A population analysis on the effects of the CYP2D6 deficiency on pharmacokinetics and exposure of esmirtazapine in healthy volunteers.

Otilia Lillin-de Vries, Thomas Kerbusch, Pieter-Jan de Kam, Neely Ivgy-May, Roberta Bursi

Schering-Plough Corporation, Oss, The Netherlands

Introduction

Esmirtazapine is the S(+)-enantiomer of mirtazapine - a well-known antidepressant. In addition to the antidepressant effect, preclinical and clinical studies have demonstrated sleep-promoting effects of mirtazapine. The S(+)-enantiomer has a shorter half-life than the R(-)-enantiomer, making it's use favorable to the R(-)-enantiomer or the racemate for the treatment of insomnia. The maleate salt of esmirtazapine is a stable pharmaceutical formulation that is currently under clinical development. Esmirtazapine is metabolized by cytochrome P450 CYP2D6 in the liver. The subjects were therefore genotyped with regard to CYP2D6.

Objectives

This is a pooled population analysis that describes the pharmacokinetics of esmirtazapine using data from three studies in healthy volunteers and assesses the effects of CYP2D6 genetic polymorphism on the pharmacokinetics of esmirtazapine.

Methods

The studies included in this analysis are:

Study 1

Design	Randomized, open-label, four period cross-over design.
Subjects	17 postmenopausal women
Treatments	1.5 mg, 7.5 mg, 18 mg of esmirtazapine maleate and 15 mg of mirtazapine SD and MD
Assessments included in the model	Blood samples for esmirtazapine plasma concentrations up to 72 h post-dose
Sampling scheme	Rich (16 samples per subject on average)

Study 2

Design	Randomized, double-blind, placebo controlled, parallel design.
Subjects	67 healthy adult volunteers (45 – 65 years)
Treatments	18 mg MD and 18 mg titrated up to 54 mg MD of esmirtazapine maleate
Assessments included in the model	Blood samples for esmirtazapine plasma concentrations up to 24 h post-dose
Sampling scheme	Rich (14 samples per subject on average)

Study 3

Design	Open-label, single-center, one-sequence cross-over design.
Subjects	20 healthy adult volunteers (18 – 45 years)
Treatments	4.5 mg of esmirtazapine maleate SD before and after blocking CYP2D6 mediated metabolism with paroxetine
Assessments included in the model	Blood samples for esmirtazapine plasma concentrations up to 72 h post-dose
Sampling scheme	Rich (16 samples per subject on average)

A pharmacokinetic model was developed using NONMEM VI with FOCE (first order conditional estimation method) to describe the concentration-time profile of esmirtazapine based on plasma samples from the three studies above.

DNA samples were processed for CYP2D6 *3, *4, *5, *6, *7, *8 and *2XN alleles. All SNPs that were tested resulted in complete inactivation of CYP2D6. The CYP2D6 genotype was coded as an integer representing the number of mutations in the CYP2D6 gene (the subjects from Study 3 receiving esmirtazapine maleate after paroxetine were included as poor metabolizers):
0 = poor metabolizer (PM, two mutations by genotyping or rendered after paroxetine);
1 = intermediate metabolizer (IM, one mutation);
2 = extensive metabolizer (EM, no mutations);
3 = ultra-rapid metabolizer (UM, one or more duplicated alleles).

Structural model:

- one-, two- and three-compartment models with first and zero-order absorption were tested on log-transformed data;
- since absolute bioavailability could not be estimated from these data, a relative bioavailability for the extensive metabolizer, F_{EM}, was fixed to 1. Subsequently a bioavailability for the poor metabolizer, F_{PM}, a bioavailability for the intermediate metabolizer, F_{IM},and a bioavailability for the ultra-rapid metabolizer, FUM, was estimated relative to F_{EM};
- the four CYP2D6 genotype categories were combined in a linear relationship (gene-dose approach) which implies the assumption that e.g. the effect of being an EM is twice the effect of being an IM on a PK parameter. Models with an effect of genotype as linear relationship were tested as well as models with genotype as a categorical covariate.

Inter-individual variability:

Random effects for inter-individual variability of the pharmacokinetic parameters were assumed to be log-normally distributed. Models with a normally distributed IIV on lag time (log-normal for the rest of the PK parameters) were also tested.

Residual variability:

The residual error was additive with log-transformed data (i.e. proportional on the normal scale). Models with a combined additive and proportional error were also tested.

Covariates:

The covariates investigated for an effect on the PK parameters were: CYP2D6 genetic polymorphism, sex, age, body weight, dose and study. Body mass index was not investigated since it is highly correlated to body weight and body height. Covariate search was done with the automated procedure SCM (stepwise covariate method) as implemented in PsN V2.2 (Pearl speaks Nonmem). Subsequently the intermediate results from SCM were also manually checked for compliance with all selection criteria (decrease in unexplained variability when the Log-Likelihood criterion is fulfilled).

Selection criteria:

Model acceptance was based on:
(a) successful minimization;
(b) successful covariance step without warning messages;
(c) no parameters estimated near the boundary;
(d) a precision of at least three digits in the parameters;
(e) correlation less than 0.95 between any two parameters;
(f) the standard error of the estimates small enough (a 95% confidence interval excludes zero);
(g) stability check performed for a selected basic model.

Model selection was based on:

(a) the comparison of full vs. reduced models is based on the Log-Likelihood Criterion;
(b) decrease in unexplained variability;
(c) weighted residuals and conditional weighted residuals vs. time are randomly spread around zero.

Model performance:

- validation has been performed with bootstrapping (n=1000);
- a comparison was made to a non-parametric estimation method to evaluate any over- or under-dispersion of the random effects;
- shrinkage was calculated to assess reliability of certain goodness-of-fit plots;
- simulations of a typical profile (by genotype) were compared to data;
- simulations of a typical profile (by genotype) with parameter uncertainty;
- comparison was performed between individual AUCs as calculated from the PK parameters and AUCs calculated with non-compartmental analysis.

Results

In total 104 healthy volunteers contributed to 2910 esmirtazapine plasma concentration samples. The dataset contained 24 PM (4 subjects by genotype and 20 subjects by phenotype), and 34 IM, 64 EM and 2 UM, all of which were by genotype.

Structural model:

The pharmacokinetics of esmirtazapine was best described by a two-compartment first-order absorption model with lag-time. Inter-individual variability (IIV) in the parameters was assumed to be log-normally distributed. Residual variability was additive with the log-transformed data (thus proportional on linear scale).

Covariates:

Previous non-compartmental analysis showed that CYP2D6 polymorphism had an effect on exposure of esmirtazapine (PMs had approximately doubled exposure in comparison to EMs). An effect of CYP2D6 polymorphism was expected a priori, so CYP2D6 genetic polymorphism was included in the model as a structural covariate. The effect of CYP2D6 polymorphism was manually tested first on the base model since we also wanted to test multivariate dependency (SCM is a univariate search algorithm). Clearance was found to depend on CYP2D6 polymorphism in a linear fashion (gene-dose). Relative bioavailability was also found to depend on CYP2D6 polymorphism: a linear relationship was tested but did not yield successful covariance step; categorical relationships were tested, and the difference between two categories, PM/IM and EM/UM, was included in the final model.

Of the covariates investigated for their effect on the PK parameters with SCM (sex, age, body weight, dose and study), the effects of dose on clearance and study on peripheral volume were selected by SCM. However, the final model chosen by SCM was unstable. Dose-proportionality has been previously demonstrated in the dose-range 1.5 – 18 mg/day. The effect of dose on clearance was small (corresponding to changes < 2% in the typical value of CL) and thus not clinically relevant.

In the final model only the effect of CYP2D6 genetic polymorphism was included on clearance and relative bioavailability.

Model performance:

Model performance was robust as indicated by bootstrapping; all PK parameters were normally distributed around their typical value. The final model was rerun with the nonparametric estimation method and the IIV estimates were similar to those obtained with the first order conditional estimation method. This indicates that the log-normal assumption for the distribution of the IIV was correct and no over- or under-dispersion was observed. Shrinkage was moderate to large indicating that diagnostic plots based on individual parameter estimates might be less adequate for detecting model misspecification.

Graphs:

Model performance was adequate as shown by simulations (after single dose administration and at steady state) and the effect of the genotype was well captured by the model (see **Figure 1**). The uncertainty in the PK parameters is visualized by simulating 1000 samples from the parameter space, based on the variance-covariance matrix (see **Figure 2**). The dose-proportionality assumption was checked with box-plots of dose vs. clearance; median clearance in the 4.5 mg dose was slightly above the medians for the other dose groups (see **Figure 3**), likely due to a study difference rather than dose-nonlinearity.

