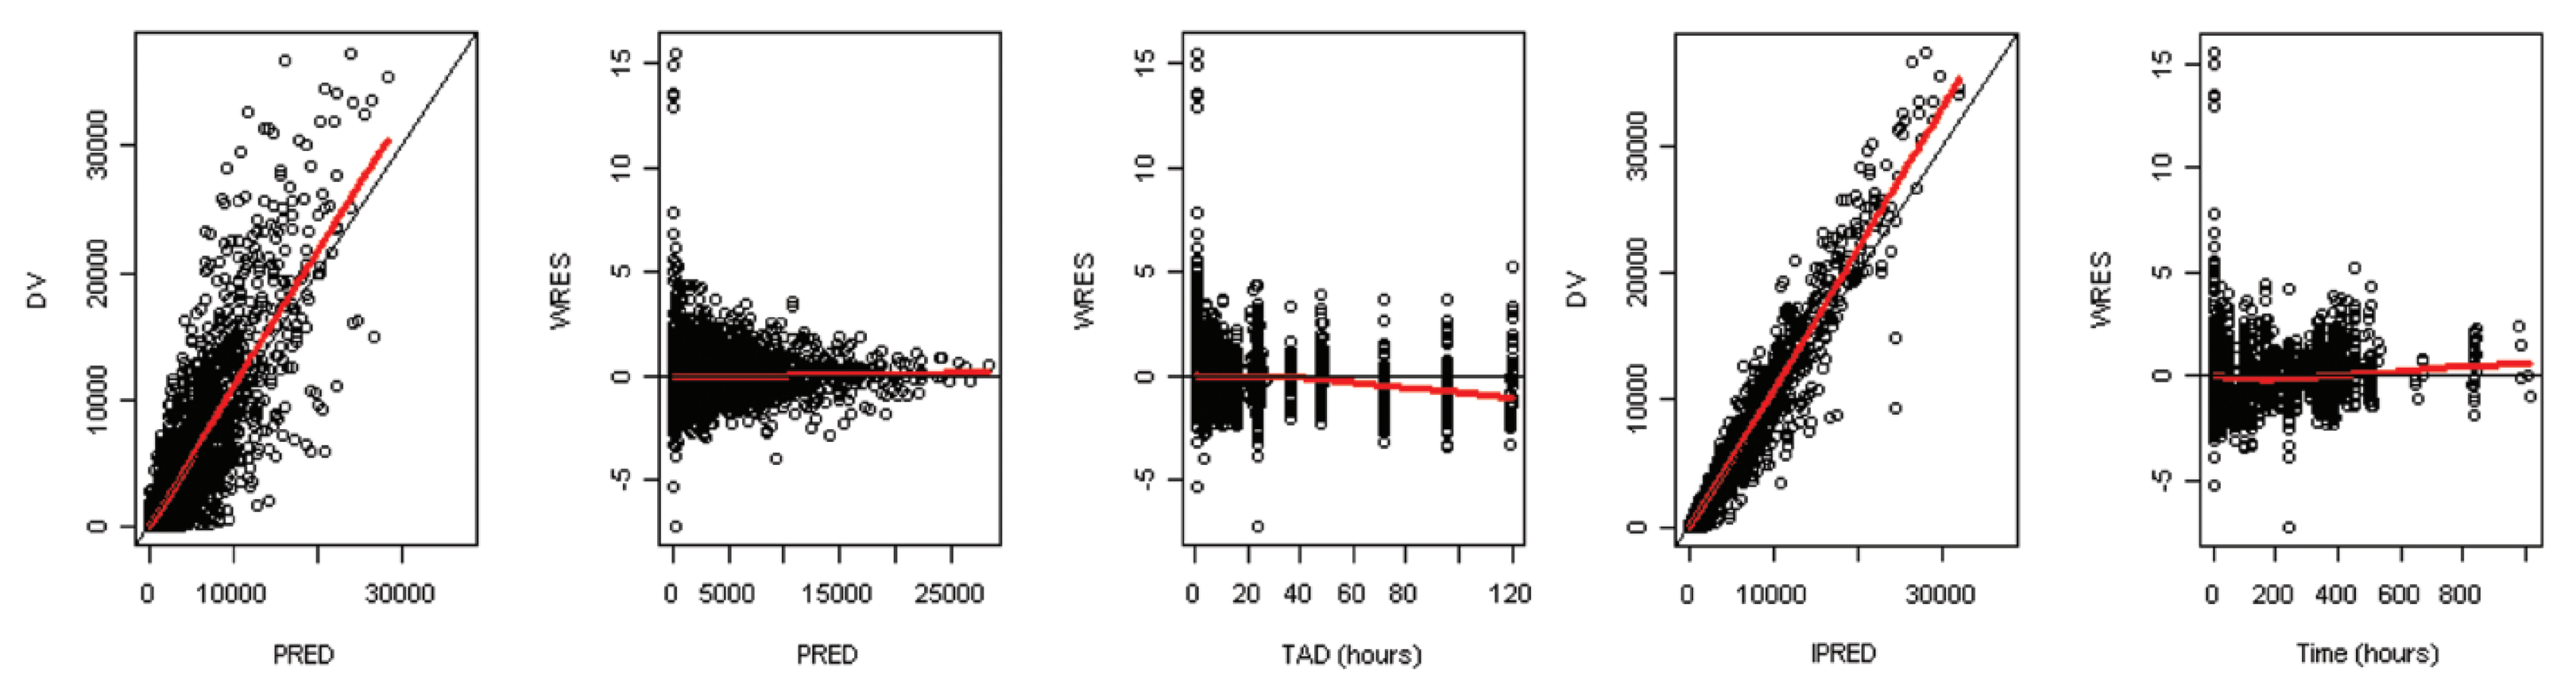
DV
WRES
PRED
TAD (hours)
IPRED
Time (hours)

Figure 3 *Inter-individual Random