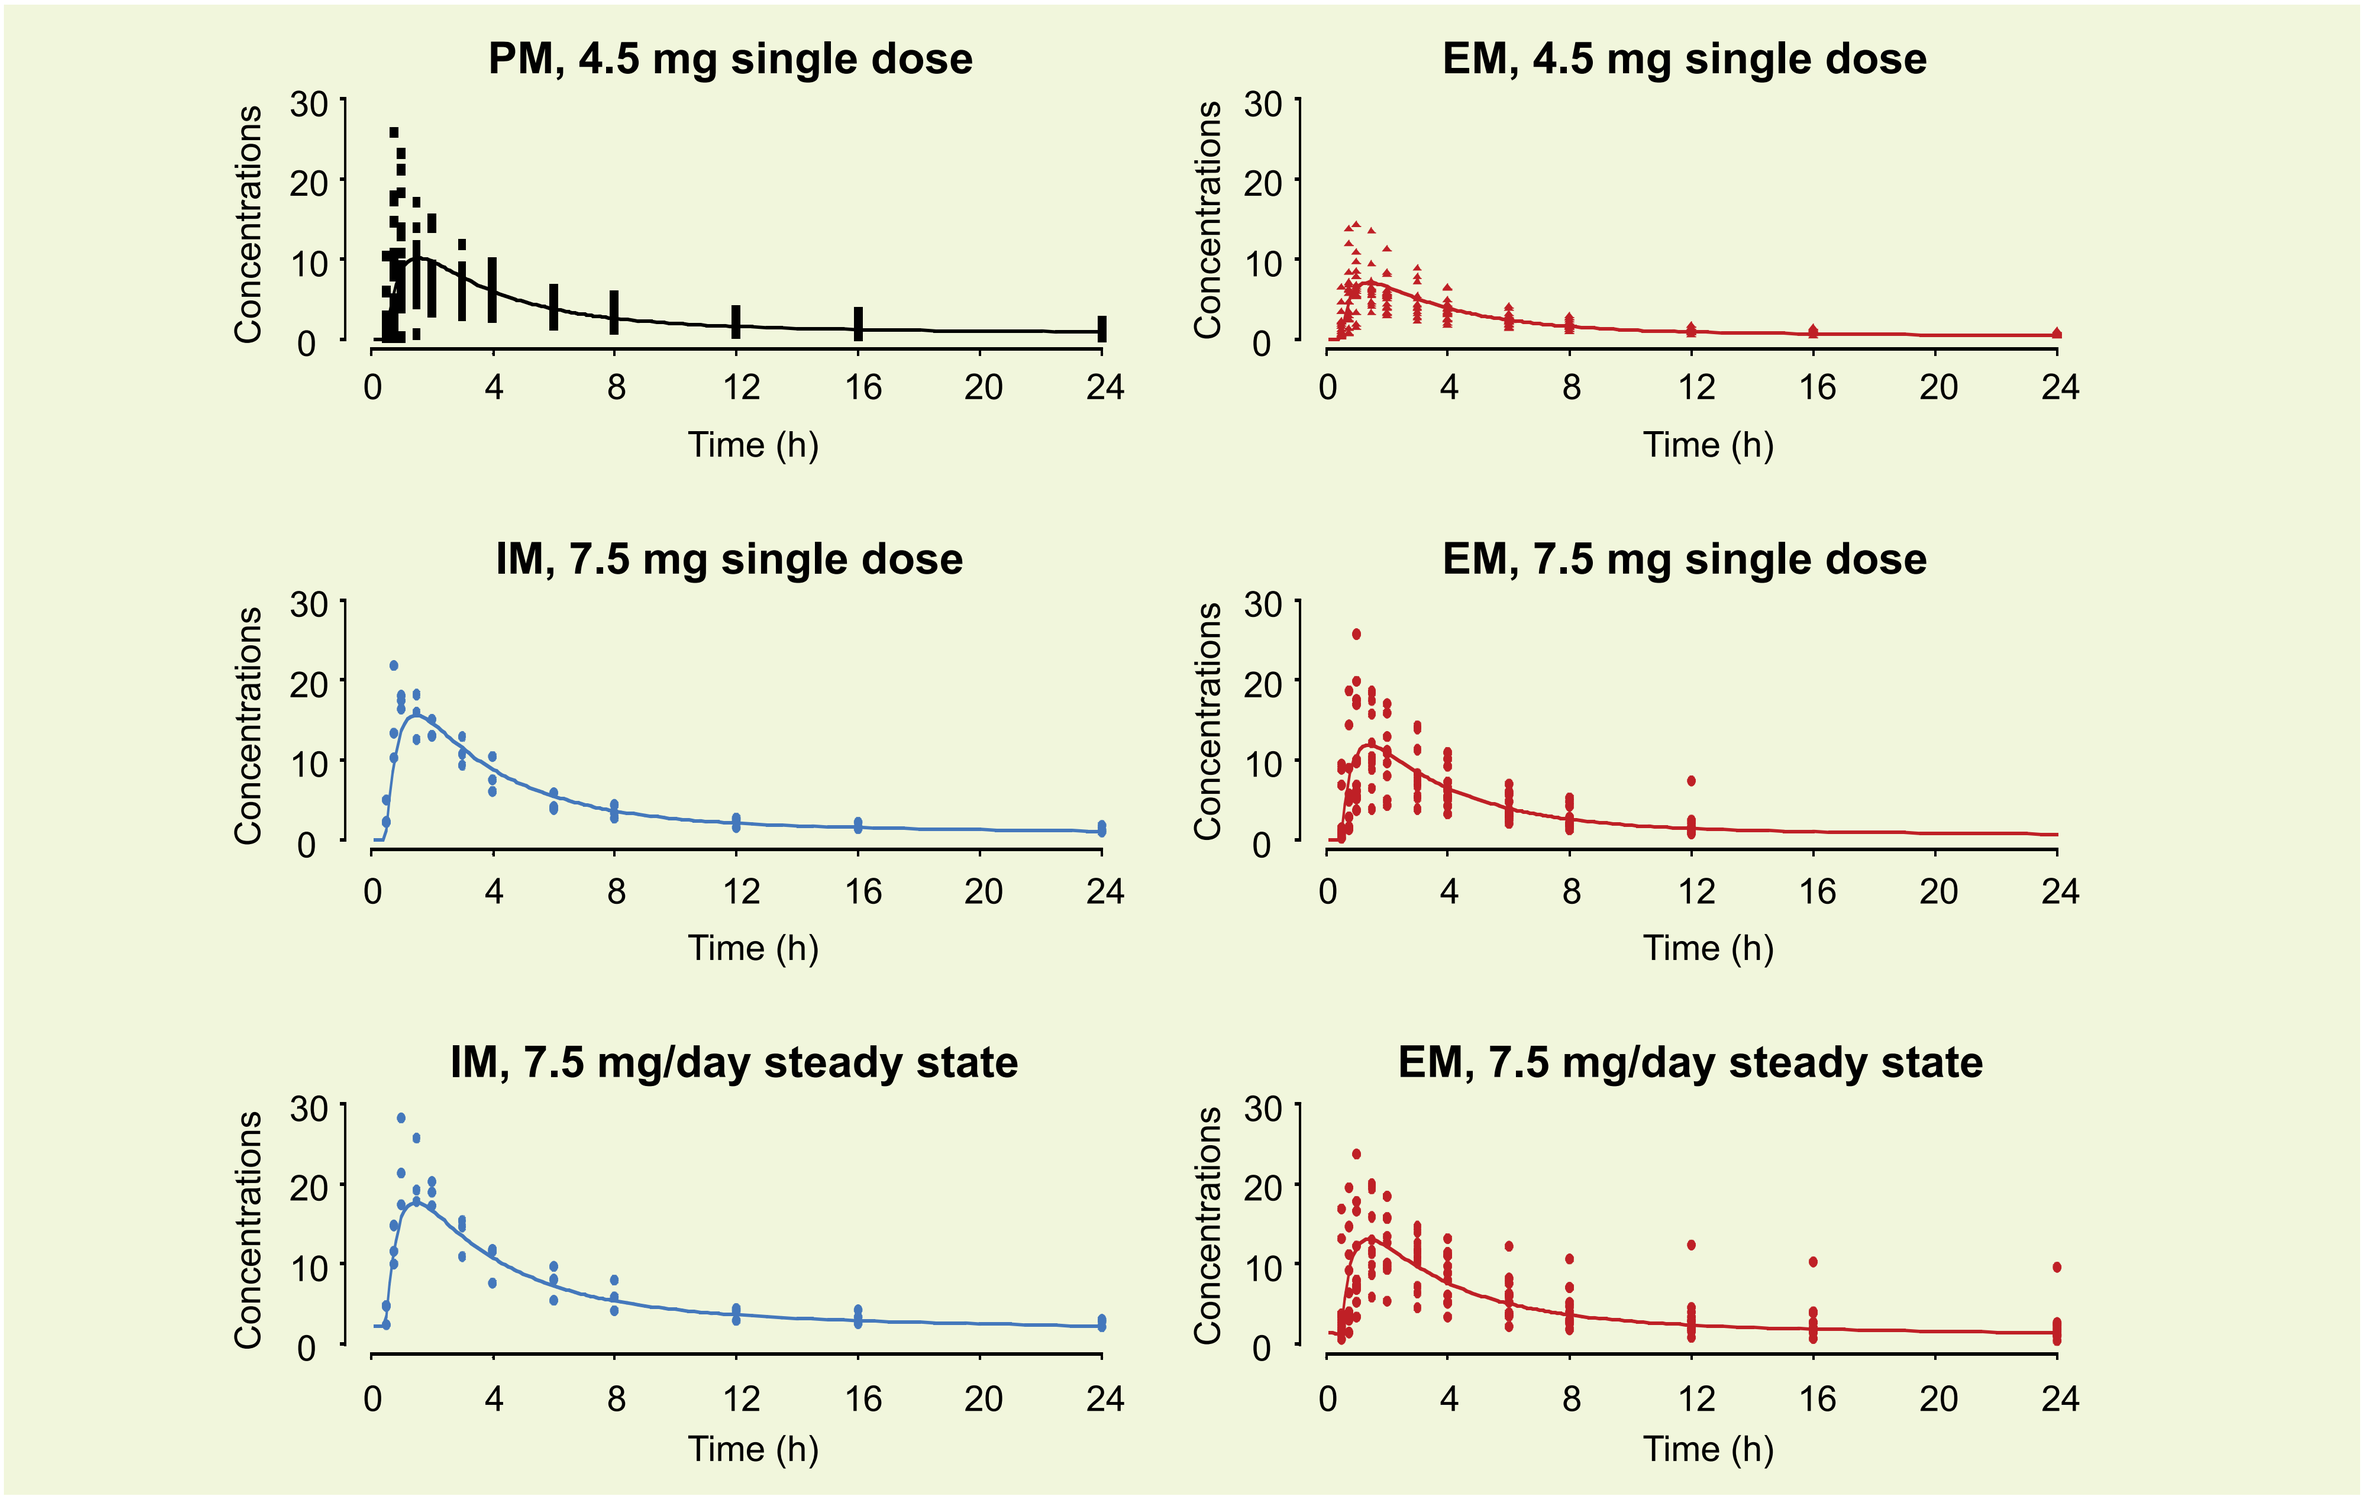
PM, 4.5 mg single dose
EM, 4.5 mg single dose
IM, 7.5 mg single dose
EM, 7.5 mg single dose
IM, 7.5 mg/day steady state
EM, 7.5 mg/day steady state
Concentrations
Time (h)

Figure 1. *Observed and simulated typical concentration-time profile after single dose administration of 4.5 mg Org 50081 for PMs and EMs (no data for UM and data for only one IM) and after administration of 7.5 mg Org 50081 for IM's and EMs (no data for UM and data for only one PM) after single dose and at steady state (Day 10)*

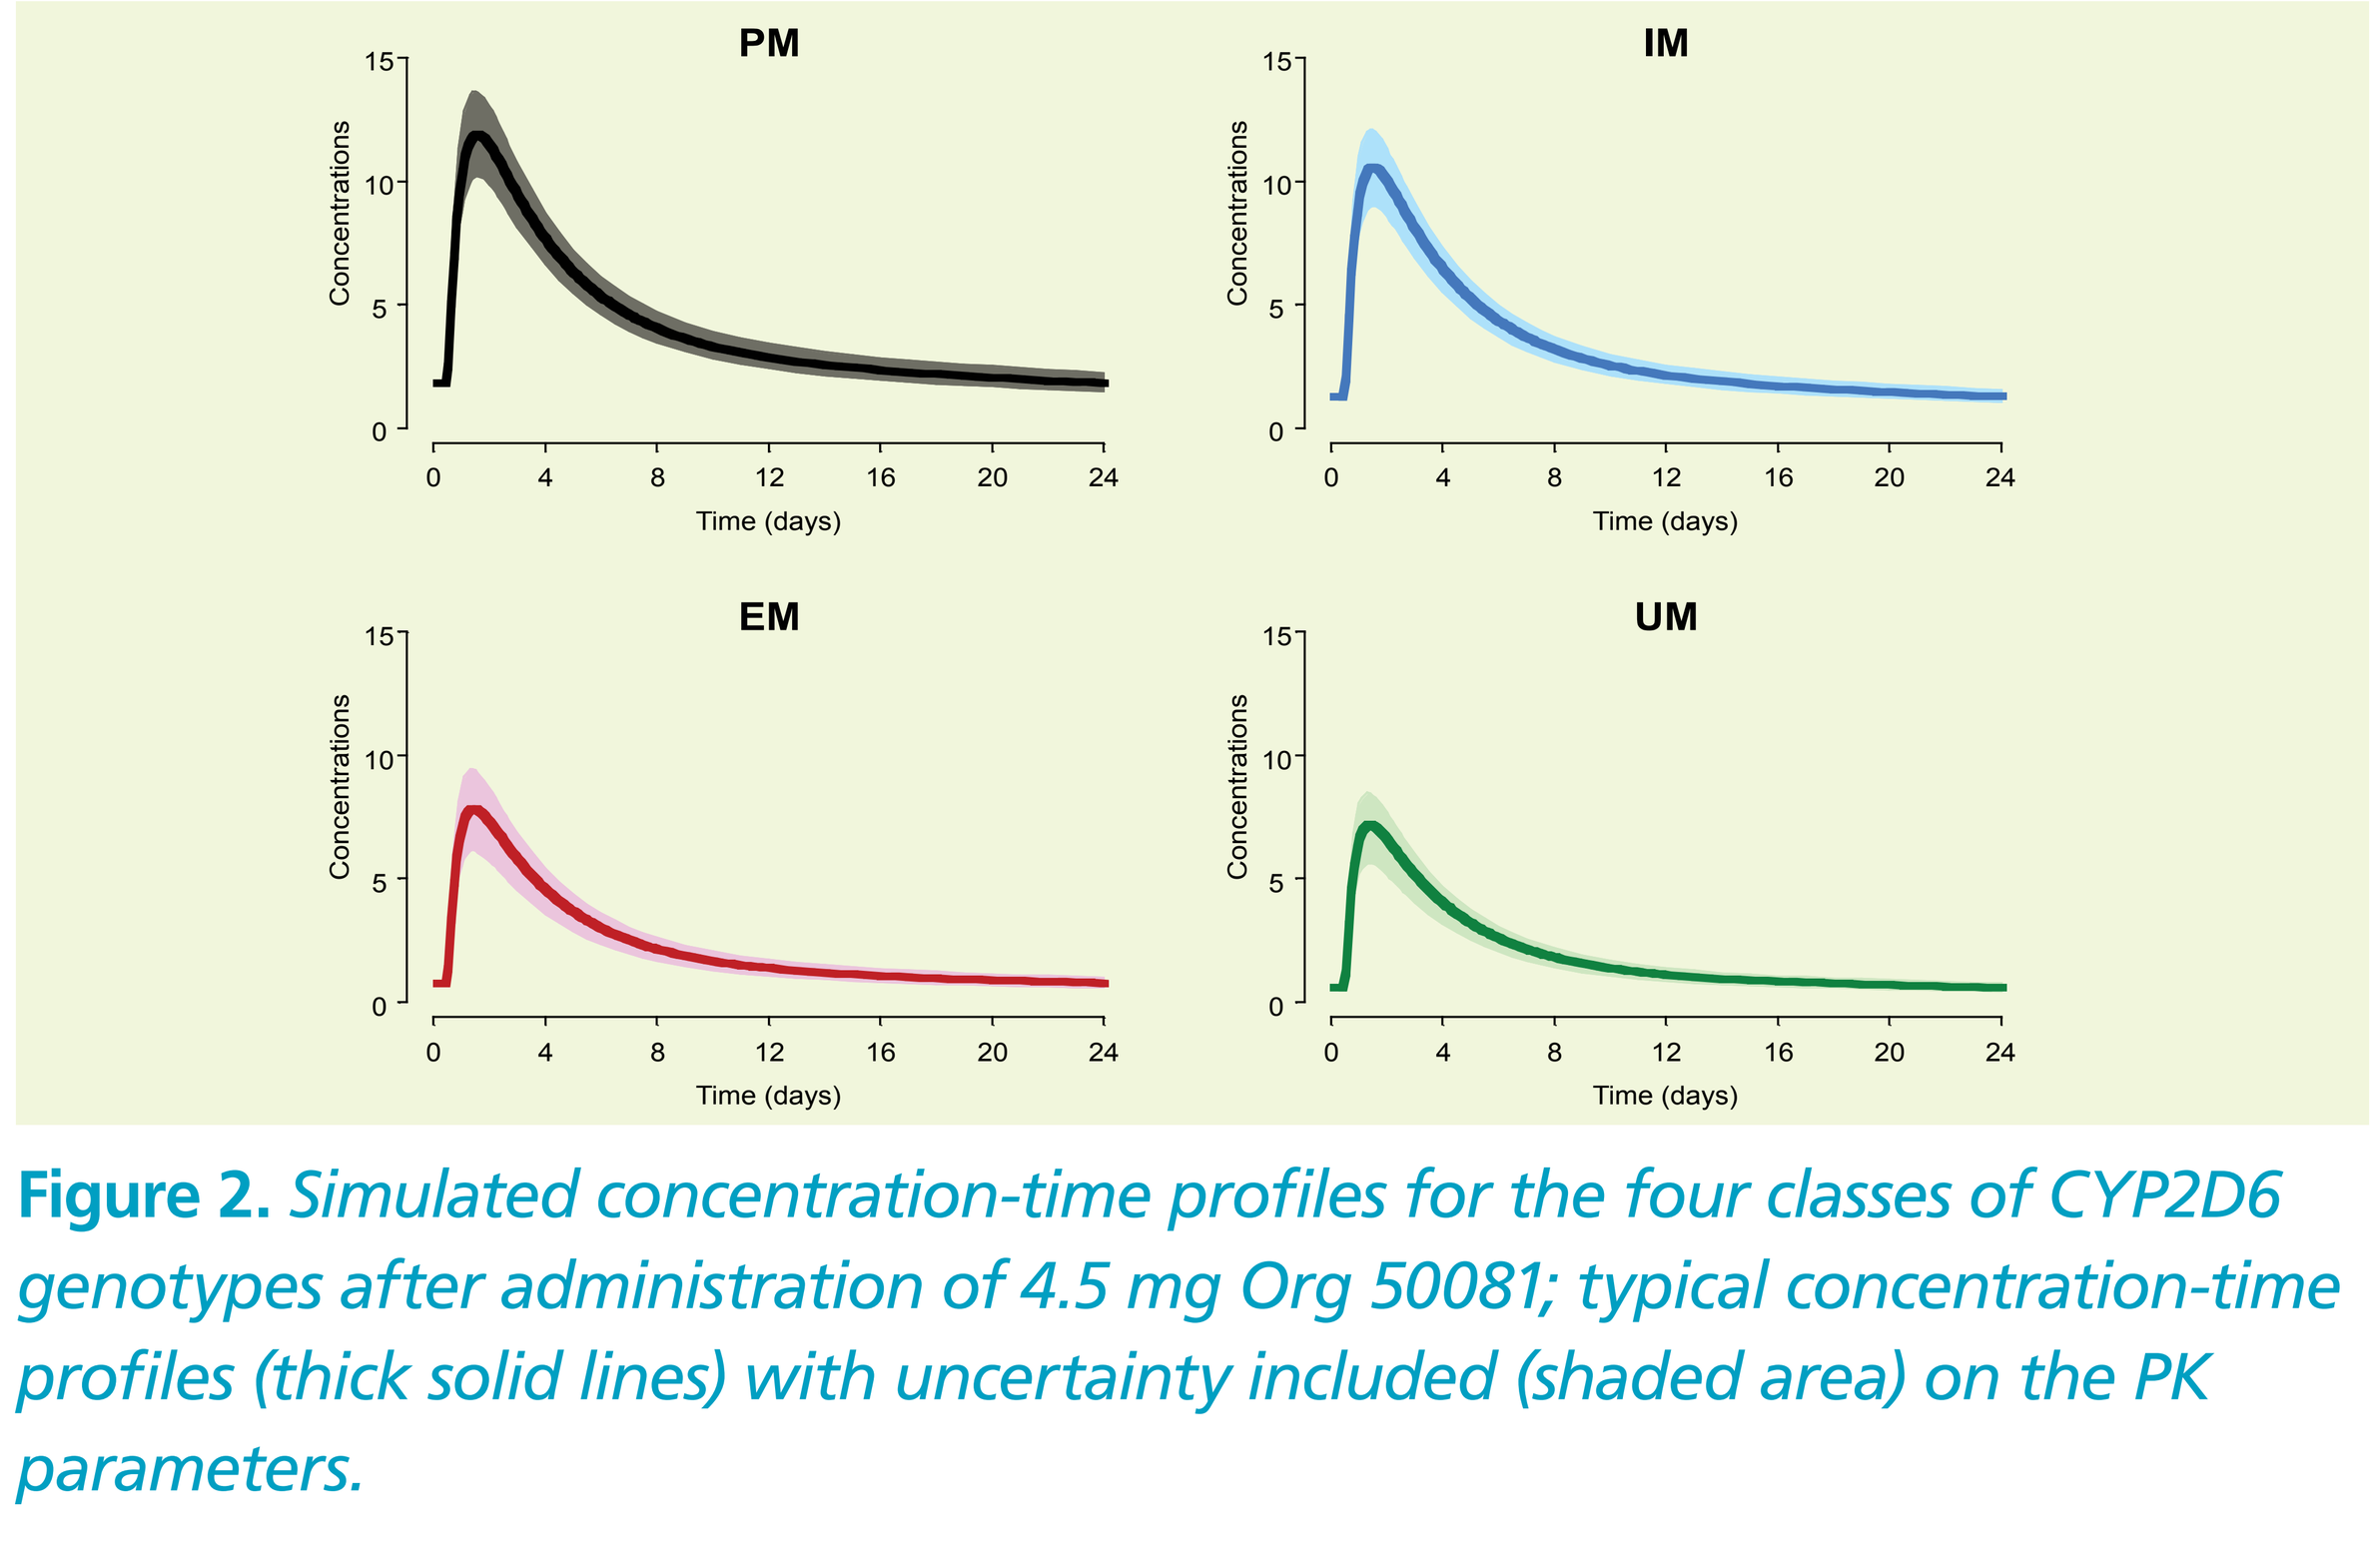
PM
IM
EM
UM
Concentrations
Time (days)

Figure 2. *Simulated concentration-time profiles for the four classes of CYP2D6 genotypes after administration of 4.5 mg Org 50081; typical concentration-time profiles (thick solid lines) with uncertainty included (shaded area) on the PK parameters.*

Table 1. *Pharmacokinetic model parameters*

Parameter (unit)	Typical value (CV %)	IIV (%)	Shrinkage (%)	(CI) $[CI_{bs}]$*
CLEM (L/h) Oral clearance of a EM (most common)	83.6 (5.8)	29.8	13.3	(74.1, 93.1) [75.2, 95.0]
CL.Geno (L/h) Effect of CYP2D6 genotype per mutation	13.7 (14.6)	-		(9.8, 17.6) [9.2, 17.1]