Effect on CL/F versus Covariates*

Base model (top row), Final Model (bottom row)

CORT: corticosteroid use, 0=No, 1=Yes; ITP: 0= healthy volunteers, 1= patients; SEX: 0=females, 1=males; RACE: 0=Japanese, 1= non-Japanese East Asian, 2=Caucasian; 3 = Black; 4 = Other

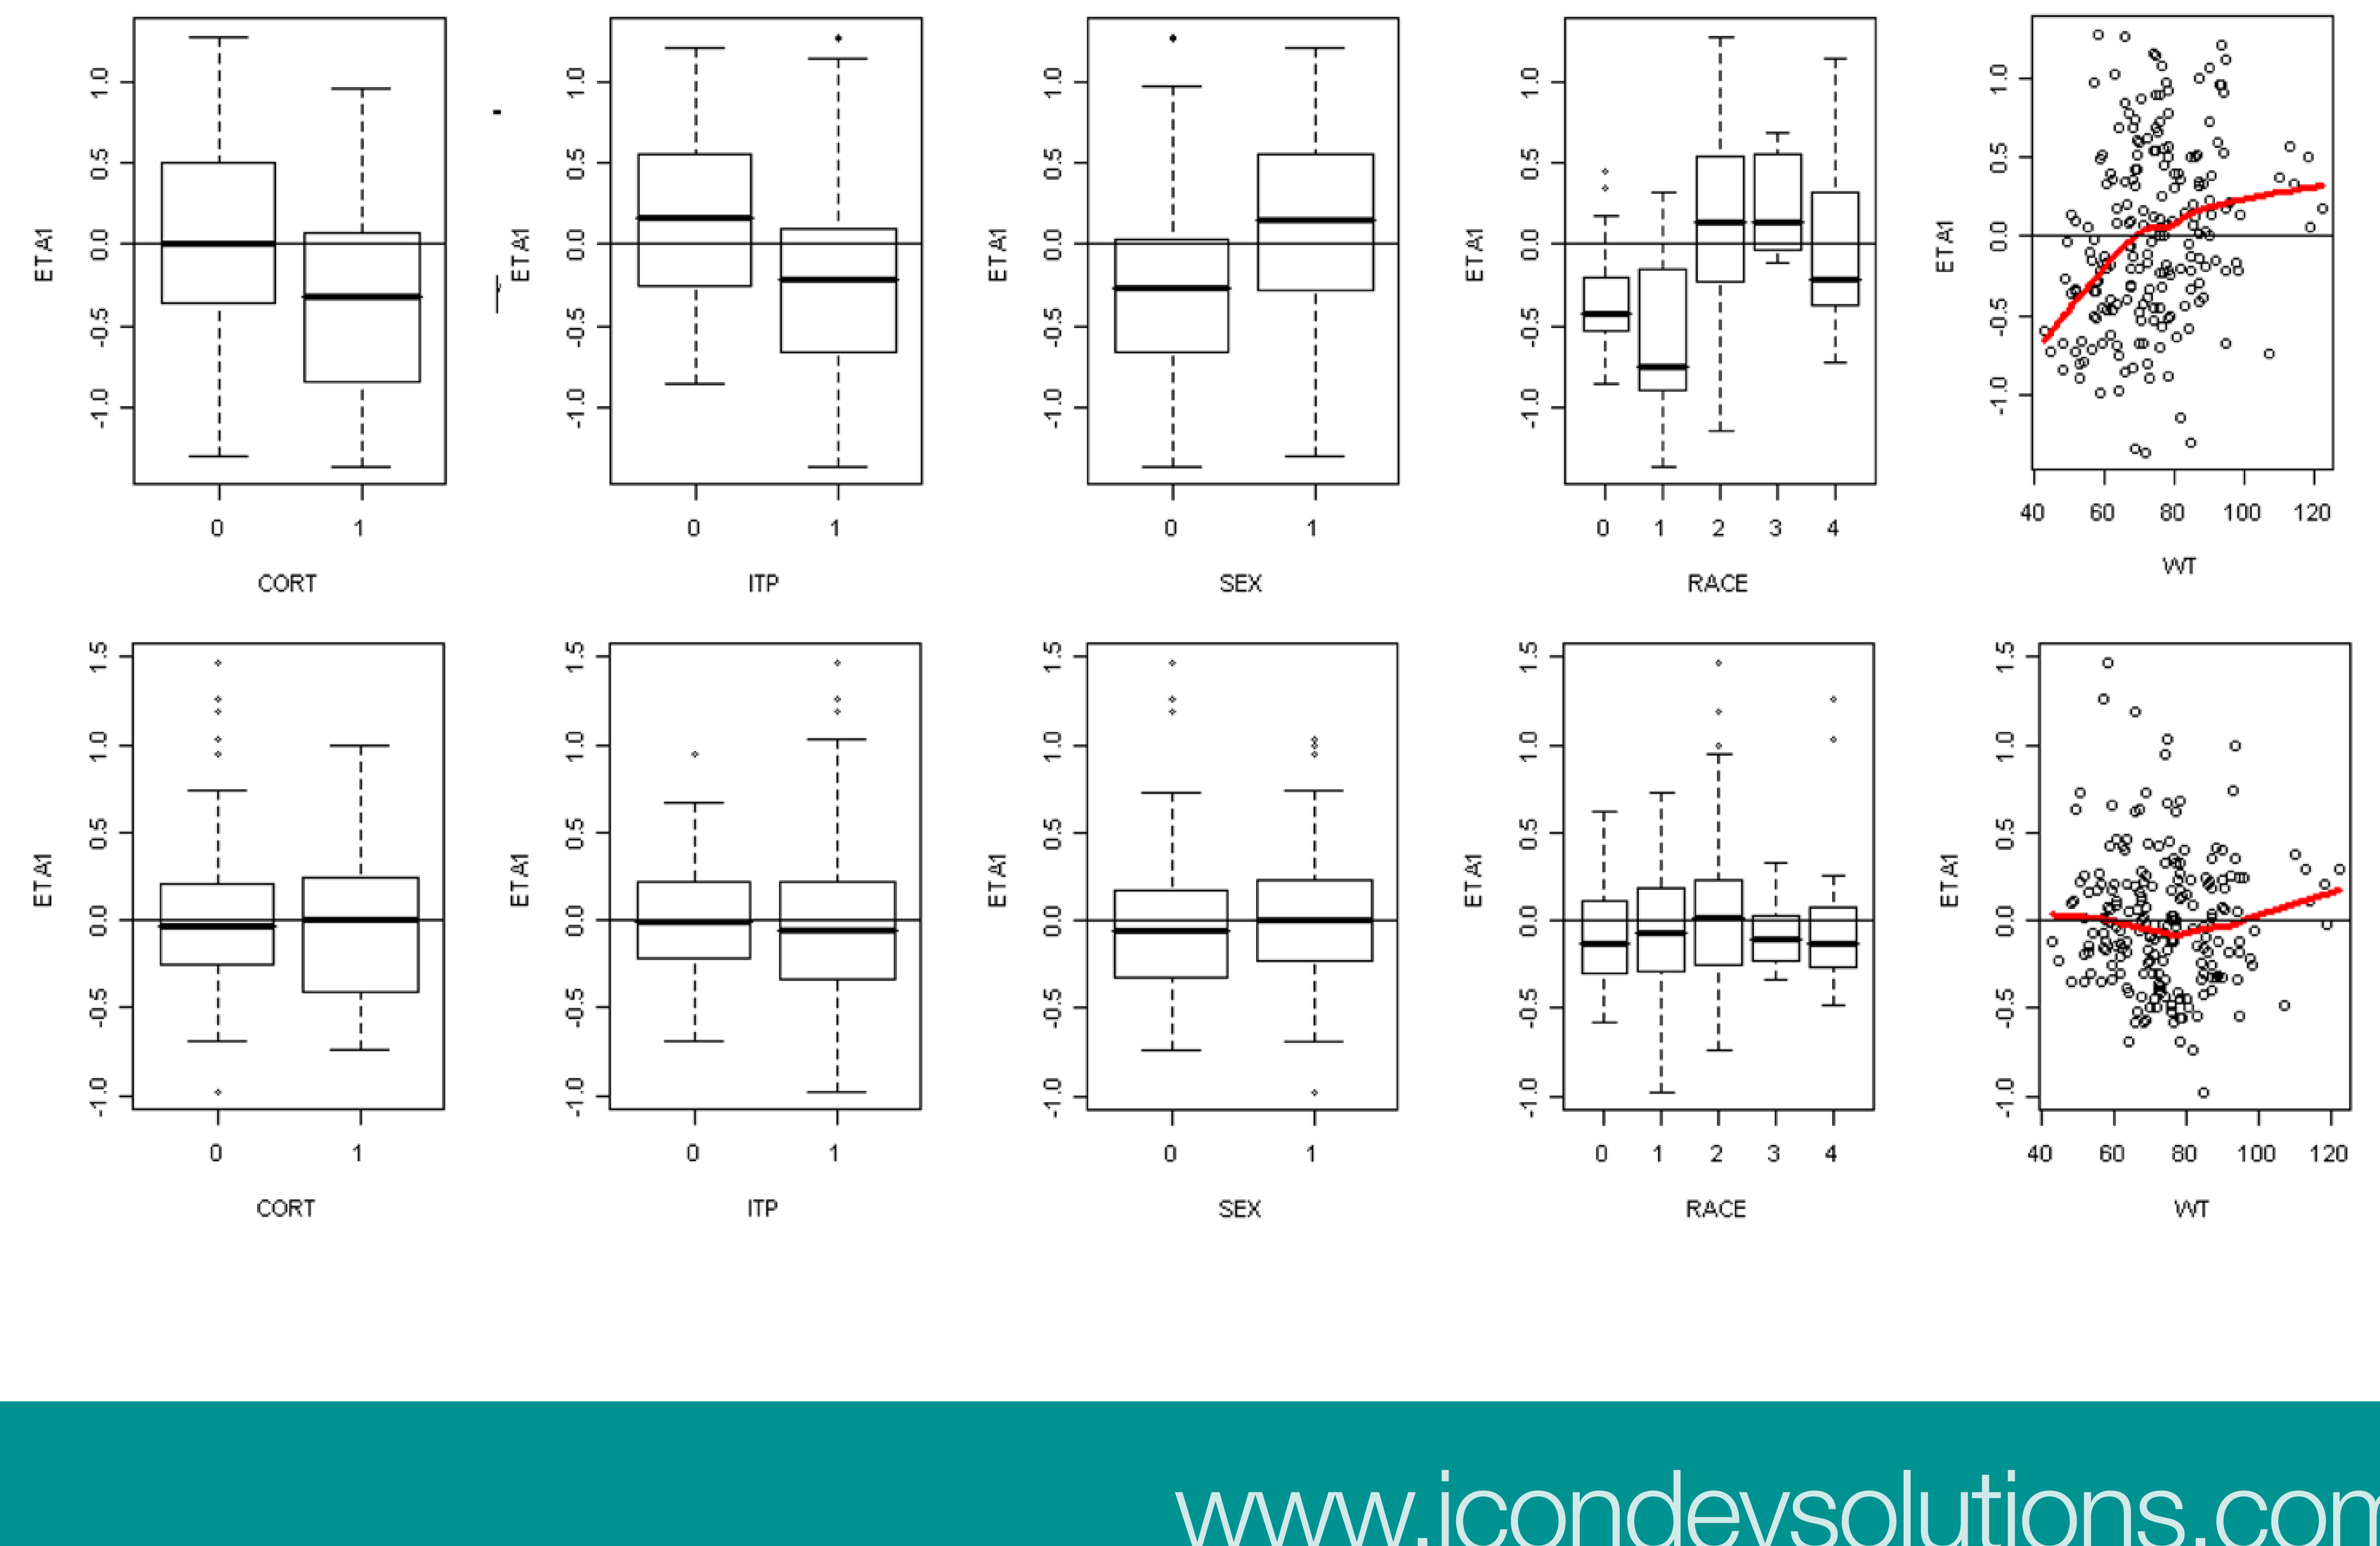
ETA1
CORT
ITP
SEX
RACE
WT

www.icondevsolutions.com