Vc (L) Central volume	59.2 (9.9)	105	9.2	(47.8, 70.6) [44.8, 79.1]
Q (L/h) Intercompartmental clearance	72 (6.4)	-		(63.0, 81.0) [62.2, 84.2]
Vp (L) Peripheral volume	1150 (6.9)	23.6	45	(994, 1306) [1000, 1340]
Ka (h-1) Absorption rate	0.327 (3.1)	-		(0.307, 0.347) [0.307, 0.348]
Alag (h) Lag time	0.475 (0.53)	2.24	55	(0.47, 0.48) [0.47, 0.48]
$F1_{EMUM}$ Bioavailability of EM and UM	1 fixed	29	17	
$F1_{PMIM}$ Bioavailability of PM and IM relative to EM and UM	1.22 (10.7)			(0.97, 1.47) [1.02, 1.53]
Residual variability				
Proportional (%)	34.3		-3.5	

Note:
*CI is 95% confidence interval for the parameter calculated from the asymptotic standard errors given by Nonmem
Clbs is 95% confidence interval for the parameter obtained with bootstrap

Oral clearance of an EM was 83.8 L/h and the effect of the CYP2D6 genotype per working allele was 13.2 L/h. This corresponds to clearance values of 97 L/h for a UM, 70.6 L/h for an IM and 57.4 L/h for a PM.

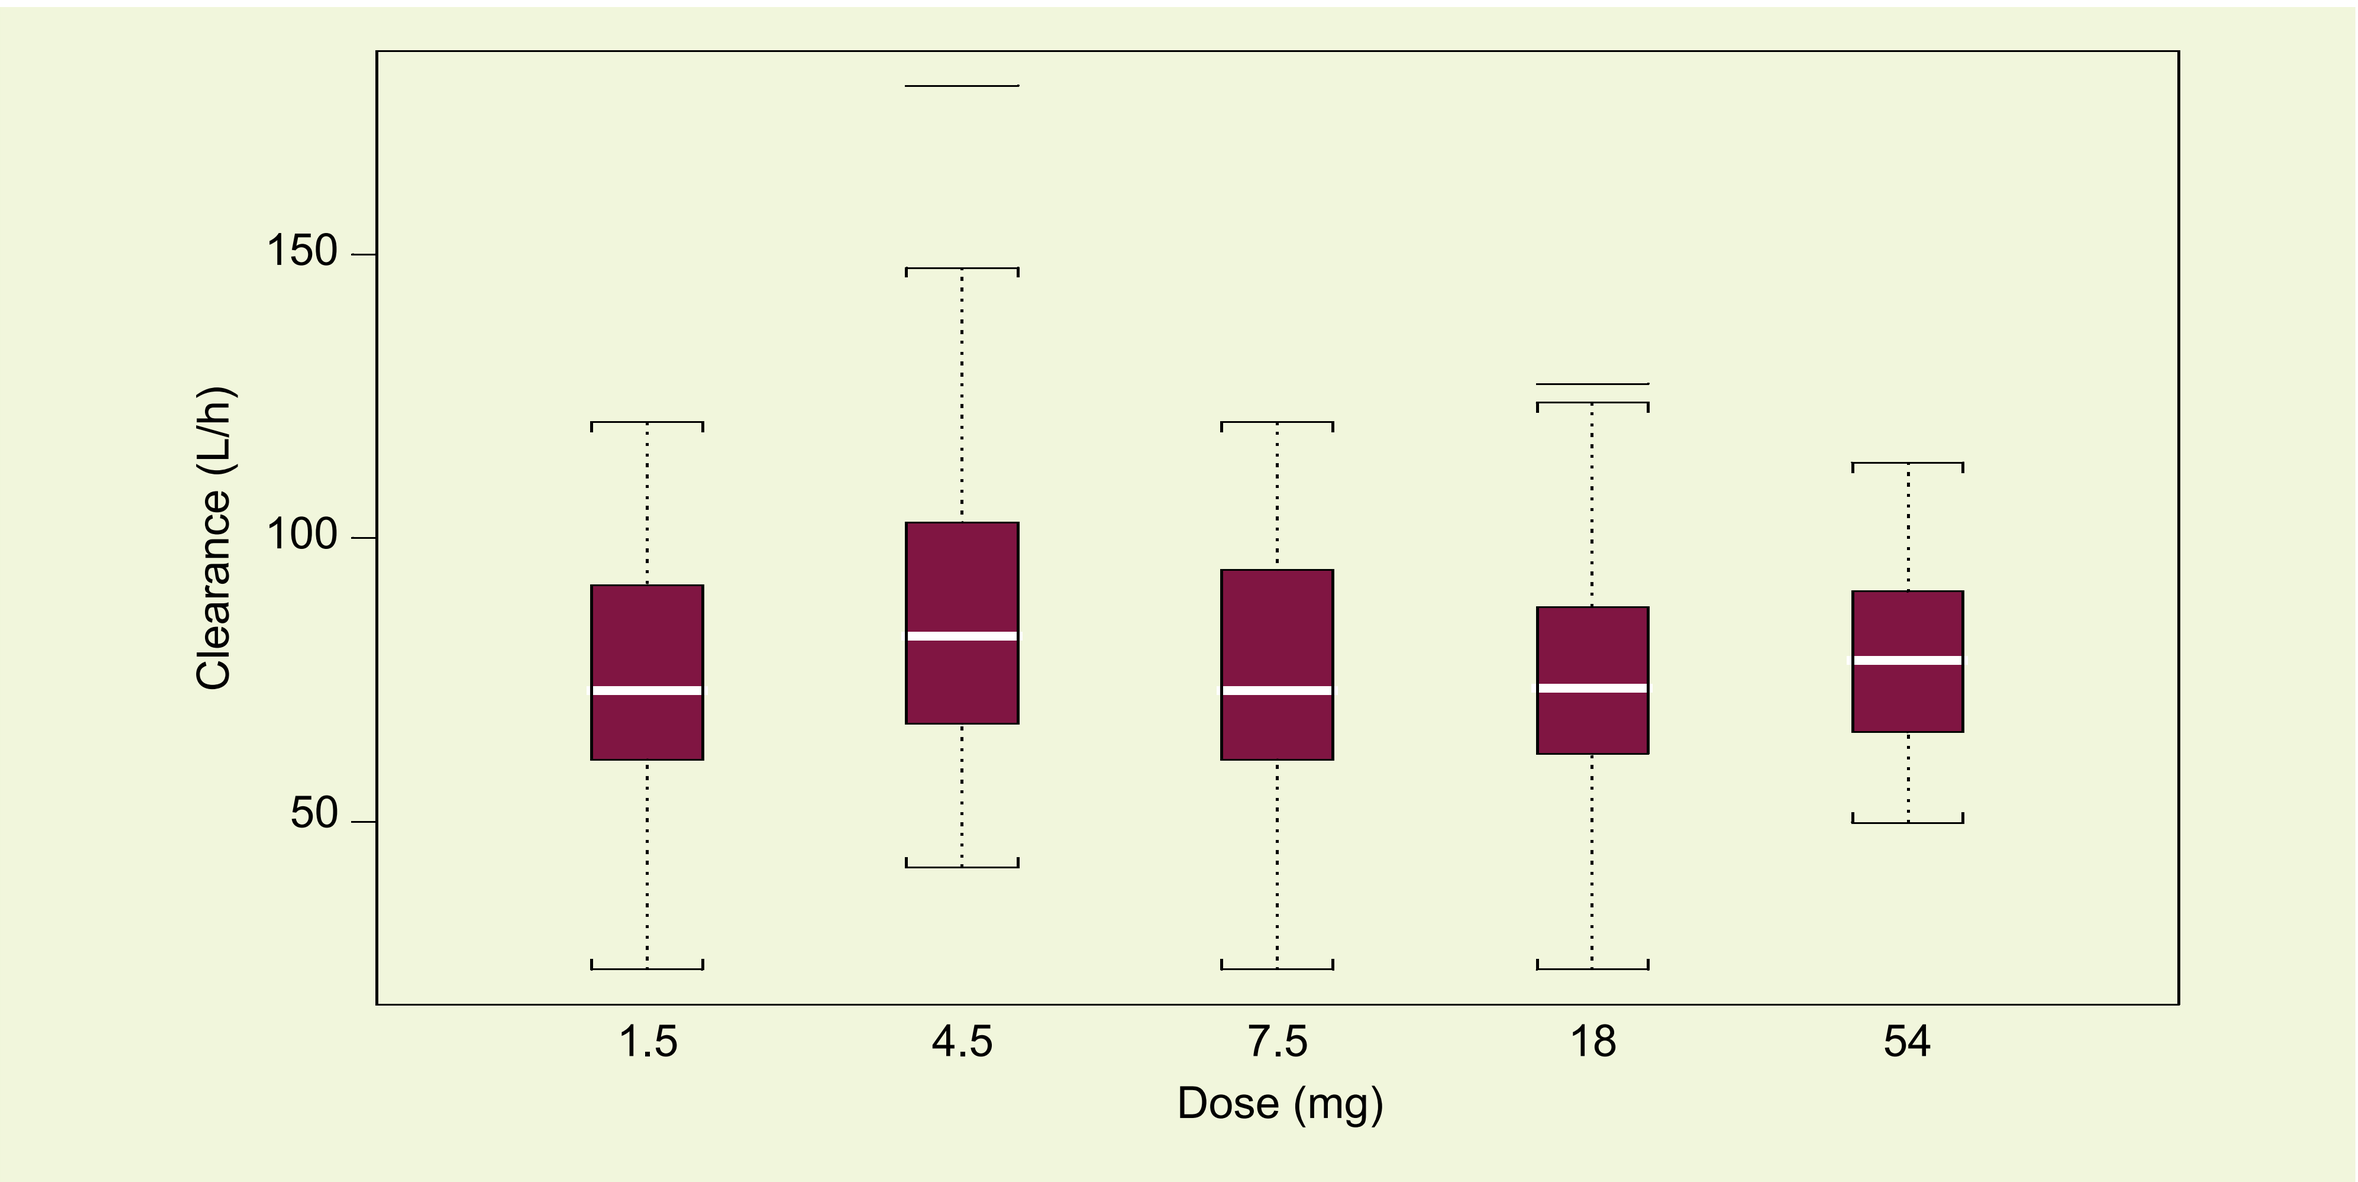
Clearance (L/h)
150
100
50
1.5
4.5
7.5
18
54
Dose (mg)

Figure 3. *Box-plots of Dose vs. Clearance*

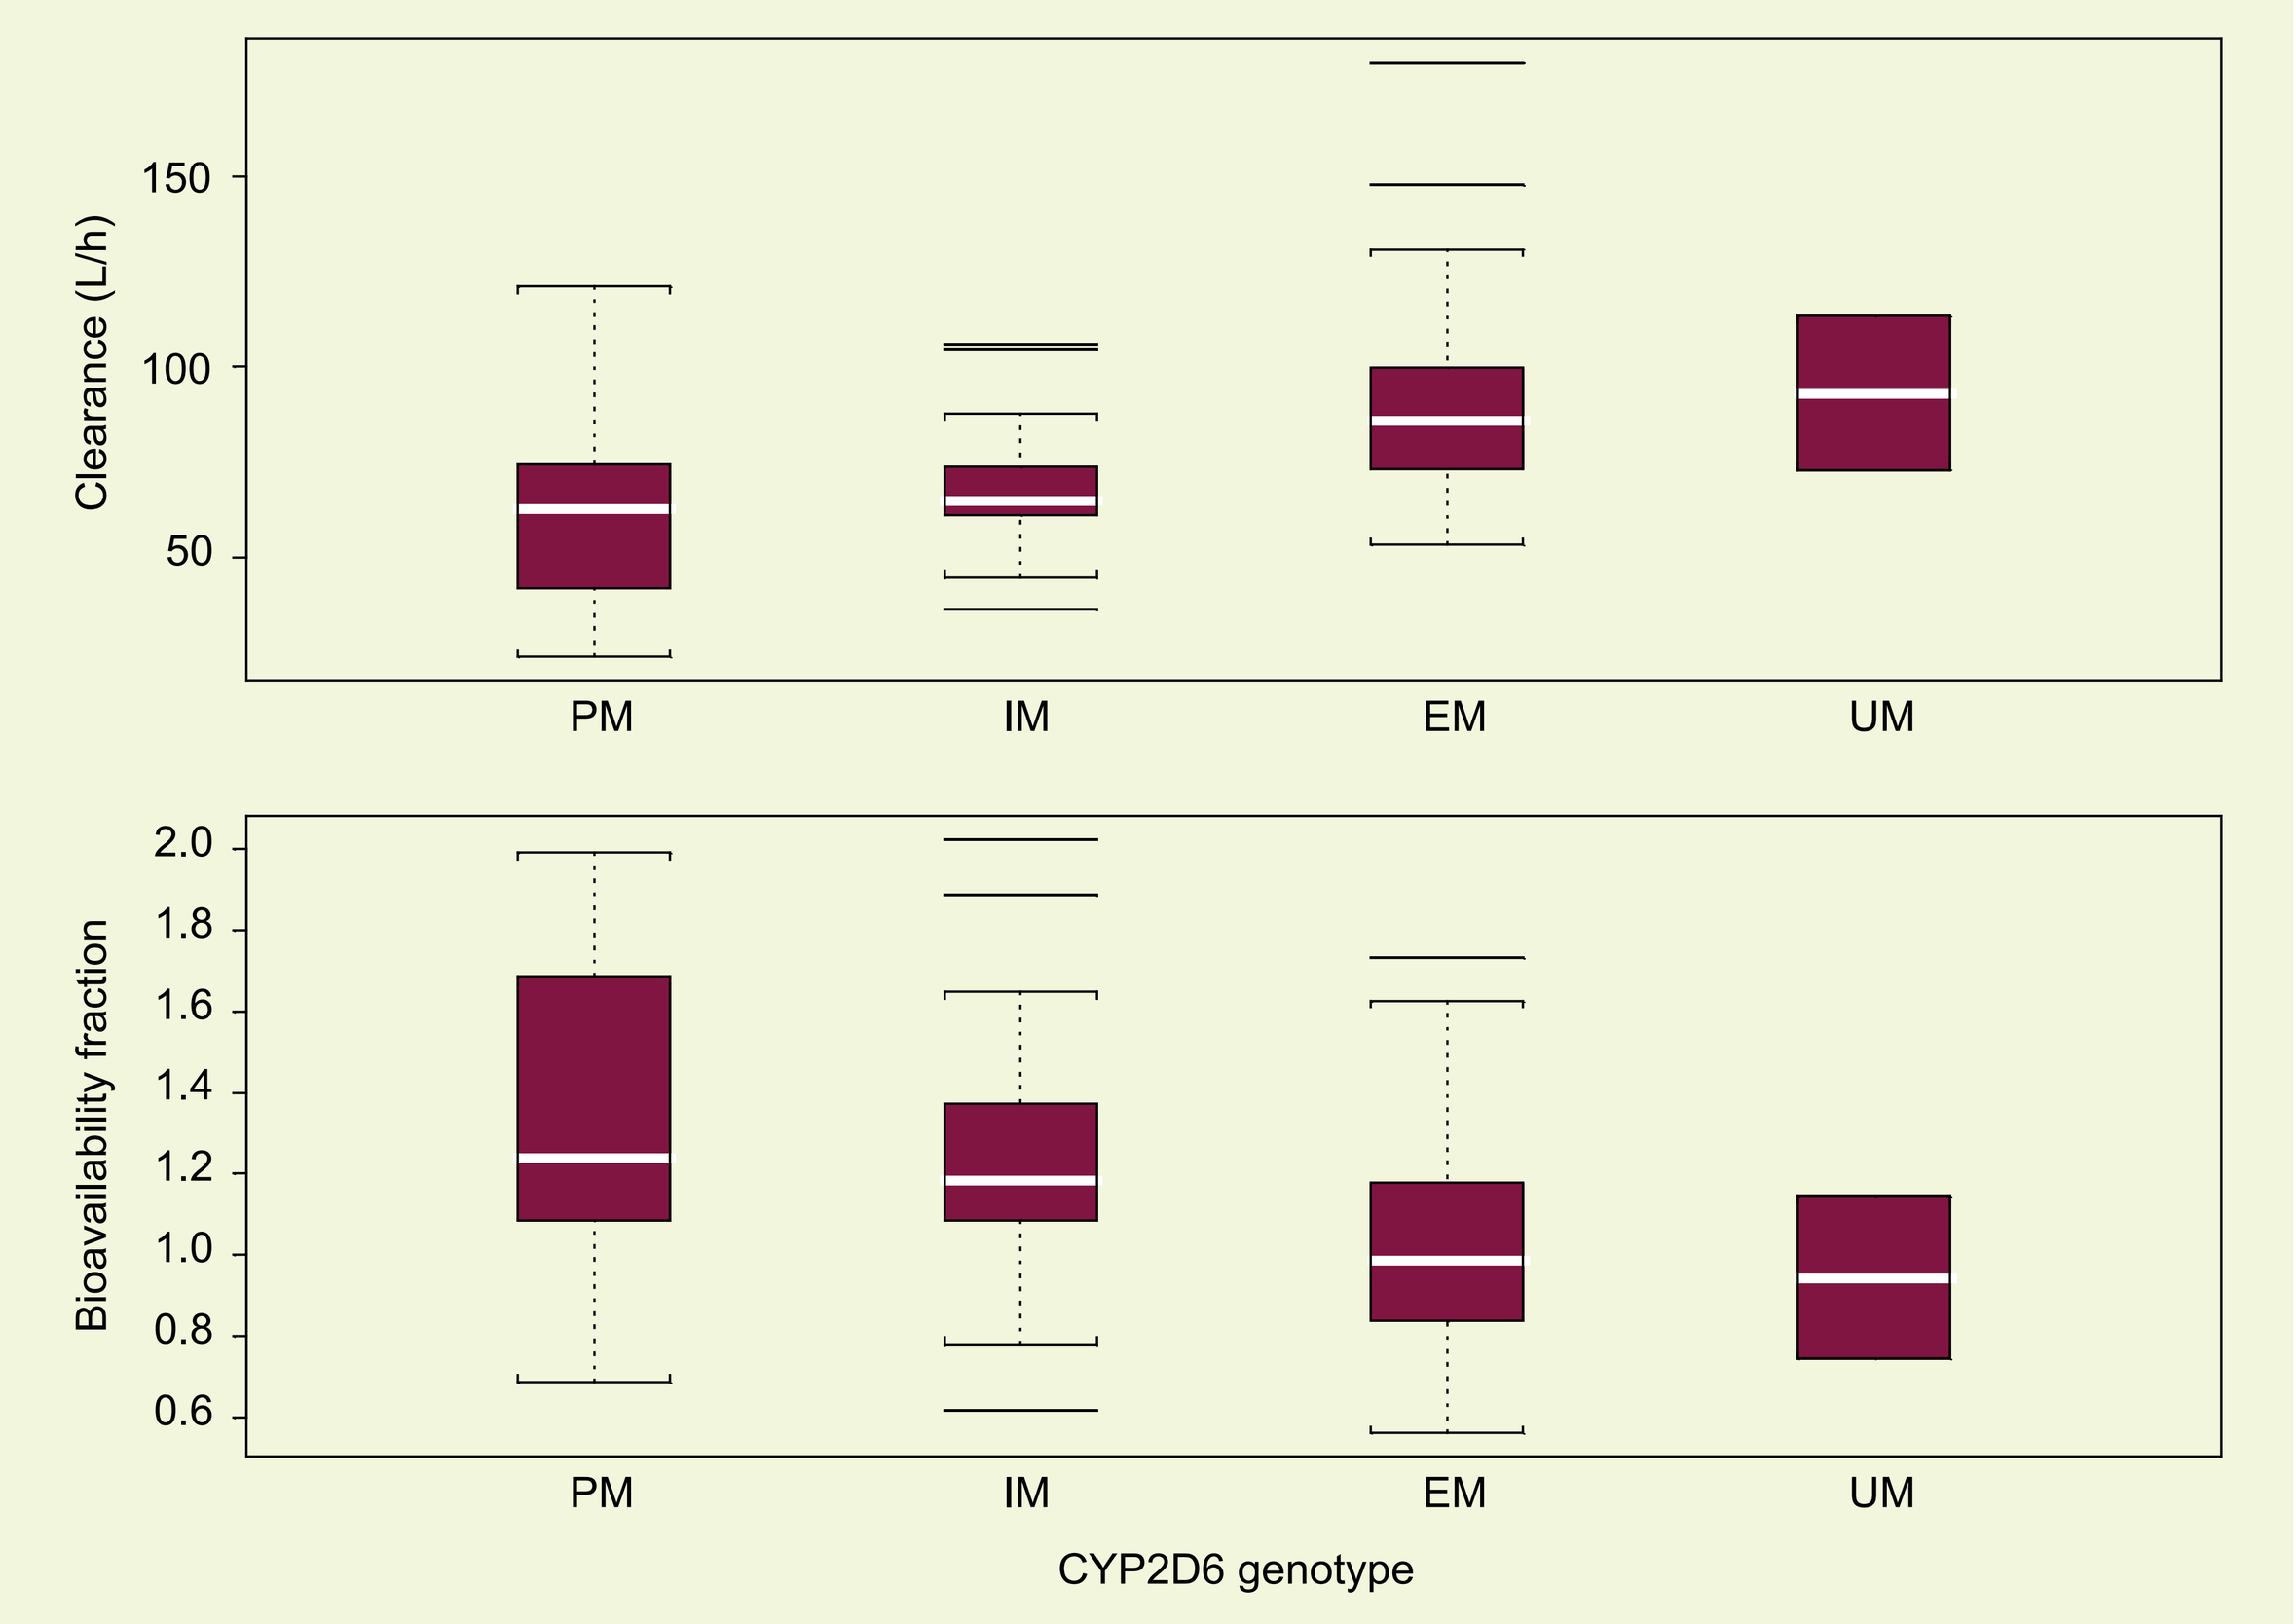
Clearance (L/h)
Bioavailability fraction
PM
IM
EM
UM
CYP2D6 genotype

Figure 4. *Box-plots of Clearance and relative bioavailability vs. CYP2D6 genotype*

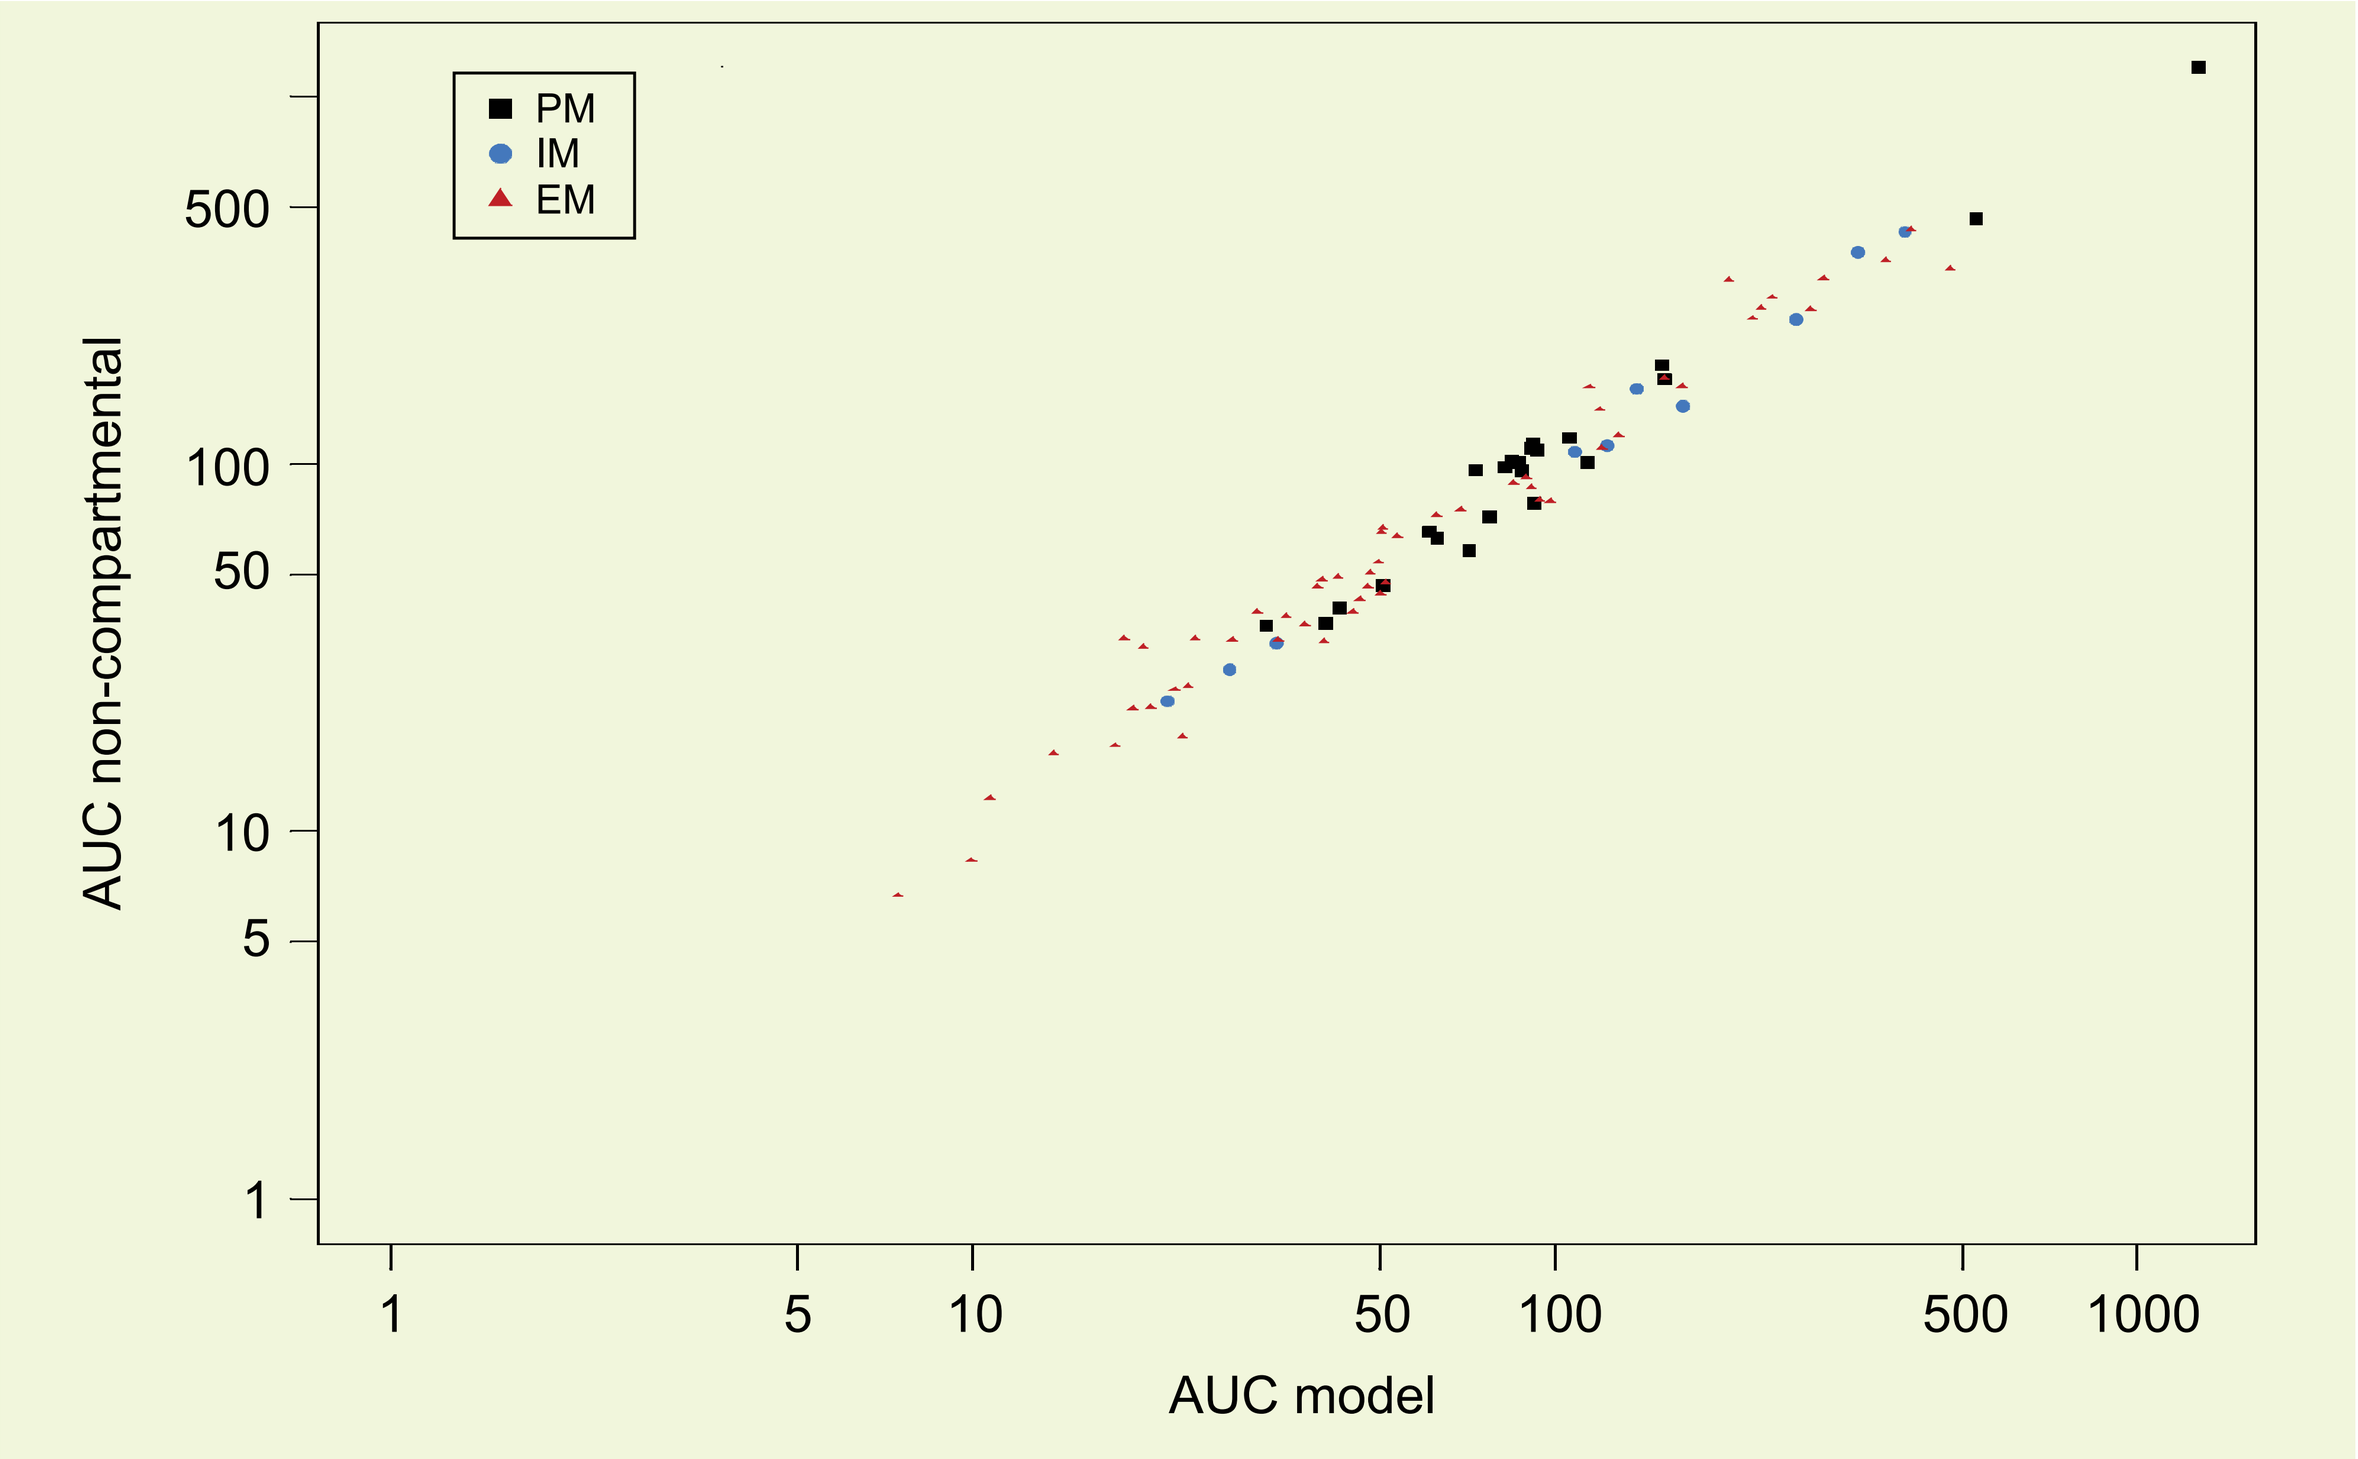
PM
IM
EM
AUC non-compartmental
AUC model

Figure 5. *Comparison of individual AUCs: model predicted AUCs vs non-compartmental AUCs*

Conclusions

A pooled population PK model successfully described the data of three Phase I trials. CYP2D6 polymorphism (PM exposure approximately 2-fold higher than EM) was identified as relevant covariate. Good concordance agreement was observed between this approach and noncompartmental analysis. This model provides a sound basis to explore the exposure-response relationship with efficacy data obtained in Phase III.

Schering-Plough

Reference

1. J. Brockmöller et al., *Clin. Pharm. And Therapeutics* **81**: 699-707 (2007)

0821003_1